Estimands in Schizophrenia

Josiassen, MK; Larsen, KG

*H. Lundbeck A/S, Ottiliavej 9, 2500 Valby, Denmark

Introduction

- The concept of **estimands**, or "what is to be estimated" has been debated widely, and in August 2017 a new draft addendum R1 to the ICH E9[1] describing a set of strategies to be used when developing estimands was published for general consultation.
- A key challenge in interpreting results from a clinical study are the so-called **intercurrent events**, defined as "Events that occur after treatment initiation and either preclude observation of the variable or affect its interpretation".
- The draft addendum introduces the following **5 strategies** for construction of estimands and addressing intercurrent events
 - Treatment policy
 - Composite
 - Hypothetical
 - Principal stratum
 - While treated
- To illustrate the theoretical concepts in practice, the strategies have been applied in a short-term **schizophrenia** setting, and potential estimators derived and applied to data from a 6-week, randomised, double-blind, study (NCT01810380) comparing the effect of flexible doses of brexpiprazole to placebo in acute schizophrenia.

Methods

- The Positive and Negative Syndrome Scale (PANSS)[2] was used as a measure of disease severity, with PANSS total score summarising the level of symptoms observed.
- The PANSS total score was observed at all visits (Screening, Baseline, Day 4, Weeks 1, 2, 3 ,4, 5, 6).
- Patients who withdrew from treatment, were requested to return at the Week 6 visit to have their efficacy and concomitant medication recorded.
- The intercurrent event that will be addressed here is: "withdrawal from study treatment"

1. **Treatment policy**
 - "*Actual values of the variable regardless of whether the intercurrent event has occurred*"
 - In the schizophrenia example this would imply
 - Follow all patients, regardless of withdrawal from treatment
 - Handle the intercurrent events by ignoring them
 - **Estimand:** Effect of being randomised to brexpiprazole compared to placebo, regardless of withdrawal from treatment
 - **Estimator:** Estimate the change from baseline in PANSS total score at Week 6 by using all observations, including those made by patients withdrawn from treatment and returning at week 6, regardless of other medication taken. Use MMRM model, including treatment, site, visit, and interaction of treatment and visit as fixed factors and baseline PANSS total score interacting with visit as covariate. The within patient variation is modelled using an unstructured covariance.
2. **Composite**
 - "*Make the intercurrent event an integrated part of the measure*"
 - In the schizophrenia example this could be implemented as
 - Define response as both obtaining an efficacy response and completing the study treatment
 - The intercurrent event is handled by the definition of the endpoint
 - **Estimand:** Effect on the composite response of seeing response and remaining on treatment
 - **Estimator:** Estimate the odds ratio of combined response of at least 30% decrease in PANSS Total score or CGI-I = 1 or 2 at Week 6 and completing the study treatment. Analyse using logistic regression including treatment as fixed effect and baseline PANSS total score as covariate.
3. **Hypothetical**
 - "*Values of the variable under a hypothetical condition where the intercurrent event would not happen*"
 - In the schizophrenia example this could be implemented as
 - Base analysis on data observed prior to the withdrawal from treatment
 - **Estimand:** The hypothetical effect seen if all patients had adhered to treatment and remained in the study.
 - **Estimator:** Estimate the change from baseline in PANSS total score at Week 6 using the same MMRM model as used for the treatment policy estimator, but excluding the Week 6 observations from patients withdrawn from treatment returning for the Week 6 visit
 - The study protocol specified the treatment effect at Week 6 from this analysis as the primary analysis
4. **Principal Stratum**
 - *"Population defined as patients that would not experience an intercurrent event, had they been treated with brexpiprazole"*
 - In the schizophrenia example this could be implemented as
 - Principal Stratification[3], and hence based on counterfactual outcomes of the intercurrent event in the placebo arm
 - An analysis targeted to estimate the "complier average causal effect"[4], i.e. effect in patients that will adhere to treatment if treated with brexpiprazole
 - In practice a comparison between
 - Patients in the brexpiprazole arm observed to adhere to treatment and
 - Patients in placebo arm weighed according to their individual likelihood of being a complier had they been treated with brexpiprazole
 - Handle intercurrent events by restricting the population to a stratum where the intercurrent event would not happen
 - **Estimand:** The effect in patients that would adhere to treatment on brexpiprazole
 - **Estimator:** Estimate the treatment difference in PANSS total score at Week 6 in patients that would adhere to treatment, if treated with brexpiprazole[5]
 - Step 1: perform a logistic regression for complier status in the brexpiprazole arm - See Figure 1
 - Step 2: use estimates from step 1 to predict the probability of complying on brexpiprazole for patients in the placebo arm
 - Step 3: calculate the weights for patients on placebo as
 - π_i, where π_i is the prediction for the i'th patient from step 2
 - Step 4: calculate the CACE using the weights in step 3:

$$CACE=\Sigma_{i:pats\ on\ brexpiprazole\ who\ comply} PANSS_i /n_{Brex} - \Sigma_{i:all\ pats\ on\ placebo}\ \pi_i PANSS_i / \Sigma_{i:all\ pats\ on\ placebo}\ \pi_i$$

 - Note that this estimator is different from the naïve estimator that compares patients who adhere to treatment on brexpiprazole with patients who adhere to treatment on placebo
5. **While on treatment**
 - "Values of the measurement up to the intercurrent event"
 - In the schizophrenia example this could be implemented as
 - Define the measure as cumulative improvement until the intercurrent event happens
 - The intercurrent event is taken into account directly via the measure
 - **Estimand:** The cumulative effect seen while on treatment
 - **Estimator:** The area under the curve of the improvement in the PANSS total score, while on treatment. Will be analysed using ANOVA, with treatment as fixed effect.

The methods use the data depending on status of the intercurrent event of withdrawal - see figure 2

Figure 1. Weights for Principal Stratum Analysis (Step 1 - Step 4)

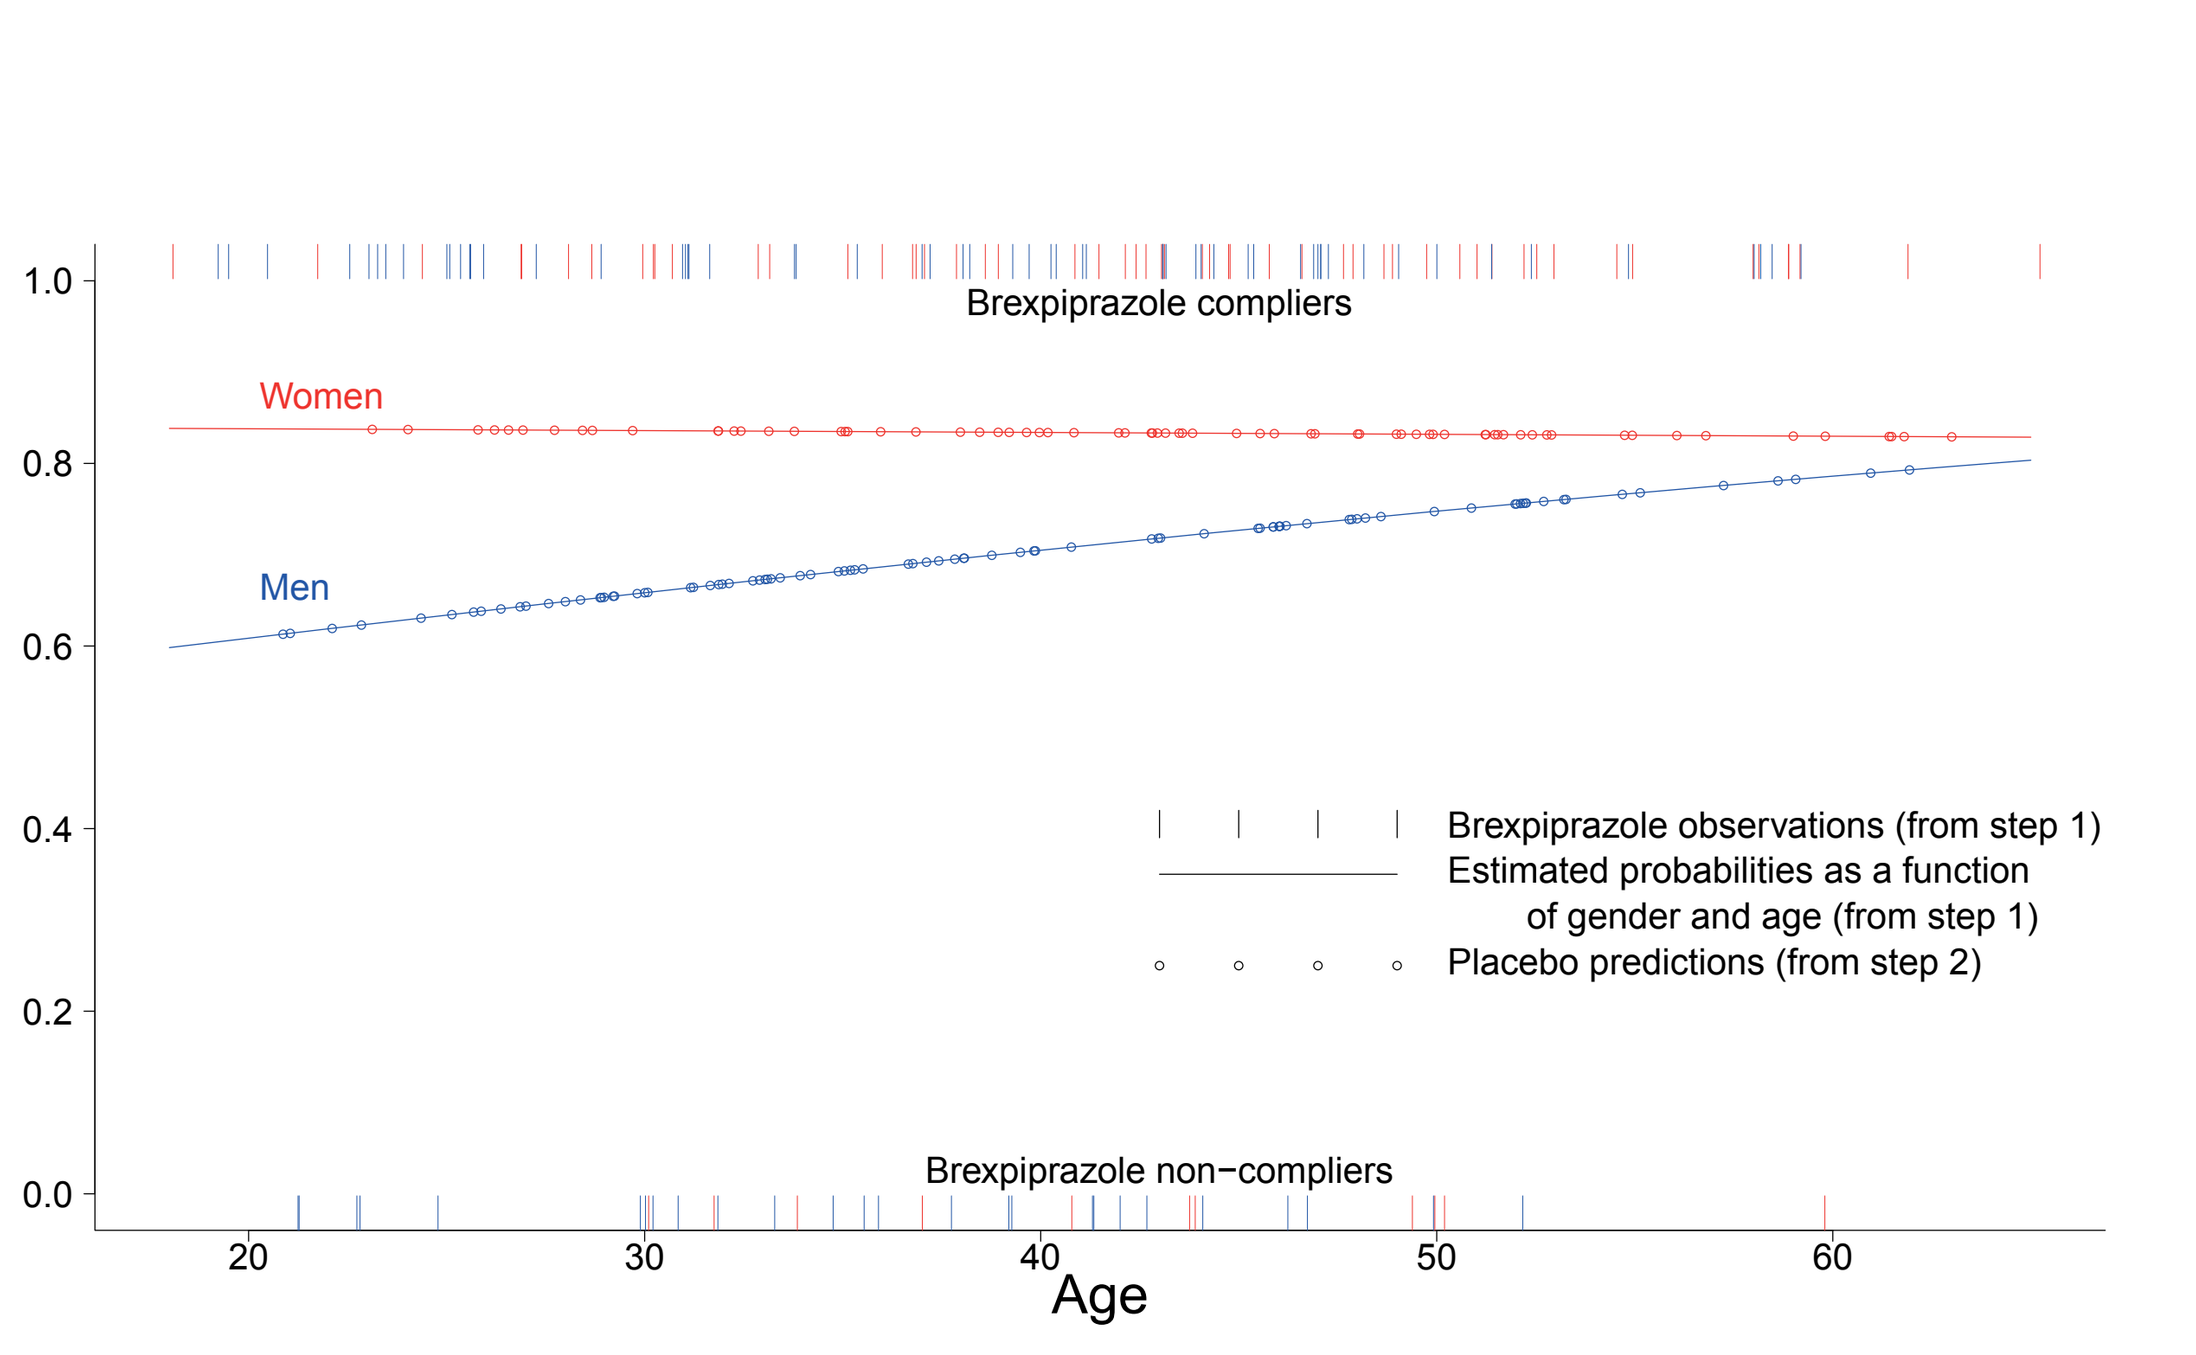
Brexpiprazole compliers
Women
Men
Brexpiprazole observations (from step 1)
Estimated probabilities as a function of gender and age (from step 1)
Placebo predictions (from step 2)
Brexpiprazole non−compliers
1.0
0.8
0.6
0.4
0.2
0.0
20
30
40
50
60
Age

Figure 2. Implications of Intercurrent Event on Individual Data Used

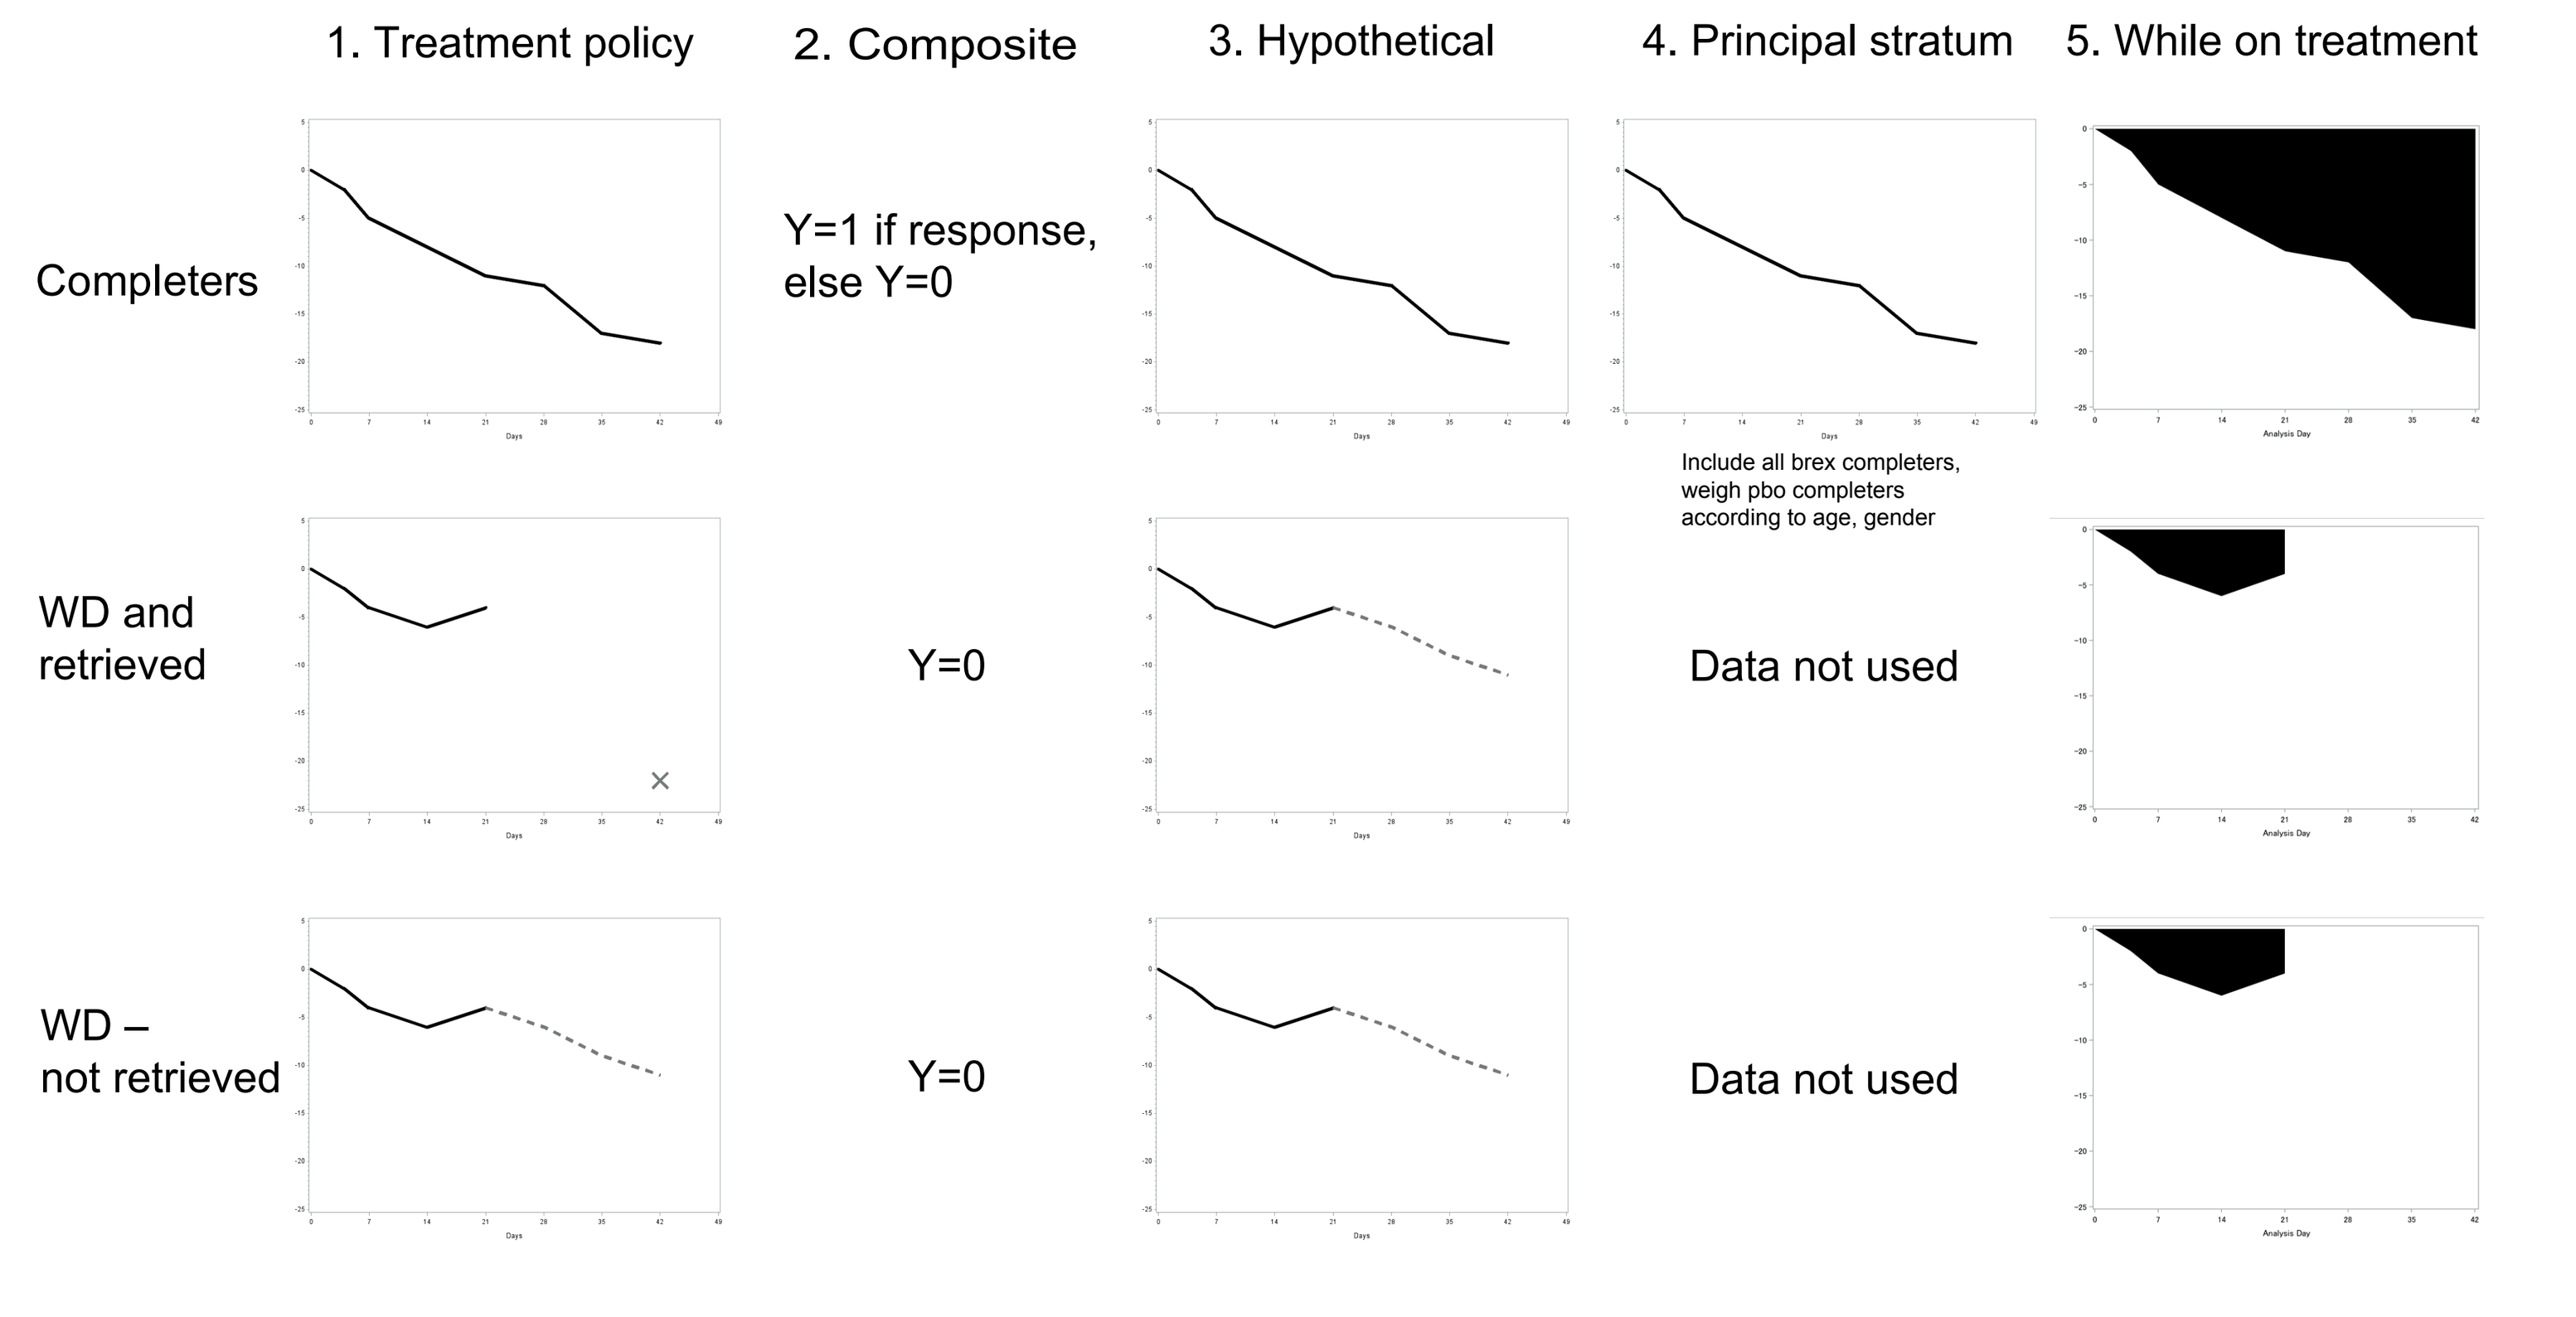
1. Treatment policy
2. Composite
3. Hypothetical
4. Principal stratum
5. While on treatment
Completers
Y=1 if response, else Y=0
Include all brex completers, weigh pbo completers according to age, gender
WD and retrieved
Y=0
Data not used
WD – not retrieved
Y=0
Data not used

Results

- The rate of patients withdrawing from treatment was 90/311 (28.9%).
- Of the patients withdrawn 46/90 (51.1%) returned for a Week 6 visit assessment. Among the patients returning for a Week 6 visit the majority 31/46 (67.4%) were on other antipsychotic treatment at the Week 6 assessment.

Figure 3. Completion and Withdrawal Data Collected

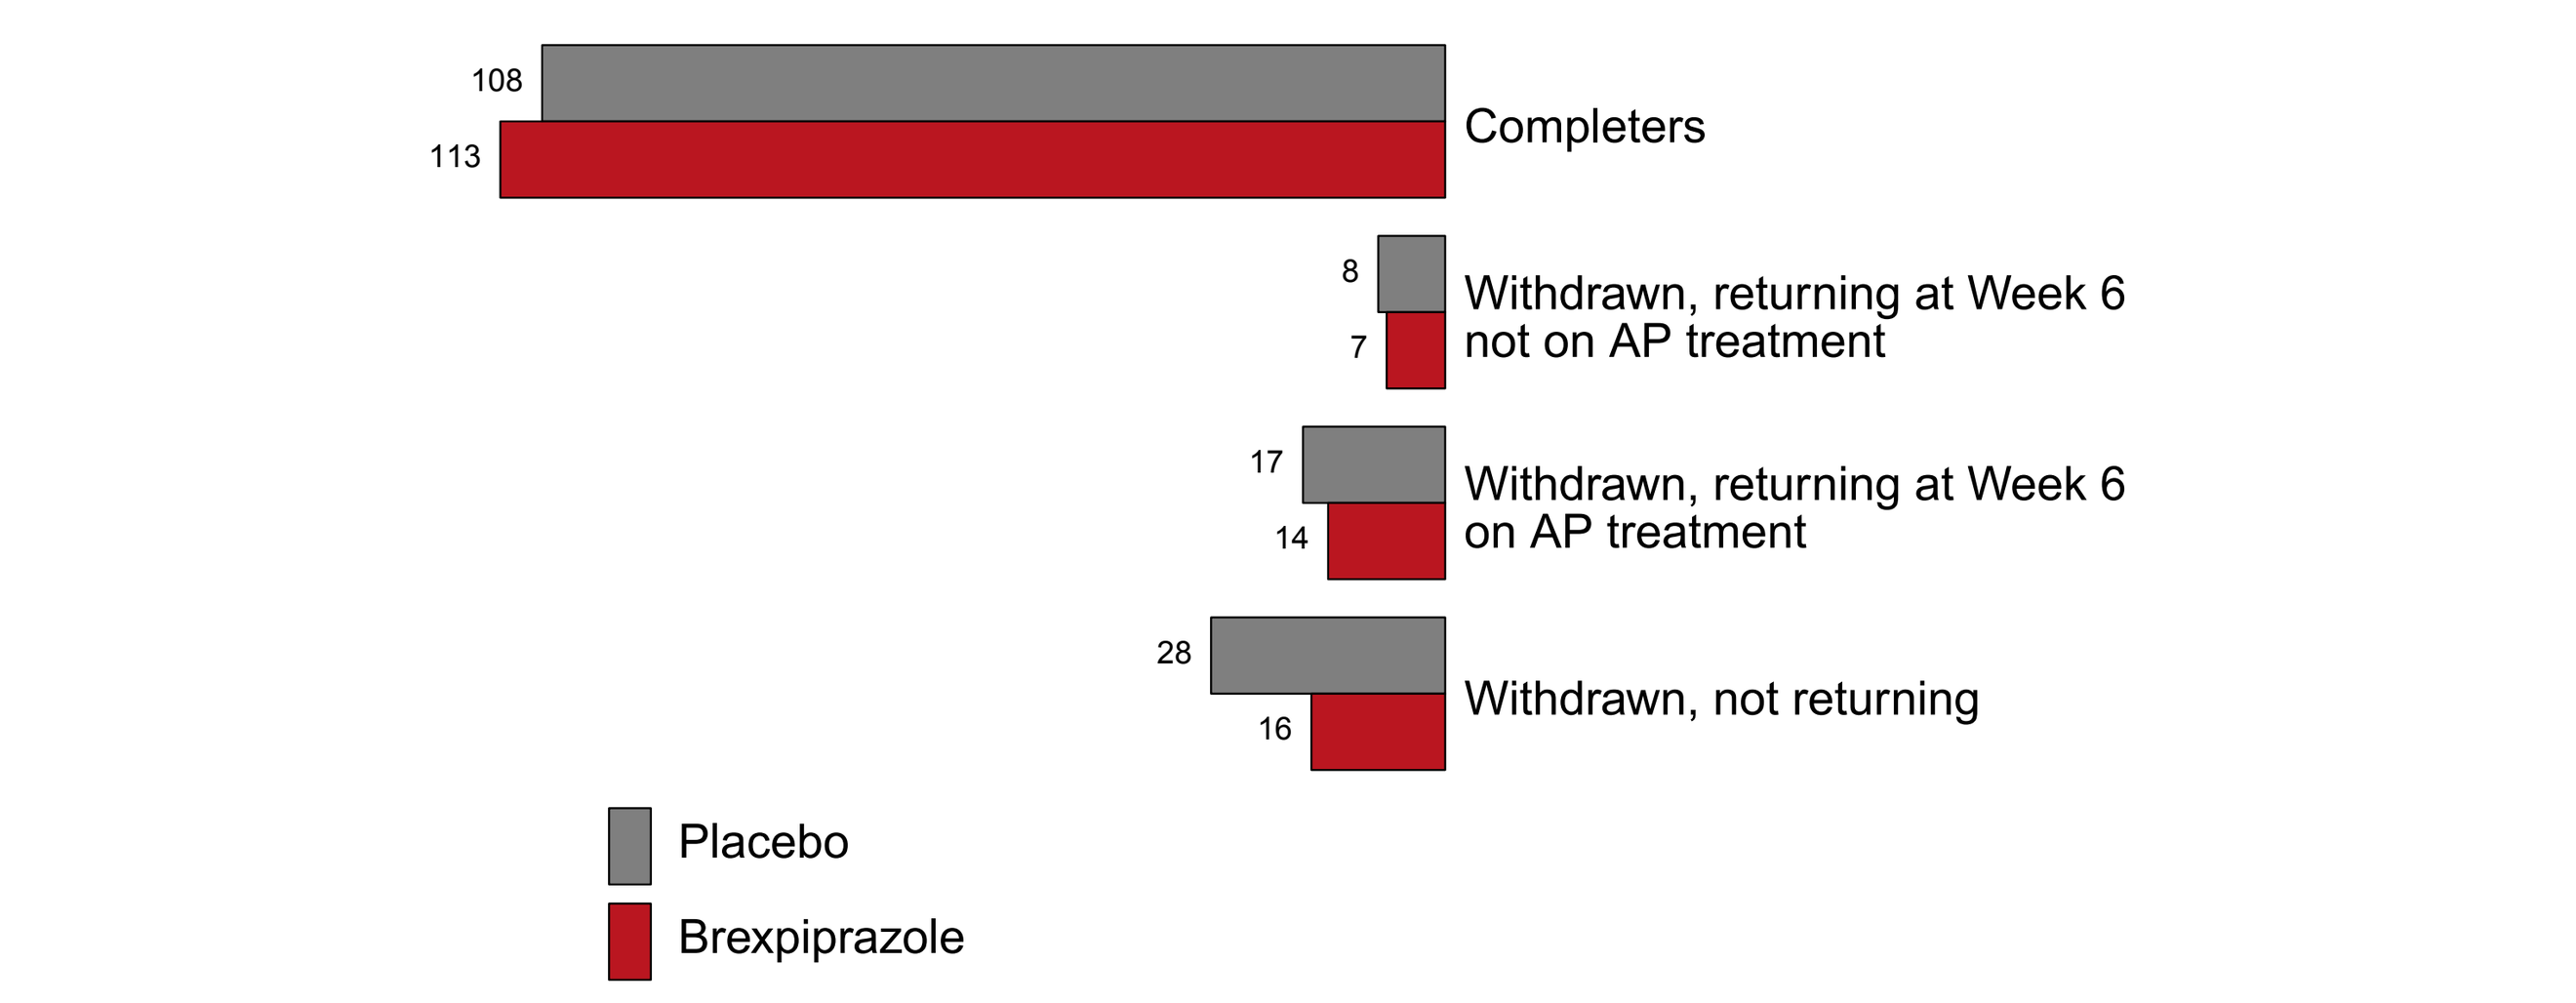
108
113
Completers
8
7
Withdrawn, returning at Week 6 not on AP treatment
17
14
Withdrawn, returning at Week 6 on AP treatment
28
16
Withdrawn, not returning
Placebo
Brexpiprazole

Table 1: Treatment Policy

Treatment	N	Change from baseline	Difference	CI	P-value
Placebo	159	-18.9			
Brexpiprazole	150	-21.3	-2.4	[-5.9;1.2]	0.1989

- Among patients randomized to brexpiprazole as compared to placebo, an advantage of 2.4 points on the PANSS total score can be expected, across completers and withdrawals.

Table 2: Composite

Treatment	N	Combined response n(%)	OR	CI	P-value
Placebo	159	49 (30.8)			
Brexpiprazole	150	70 (46.7)	1.96	[1.23;3.12]	0.0046

- The OR of combined completion and response for brexpiprazole compared to placebo is 1.96. The observed frequencies reflects a NNT of 7.

Table 3: Hypothetical

Treatment	N	Change from baseline	Difference	CI	P-value
Placebo	159	-15.9			
Brexpiprazole	150	-20.0	-4.1	[-8.2;0.1]	0.0560

- In the hypothetical scenario where all patients adhere to and complete the treatment, there is an advantage of 4.1 points on the PANSS total score of brexpiprazole compared to placebo.

Table 4: Principal Stratum

Treatment	N	Change from baseline	Difference	CI	P-value
Placebo	159	-20.5			
Brexpiprazole	150	-24.5	-4.0	[-7.9;-0.1]	0.0428

- Among patients that can complete treatment with brexpiprazole, there is an advantage of 4.0 points on the PANSS total score for brexpiprazole compared to placebo.

Table 5: While on Treatment

Treatment	N	Average -AUC [PANSS x days]	Difference	CI	P-value
Placebo	159	439			
Brexpiprazole	150	556	117	[18;216]	0.0213

- The cumulated treatment benefit until withdrawal from study treatment is 117 PANSS total score points x days better for brexpiprazole than for placebo. Divided by 6 weeks, this is an advantage of 2.8 points on the PANSS total score across the full period - See figure 4.

Figure 4. Summary of While on Treatment Data

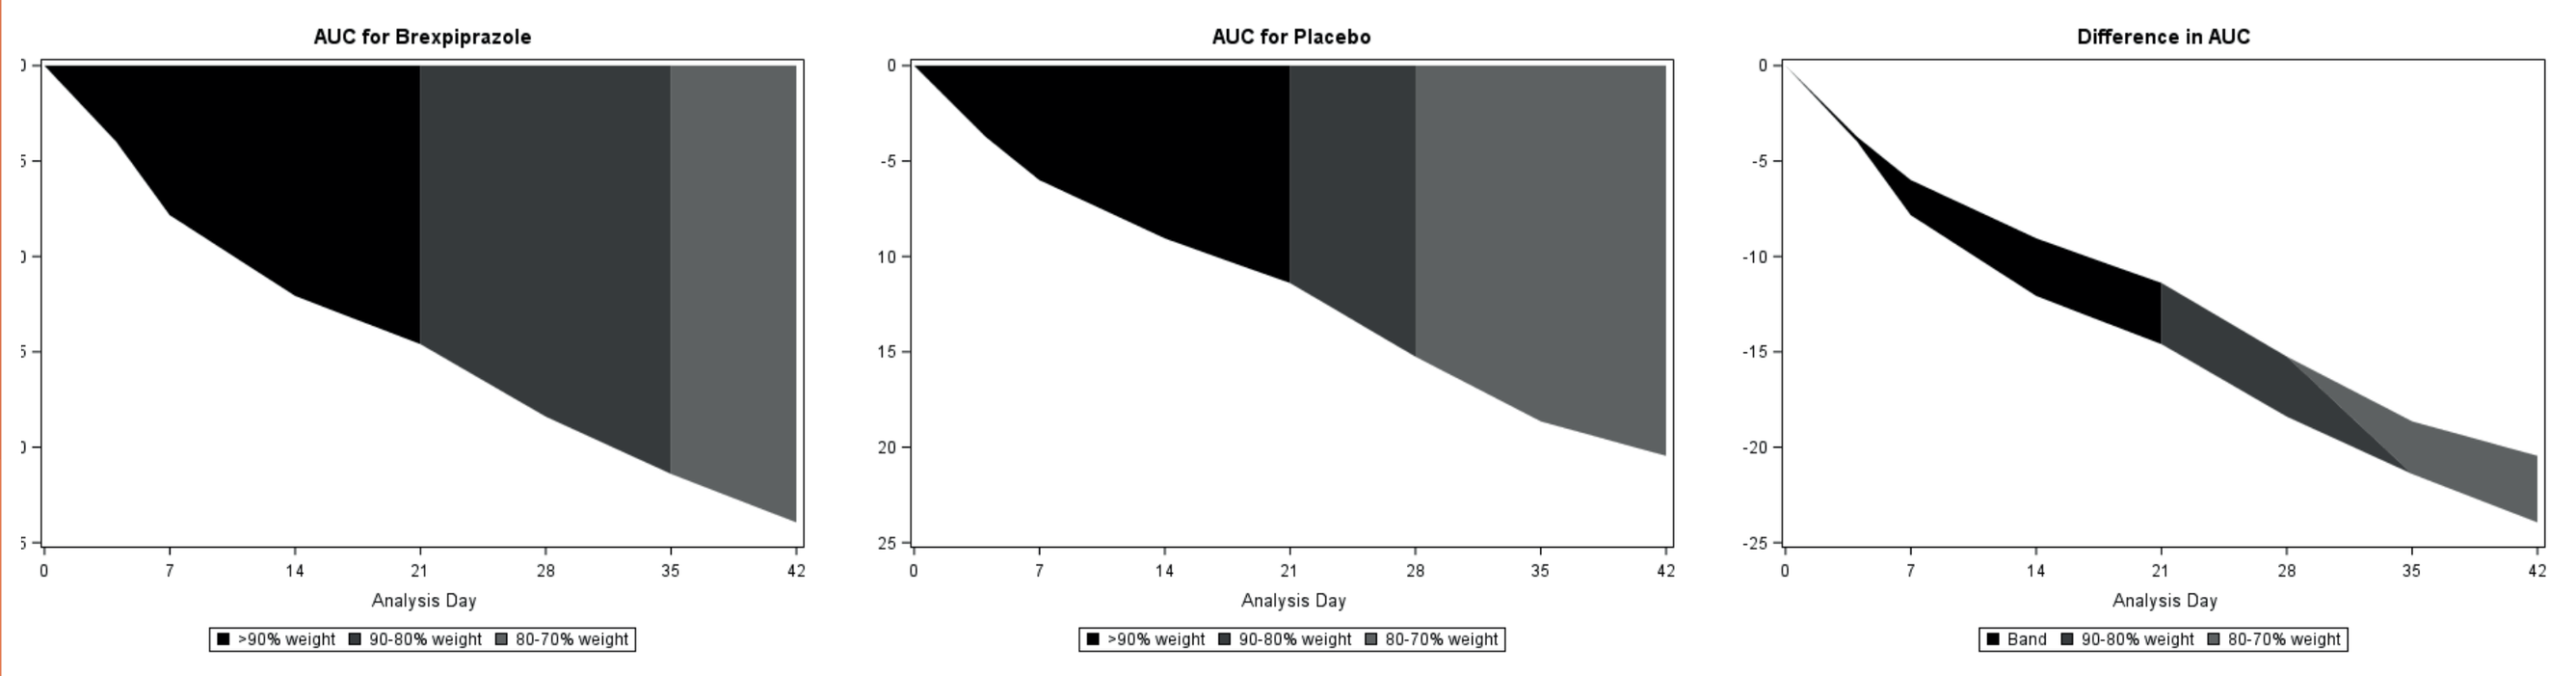
AUC for Brexpiprazole
AUC for Placebo
Difference in AUC
Analysis Day
>90% weight
90-80% weight
80-70% weight
Band

Discussion

- The preferred choice of estimand depends on the objective of the study. In this case, the study was aimed at regulatory confirmation of efficacy, and the primary endpoint of the study was the presented estimator for the hypothetical strategy.
- With an aim of regulatory confirmation, response have been accepted as a regulatory endpoint previously, so it is likely that the composite strategy and the chosen estimator could have worked, and the treatment policy may also have been accepted. But so far, the majority of regulatory approvals in schizophrenia have not been based on either causal inference (Principal stratum), or improvement while treated, so pre-specifying such endpoints in a confirmatory study would require good arguments why these would be the most appropriate
- Had the aim been to establish proof of concept, in a phase 2 study, a positive result on either type of endpoint would likely had enforced the believe in the drug as being efficacious. But using the hypothetical strategy to get an estimate of "the effect in patients that adheres to treatment and remain in the study" could be argued to be the most relevant.
- To ensure that releasing the treatment does not lead to a detrimental effect in the population, the treatment policy estimand could be considered.
- Comparison of the analysis with (Treatment policy) and without (Hypothetical) the additional data on 46 patients retrieved after withdrawal from study treatment, shows an estimated difference in change from baseline with 3 points for placebo and 1.3 points for brexpiprazole.
- The difference between the treatment policy and hypothetical estimators could be due to the fact that 2/3 of the withdrawn patients were on another antipsychotic by the time they returned. Thus, the lower level of treatment difference seen in the Treatment policy analysis, than what is seen in the other analyses is likely reflecting that more placebo patients were withdrawn, and benefitted more from changing treatment to an active treatment.

Figure 5. Comparison Across Analyses Based on Change from Baseline in PANSS Total Score

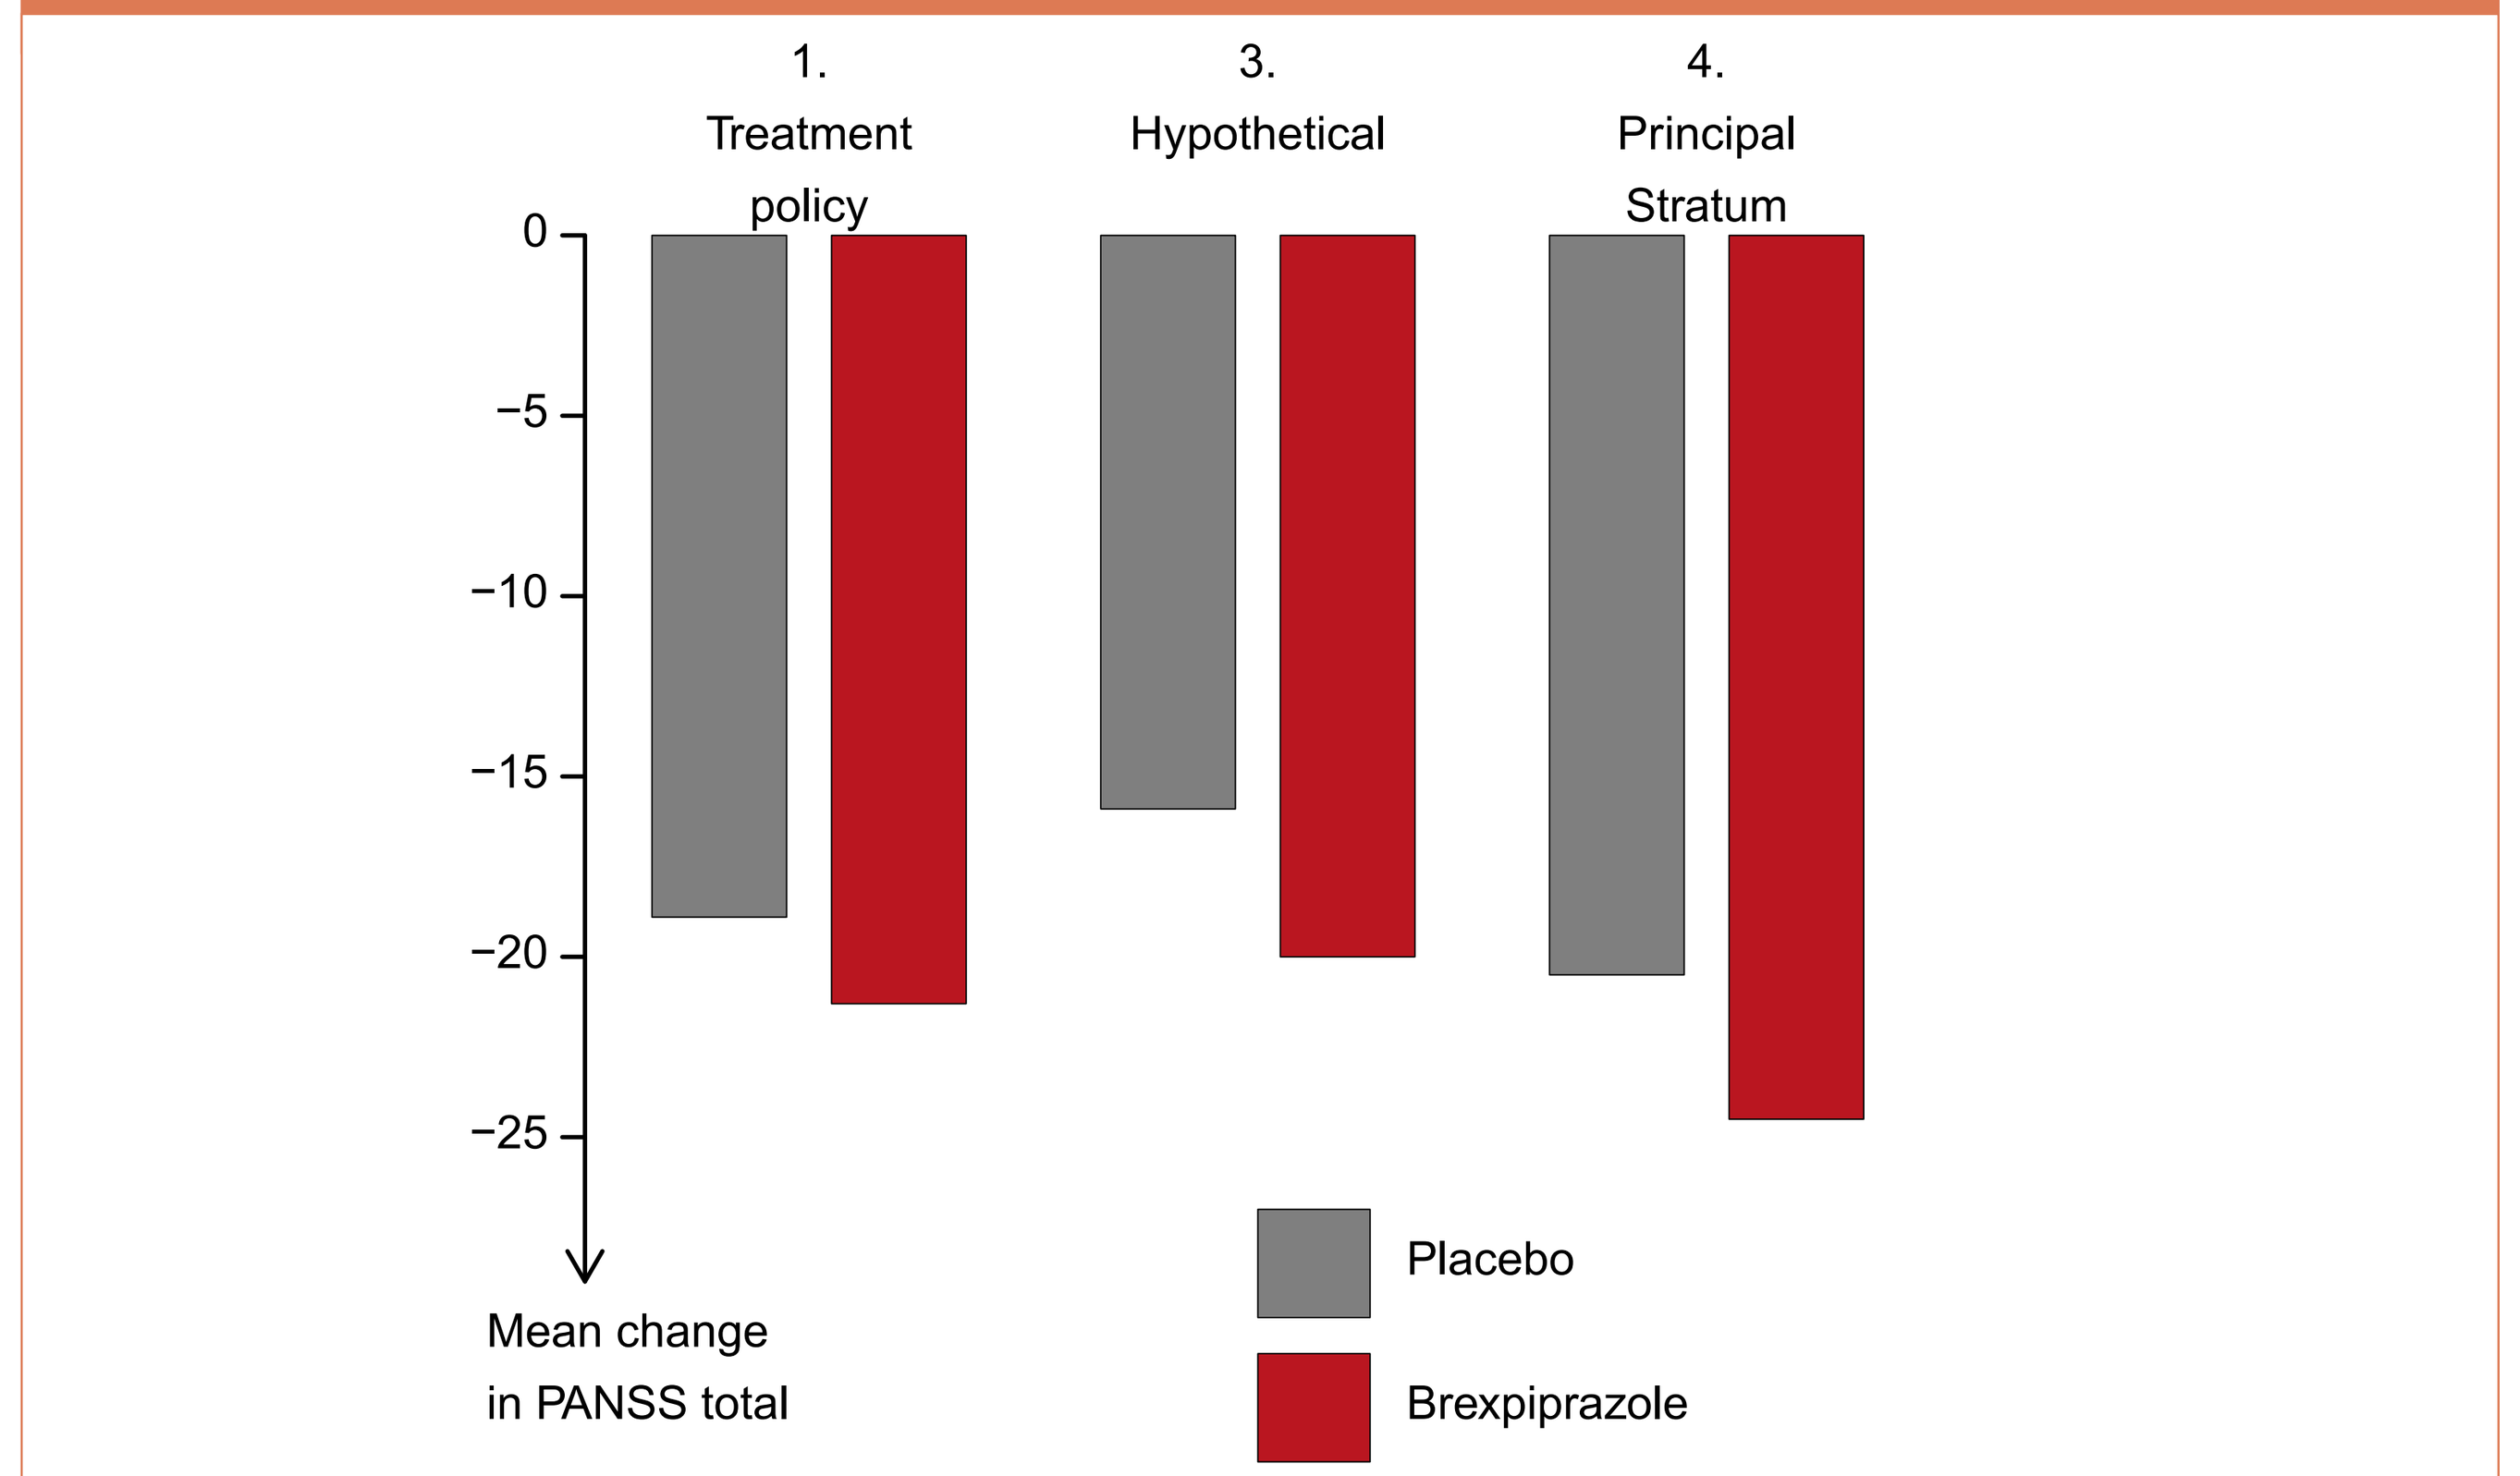
1. Treatment policy
3. Hypothetical
4. Principal Stratum
0
−5
−10
−15
−20
−25
Mean change in PANSS total
Placebo
Brexpiprazole

Conclusion

- When combining the different vantage points of the 5 strategies applied, two points emerge; in general, a treatment difference is seen between brexpiprazole and placebo in this study, and not all of that treatment difference is observed if a non-neglible proportion of the patients stop the study treatment and start on other antipsychotics and data on this is included in the analysis.
- A similar pattern could be envisioned in other studies with high withdrawal rate, and where patients are likely to switch to a new medication upon withdrawal from study, which questions the potential use of the Treatment policy strategy in such settings.

Author Disclosures:
MK Josiassen and KG Larsen are full-time employees of H. Lundbeck A/S

Presented at the ISCTM 14th Annual Scientific Meeting, 20-22 February 2018, The Fairmont - Washington, DC.

References

1. E9(R1) Statistical Principles for Clinical Trials: Addendum: Estimands and Sensitivity Analysis in Clinical Trials. Draft from FDA: https://www.fda.gov/ucm/groups/fdagov-public/@fdagov-drugs-gen/documents/document/ucm582738.pdf
2. Kay SR, Fiszbein A, Opler LA. The positive and negative syndrome scale (PANSS) for schizophrenia. Schizophr Bull. 1987; 13(2) : 261-276.
3. Frangakis CE, Rubin DB. Principal stratification in causal inference. Biometrics 2002, 58 : 20-29.
4. Imbens GW, Rubin DB. Bayesian inference for causal effects in randomized experiments with non-compliance. The Annals of Statistics 1997, 25(1), 305-327.
5. Stuart EA, Jo B. Assessing the sensitivity of methods for estimating principal causal effects. Statistical methods in medical research 2015, 24(6), 657-674.