Empirical Evaluation of Suicidal Ideation and Behavior Categories Established by the Columbia Classification Algorithm of Suicide Assessment (C-CASA)

Alphs L,[1] Fu DJ,[2] Williamson D,[1] Turkoz I,[2] Canuso C,[2] Wang Y,[2] Joshi U,[3] Jin D,[2] O'Neill J,[2] Jamieson C,[4] Smith K[4]

Janssen Scientific Affairs LLC[1], Janssen Research and Development LLC[2], Titusville, NJ, USA; PRA Health Sciences – (Performing Services for Janssen R&D), Raleigh, NC, USA[3]; Janssen Global Services, LLC, Raritan NJ, USA[4]

INTRODUCTION

- Suicidal ideation and behaviors are growing public health problems.[1]
- The Columbia Classification Algorithm of Suicide Assessment (C-CASA) has been developed as an approach for systematically classifying suicidal ideation and behavior (SIB) that has been adopted by the FDA to characterize SIB in clinical trials.
- The FDA requires that data collected using alternatives to the Columbia Suicide Severity Rating Scale (C-SSRS) be mappable to the C-CASA.
- The categories identified and defined by the C-CASA were initially developed to categorize safety events for regulatory studies and have evolved over time, with versions produced in 2010 and 2012.[2,3]
- The 2012 version of the C-CASA omits important considerations commonly observed in clinical trials, e.g., no evidence of suicide ideation or behavior. For this reason, an alternative version (C-CASA 2012 Plus) has been created for use in clinical trials.
- Questions remain as to whether any of these C-CASA versions validly and reliably capture all variations of SIB that are experienced in real world clinical trials.
- Several scales have been developed to capture information on suicidal ideation and behavior. Among these are the C-SSRS and the Suicide Ideation and Behavior Assessment Tool (SIBAT).
- This work summarizes the ability to use information captured by these 2 scales to classify participants on 3 different versions of the C-CASA.

Objective of this Work

To characterize the convergent validity of mapping of the SIBAT and C-SSRS to three version of the C-CASA with the following approach:

- Collect SIB information on participants with varying degrees of SIB using the SIBAT and C-SSRS during a single visit by different raters
- Compare concordance of different mapping structures of the C-CASA with data collected on participants using SIBAT and C-SSRS

METHODS

Study Design

- This is a prospective, cross-sectional validation study using data from adolescents, adults, and geriatric participants with various levels of suicidality.
- Independent Review Boards approved the study protocol and its amendments. The study was conducted in accordance with the ethical principles that have their origin in the Declaration of Helsinki, consistent with Good Clinical Practices and applicable regulatory requirements.
- Adult participants provided written informed consent before participation. Adolescents were enrolled only after consent was obtained from a legally acceptable representative and the adolescent had provided written assent.
- The study is registered at clinicaltrials.gov, NCT03085108.

Participants

- Male or female 12 to 85 years of age, inclusive.
- Participants with various levels of suicidality (not suicidal to extremely suicidal) were included based on the Clinical Global Impression of Severity of Suicidality Revised version (CGI-SS-R) derived from SIBAT (shown in Table 1). Participants were selected from emergency room, acute inpatient psychiatric hospital unit, or psychiatry clinical research office settings.

Evaluations

Participants agreed to complete questionnaires and to be interviewed regarding their symptoms of suicidality. Separate interviews were conducted using the SIBAT and C-SSRS.

SIBAT

- Participants completed the following 5 patient-rated SIBAT modules (**Figure 1**):
 - o About Me – patient-reported demographics, suicide history and stable suicide risk factors (up to 84 items)
 - o My Risk/Protective Factors – patient-reported dynamic suicide risk characteristics (up to 31 items)
 - o My Current Thinking – patient-reported current suicide ideation (48 items)
 - o My Actions – patient-reported rating of suicide behavior (up to 44 items)
 - o My Risk – patient-reported global risk of suicide risk (up to 6 items)
- Thereafter, a trained clinician completed the following 3 modules (**Figure 1**):
 - o Semi-structured interview-approximately 25 questions
 - o Clinical Global Impression Ratings (4 items), including the Clinical Global Impression of Severity of Suicidality (Revised) (CGI-SS-R) that provided the clinicians assessment of severity of suicidality from none to extremely severe based on data collected using the SIBAT
 - o Treatment-management – clinician-reported Clinical Judgment of Optimal Suicide Management (up to 6 items)
- Results were mapped to the various versions of the C-CASA using a computerized algorithm based on the most severe SIB classification.

Figure 1: SIBAT Structure

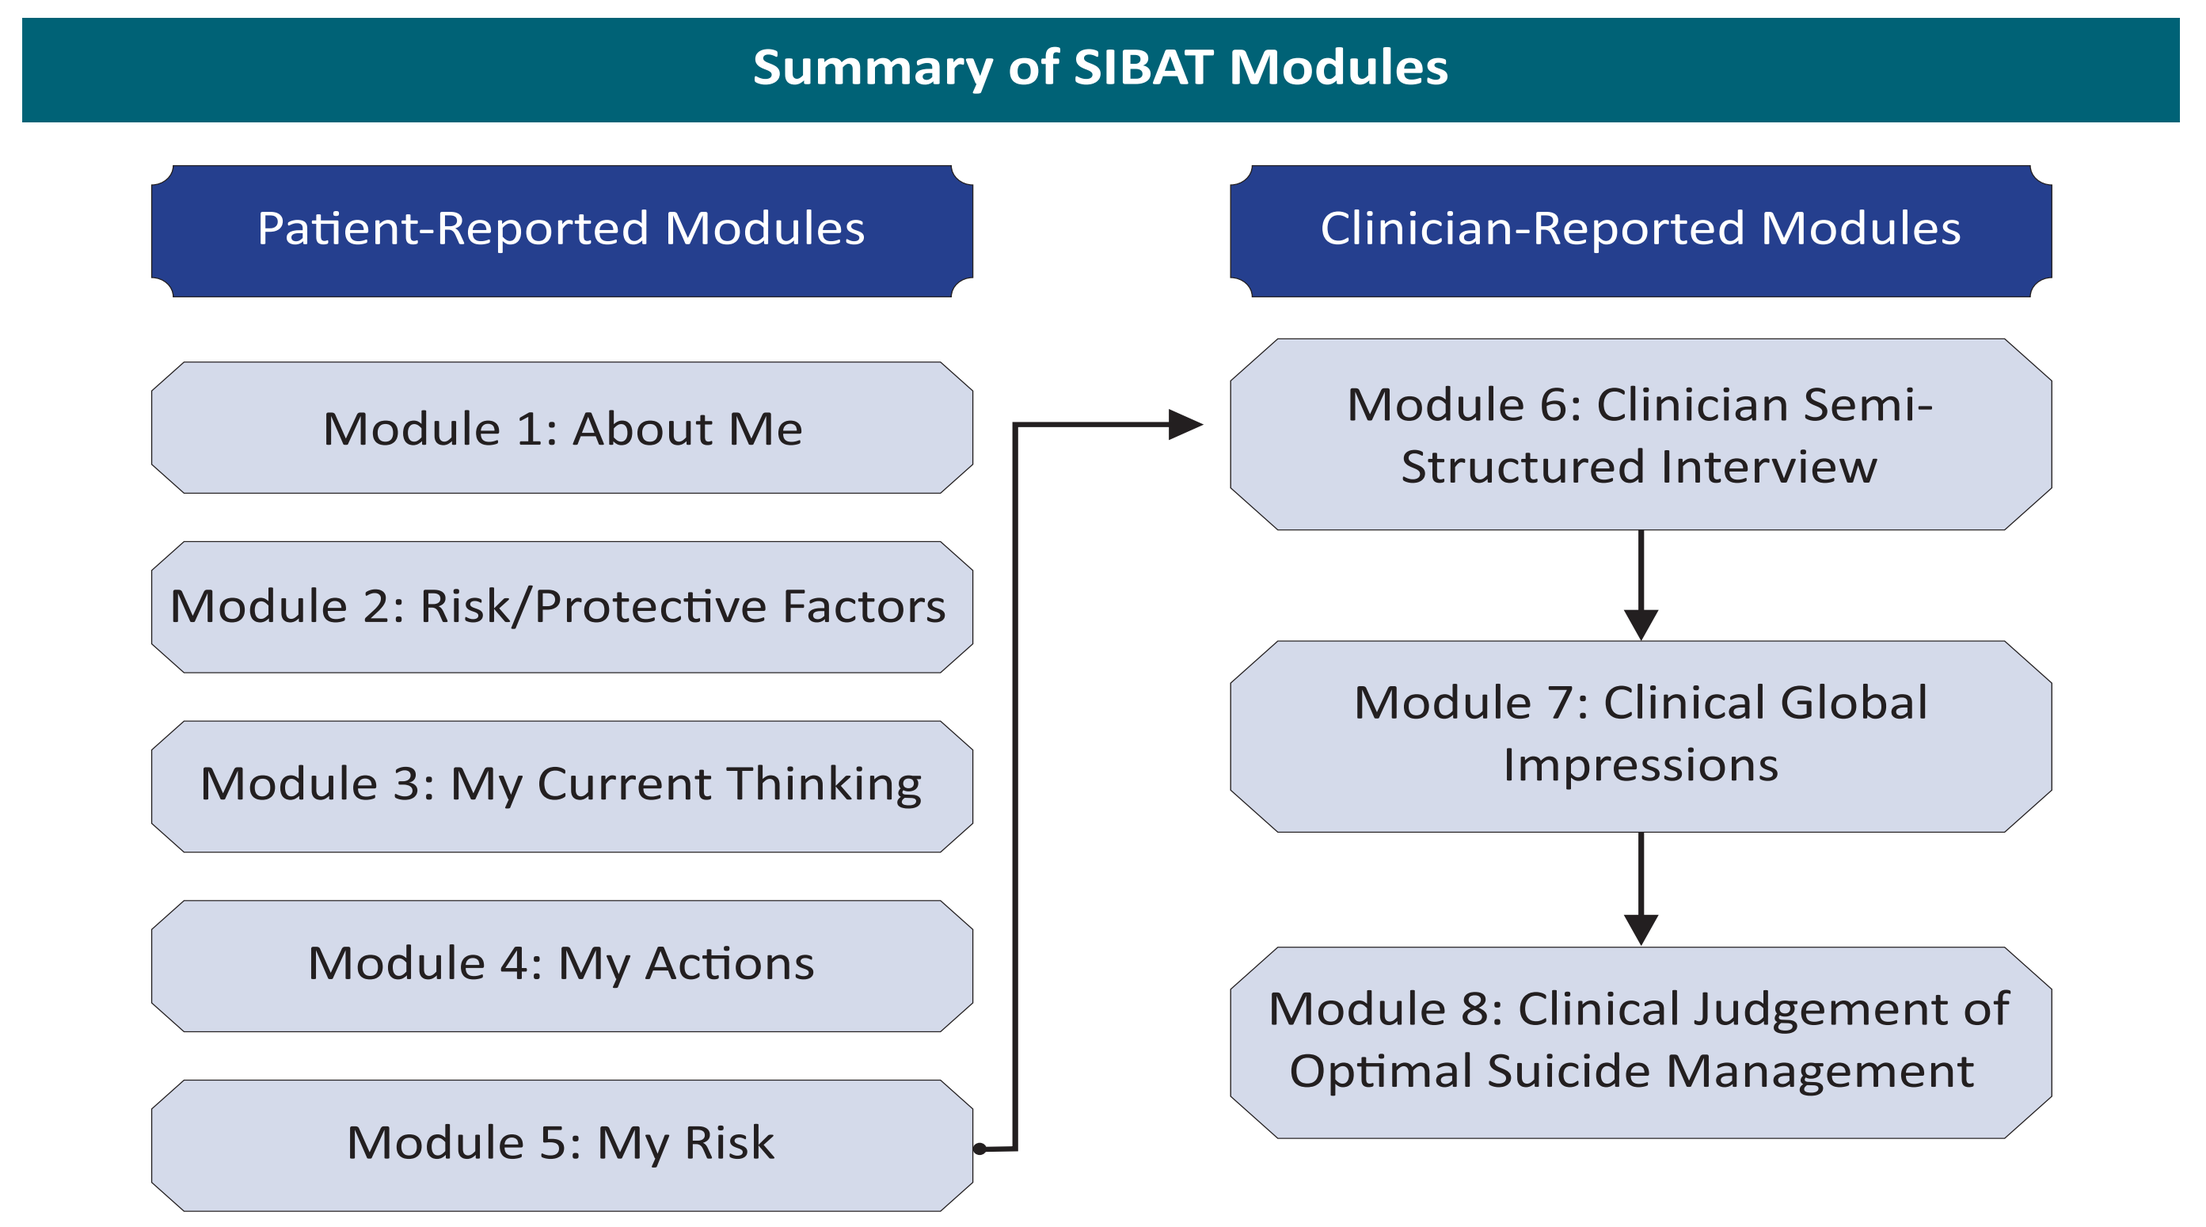
Summary of SIBAT Modules
Patient-Reported Modules
Clinician-Reported Modules
Module 1: About Me
Module 2: Risk/Protective Factors
Module 3: My Current Thinking
Module 4: My Actions
Module 5: My Risk
Module 6: Clinician Semi-Structured Interview
Module 7: Clinical Global Impressions
Module 8: Clinical Judgement of Optimal Suicide Management

C-SSRS

- Consenting participants were interviewed using the semi-structured C-SSRS interview.
- Trained clinician completed the C-SSRS ratings and the CGI-SS-R, based on data collected using the C-SSRS.
- The same trained clinician mapped their ratings to the most severe SIB classification of the various versions of the C-CASA.

C-CASA

- Three different versions of the C-CASA were used in this study (**Figure 2**):
 - o Original version (2010 C-CASA)[2]
 - o Current version (2012 C-CASA)[3]
 - o Expanded version (2012 C-CASA-PLUS) (This modified version includes items absent from C-CASA 2012 but present in the C-CASA 2010 that are needed for clinical evaluation of participants in clinical trials)
- The level of agreement between SIBAT and C-SSRS-generated data for each C-CASA mapping category was summarized manually across the 3 versions of the C-CASA.

Requirements for C-CASA Mapping Concordance

- Overall: Suicidal ideation ≠ Suicidal behavior ≠ any 'Other' category
- Within Suicidal Ideation: Passive SI ≠ Active SI with method, but no plan or intent ≠ Active SI with method, intent, but no plan ≠ Active SI with method, intent and plan
- Within Suicidal Behavior: Completed suicide ≠ Suicide attempt ≠ Aborted suicide attempt ≠ Interrupted suicide attempt
- Other Events: Fatal event ≠ Self-injurious behavior ≠ Any other response

Figure 2: Comparison of 3 C-CASA Categorizations

C-CASA 2010	C-CASA 2012	C-CASA 2012 PLUS
Completed Suicide	Completed Suicide	Completed Suicide
Suicide Attempt	Suicide Attempt	Suicide Attempt
Preparatory acts towards imminent suicidal behavior	Preparatory acts towards imminent suicidal behavior Interrupted Suicide Attempt Aborted Suicide Attempt	Preparatory acts towards imminent suicidal behavior Interrupted Suicide Attempt Aborted Suicide Attempt
Suicidal Ideation (SI)	Passive SI Active SI: Non-specific (no method, intent or plan) Active SI: method, but no intent or plan Active SI: method and intent, but no plan Active SI: method, intent and plan	Passive SI Active SI: Non-specific (no method, intent or plan) Active SI: method, but no intent or plan Active SI: method and intent, but no plan Active SI: method, intent and plan
Self-Injurious Behavior, Intent unknown	Self-Injurious Behavior, Intent unknown*	Self-Injurious Behavior, Intent unknown
Self-Injurious Behavior Without Suicidal Intent	Self-Injurious Behavior Without Suicidal Intent	Self-Injurious Behavior Without Suicidal Intent
Other (accidental, psychiatric medical), no deliberate self-harm		Other (accidental, psychiatric medical), no deliberate self-harm
Not enough information (fatal)		Not enough information (fatal)
Not enough information (non-fatal)		Not enough information (non-fatal)
		No SI or behavior
Not Mapped*	Not Mapped	Not mapped*

*These categories were not included in the corresponding version of C-CASA to C-SSRS raters for their mapping; SI: Suicidal ideation

RESULTS

A sample of 47 participants were identified from 4 clinical sites in the United States (University of Alabama at Birmingham, AL; CNS Network, Garden Grove, CA; Rutgers University, Piscataway, NJ; University of Cincinnati, OH).

Table 1. Demographics and SIB Severity of Study Participants

Characteristic	n=47
Age (years), mean (SD)	33 (16.1)
Sex, n (%)	
Male	16 (34%)
Female	31 (66%)
Race	
White	32 (68.1%)
Black/African American	10 (21.3%)
Multiple	3 (6.4%)
American Indian or Alaska Native	1 (2.1%)
Not reported	1 (2.1%)
CGI-Severity of Suicidality Rating (SIBAT)	
Normal, not at all suicidal	7 (15%)
Questionably suicidal	8 (17%)
Mildly suicidal	15 (32%)
Moderately suicidal	8 (17%)
Markedly suicidal	4 (9%)
Severely suicidal	4 (9%)
Among the most extremely suicidal patients	1 (2%)

Table 2. Mapping to 3 versions of the C-CASA with the C-SSRS and SIBAT for Most Severe Level of Suicide Ideation and Behavior Captured (10 Illustrative Cases) and Corresponding CGI-SS-R*

Participant	Scale	C-CASA 2010	C-CASA 2012	C-CASA 2012 Plus	CGI-SS-R Rating*
1 54135419SUI0001-001001012	**SIBAT**	Suicidal ideation (SI)	Active SI: Non- specific (no method, intent or plan)	Active SI: Non-specific (no method, intent or plan)	**Questionably Suicidal**
	C-SSRS	Nonfatal, not enough information	Not ratable	No SI or behavior	
2 54135419SUI0001-001001017	**SIBAT**	SI	Active SI: Non-specific (no method, intent or plan)	Active SI: Non-specific (no method, intent or plan)	**Mildly Suicidal**
	C-SSRS	Other, no deliberate self-harm	Not ratable	No suicidal ideation or behavior	
3 54135419SUI0001-001001015	**SIBAT**	SI	Active SI: method and intent, but no plan	Active SI: method and intent, but no plan	**Severely Suicidal**
	C-SSRS	SI	Interrupted Suicide Attempt	Interrupted Suicide Attempt	
4 54135419SUI0001-001001028	**SIBAT**	Preparatory acts toward imminent suicide behavior	Interrupted Suicide Attempt	Interrupted Suicide Attempt	**Questionably Suicidal**
	C-SSRS	Other, no deliberate self-harm	Not ratable	No SI or behavior	
5 54135419SUI0001-001001034	**SIBAT**	SI	Active SI: method, but no intent or plan	Active SI: method, but no intent or plan	**Severely Suicidal**
	C-SSRS	SI	Active SI - method and intent, but no plan	Active SI - method and intent, but no plan	
6 54135419SUI0001-001002002	**SIBAT**	Preparatory acts toward imminent suicide behavior	Interrupted Suicide Attempt	Interrupted Suicide Attempt	**Markedly Suicidal**
	C-SSRS	SI	Active SI - method, intent and plan	SI - method, intent and plan	
7 54135419SUI0001-001002003	**SIBAT**	SI	Active SI - method, intent and plan	Active SI- method, intent and plan	**Markedly Suicidal**
	C-SSRS	Suicide attempt	Suicide behavior - interrupted suicide attempt	Suicide behavior - interrupted suicide attempt	
8 54135419SUI0001-001002004	**SIBAT**	Suicide attempt	Suicide Attempt	Suicide Attempt	**Mildly Suicidal**
	C-SSRS	Preparatory suicide behavior	SI - active: method, but no intent or plan	SI - active: method, but no intent or plan	
9 54135419SUI0001-001002007	**SIBAT**	Preparatory acts toward imminent suicide behavior	Aborted Suicide Attempt	Aborted Suicide Attempt	**Moderately Suicidal**
	C-SSRS	SI	SI - passive: wish to be dead	SI - passive: wish to be dead	
10 54135419SUI0001-001004020	**SIBAT**	Preparatory suicide behavior	Preparatory acts towards imminent suicidal behavior-not counting aborted or interrupted	Preparatory acts towards imminent suicidal behavior-not counting aborted or interrupted	**Mildly Suicidal**
	C-SSRS	SI	Active SI - method, intent and plan	Active SI- method, intent and plan	

*Obtained from the SIBAT; SI: Suicidal ideation

Table 3. Concordant SIBAT/C-SSRS-Generated C-CASA Ratings

Concordance	SIBAT	C-SSRS
Internal Concordance of C-CASA 2010 with C-CASA 2012	46/47	40/45
Internal Concordance of C-CASA 2010 with C-CASA 2012 Plus	46/47	40/45
Internal Concordance of C-CASA 2012 with C-CASA 2012 Plus	47/47	45/45
Number of 'Not Mappable' Ratings C-CASA 2010	0/47	0/46*
Number of 'Not Mappable' Ratings C-CASA 2012	9/47	9/45
Number of 'Not Mappable' Ratings C-CASA 2012 Plus	0/47	0/45*

* This category was not included in the C-CASA to the raters.

Concordance of SIBAT mappings to C-SSRS mappings C-CASA 2010

Overall	30/45
- SIBAT identifies higher level of C-CASA classification	13/15
- C-SSRS identifies higher level of C-CASA classification	2/15

Concordance of SIBAT mappings to C-SSRS mappings C-CASA 2012

Overall	20/45
- SIBAT identifies higher level of suicidality	19/25
- C-SSRS identifies higher level of suicidality	6/25

Concordance of SIBAT mappings to C-SSRS mappings C-CASA 2012 Plus

Overall	20/45
- SIBAT identifies higher level of suicidality	19/25
- C-SSRS identifies higher level of suicidality	6/25

Limitations

- Relatively small study sample did not exhaust all C-CASA mapping possibilities.
- One C-CASA category was omitted in each version of the C-CASA for C-SSRS raters (Figure 2).
- The results presented here are preliminary and analyses are ongoing for this study.

DISCUSSION AND CONCLUSIONS

- Results from initial mapping of the 2 scales to the 3 C-CASA versions found that most participants could be reliably mapped to the C-CASA 2010 and C-CASA 2012 Plus versions of the C-CASA, but a meaningful number could not be mapped to the extant C-CASA 2012 version.
- Discordance between clinician ratings of suicide severity as rated by the CGI-SS-R and C-CASA ratings was notable. This may relate to differences in intent of the two rating systems. The C-CASA classifies recent historical information about the patient. The CGI-SS-R captures the clinician's impression of the patient's current suicidality.
- Although the SIBAT and C-SSRS mappings agree most of the time, they disagree on the primary most severe category for C-CASA classification at least 30% of the time. Across all three versions of the C-CASA, most of these disagreements were one of three types:
 - o Participants classified as having attempted suicide by the SIBAT were often classified as having an interrupted or aborted attempt by C-SSRS.
 - o Participants classified as having an interrupted or aborted suicide attempt by SIBAT were often classified as having suicide ideation by C-SSRS.
 - o Participants classified as having active suicide ideation by SIBAT were often classified as having passive suicide or without suicide ideation or behavior by the C-SSRS.
- Discordance was greatest when mapping to the C-CASA 2012 and C-CASA 2012 Plus, suggesting that increasing specificity of suicidal ideation in 2012 version of the C-CASA increases discordance of ratings across scales.
 - o Discordance of one SIBAT rating occurred because the C-CASA Suicide Ideation classification algorithm did not accommodate the participant's ratings.
 - o Discordance may have arisen from limitations in scale, rater or C-CASA rating algorithm.
- Discordance of mappings to versions of the C-CASA for the SIBAT and C-SSRS were non-random, with greater severity of suicidality general captured by the SIBAT. This may be due to the greater comprehensiveness of the SIBAT; however, this remains to be empirically evaluated.
- SIBAT-generated C-CASA ratings come from computerized algorithms based on the participant self-report modules of the SIBAT, whereas those C-CASA ratings from the C-SSRS were determined by a clinician rater based on their interview of the participant.
- This work is a step toward the development of better instruments for use in clinical trials evaluating SIB.

References:

1. Olin B, Jayewardene AK, Bunker M, Moreno F (2012). Mortality and suicide risk in treatment-resistant depression: an observational study of the long-term impact of intervention. PLoS ONE 7(10): e48002. **2.** FDA/CDER. Guidance for Industry: Suicidality: Prospective Assessment of Occurrence in Clinical Trials (issued in September 2010). Available at: https://www.federalregister.gov/documents/2010/09/09/2010-22404/draft-guidance-for-industry-on-suicidality-prospective-assessment-of-occurrence-in-clinical-trials **3.** FDA/CDER. Guidance for Industry: Suicidal Ideation and Behavior: Prospective Assessment of Occurrence in Clinical Trials (issued in August 2012). Available at: https://www.fda.gov/downloads/drugs/guidances/ucm225130.pdf. **4.** Posner K, Brown GK, Stanley B, et al. The Columbia—Suicide Severity Rating Scale: Initial validity and internal consistency findings from 3 multi-site studies with adolescents and adults. Am J Psychiatry. 2011;166:1266-1277.

Disclosures:

This study was funded by Janssen Research & Development LLC, Titusville, NJ USA. One or more authors report potential conflicts, which are described in the program.

Acknowledgements:

We acknowledge Sandra Norris, PharmD of the Norris Communications Group LLC for medical writing assistance. We also acknowledge raters: Razzya Abdulmomin, David Barkely II, Stephen Benoit, Allison Borges, Yoav Domany, Kiki Fehling, David Fleck, Sybil Fortner, Haig Goenjian, Steven Hoover, Chris Hughes, Cynthia Keenan, Austin Luker, Clinton Martin, Cheryl McCullumsmith, Michael Meader, Janaki Nimmagadda, Taylor Preston, Karen Seif, Edward Selby, Samanthan Shiney, Yushi Wang

Presented at the 14th Annual Scientific Meeting of the International Society for CNS Clinical Trials and Methodology (ISCTM), 20-22 February 2018, Washington DC.