ISCTM Autumn Conference | September 16th - 17th, 2026 | Philadelphia, PA

Enrichment for Canonical Symptom Presentation in Generalized Anxiety Disorder

Xingmei Wang[1], Alan Kott[1], Gary Sachs[1], Federico Bolognani[2], Seth C. Hopkins[2]
[1]Signant Health | [2]Newleos | ISCTM 2026

WHAT IS THE METHODOLOGICAL ISSUE BEING ADDRESSED?

- Clinical trials in psychiatry are challenged by heterogeneous patient populations and uncertainty in measuring neuropsychiatric constructs with symptom rating scales. Conventional inclusion criteria specify symptom severity, but do not determine whether an individual subject's item-level symptom presentation is well-suited to the psychometric structure of the selected instrument. Variance–covariance difference (VCD) methods were previously developed to identify canonical versus anomalous symptom structure at the individual-subject level, with the goal of reducing nondrug-related variability and increasing the ability to detect a drug effect if one is present. This analysis evaluates whether this symptom-structure enrichment concept can be applied to the Hamilton Anxiety Rating Scale (HAM-A).

BACKGROUND

- The HAM-A is widely used in anxiety clinical trials, but total score severity may obscure important differences in item structure.
- We hypothesized that subjects whose screening and baseline HAM-A item patterns conform more closely to a prespecified psychic/somatic anxiety construct would provide greater measurement certainty than subjects with anomalous HAM-A item structure.
- In effect, this method reframes the usual question of "what instrument is best for my study?" as "what symptom presentation is best for my instrument?"

METHODS

Dataset: 635 subjects, 3 studies, HAM-A 14-item scores (screening + baseline)

VCD Construction: 287-element vector comprising:

- Variances (14) – item change across visits
- Covariances (91) – item co-variation
- Screening differences (91) – within-visit item pairwise difference
- Baseline differences (91) – within-visit item pairwise difference

Analysis:

1. Isolation Forest → anomaly scores
2. Stratification by anomaly-score percentiles
3. Confirmatory Factor Analysis (CFA) of 2-factor model (psychic & somatic anxiety)
4. Validation via fit indices: CFI and RMSEA

RESULTS

- Distribution of the anomaly scores
- Theright-skewedanomalyscoredistributionreflectsisolationforestpathlengths:theleft-concentratedbulkrepresentscanonicalsubjectsrequiringlongerpathstoisolate,whiletheextended right tail identifies anomalous subjects with short path lengths and atypical symptom structures.

FIGURE 1. ANOMALY SCORE DISTRIBUTION

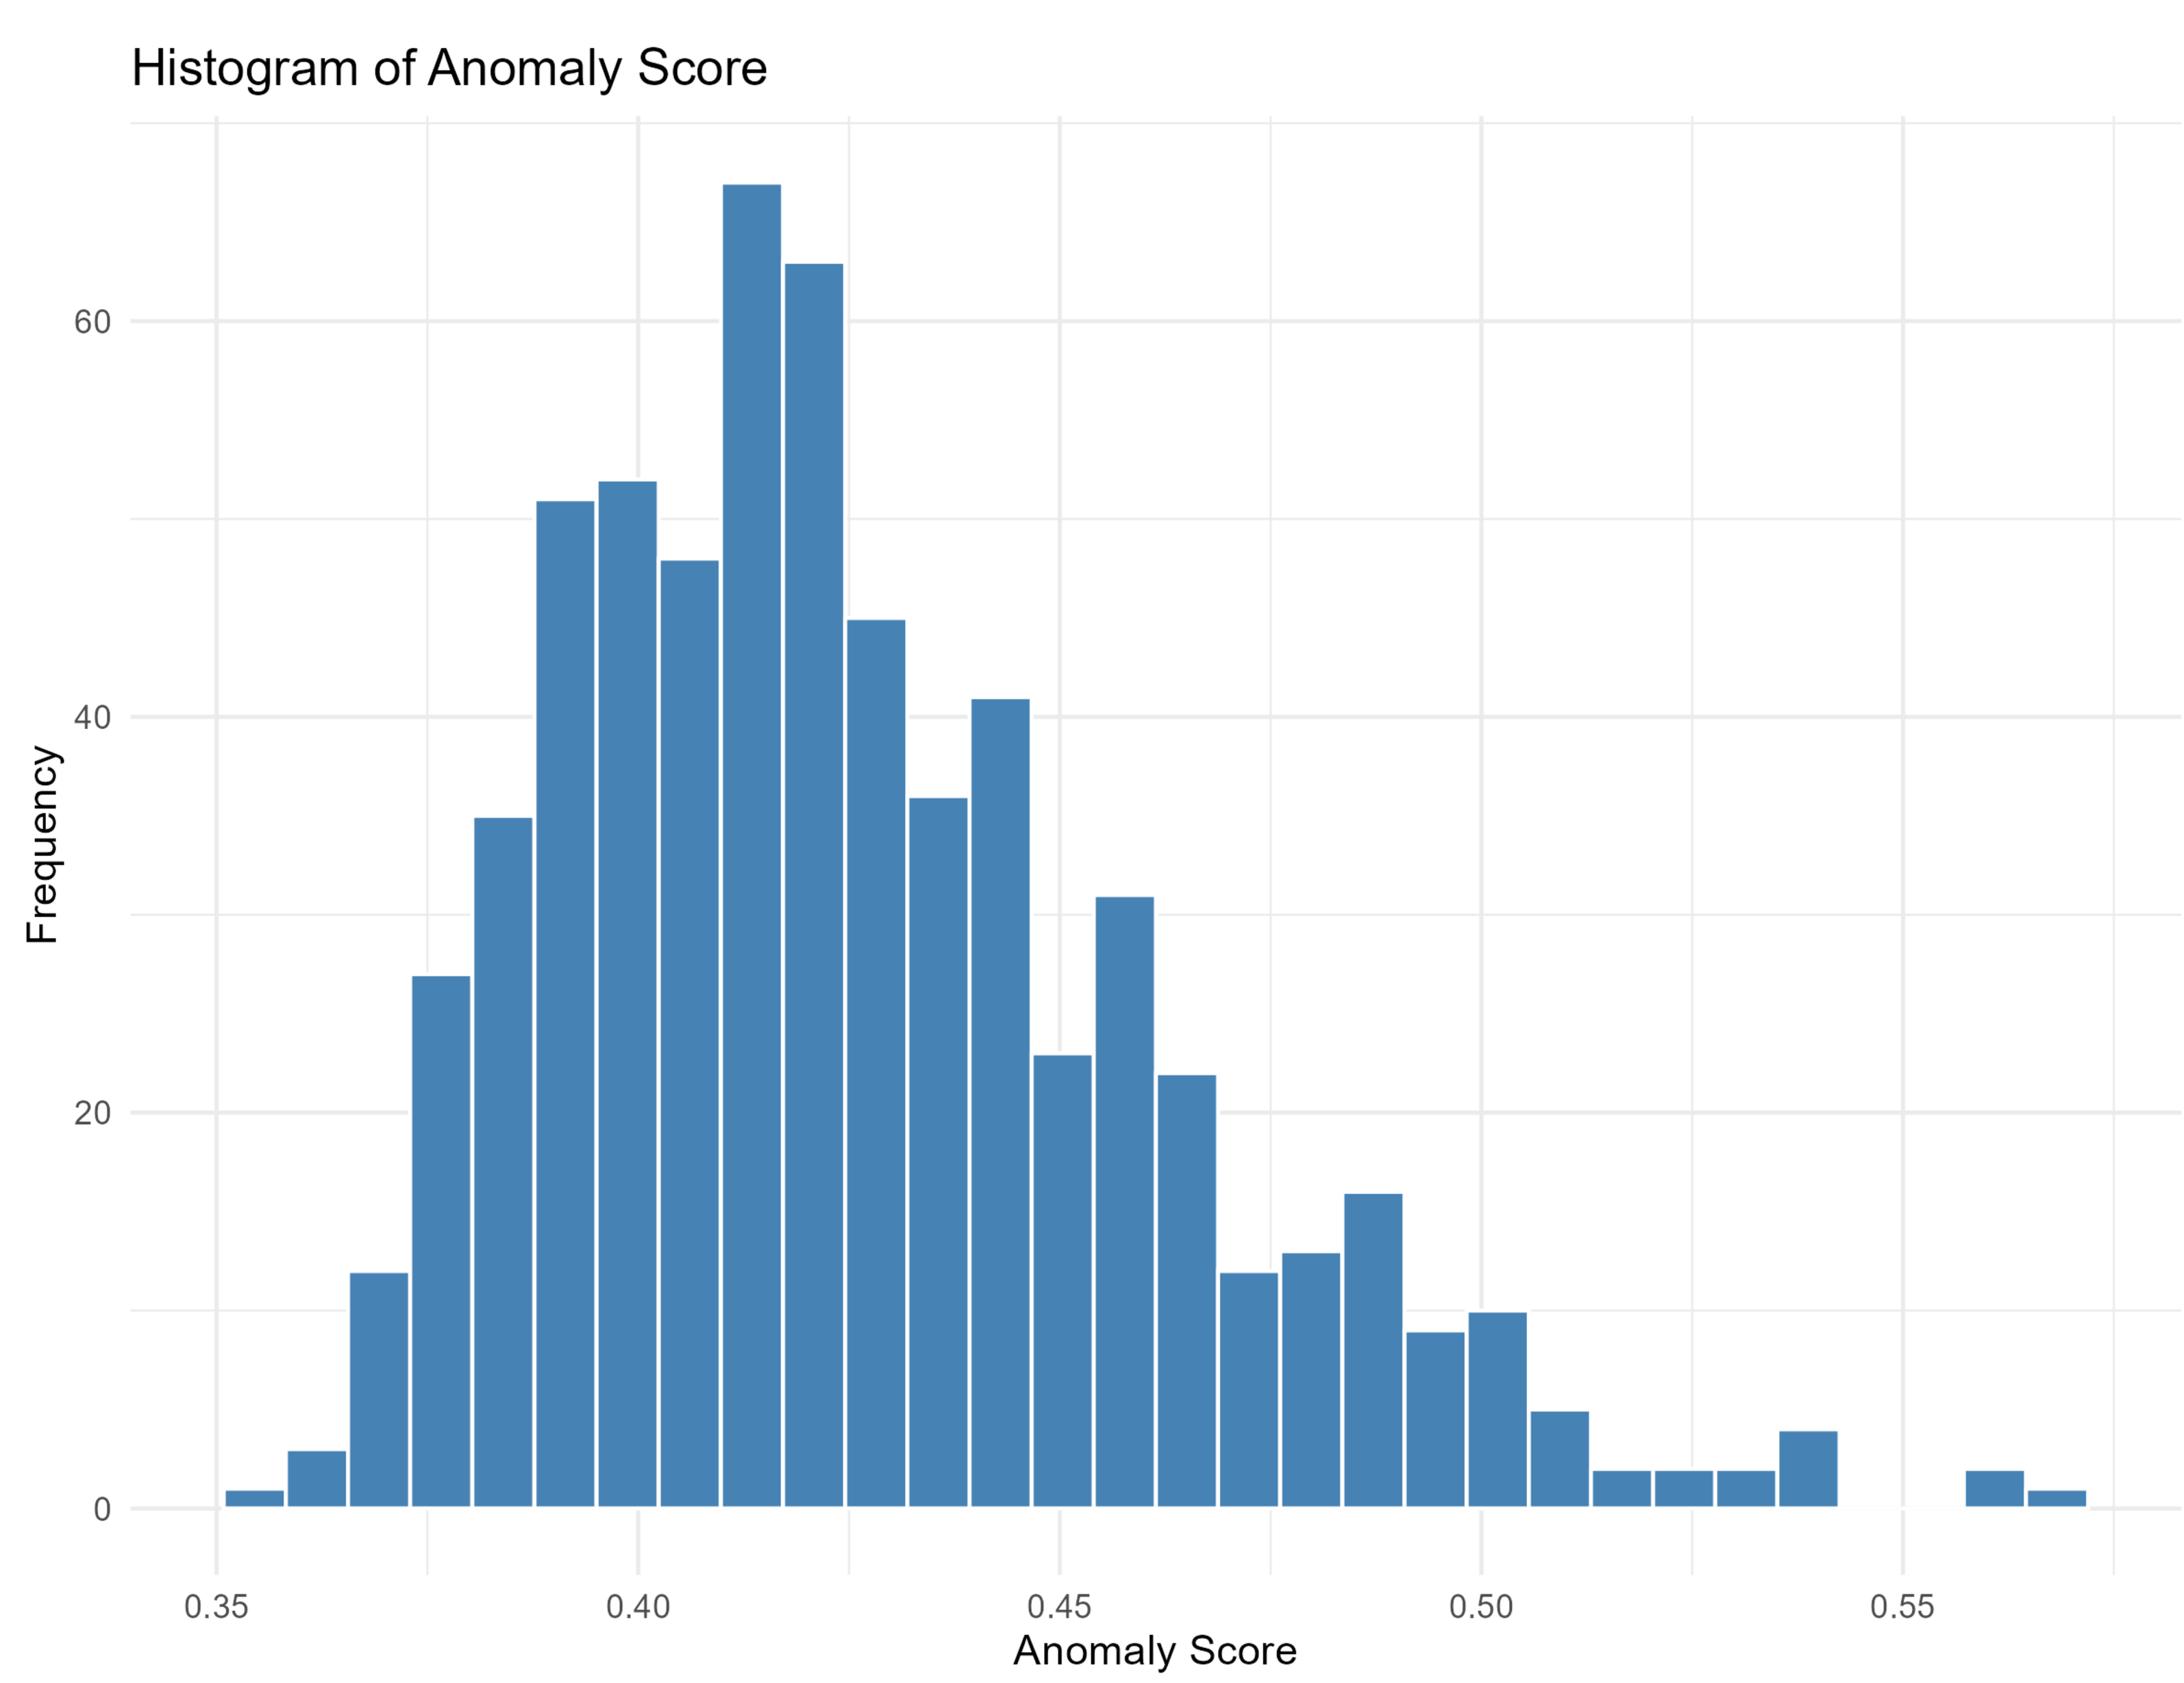
Histogram of Anomaly Score
Frequency
0
20
40
60
0.35
0.40
0.45
0.50
0.55
Anomaly Score

FIGURE 2. TWO-FACTOR MODEL FIT ACROSS ANOMALY SCORE DISTRIBUTION

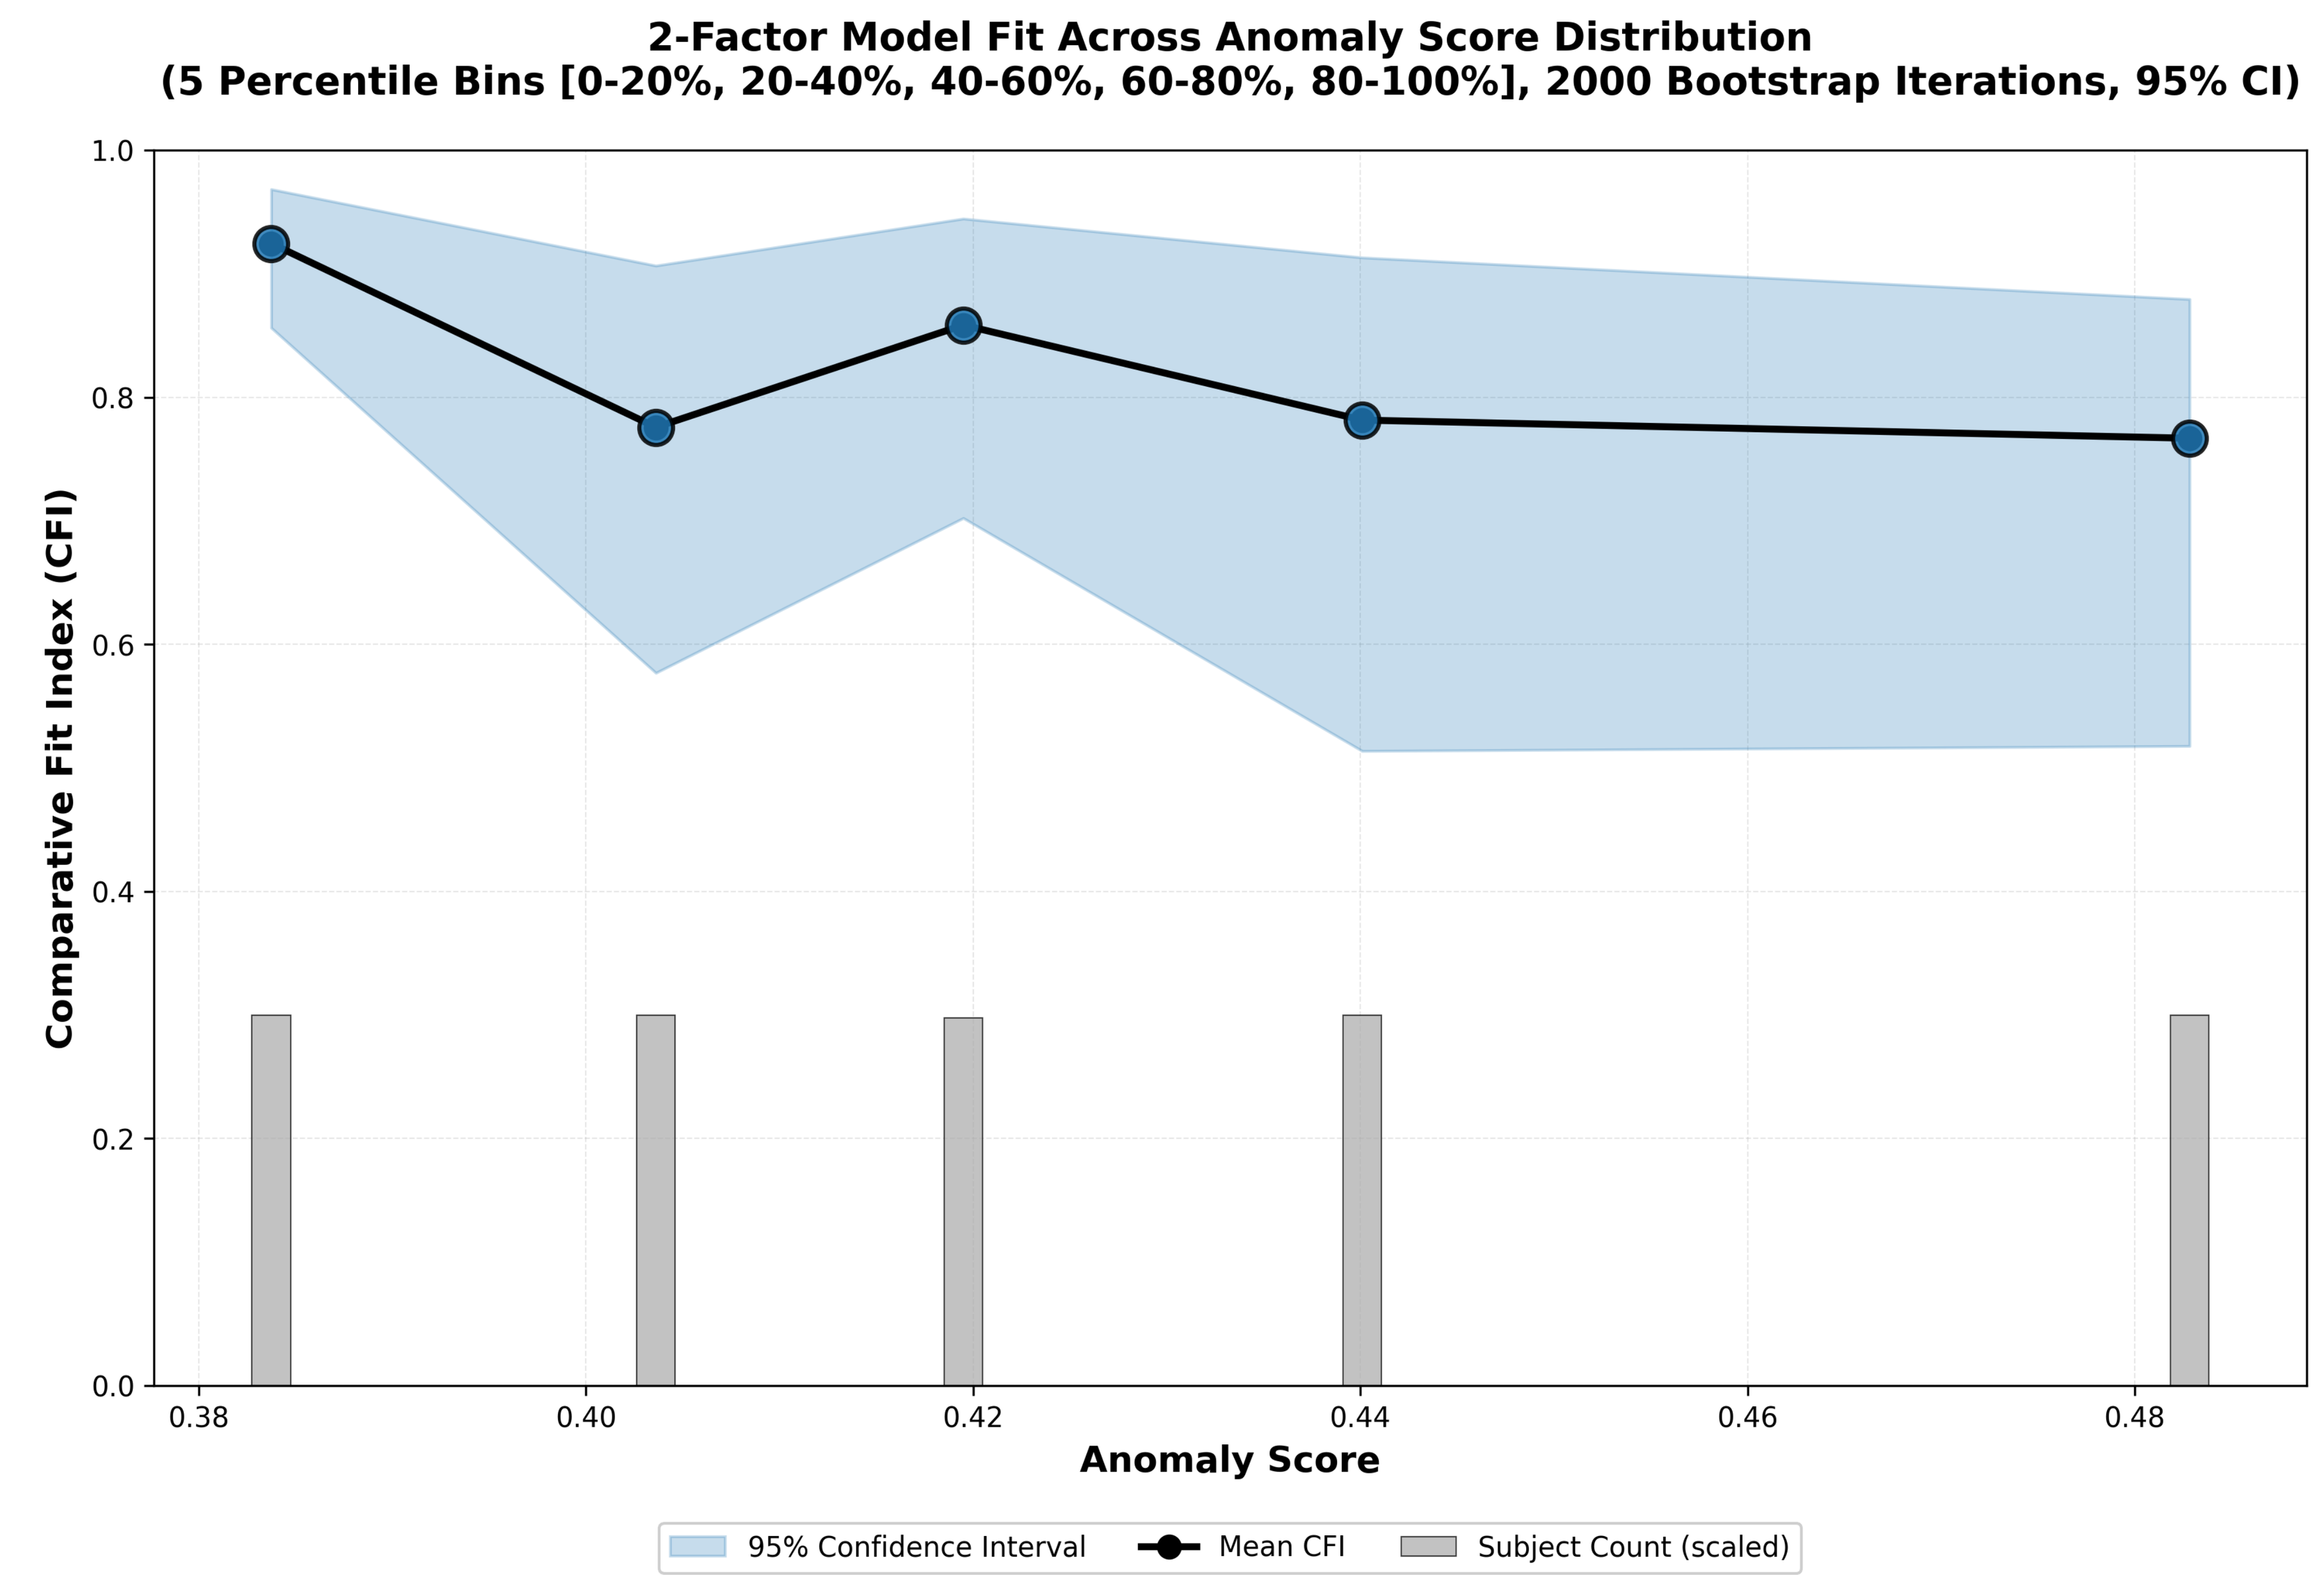
2-Factor Model Fit Across Anomaly Score Distribution
(5 Percentile Bins [0-20%, 20-40%, 40-60%, 60-80%, 80-100%], 2000 Bootstrap Iterations, 95% CI)
Comparative Fit Index (CFI)
0.0
0.2
0.4
0.6
0.8
1.0
0.38
0.40
0.42
0.44
0.46
0.48
Anomaly Score
95% Confidence Interval
Mean CFI
Subject Count (scaled)

CANONICAL SUBJECTS DEMONSTRATED SUPERIOR FIT TO THE 2-FACTOR HAM-A MODEL ACROSS ALL PERCENTILE DEFINITIONS:

	Non-Canonical				Canonical			
Canonical Definition	N	Cut-off	CFI	RMSEA	N	Cut-off	CFI	RMSEA
0% - 90%	63	>=0.477	**0.895**	**0.136**	572	<0.477	**0.827**	**0.118**
0% - 80%	127	>=0.455	**0.792**	**0.154**	508	<0.455	**0.837**	**0.114**
0% - 70%	190	>=0.44	**0.785**	**0.146**	445	<0.44	**0.877**	**0.102**
0% - 60%	254	>=0.428	**0.816**	**0.134**	381	<0.428	**0.886**	**0.098**
0% - 50%	315	>=0.42	0.824	0.136	320	<0.42	0.869	0.099
0% - 40%	397	>=0.412	0.815	0.133	256	>0.412	0.909	0.091
0% - 20%	507	>=0.395	0.80	1.134	128	<0.395	0.959	0.082

Key Observations

- Canonical subjects consistently show higher CFI values for most cut-offs (range: 0.827-0.959) compared to non-canonical subjects (range: 0.792-0.895)
- Canonical subjects consistently show lower RMSEA values (range: 0.098-0.118) compared to non-canonical subjects (range: 0.134-0.154)

DISCUSSION

- These preliminary results support the portability of VCD symptom-structure enrichment from prior PANSS and MADRS applications to HAM-A.
- A subject-level VCD vector derived from screening and baseline HAM-A item scores, coupled with unsupervised anomaly detection, identified subjects whose anxiety symptoms better conformed to a prespecified psychic/somatic HAM-A construct.
- The method is not intended as a responder-enrichment strategy or a diagnosis classifier; rather, it is an instrument-validity enrichment strategy designed to reduce nondrug-related variability by selecting subjects whose symptom presentation is more canonical for the rating scale used as an endpoint.
- Further work is needed to prespecify an operational threshold, evaluate cross-study robustness, examine demographic and clinical representativeness, test item functioning with item response theory, and determine whether canonical HAM-A symptom structure improves endpoint signal under a locked analysis plan.
- These early findings suggest a path toward reducing Type II risk from measurement variability in anxiety trials, while guarding against Type I error by anchoring enrichment to pre-randomization psychometric validity rather than treatment separation.
- Prior research shows that this approach reduces measurement noise and nondrug-related variability and increases statistical power to detect drug effects.

REFERENCES

1. Hopkins SC, Tomioka S, Szabo ST, Koblan KS. A clinical trial inclusion criteria to enrich for patients presenting with canonical symptom structure in bipolar depression. Contemporary Clinical Trials. 2024;145:107644.
2. Hopkins SC, et al. Assessment of negative symptoms in clinical trials of acute schizophrenia: test of a novel enrichment strategy. Schizophrenia Bulletin Open. 2022;3(1) :sgac027.
3. Liu FT, Ting KM, Zhou ZH. Isolation Forest. Proceedings of the 8th IEEE International Conference on Data Mining (ICDM). 2008.

SIGNANT HEALTH
THE EVIDENCE GENERATION COMPANY®