Association Between SCID Screening Question Flags and Screen Failure Outcomes in Major Depressive Disorder Trials

Authors: Sharp, IR[1]., Echevarria, B[1]., Negash, S[1]., Blattner, R[1]., and Opler, M[1].

[1]Clario, a part of Thermo Fisher Scientific, Philadelphia, PA USA

Methodological Issue Being Addressed

Clinical trial screening for major depressive disorder (MDD) relies heavily on structured diagnostic interviews and symptom severity scales, which may not fully capture contextual or exclusionary clinical features relevant to eligibility. The SCID Screening Questions (SSQ) are a brief adjunctive tool to identify diagnostic inconsistencies, comorbidities, and protocol-specific exclusion criteria early in screening. However, their utility as an independent screening instrument when administered by independent raters has not been systematically evaluated.

This study examines whether the SSQ can serve as a reliable, informative adjunct to traditional screening methods in large, multi-site clinical trials.
We evaluated the frequency, type, and distribution of SSQ flags and their relationship to screen failure outcomes, including MADRS-based and non-MADRS-based exclusions.

Introduction

The SSQ is a modifiable, short screening tool used to assess DSM disorders and inclusion/exclusion criteria, with items derived from the SCID for the DSM (First et al., 2016). It is intended as an independent assessment of diagnostic inclusion/exclusion criteria and overall appropriateness for trial participation (e.g., cognitive impairment), used in addition to a site-based diagnostic interview.

The SSQ generates “flags” that prompt re-evaluation of a subject’s suitability for the trial. We evaluated the usefulness of the SSQ, administered by independent raters at the screening visit, to identify participants who may meet protocol-exclusionary diagnostic criteria leading to screen failure, as well as the type, frequency, and number of SSQ flags and their relation to inclusion/exclusion outcomes.

References

First, M. B., Williams, J. B. W., Karg, R. S., & Spitzer, R. L. (2016). Structured Clinical Interview for DSM-5 Disorders, Clinician Version (SCID-5-CV). American Psychiatric Association.

Methods

We analyzed aggregated data from three global, multi-site Phase III MDD trials that included the SSQ (N = 1,802) administered by independent, remote raters as a separate supplement to the SCID done at the research sites. Logistic regressions examined associations between number and type of SSQ flags and screen failure. Of 1,802 subjects, 925 (51.3%) were enrolled and 877 (48.7%) failed screening; 211 screen failures were due to MADRS criteria and 666 to non-MADRS reasons.

Results

Across the cohort, 1,550 SSQ flags were triggered. Subjects who failed screening had more SSQ flags than enrolled subjects (1.17 ± 1.75 vs. 0.57 ± 1.10); this difference remained when subjects who failed solely on MADRS criteria were excluded (1.21 ± 1.90 vs. 0.57 ± 1.10).

Mean Number of Flags by Enrollment Condition

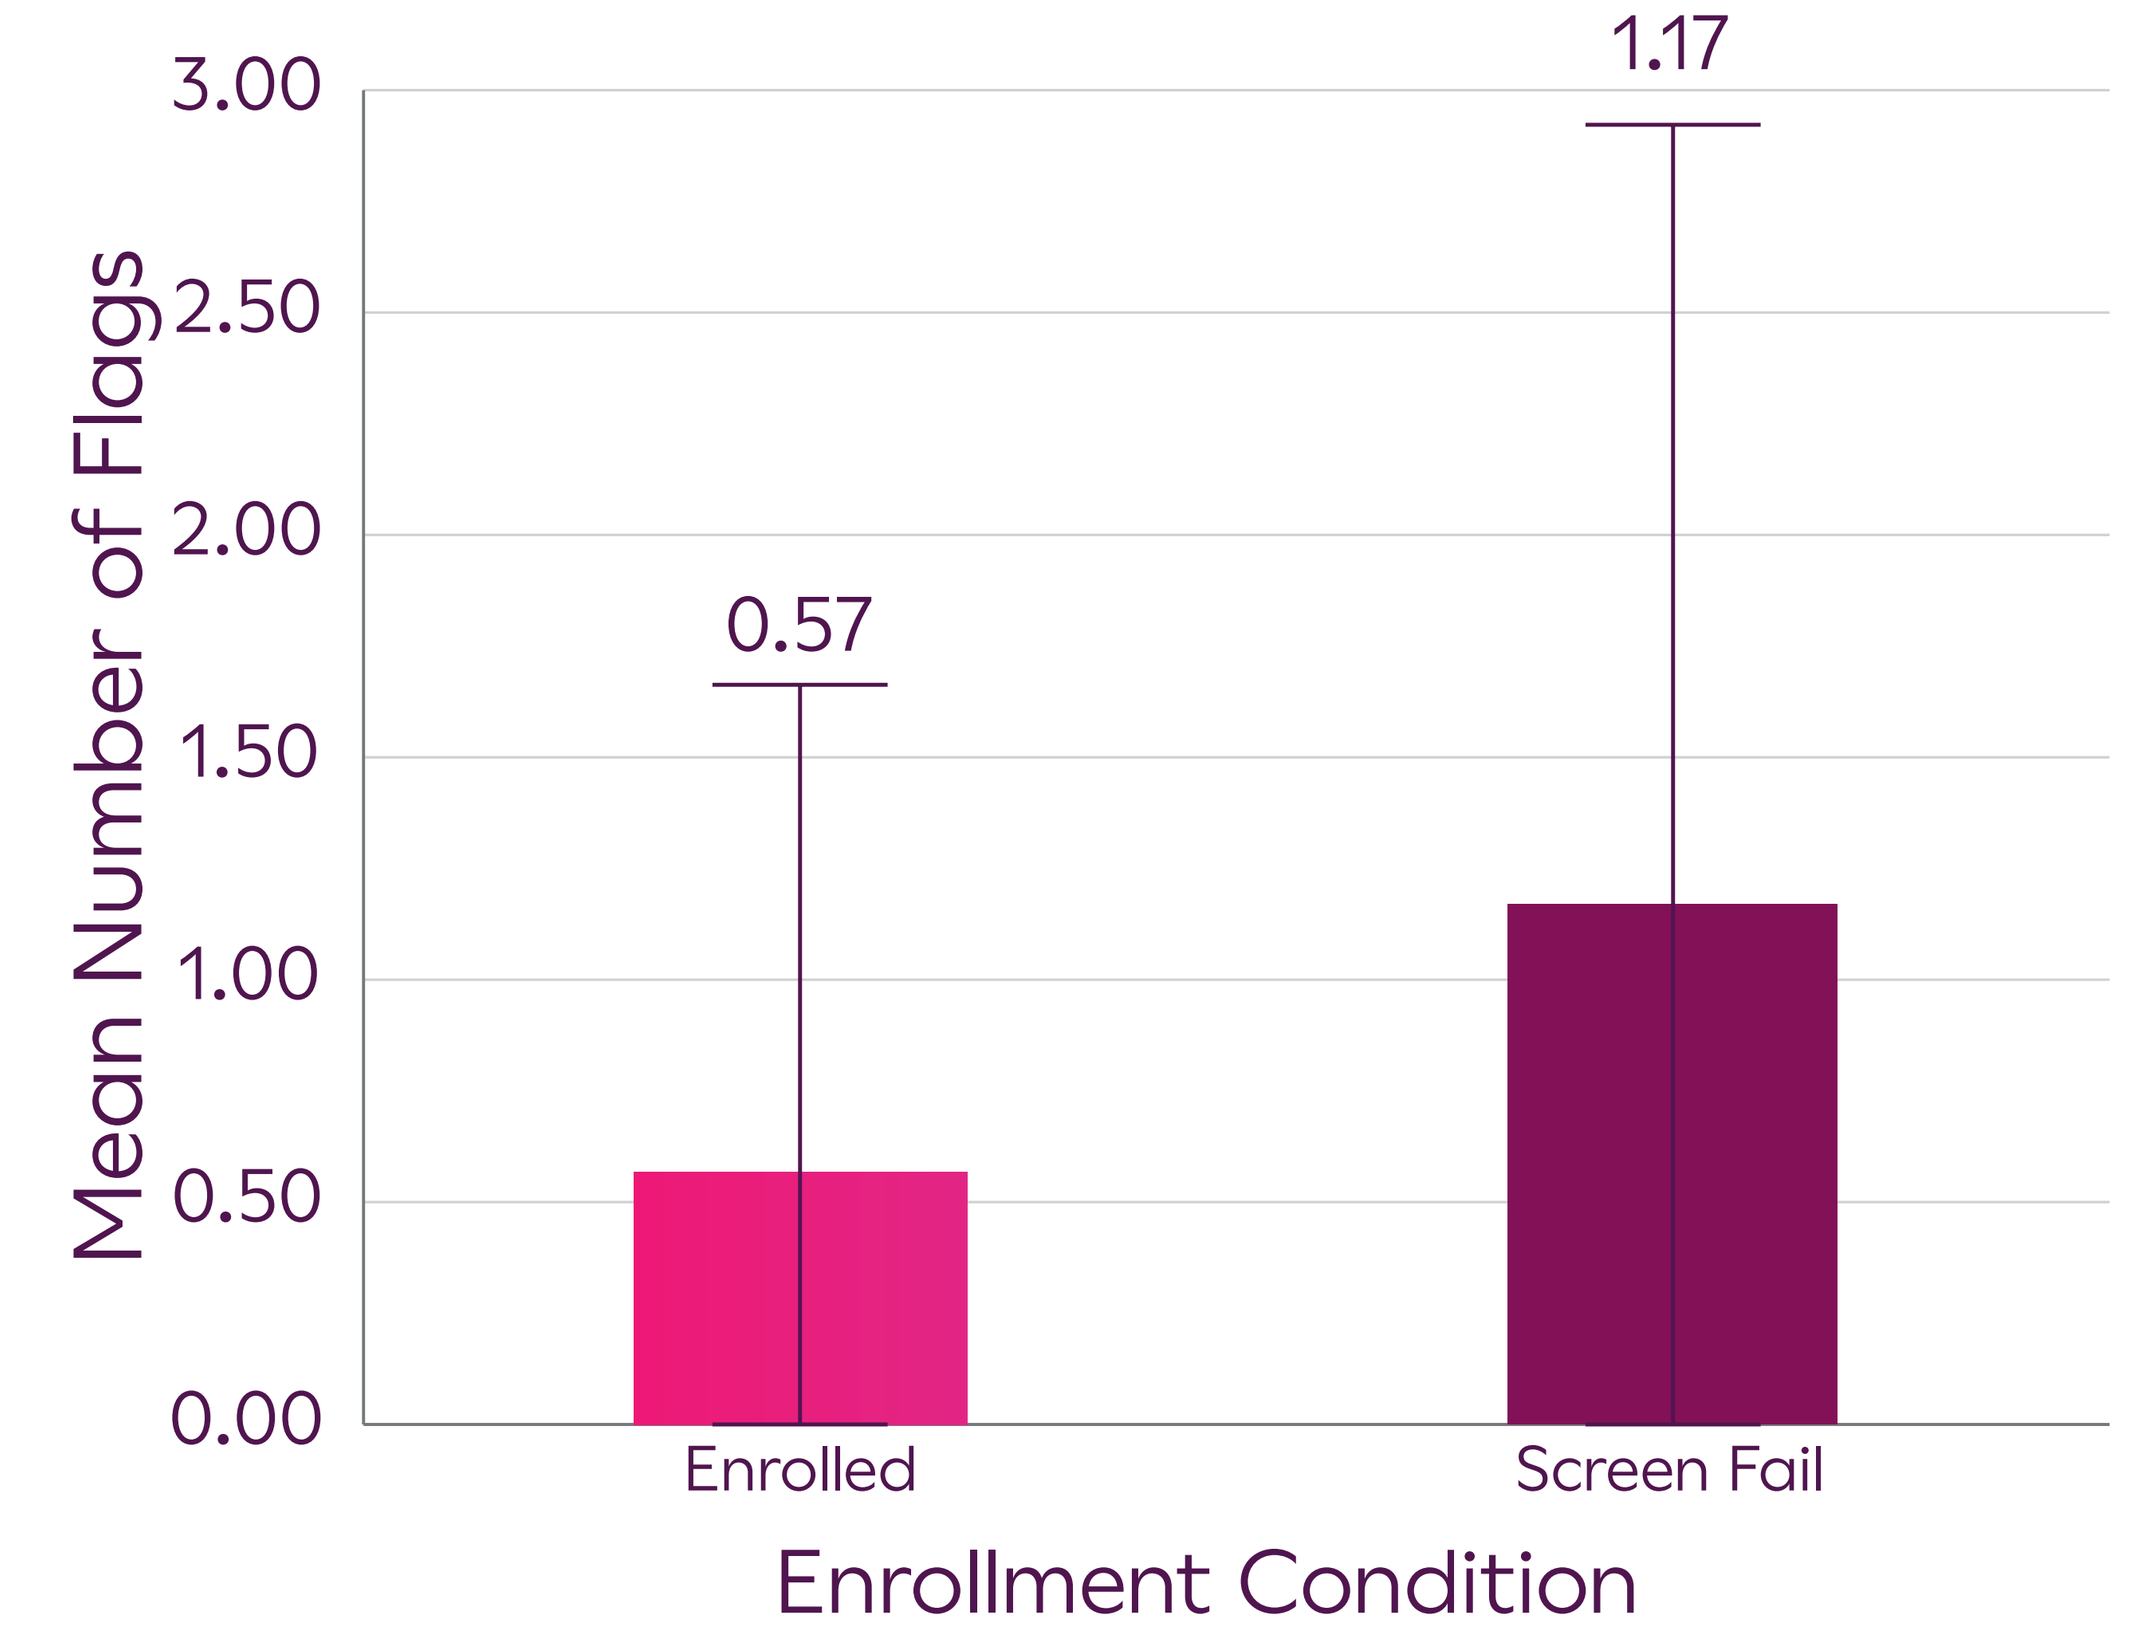
3.00
2.50
2.00
1.50
1.00
0.50
0.00
Mean Number of Flags
0.57
1.17
Enrolled
Screen Fail
Enrollment Condition

Number of Flags by Item

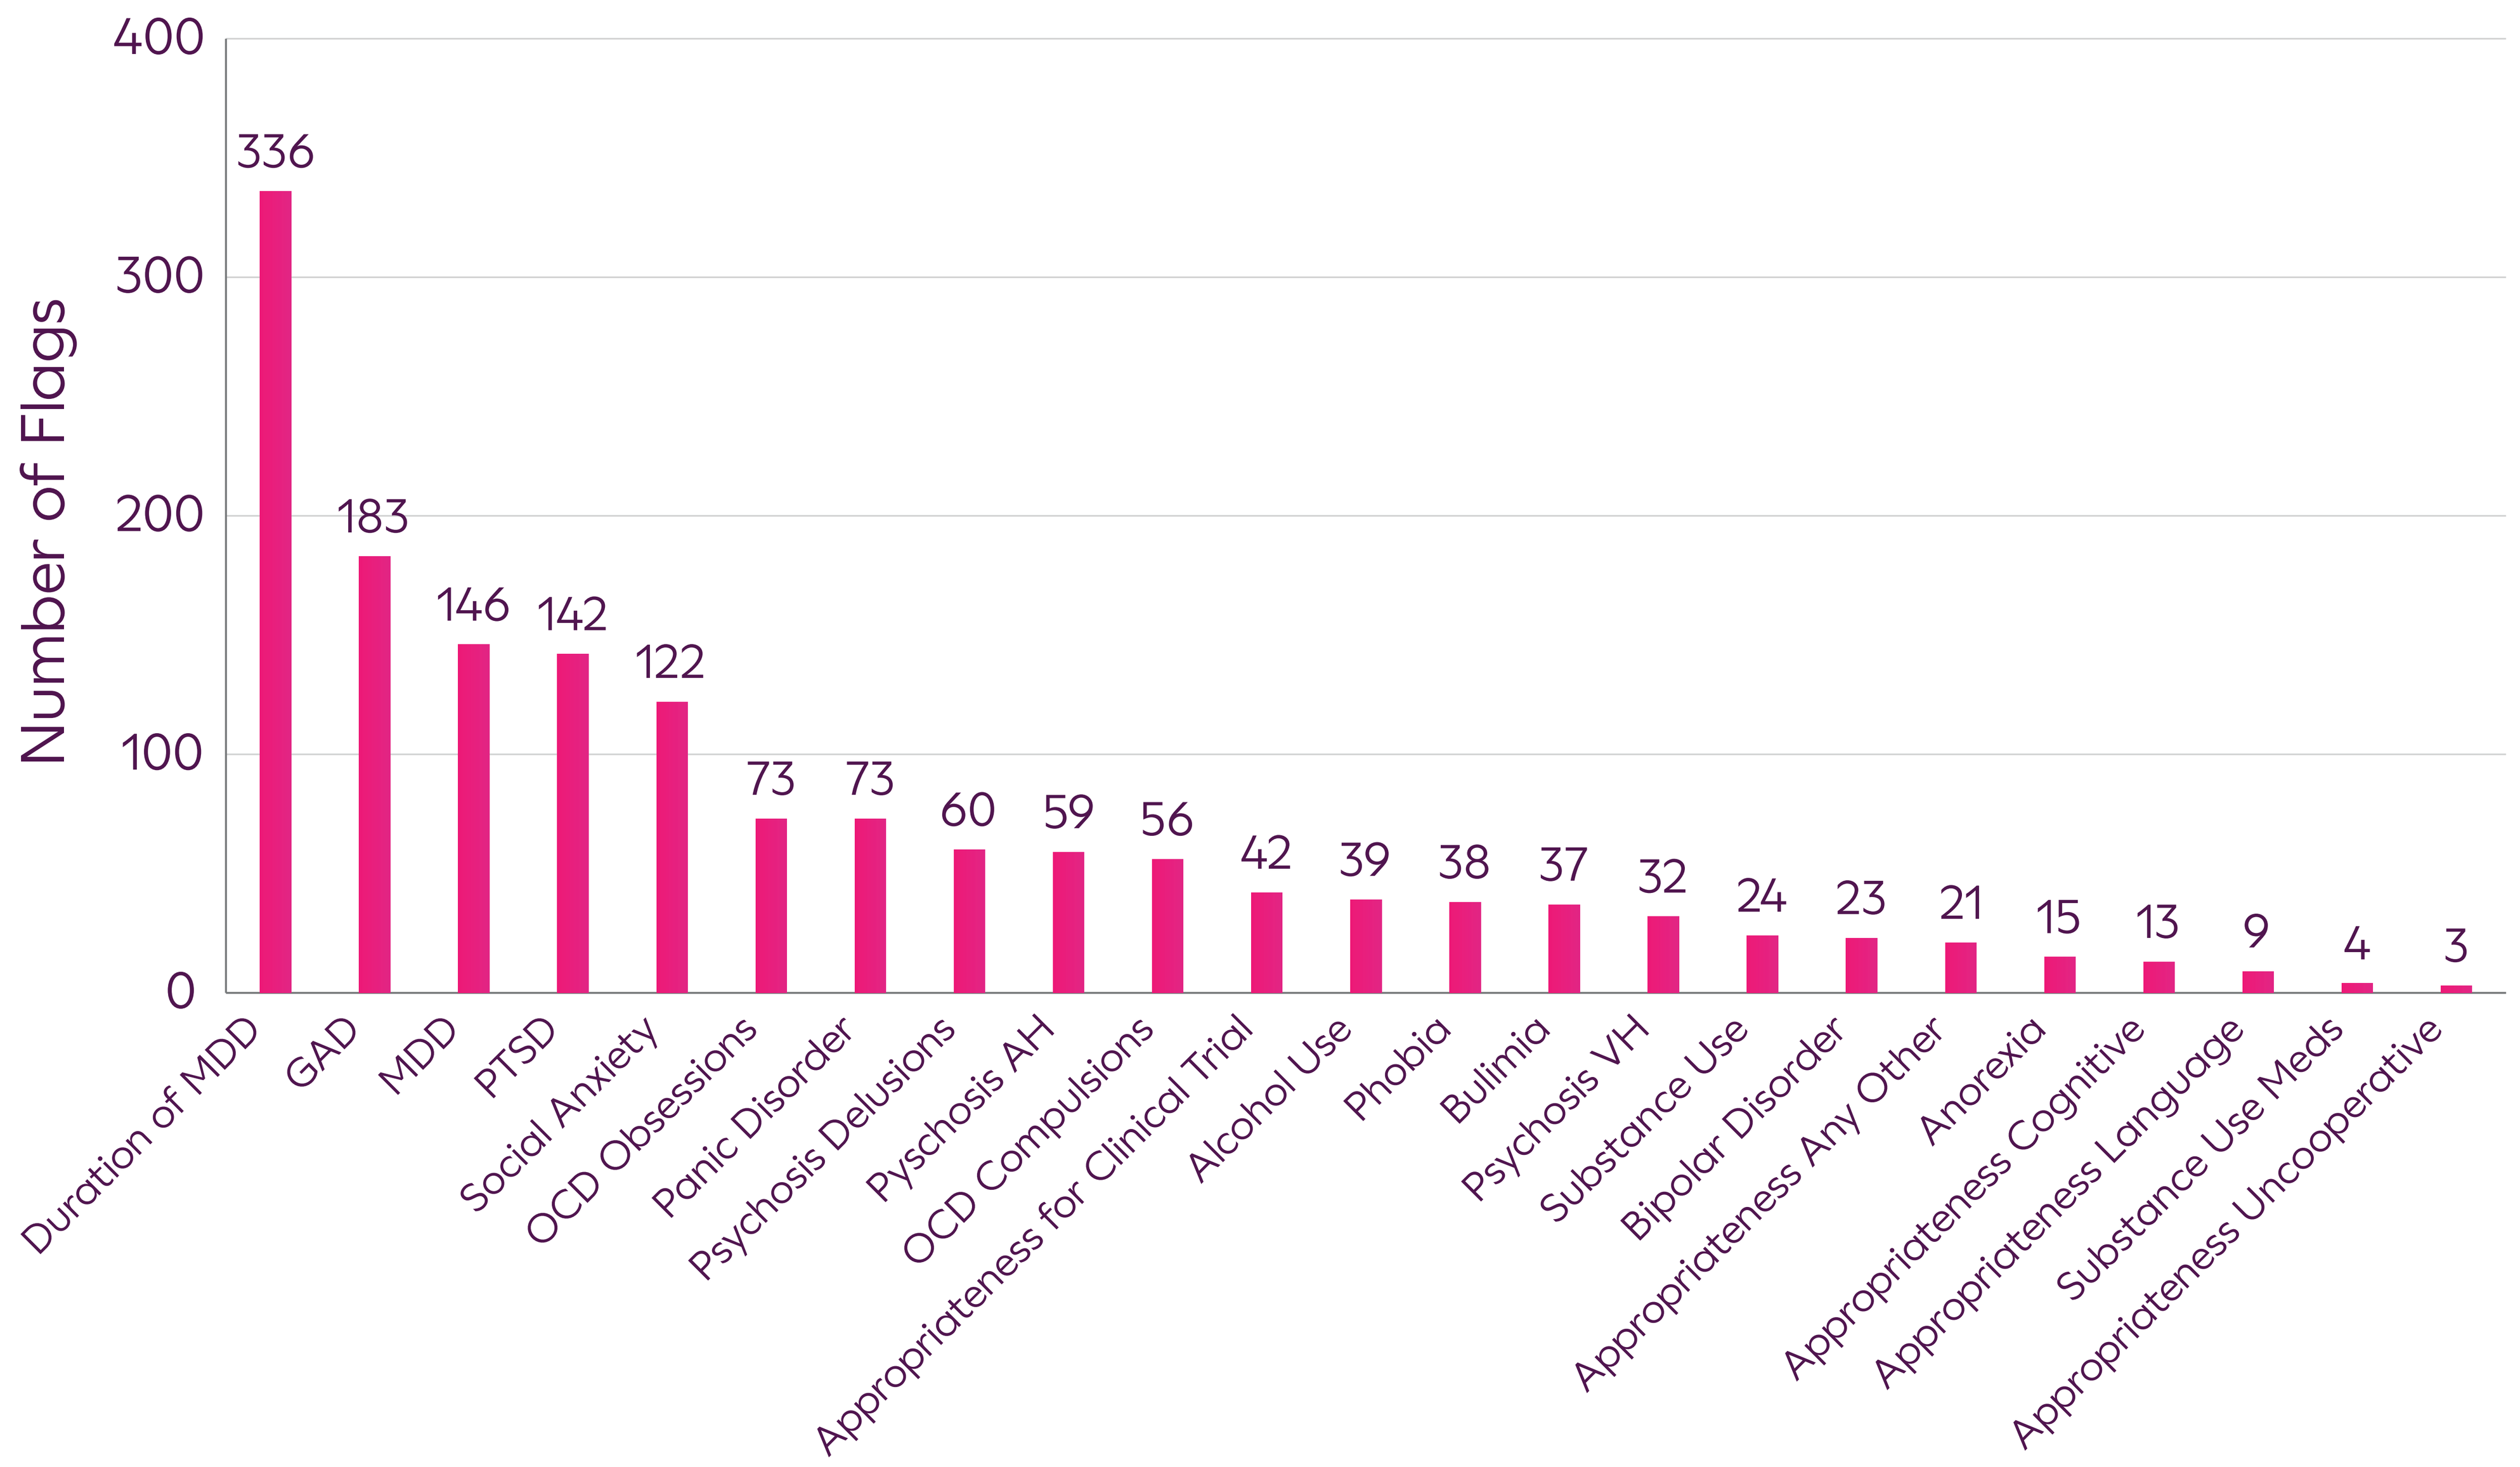
400
300
200
100
0
Number of Flags
336
183
146
142
122
73
73
60
59
56
42
39
38
37
32
24
23
21
15
13
9
4
3
Duration of MDD
GAD
MDD
PTSD
Social Anxiety
OCD Obsessions
Panic Disorder
Psychosis Delusions
Pyschosis AH
OCD Compulsions
Appropriateness for Clinical Trial
Alcohol Use
Phobia
Bulimia
Psychosis VH
Substance Use
Bipolar Disorder
Appropriateness Any Other
Anorexia
Appropriateness Cognitive
Appropriateness Language
Substance Use Meds
Appropriateness Uncooperative

Logistic regression showed that the total number of SSQ flags was significantly associated with increased likelihood of screen failure (estimate = 0.3255, $p < .001$), and this association remained when MADRS-based exclusions were removed (estimate = 0.3089, $p < .001$).

Item-level analyses identified several SSQ domains associated with higher screen failure risk, including MDD diagnostic reconsideration (estimate = 1.704), duration of MDD (0.51–0.57), PTSD (0.72–0.79), and panic disorder (0.72–0.73). The most frequent SSQ flags involved duration of MDD, PTSD, and MDD diagnostic reconsideration, suggesting that diagnostic clarification and comorbidity assessment are key drivers of screening outcomes.

Conclusions

In summary, SSQ-derived flags are common in MDD trial screening and provide information beyond site-based diagnostic assessments and symptom scales. Both total and item-level flags were strongly associated with screen failure, including non-MADRS exclusions, indicating that the SSQ captures clinically relevant exclusion factors not reflected in severity thresholds. These findings support the SSQ as a complementary tool to improve early identification of ineligible participants and enhance screening efficiency in MDD trials.