Baseline PANSS Symptoms Predict Relapse Risk in Placebo-Treated Patients

Jonathan Rabinowitz[1,3] Michael Davidson[2,3] Helene Speyer[4] Mark Weiser[5] Remy Luthringer[3]

[1]Bar-Ilan University, Israel | [2]University of Nicosia Medical School, Cyprus | [3]Minerva Neurosciences, USA | [4]Mental Health Services in the Capital Region of Denmark | [5]Tel Aviv University, Israel

5
placebo-controlled relapse-prevention trials

557
placebo-randomized participants

HR 1.06 p = .05
per 1-point higher baseline PANSS Positive score

HR 0.94 p = .01
per 1-point higher baseline PANSS Negative score

METHODOLOGICAL ISSUE ADDRESSED

Identifying baseline symptom criteria that predict relapse is important for both schizophrenia trial design and clinical practice.

Depending on a trial's objective, enrollment criteria should either:

- select patients at **sufficient relapse risk** to detect a relapse-prevention effect, or
- select **lower-risk patients** in negative symptom or cognition studies.

Clarifying which baseline symptoms predict time to relapse could refine eligibility criteria and improve trial efficiency.

INTRODUCTION

We aimed to determine whether baseline **psychopathology**, measured on entry into double-blind treatment, predicts **survival time to relapse** among patients randomized to **placebo** in antipsychotic relapse-prevention studies.

We examined the independent associations of positive, negative, and general psychopathology symptoms with relapse risk.

METHODS

Data source
Individual-patient data on placebo-randomized participants from **five placebo-controlled relapse-prevention studies** with a double-blind phase of at least **6 months**, obtained through **Vivli** and **YODA**.

Sample
Placebo-randomized participants, **n = 557**.

Outcome
Survival time to relapse. Relapse was defined as a **12-point increase on PANSS total score** as per Siafis et al., 2024.

Statistical analysis
Cox proportional-hazards regression. Predictors were PANSS Positive, Negative, and General Psychopathology subscale scores, age, and sex.

RESULTS

- Higher baseline **positive** symptoms were associated with **increased** relapse risk (HR = 1.06, p = .05).
- Higher baseline **negative** symptoms were associated with **decreased** relapse risk (HR = 0.94, p = .01).
- Age and sex were retained as covariates.

Baseline predictor	HR	p
PANSS Positive	1.06	.05
PANSS Negative	0.94	.01

Table 1. Cox proportional-hazards model: baseline PANSS subscale scores and time to relapse. Hazard ratios are per 1-point higher baseline score. Age and sex were retained as covariates.

Hazard ratio per 1-point higher baseline score (95% CI)

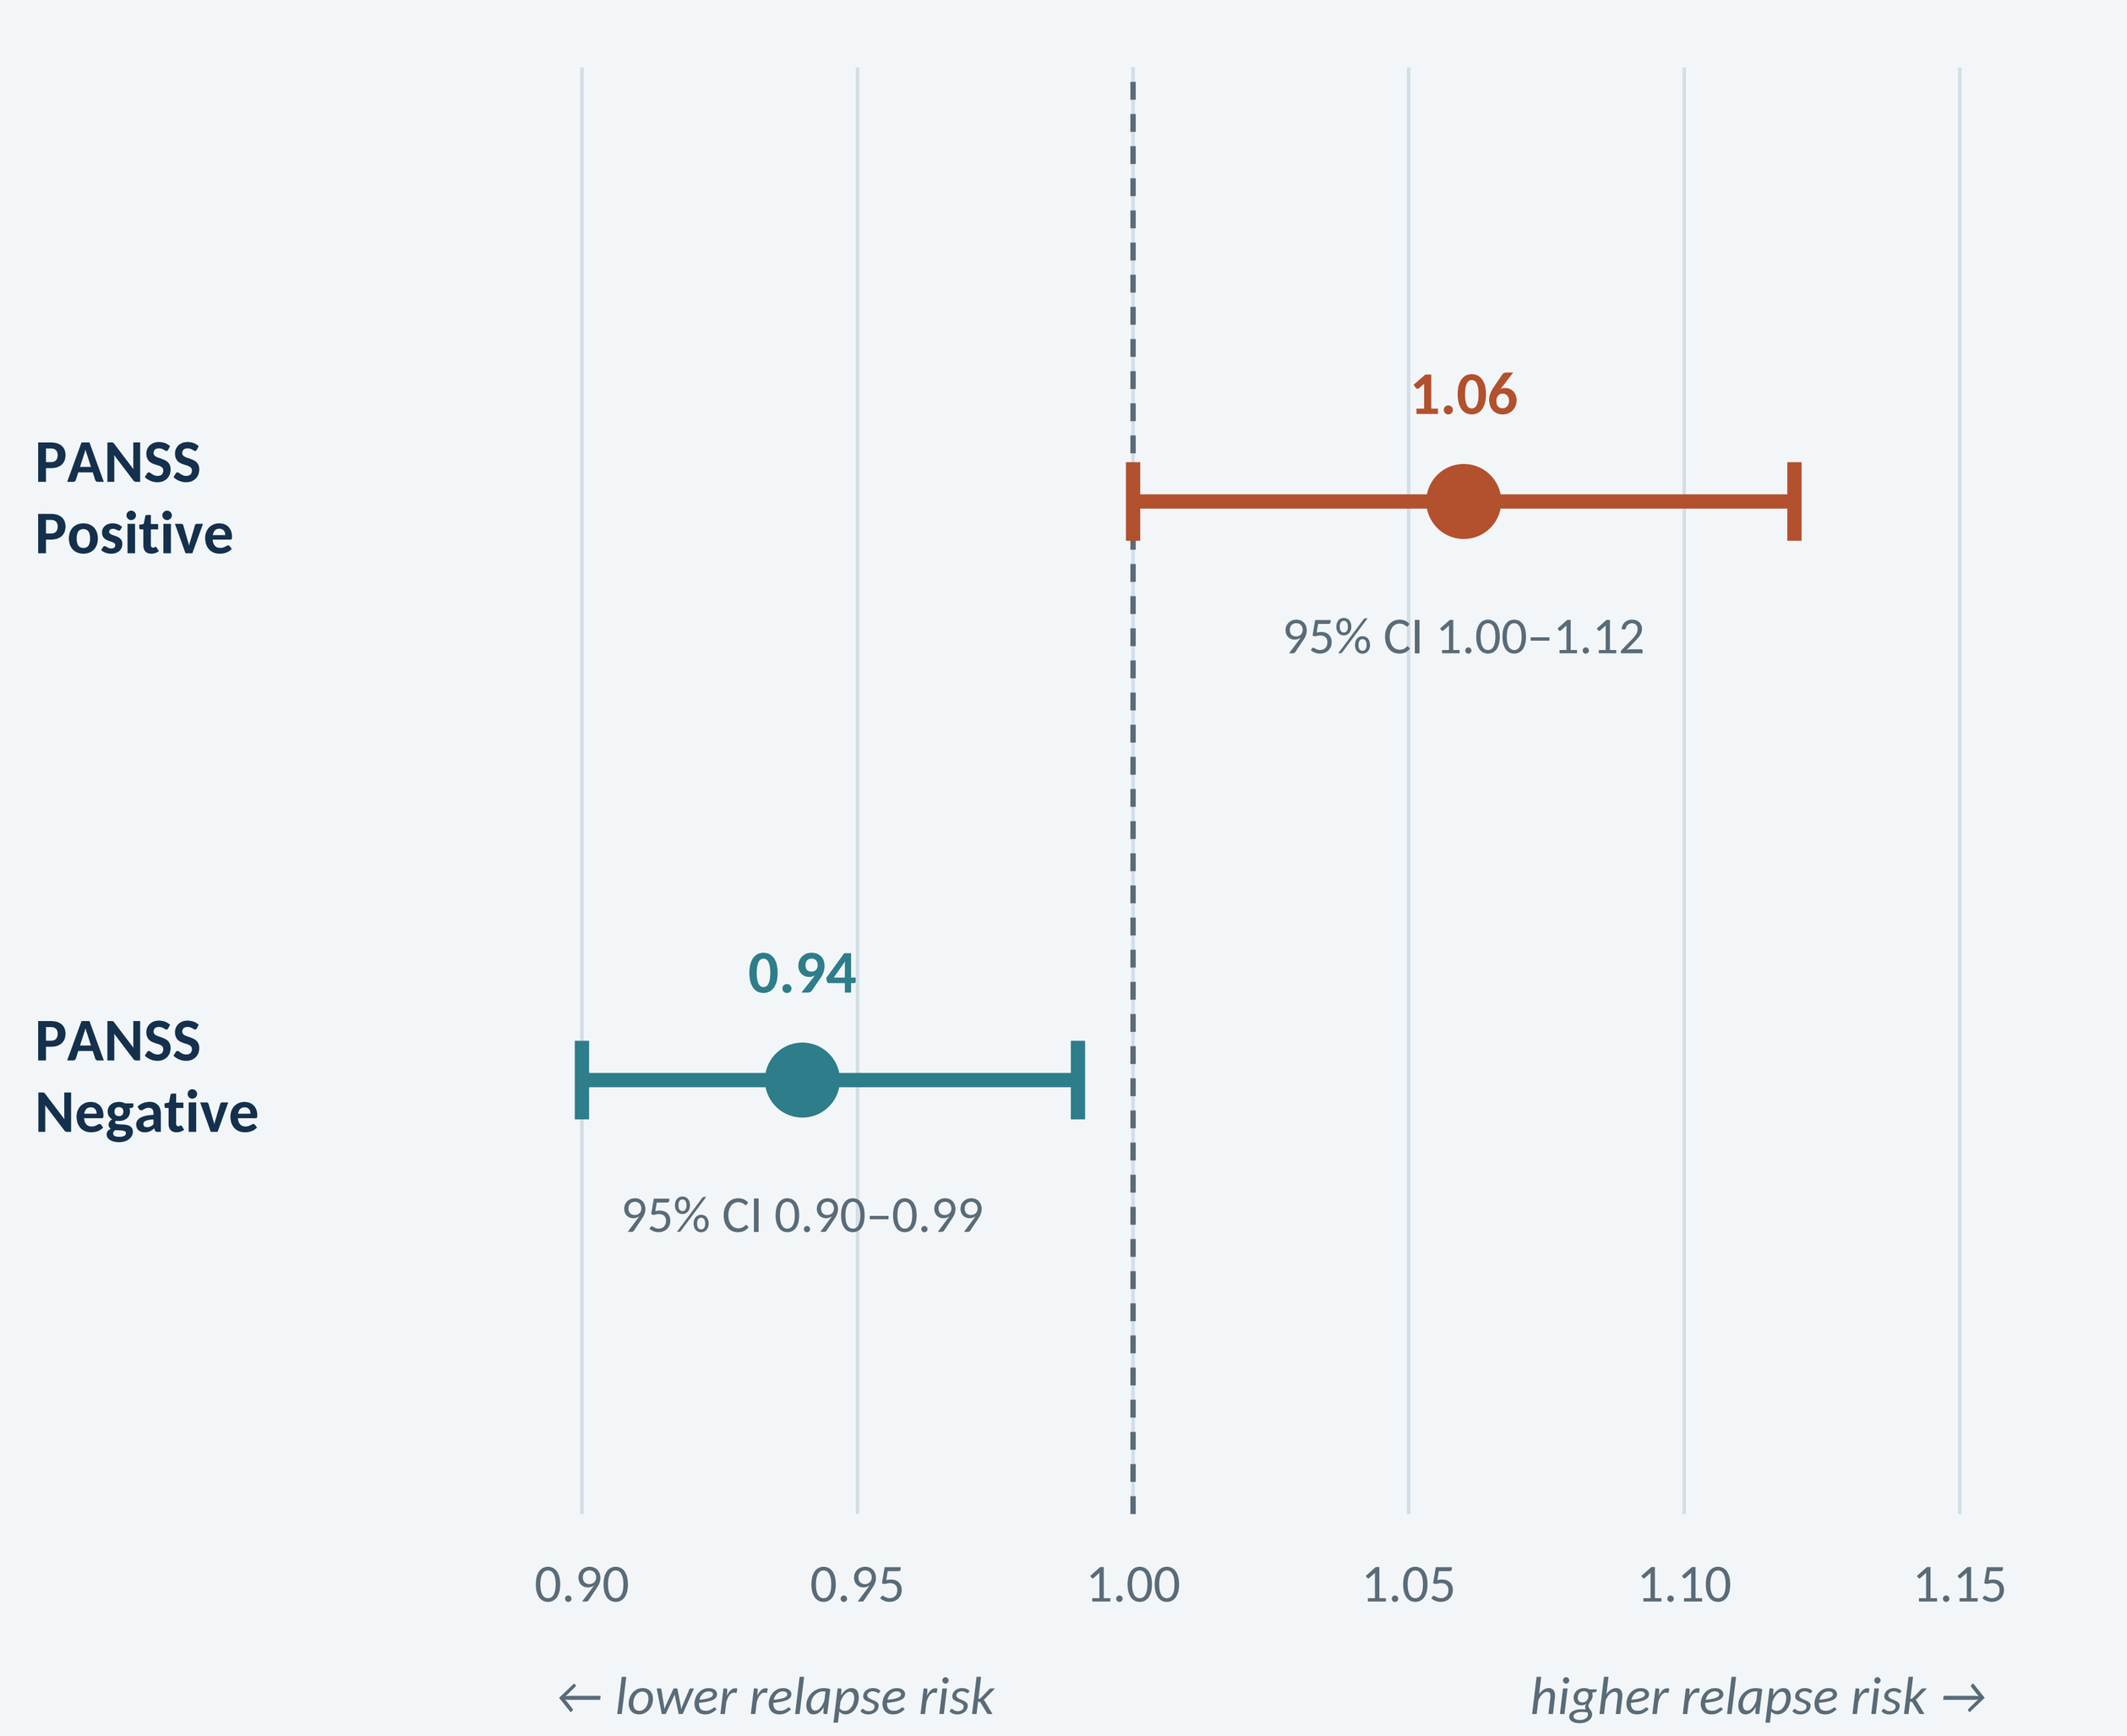
PANSS Positive
1.06
95% CI 1.00–1.12
PANSS Negative
0.94
95% CI 0.90–0.99
0.90
0.95
1.00
1.05
1.10
1.15
← lower relapse risk
higher relapse risk →

Figure 1. Hazard ratios with 95% confidence intervals. Points to the right of 1.0 indicate higher relapse risk; points to the left indicate lower relapse risk. Confidence intervals are derived from the reported hazard ratios and p-values.

MODEL-IMPLIED EFFECT SIZE

Model-implied hazard ratio by score difference

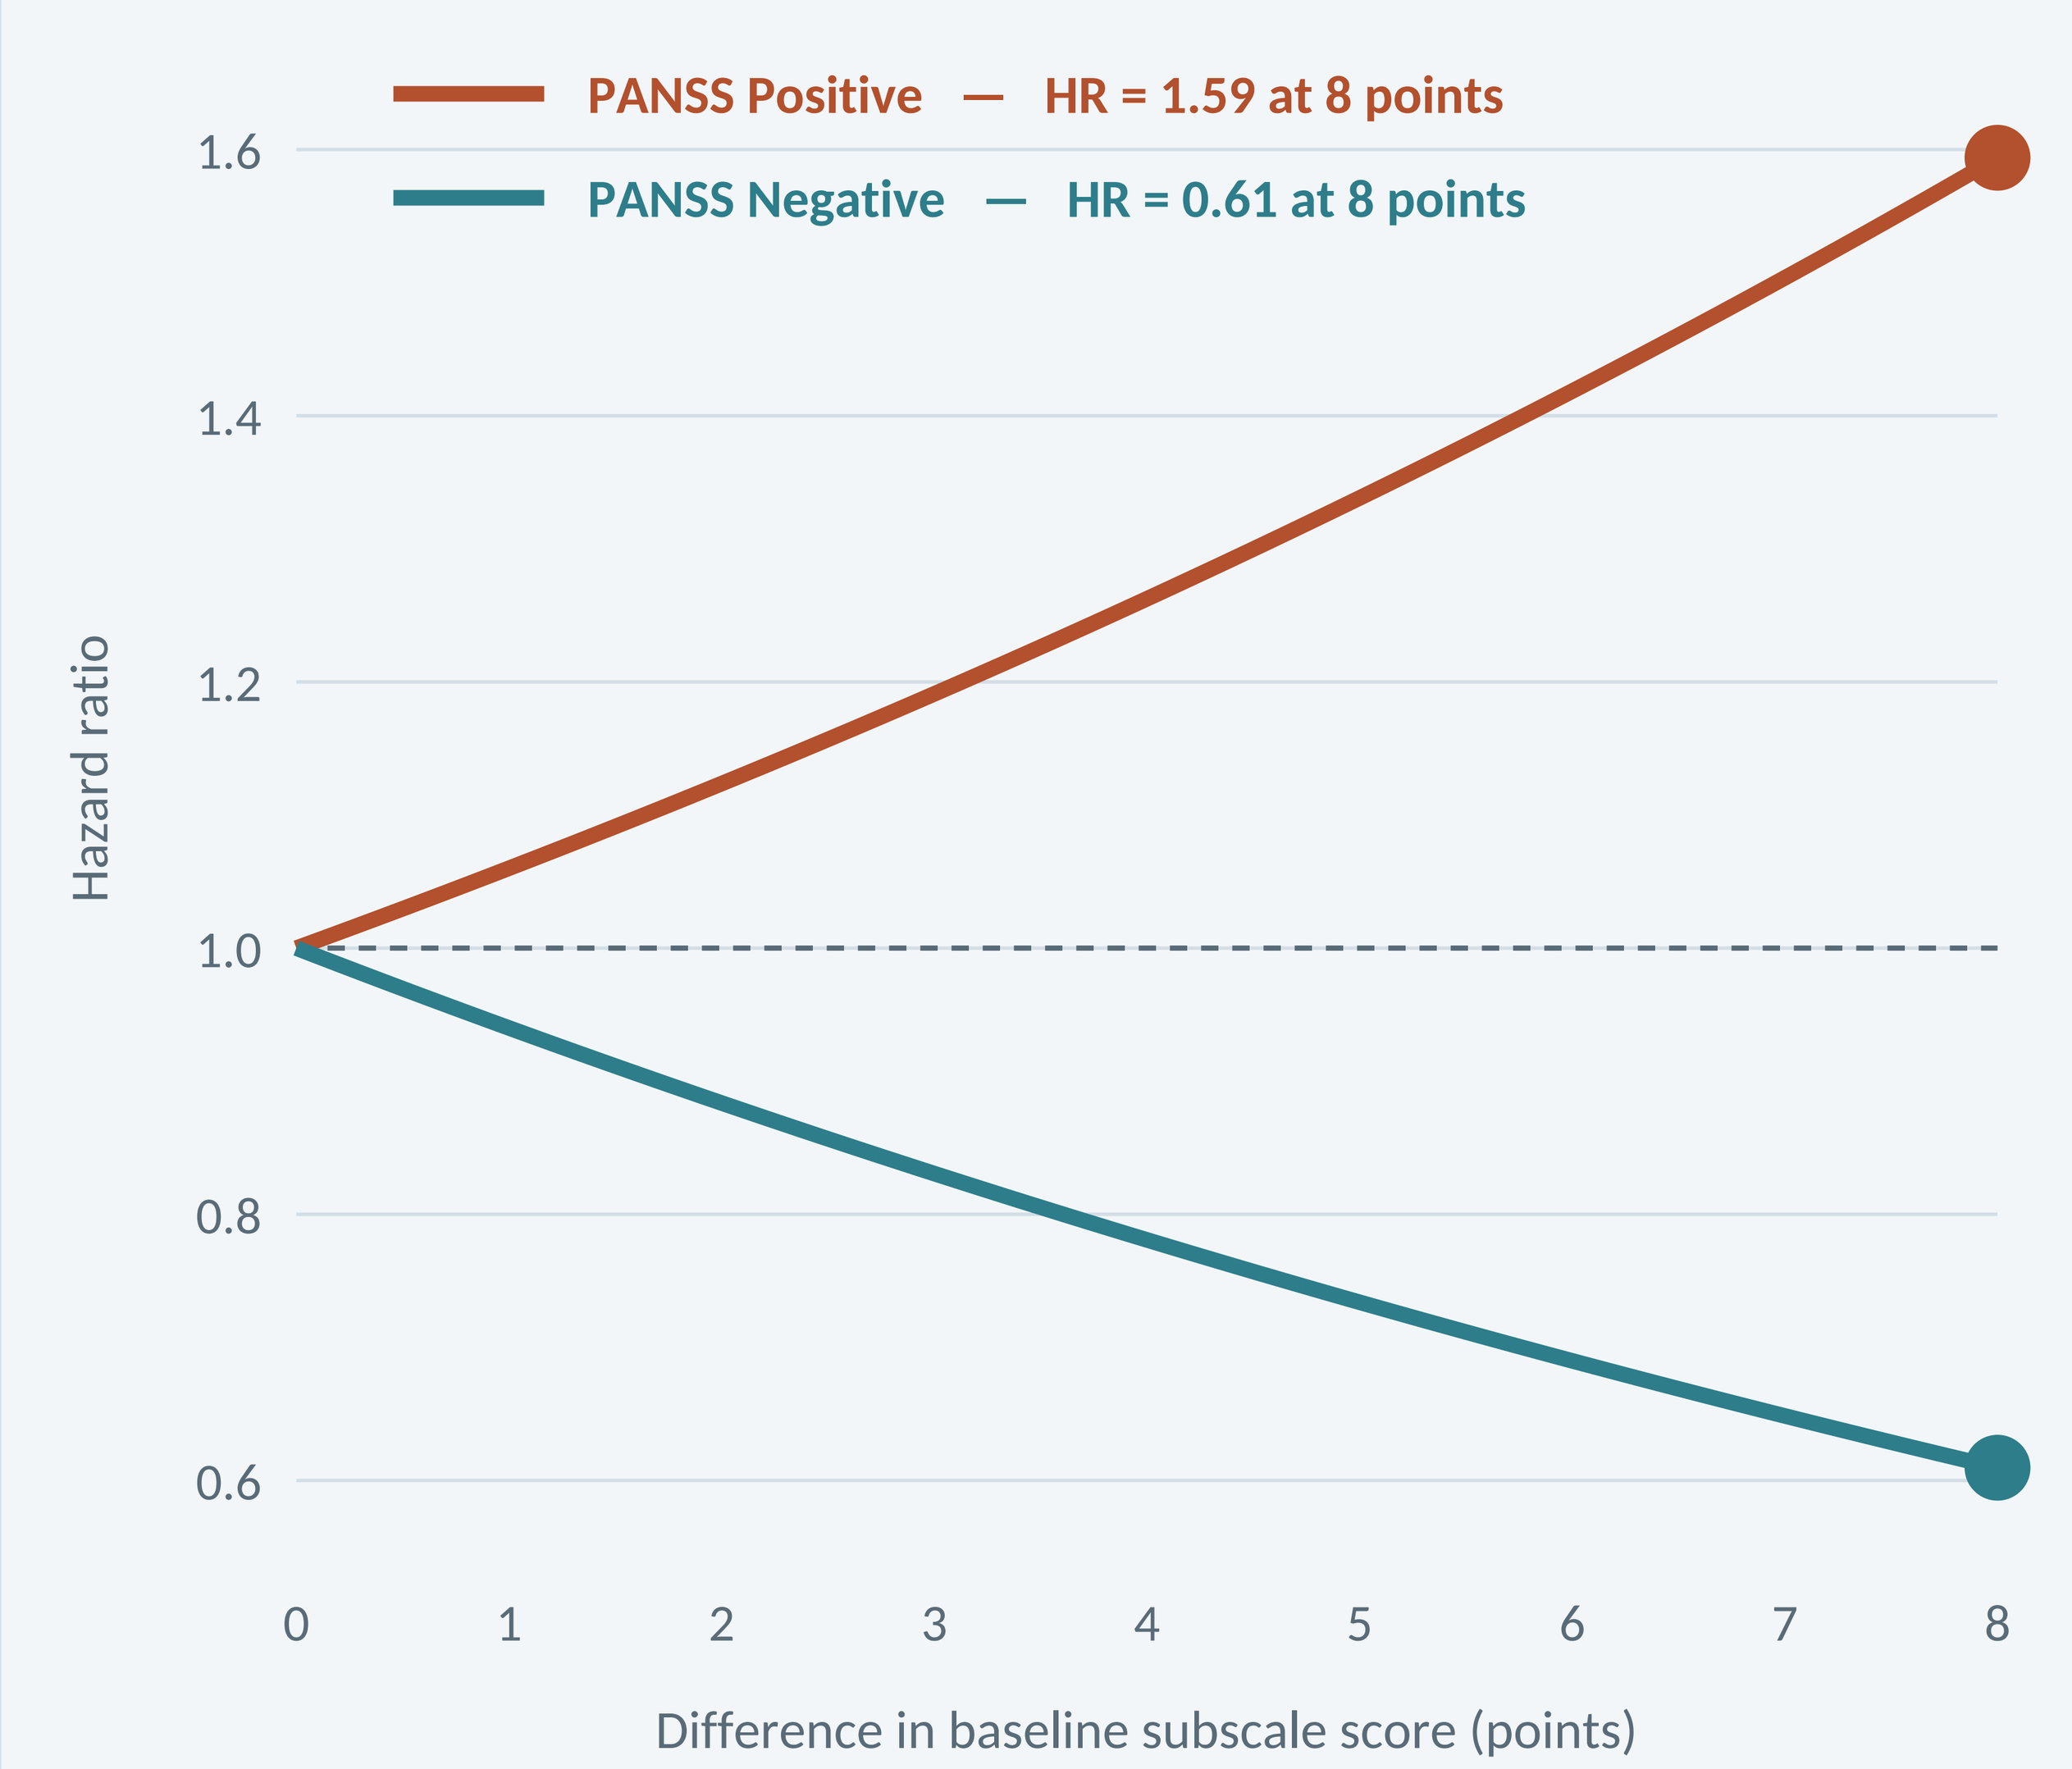
PANSS Positive — HR = 1.59 at 8 points
PANSS Negative — HR = 0.61 at 8 points
Hazard ratio
1.6
1.4
1.2
1.0
0.8
0.6
0
1
2
3
4
5
6
7
8
Difference in baseline subscale score (points)

Figure 2. Hazard ratios implied by the model in Table 1 for between-patient differences of 1–8 points on a baseline subscale, holding other covariates constant.

CONSIDERATIONS FOR TRIAL DESIGN

Relapse-prevention trials
Selecting patients with **higher baseline positive** and **lower baseline negative** symptoms may yield a sample at greater relapse risk, which is needed to detect a relapse-prevention effect.

Negative symptom or cognition trials
The opposite baseline profile may identify a **lower-risk sample**, in which relapse is a competing event rather than the outcome of interest.

These directions are hypothesis-generating and require prospective testing before they are used as eligibility criteria.

CONCLUSIONS

- Among placebo-treated patients, baseline symptom profiles predicted relapse risk in **opposing directions**: positive symptoms increased risk, whereas negative symptoms appeared protective.
- Baseline PANSS Positive and Negative subscale scores may help refine **eligibility and enrichment criteria** in clinical trials and improve trial efficiency.
- **Prospective validation is warranted.**

LIMITATIONS

- Post-hoc analysis of pooled data from previously completed trials that differed in design, duration, and relapse ascertainment.
- Relapse was operationalized by a single PANSS-based criterion (a 12-point increase on the total score).
- Analyses were restricted to the placebo-randomized subsample; findings may not generalize to actively treated patients.
- Associations are observational within randomized arms and are not causal.

ACKNOWLEDGMENTS AND DATA SOURCE

This poster is based on research using data from: Johnson & Johnson, Eli Lilly, Otsuka Pharmaceuticals that has been made available through Vivli, Inc. Vivli has not contributed to or approved, and is not in any way responsible for, the contents of this publication.

REFERENCE

Siafis, S., Brandt, L., McCutcheon, R.A., Gutwinski, S., Schneider-Thoma, J., Bighelli, I., Kane, J.M., Arango, C., Kahn, R.S., Fleischhacker, W.W., McGorry, P., Carpenter, W.T., Falkai, P., Hasan, A., Marder, S.R., Schooler, N., Engel, R.R., Honer, W.G., Buchanan, R.W., Davidson, M., Weiser, M., Priller, J., Davis, J.M., Howes, O.D., Correll, C.U., Leucht, S., 2024. Relapse in clinically stable adult patients with schizophrenia or schizoaffective disorder: evidence-based criteria derived by equipercentile linking and diagnostic test accuracy meta-analysis. The Lancet Psychiatry 11(1), 36-46.

Disclosures

One or more authors report potential conflicts which are described in the program