Comparison of CAINS and PANSS as Assessments of Negative Symptoms in Outpatients with Schizophrenia in a Clinical Trial Setting

Longenecker, J[1]; Snipes, C[1]; Livezey, V[1]

[1]Click Therapeutics, Inc., New York, NY, USA

Methodological Issue Addressed

The Positive and Negative Syndrome Scale (PANSS) - the established outcome for clinical trials in schizophrenia - emphasizes observable behaviors, has incomplete coverage of negative symptoms, and may be less sensitive to change, particularly in populations with stable positive symptoms, compared to more contemporary assessments.

Introduction

- Negative symptoms are the primary source of functional impairment in schizophrenia, making accurate detection of therapeutic signal essential.
- While the PANSS has long served as the gold standard, its Marder Negative Factor Score (NSFS) heavily emphasizes observable behavioral deficits and the expressive domain.
- The CAINS was developed to capture contemporary models of negative symptoms with greater sensitivity and less confounding variance. It operationalizes the NIMH consensus two-factor model, differentiating the Motivation and Pleasure (MAP) and Expression (EXP) domains.
- This study provides a descriptive comparison of the convergent validity and longitudinal sensitivity of both scales.

Methods

Exploratory, post hoc analysis of the **CONVOKE RCT** evaluating CT-155 (a 16-week digital therapeutic for negative symptoms) vs. digital control in schizophrenia outpatients (n=457) and data from the **Open Label Extension** study in a subset of participants (n=73) who completed a second CT-155 course.

Measures

- **PANSS:** 7-item NSFS, 2-item proxy MAP, 2-item proxy EXP
- **CAINS:** MAP, EXP, total score

Statistical Analysis

- **Convergent validity** between CAINS and PANSS was compared using Spearman correlations, with 95% confidence intervals (CIs) calculated using a non-parametric bootstrap (based on the 2.5th and 97.5th percentiles of 5000 bootstrap samples).
- **Longitudinal sensitivity** of CAINS-MAP and PANSS NSFS was explored using Mixed Models for Repeated Measures (MMRM), including the fixed effects of week as a categorical variable and baseline value.

CT-155 Studies and Assessment Timeline

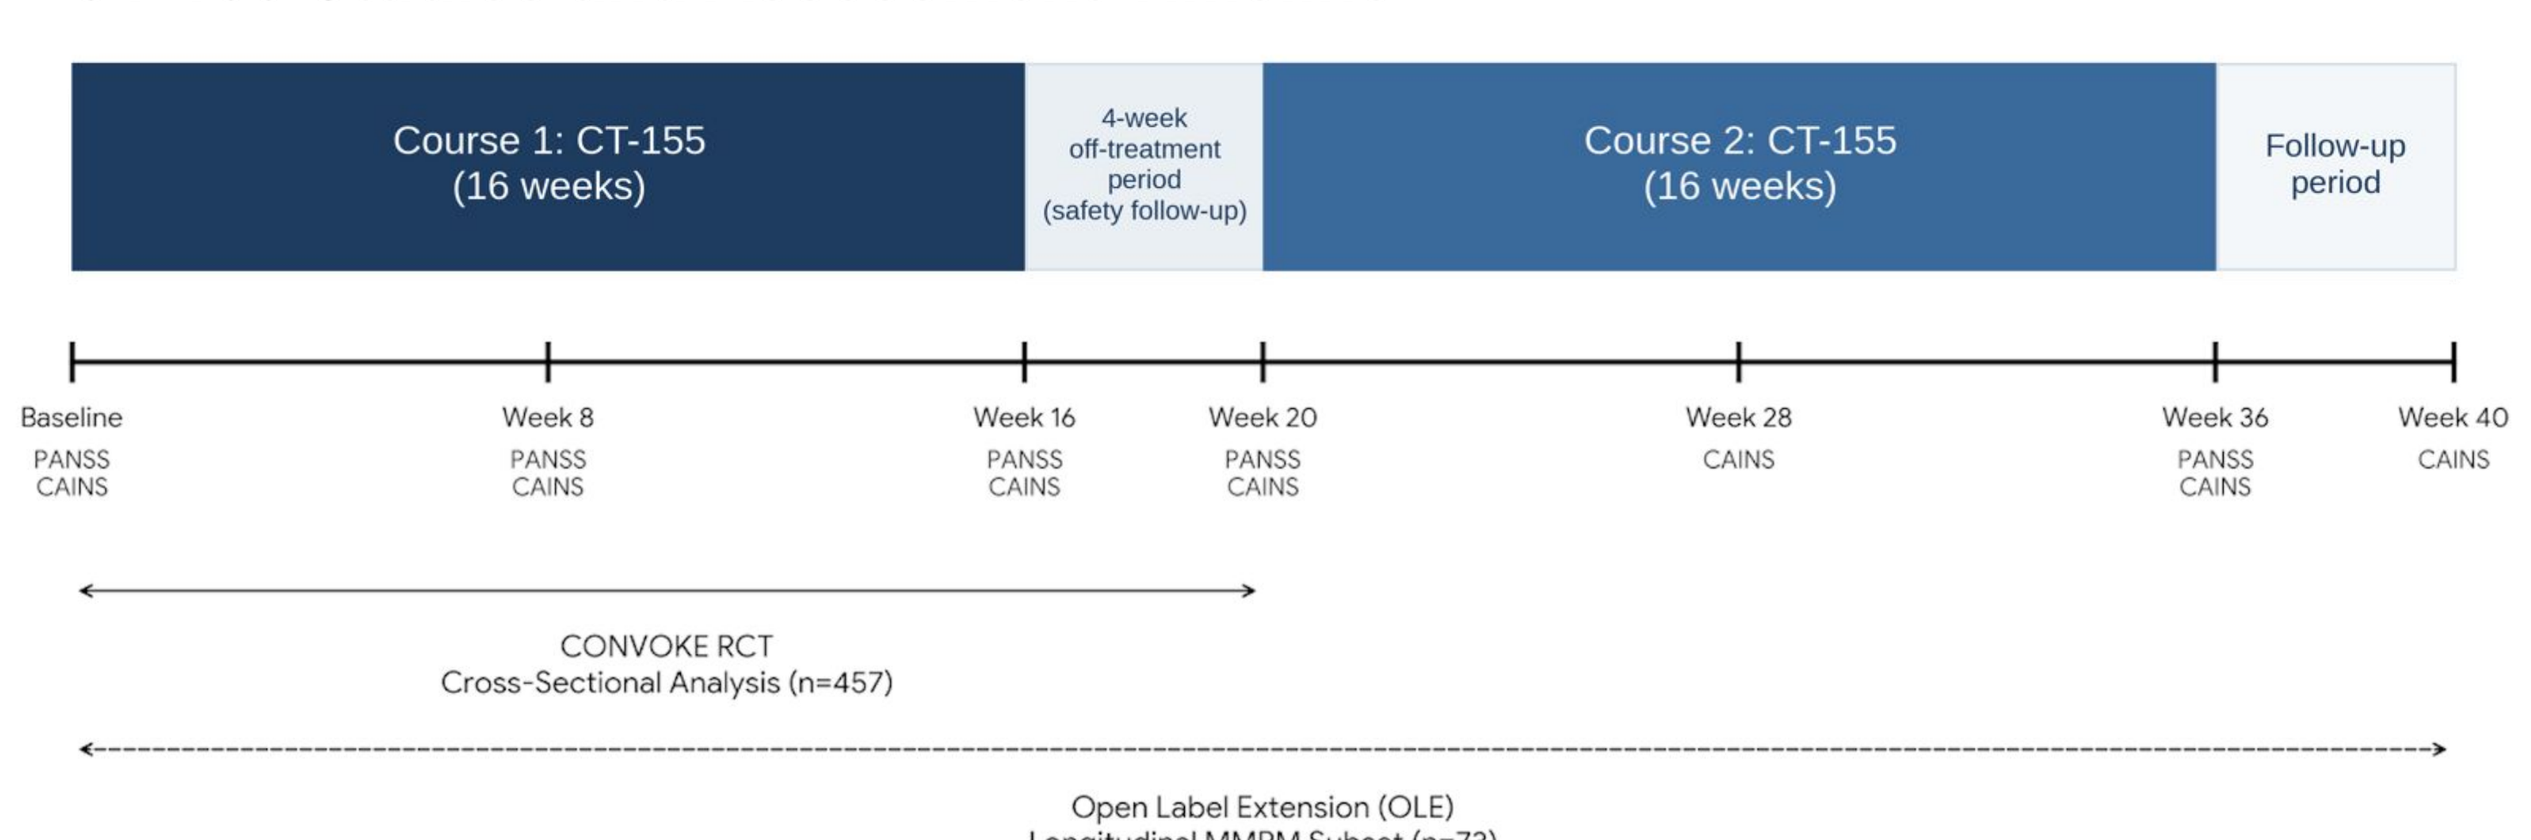
Course 1: CT-155 (16 weeks)
4-week off-treatment period (safety follow-up)
Course 2: CT-155 (16 weeks)
Follow-up period
Baseline PANSS CAINS
Week 8 PANSS CAINS
Week 16 PANSS CAINS
Week 20 PANSS CAINS
Week 28 CAINS
Week 36 PANSS CAINS
Week 40 CAINS
CONVOKE RCT Cross-Sectional Analysis (n=457)
Open Label Extension (OLE) Longitudinal MMRM Subset (n=73)

Results

Convergent validity (cross-sectional correlations)

Correlations with PANSS NSFS were stronger, and statistically significant, for CAINS-EXP (ρBL=0.19, 95% CI [0.10, 0.28]; ρw16=0.29, 95% CI [0.19, 0.38]) than CAINS-MAP (ρBL=0.01, 95% CI [-0.08, 0.11], not significant; ρw16=0.12, 95% CI [0.02, 0.22]) or Total (ρBL=0.10, 95% CI [0.01, 0.19]; ρw16=0.21, 95% CI [0.10, 0.30]) scores at Baseline and Week 16, consistent with the known overemphasis of expressive negative symptoms in PANSS.

However, NSFS and CAINS-EXP change scores showed a non-significant, lower convergence (ρ=0.04, 95% CI [-0.07, 0.14]) than cross-sectional correlations at each time point.

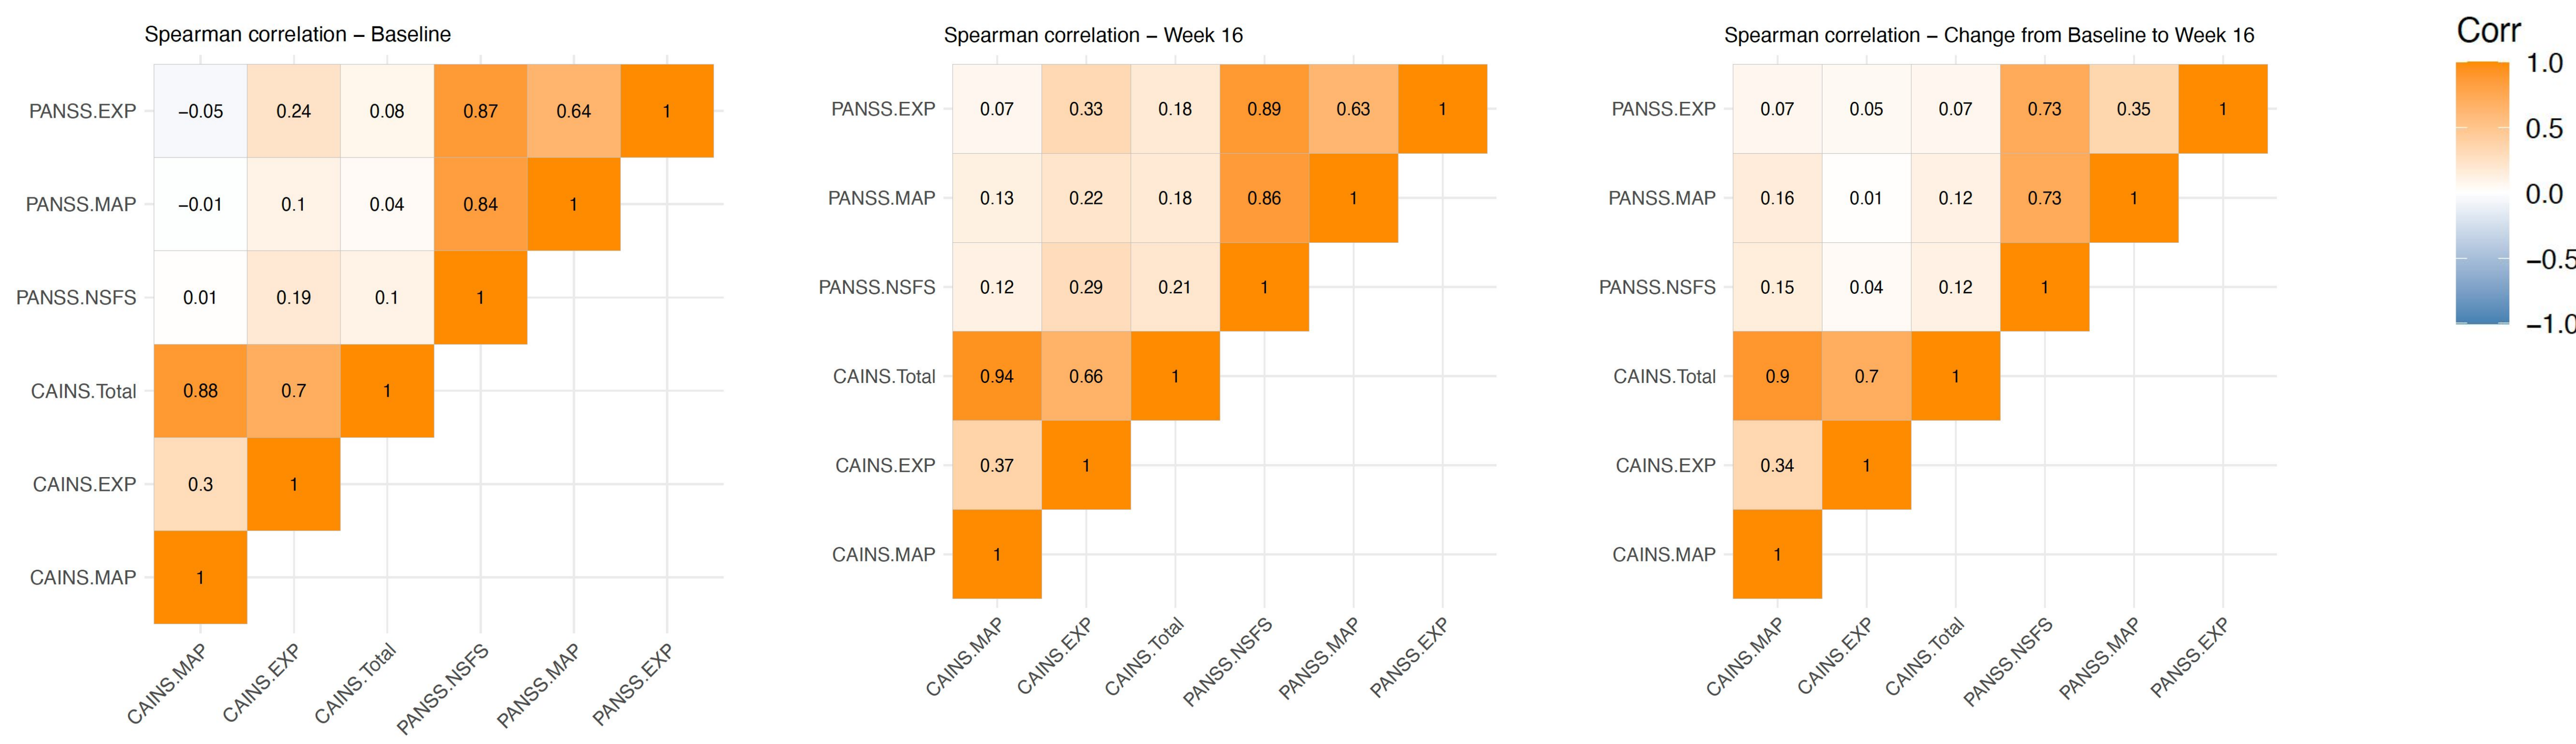
Spearman correlation – Baseline
PANSS.EXP −0.05 0.24 0.08 0.87 0.64 1
PANSS.MAP −0.01 0.1 0.04 0.84 1
PANSS.NSFS 0.01 0.19 0.1 1
CAINS.Total 0.88 0.7 1
CAINS.EXP 0.3 1
CAINS.MAP 1
CAINS.MAP CAINS.EXP CAINS.Total PANSS.NSFS PANSS.MAP PANSS.EXP
Spearman correlation – Week 16
PANSS.EXP 0.07 0.33 0.18 0.89 0.63 1
PANSS.MAP 0.13 0.22 0.18 0.86 1
PANSS.NSFS 0.12 0.29 0.21 1
CAINS.Total 0.94 0.66 1
CAINS.EXP 0.37 1
CAINS.MAP 1
CAINS.MAP CAINS.EXP CAINS.Total PANSS.NSFS PANSS.MAP PANSS.EXP
Spearman correlation – Change from Baseline to Week 16
PANSS.EXP 0.07 0.05 0.07 0.73 0.35 1
PANSS.MAP 0.16 0.01 0.12 0.73 1
PANSS.NSFS 0.15 0.04 0.12 1
CAINS.Total 0.9 0.7 1
CAINS.EXP 0.34 1
CAINS.MAP 1
CAINS.MAP CAINS.EXP CAINS.Total PANSS.NSFS PANSS.MAP PANSS.EXP
Corr
1.0
0.5
0.0
−0.5
−1.0

Longitudinal sensitivity (MMRM)

Within the subset of participants who completed two cycles of treatment, MMRM analysis demonstrated that CAINS-MAP captured an earlier, sustained reduction in negative symptoms, with no significant difference in scores between Week 16 and Week 36 (LS Mean diff = -0.05, 95% CI [-1.48, 1.39], p=0.95). Conversely, NSFS exhibited slower detection of symptom reduction, capturing a statistically significant continued reduction between Week 16 and Week 36 (LS Mean diff = -1.56, 95% CI [-2.56, -0.57], p=0.003).

CAINS-MAP Change from Baseline - LS Means (SE) vs Week

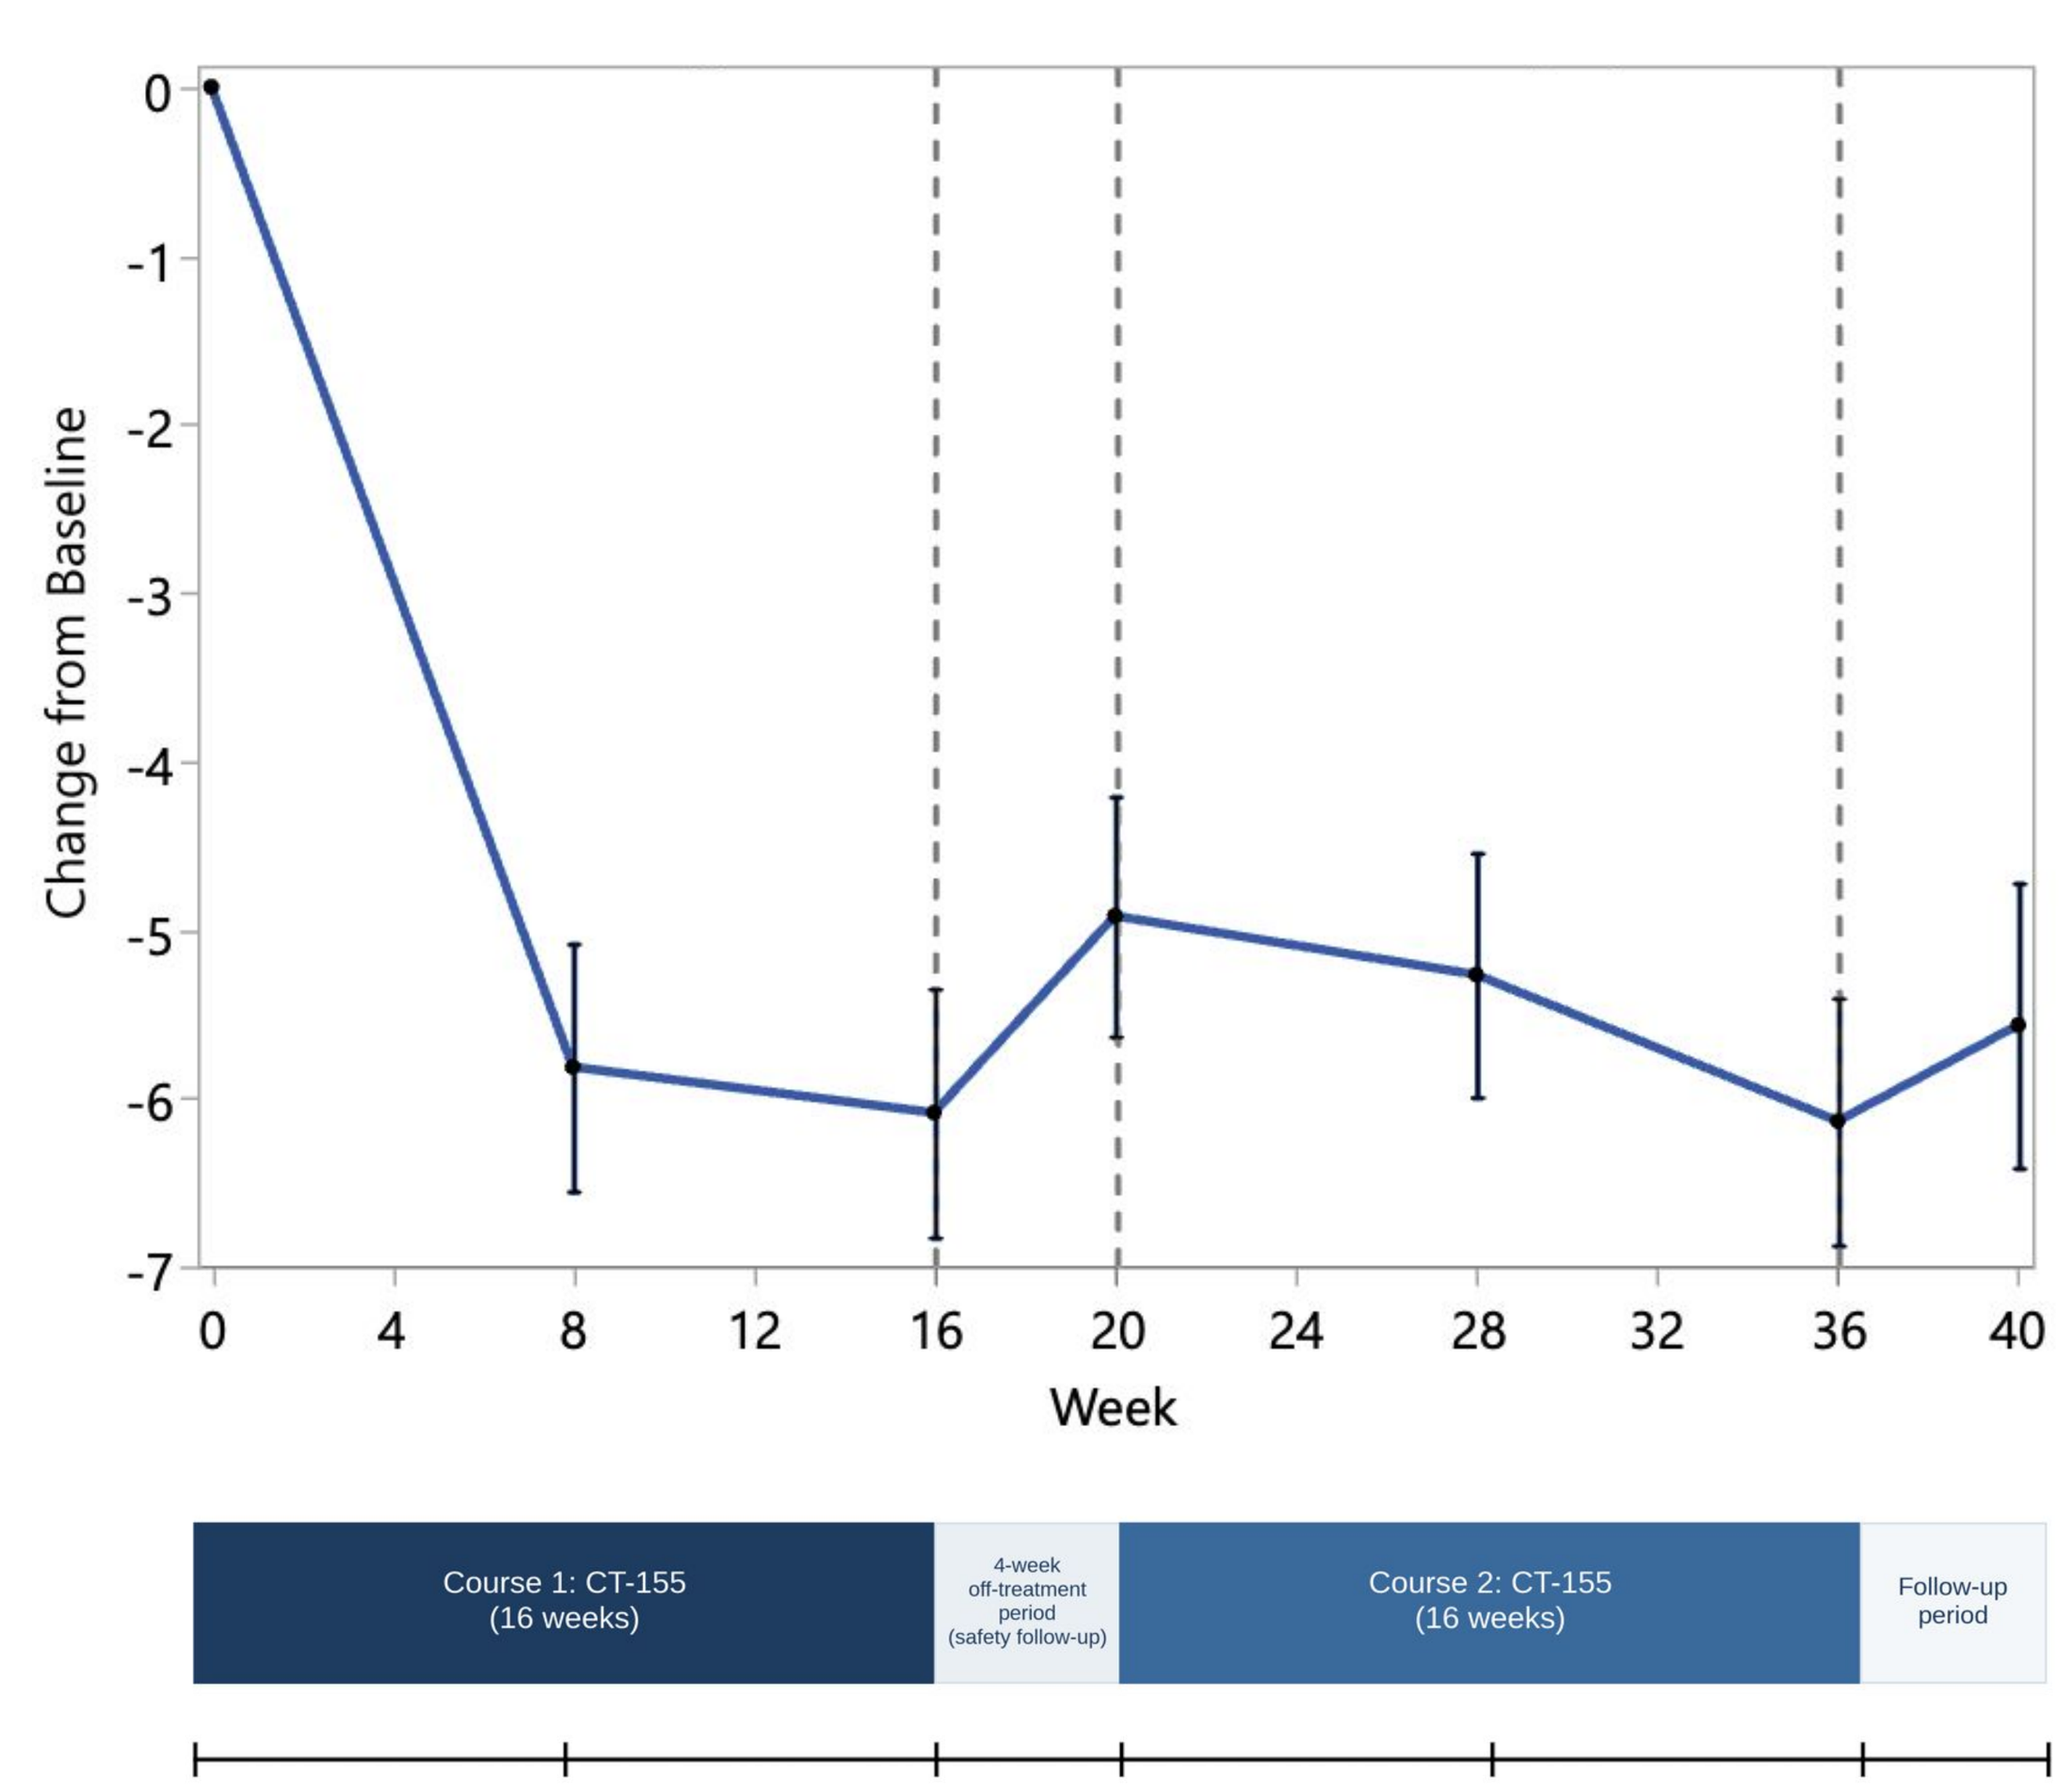
Change from Baseline
0
-1
-2
-3
-4
-5
-6
-7
0 4 8 12 16 20 24 28 32 36 40
Week
Course 1: CT-155 (16 weeks)
4-week off-treatment period (safety follow-up)
Course 2: CT-155 (16 weeks)
Follow-up period
Baseline
Week 8
Week 16
Week 20
Week 28
Week 36
Week 40

PANSS NSFS Change from Baseline - LS Means (SE) vs Week

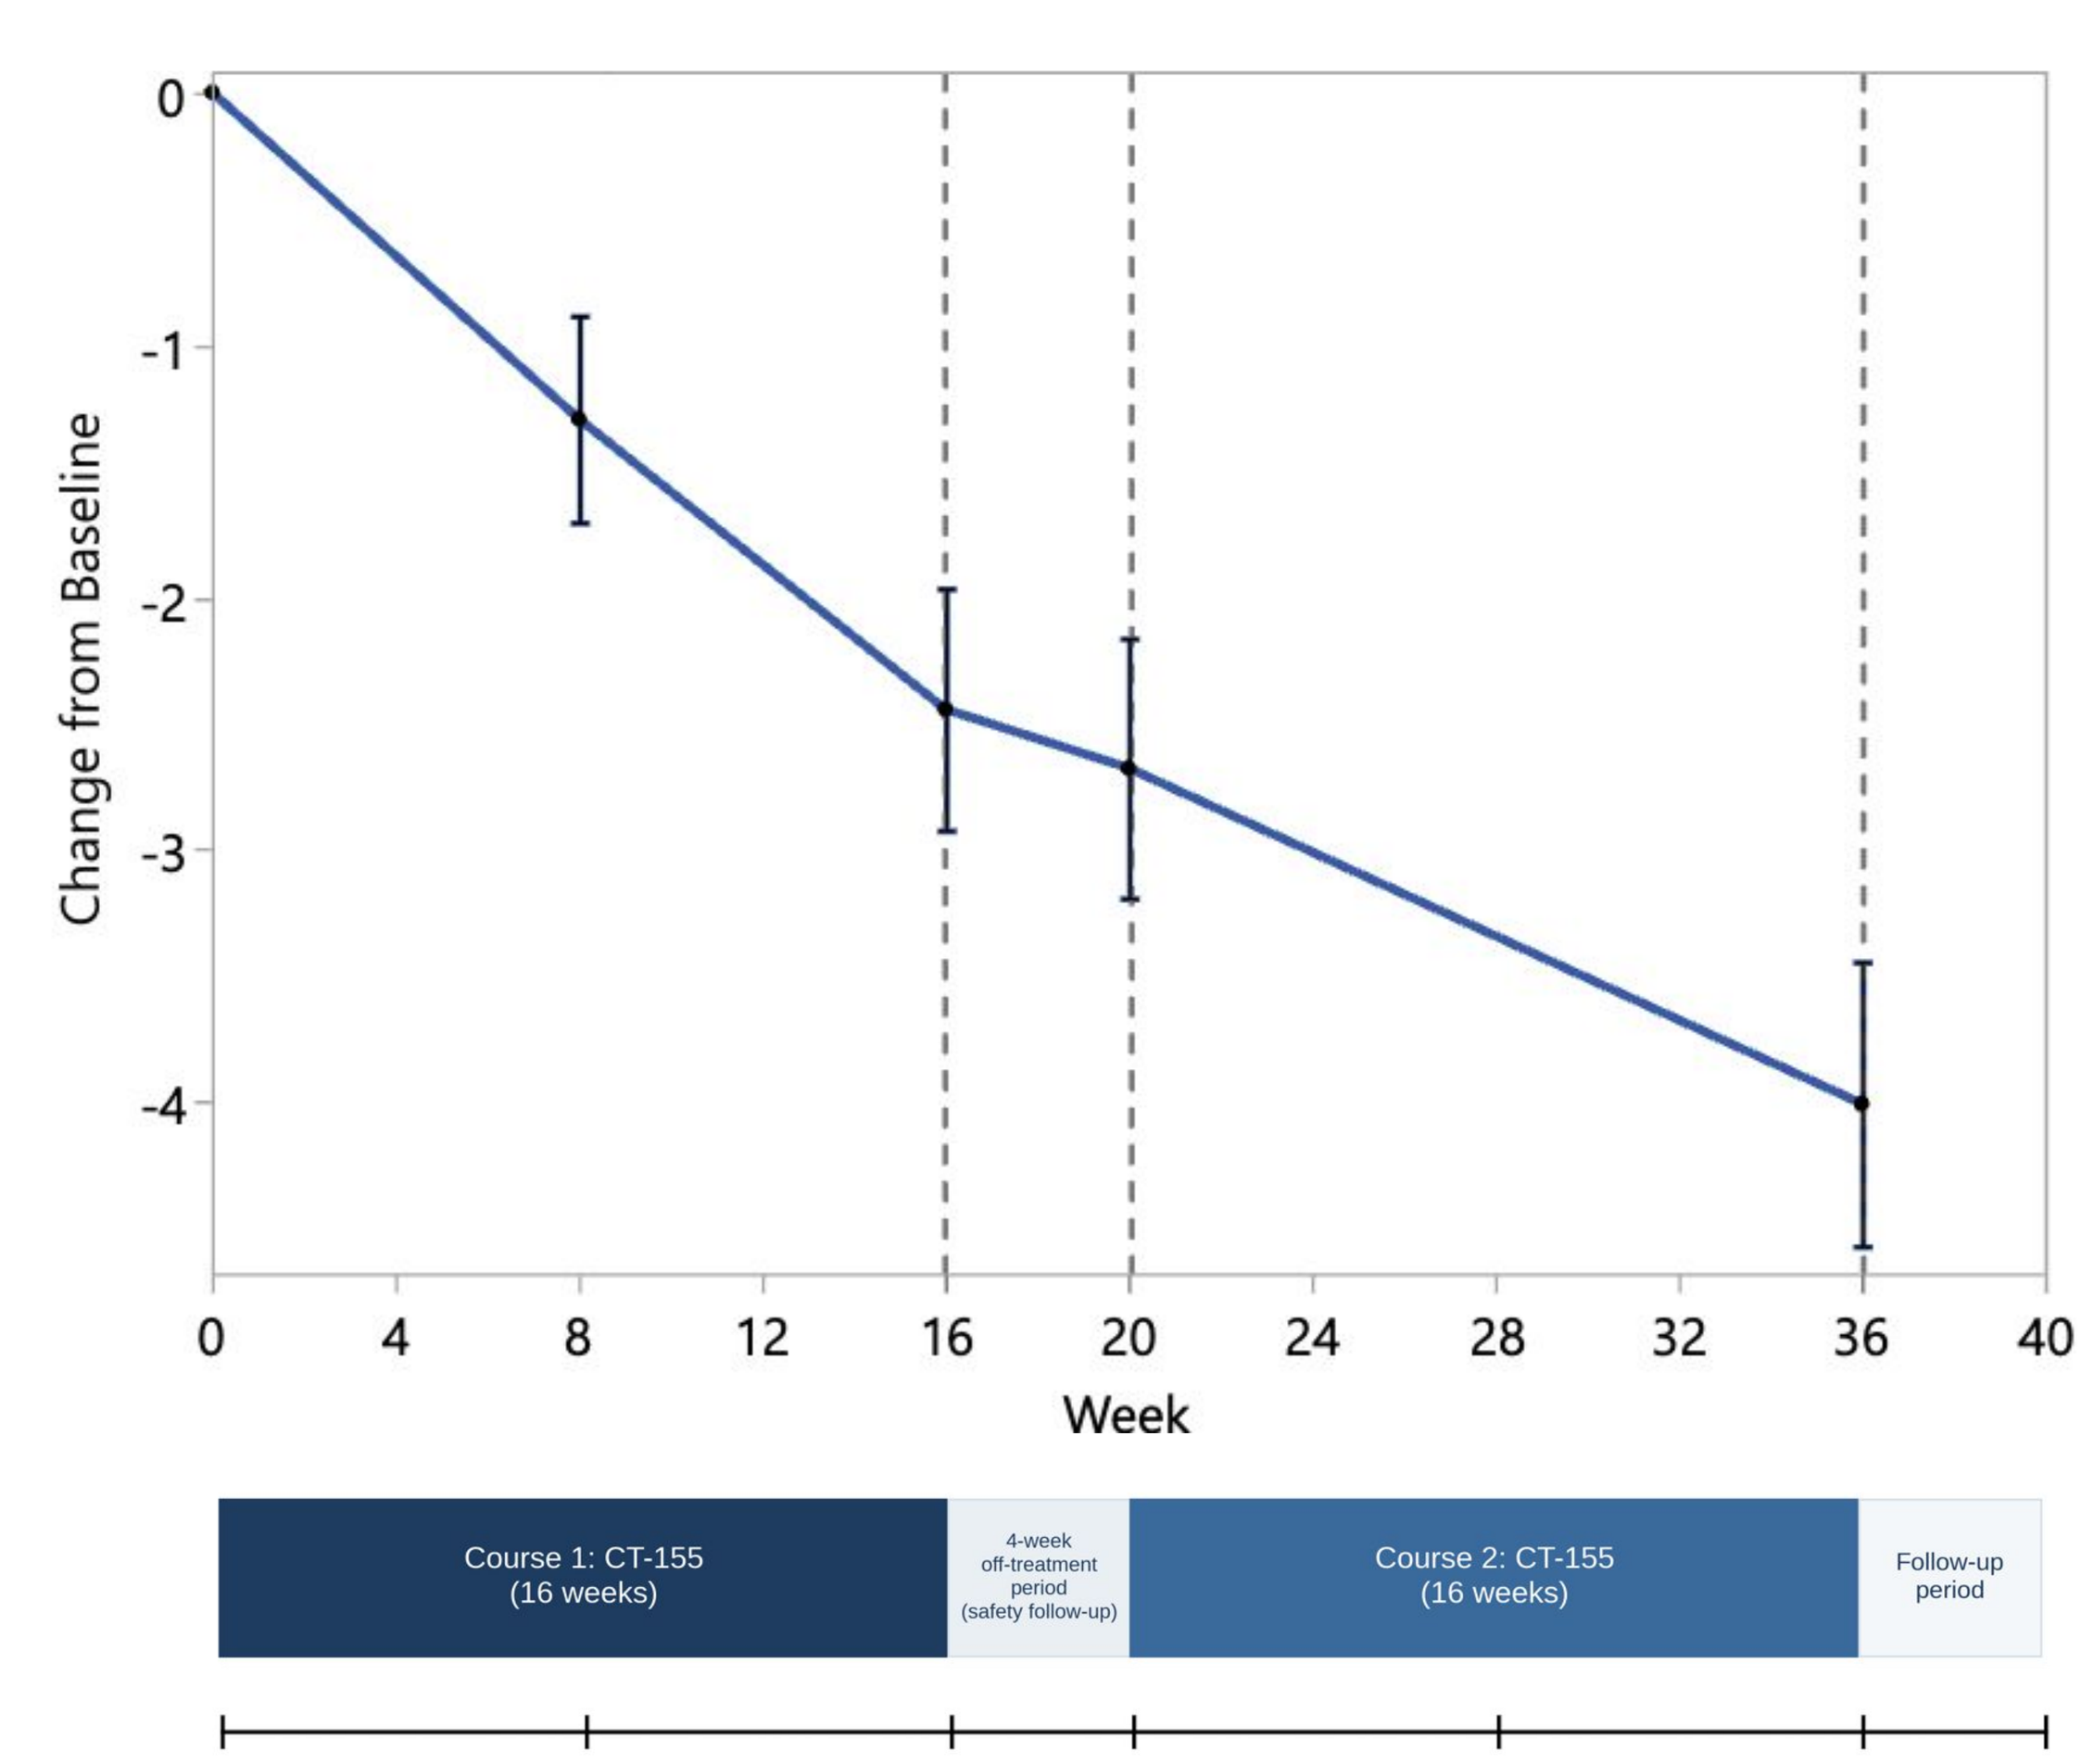
Change from Baseline
0
-1
-2
-3
-4
0 4 8 12 16 20 24 28 32 36 40
Week
Course 1: CT-155 (16 weeks)
4-week off-treatment period (safety follow-up)
Course 2: CT-155 (16 weeks)
Follow-up period
Baseline
Week 8
Week 16
Week 20
Week 28
Week 36
Week 40

Click Therapeutics

Conclusions

- Newer endpoints like the CAINS may offer advantages in accurately detecting the therapeutic effects of interventions targeting negative symptoms.
- The differences in cross-sectional convergence and longitudinal sensitivity between CAINS and PANSS provide further evidence of the partial coverage of the PANSS instrument.
- Longitudinal results suggest the PANSS may show lower sensitivity to treatment-induced clinical changes, particularly in outpatients with well-controlled positive symptoms.
- These analyses provide methodological considerations for future psychosis trials, suggesting that leveraging contemporary instruments may be essential to optimizing signal detection.

References

Kring, A. M., Gur, R. E., Blanchard, J. J., Horan, W. P., & Reise, S. P. (2013). The Clinical Assessment Interview for Negative Symptoms (CAINS): final development and validation. American Journal of Psychiatry, 170(2), 165–172.

Marder, S. R., Davis, J. M., & Chouinard, G. (1997). The effects of risperidone on the five dimensions of schizophrenia derived by factor analysis: combined results of the North American trials. Journal of Clinical Psychiatry, 58(12), 538–546.

Wolpe, N., Perrottelli, A., Giuliani, L., Yang, Z., Rekhi, G., Jones, P. B., … & Fernandez-Egea, E. (2025). Measuring the clinical dimensions of negative symptoms through the Positive and Negative Syndrome Scale. European Neuropsychopharmacology, 93, 68–76.

Acknowledgement

This work was supported by Click Therapeutics, Inc. The authors thank the CONVOKE participants, investigators and site staff.

One or more authors report potential conflicts which are described in the program.