ISCTM Autumn Conference | September 16th - 17th, 2026 | Philadelphia, PA

Prevalence and Severity of PANSS Worsening Across Schizophrenia Clinical Trial Populations: An Analysis of 33 Studies

Kott, Alan; Wang, Xingmei; Daniel, David G.
Signant Health

WHAT IS THE METHODOLOGICAL ISSUE BEING ADDRESSED?

- How do worsening rates, operationalized via PANSS deterioration thresholds, differ by trial population in schizophrenia clinical trials?

INTRODUCTION

- Clinical trials in schizophrenia typically emphasize mean symptom improvement as the primary measure of efficacy.
- However, a meaningful and heterogeneous subset of participants worsens during trials, irrespective of treatment assignment, and this proportion is rarely reported.
- In this retrospective analysis of a large pooled dataset, we estimated the prevalence of worsening using progressively stringent PANSS-based thresholds and assessed differences across three patient populations: acutely exacerbated, predominant negative symptoms, and suboptimally controlled.

METHODS

- Data were pooled from 33 double-blind, placebo-controlled trials (21 acute exacerbation, 8 negative symptom, 4 suboptimally controlled), comprising 14,281 subjects.
- Worsening was defined using progressively stringent PANSS-based thresholds as an increase in PANSS score at any time during the primary treatment period compared to baseline of
 - ≥1 point – captures any level of worsening irrespective of clinical relevance,
 - ≥10 points – lowest used clinically relevant threshold in published literature, e.g. CATIE Sz (Glick, 2015),
 - ≥12 points – proposed relapse threshold in recent meta-analysis (Siafis, 2024),
 - ≥20 points – to capture substantial clinical deterioration
- For each threshold, mixed-effects logistic regression models were fitted with fixed effects for study type, baseline PANSS total, centered study start year, and proportion of planned follow-up completed.
 - A protocol-level random intercept accounted for between-study heterogeneity.
 - Models were estimated using residual pseudo-likelihood with containment-based degrees of freedom.

RESULTS

- In acute exacerbation trials, mean observed proportions meeting worsening thresholds (≥1, ≥10, ≥12, ≥20) were 34%, 10%, 8%, and 3%, respectively.
- Corresponding rates in negative symptom trials were 33%, 7%, 5%, and 1%, and in suboptimally controlled trials were 23%, 3%, 2%, and <1%.
- Modeled worsening rates can be found in Figure 1.
- Protocol-level random effects were consistently significant, indicating substantial between-study heterogeneity.
- For the ≥10-point threshold, both acute and negative symptom studies showed higher odds of worsening compared with suboptimally controlled trials (OR = 3.0, 95% CI: 1.7–5.4, p = 0.0002; OR = 2.4, 95% CI: 1.3–4.6, p = 0.0068).
- Similar patterns were observed for ≥12 and ≥20 thresholds (Figure 2).
- Baseline PANSS total showed a non-significant trend toward lower odds of worsening, and no temporal trend was observed by study year.

FIGURE 1. MODELED RATES OF WORSENING

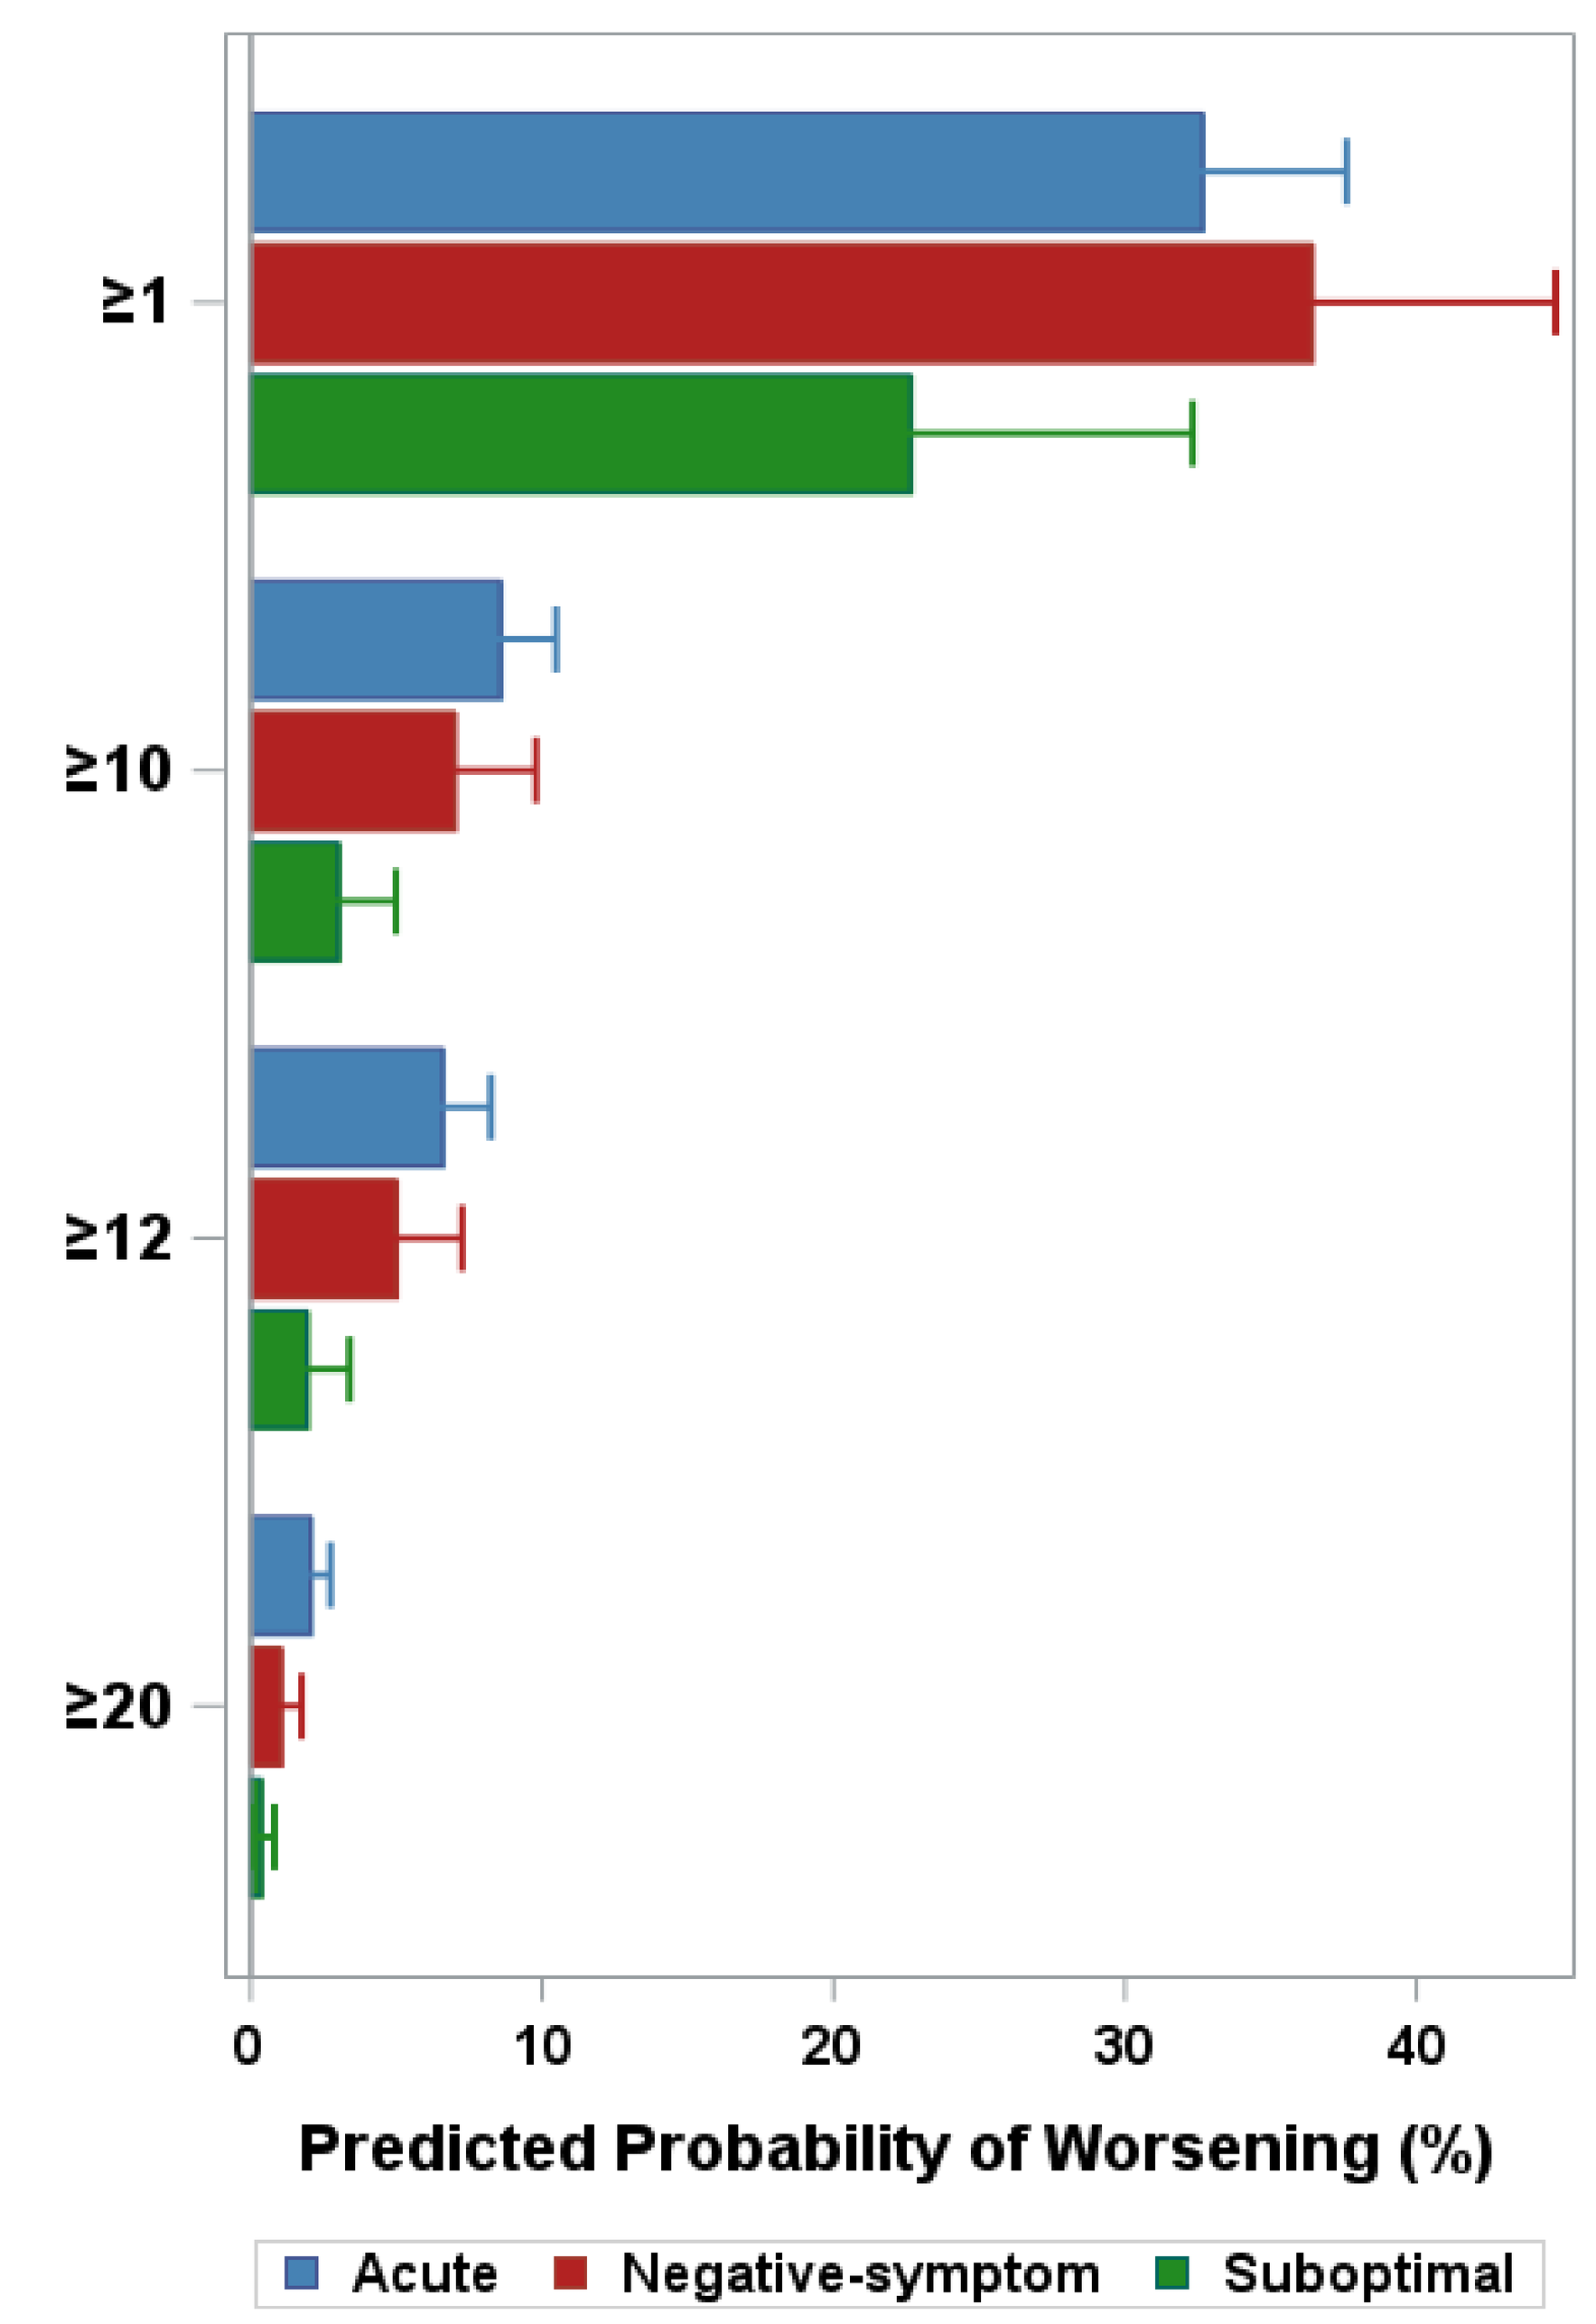
≥1
≥10
≥12
≥20
0
10
20
30
40
Predicted Probability of Worsening (%)
Acute
Negative-symptom
Suboptimal

FIGURE 2. ODDS OF WORSENING COMPARED WITH SUBOPTIMALLY CONTROLLED TRIALS

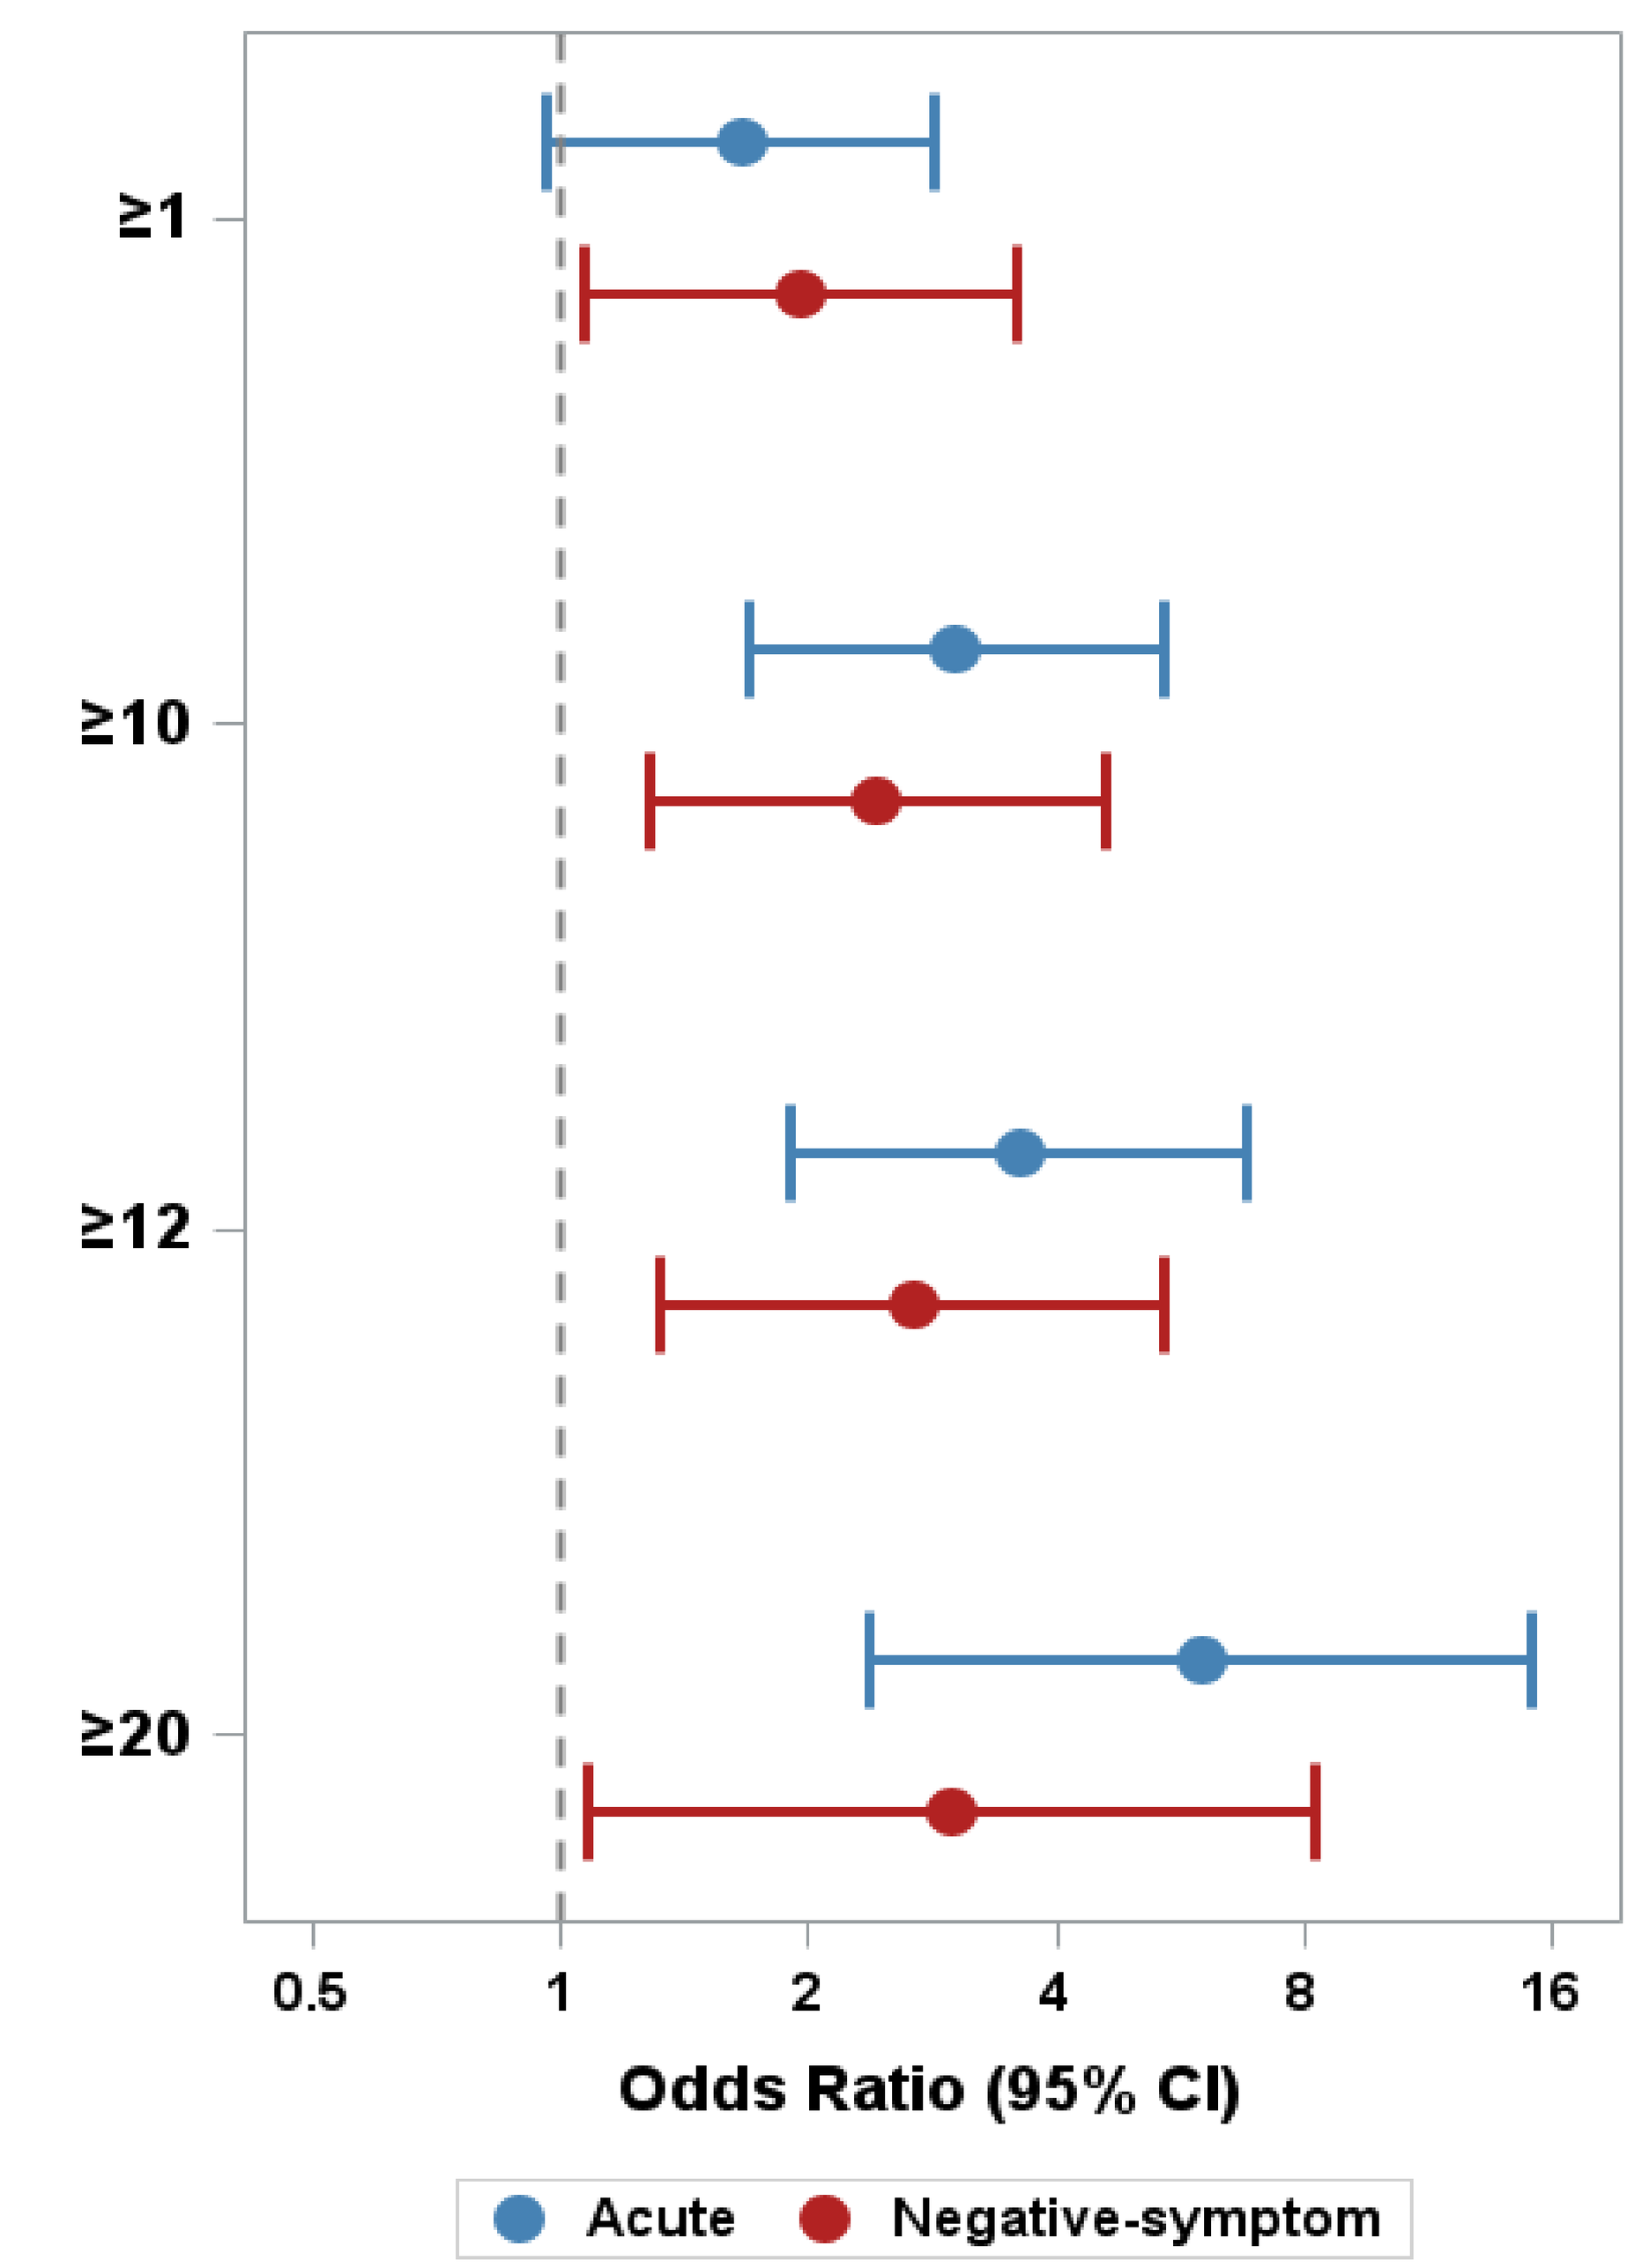
≥1
≥10
≥12
≥20
0.5
1
2
4
8
16
Odds Ratio (95% CI)
Acute
Negative-symptom

CONCLUSION

- Although mild deterioration is relatively common, clinically meaningful worsening during the primary treatment period is infrequent.
- This is somewhat unexpected in acute exacerbation trials, where greater deterioration might be anticipated.
- Lower worsening rates in suboptimally controlled populations—despite longer follow-up—likely reflect study design, as these trials evaluated adjunctive treatments added to stable antipsychotic therapy rather than monotherapy.
- A similar pattern was observed in negative symptom trials, with higher worsening rates in monotherapy designs.
- Higher worsening rates in acute versus negative symptom trials may also reflect differences in patient selection, with acute trials enriching for positive symptoms and instability, versus relative stability in negative symptom populations.
- Key limitations include lack of treatment assignment, precluding arm-level comparisons, and absence of sensitivity analyses imputing dropouts as worsened, which may underestimate true deterioration rates.
- Strengths include the large pooled sample and use of mixed-effects models to account for protocol-level variability.
- Overall, clinically meaningful worsening appears relatively infrequent in schizophrenia trials and influenced by design features.
- Routine reporting of worsening rates alongside mean improvements would provide a more complete and clinically relevant assessment of treatment outcomes.

REFERENCES

1. Glick, Henry A.; Li, Pengxiang; Harvey, Philip D. (2015): The relationship between Positive and Negative Syndrome Scale (PANSS) schizophrenia severity scores and risk for hospitalization. An analysis of the CATIE Schizophrenia Trial. In Schizophr Res 166 (1–3), pp. 110–114. DOI: 10.1016/j.schres.2015.05.021.
2. Siafis, Spyridon; Brandt, Lasse; McCutcheon, Robert A.; Gutwinski, Stefan; Schneider-Thoma, Johannes; Bighelli, Irene et al. (2024): Relapse in clinically stable adult patients with schizophrenia or schizoaffective disorder. Evidence-based criteria derived by equipercentile linking and diagnostic test accuracy meta-analysis. In The Lancet Psychiatry 11 (1), pp. 36–46. DOI: 10.1016/S2215-0366(23)00364-4.

All authors are Signant Health employees.

SIGNANT HEALTH
THE EVIDENCE GENERATION COMPANY®