Single Dose LSD for the Treatment of GAD: A Phase 2, Multicenter, Randomized, Double-blind, Parallel-group, Dose-finding Study to Assess the Effect of Four Doses of MM-120 for the Treatment of Generalized Anxiety Disorder: Rationale and Methods

Authors: Sarah Karas, Jamie Freedman, Craig Conant, Jamileh Jemison, Todd Solomon, Michelle Matvey, Rob Silva, Dan Karlin, Rob Barrow,

[1]Mind Medicine, Inc.,

Introduction

MM-120, D-lysergic acid diethylamide (LSD) D-tartrate, is being developed to treat Generalized Anxiety Disorder (GAD). MM-120 is a synthetic tryptamine belonging to the group of serotonergic hallucinogens broadly known as classic psychedelics. LSD has been shown to be efficacious in treating anxiety[1–4]. Specifically, a recent phase 2a LSD clinical trial produced long-lasting and notable reductions in anxiety and comorbid depression symptoms for participants up to 16 weeks[3]. Previous studies in healthy volunteers show LSD to be well-tolerated physiologically5, and in controlled clinical settings, LSD has been psychologically well-tolerated.[1–4] The MMED008 Phase 2 trial is the first randomized placebo controlled trial of LSD in modern history.

There are a limited number of medications to treat GAD. First line treatments often include benzodiazepines and SSRI/SNRIs. Benzodiazepines, while acutely effective, are not indicated for long term use due to a risk profile including tolerance, dependence, cognitive impairment, and sedation. SSRIs/SNRIs have variable and often underwhelming efficacy, intolerable side effects, and contraindications, leaving patients with inadequate treatment options. Historical and modern investigator-initiated and academic studies support the investigation of LSD as an alternative efficacious treatment for GAD. There is evidence to suggest that a single high dose (100-200 μg) administration of LSD may have rapid and durable clinical effects for anxiety[2–4].

Dose-range finding studies of psychedelics have largely been absent from the literature and are recommended by regulatory agencies for the development of new drugs[6]. As such, this trial includes 5 arms: placebo and four doses of MM-120. The proposed doses for use have been characterized in healthy participants5, but not in patient populations. The current literature lacks supportive data on the relationship between various doses of LSD and subsequent clinical benefit in patient populations. While there is preliminary evidence of correlation between acute pharmacodynamic effects and clinical benefit at a single dose level, this study is the first investigation into the acute (magnitude) and durability of different doses' clinical response. Previous studies assessing psychedelics have used an active comparator such as niacin, or a low dose comparator containing the investigational product under study. The inclusion of these types of comparators have been utilized to try and remit the possibility of functional unblinding. However, there has been no data to suggest that these active comparators have worked to this effect. The MMED008 study has taken the novel approach of using a true inactive placebo, which is the gold standard for conducting randomized controlled trials in psychiatric disorders. In addition, a dose-finding study with four active arms further mitigates the possibility of participants from becoming unblinded to their assigned treatment arm.

To date, published randomized, placebo-controlled psychedelic clinical trials, were mainly trials investigating compounds such as 3,4-Methyl enedioxymethamphetamine (MDMA) and psilocybin, which have used a psychedelic-assisted therapy component in conjunction with the investigational product[7–9]. However, establishing the efficacy of the investigational product independent of concurrent psychotherapy is not possible when used in conjunction. The MMED008 trial does not include psychotherapy.

This is the first, double blind, randomized, placebo-controlled trial of a classic psychedelic (MM-120) that has taken these approaches and will serve to further elucidate the relationship between the pharmacology of psychedelics and psychiatric illness generally; and MM-120 and Generalized Anxiety Disorder (GAD) specifically.

Table 1: MM-120 Dose Rationale

Dose of MM-120 (μg) (Freebase-equivalent)	Rationale
0	Placebo control – an inactive placebo is the most appropriate control for the Multiple Comparison Procedure-Modelling (MCP-Mod) statistical approach to be used in the current study and for characterization of dose-response
25	Threshold dose – minimum dose at which psychoactive effects are perceivable by patients on average
50	Dose that is above threshold but unlikely to result in significant "psychedelic effects"
100	Lower of two doses that reliably results in a "psychedelic effect" while minimizing "bad drug effect"
200	Higher of two doses that reliably results in a "psychedelic effect"

Table 2: Schedule of Key Activities

STUDY PERIOD	SCREENING	BASELINE & DOSING			FOLLOW-UP					
Visit	Screening	Baseline	Day 1 Randomization (3A) & Dosing Session (3B)		Day 2	Week 1	Week 2	Week 4	Week 8	Week 12 (or Early WD)/EOS
Visit Number	1	2	3A	3B	4	5	6	7	8	9
Timing or Permitted Window	Up to 30 days prior to Baseline	1-5 days prior to Day 1	Pre-dose	Dosing & post-dose	1 day after dosing	Day 8 ± 1 day	Day 15 ± 3 days	Day 29 ± 3 days	Day 57 ± 5 days	Day 85 ± 5 days
Informed consent	X									
Eligibility assessment	X	X	X							
Randomization			X							
Physical exam	X				X					X
Neuropsychiatric exam		X			X	X	X	X	X	X
Vital signs	X	X	X		X	X	X	X	X	X
12-lead safety ECG	X	X			X					
Blood sample collection	X	X			X	Opt	Opt	X	X	X
Urinalysis	X	X			X					
Urine drug screen	X[i]	X[i]	X			X	X	X	X	X
Placebo Script Review		X						X	X	
MINI	X									
C-SSRS	X	X	X	X	X	X	X	X	X	X
HAM-A (central rater)	X	X				X	X	X	X	X
MADRS (central rater)	X	X				X	X	X	X	X
CGI-S	X	X			X	X	X	X	X	X
CGI-I					X	X	X	X	X	X
PGI-S	X	X			X	X	X	X	X	X
PGI-C					X	X	X	X	X	X
SDS	X	X				X	X	X	X	X
EQ-5D-5L		X				X	X	X	X	X
PSQI		X						X	X	X
ASEX		X				X	X	X	X	X
Drug Effect VAS					X					
MEQ30					X					
5D-ASC					X					
Treatment blinding question					X					
Administration of study drug				X						
Subject education / follow-up session with both DSMs		X	X		X	X	X			

5D-ASC: 5-Dimensional Altered States of Consciousness Rating Scale; ASEX: Arizona Sexual Experiences Questionnaire; CGI-I: Clinical Global Impression – Improvement; CGI-S: Clinical Global Impression – Severity; C-SSRS: Columbia-Suicide Severity Rating Scale; ECG: electrocardiogram; EOS: End of Study; MADRS: Montgomery-Åsberg Depression Rating Scale ; MEQ30: Mystical Experience Questionnaire; MINI: Mini-International Neuropsychiatric Interview; PGI-C: Patient Global Impression – Change; PGI-S: Patient Global Impression – Severity; PSQI: Pittsburgh Sleep Quality Index; SDS: Sheehan Disability Scale; HAM-A: Structured Interview Guide for the Hamilton Anxiety Rating Scale; VAS, visual analogue scale.

Methods

Study MMED008 seeks to enroll at least 180 male and female participants 18 years to 74 years of age who meet DSM-5 criteria for GAD and have a minimum Hamilton Anxiety Rating Scale (HAM-A) score of 20 at both Screening and Baseline visits. Participants will be recruited in the United States from approximately 20 research sites.

Potential participants who have contraindicated medical or psychiatric conditions or are taking psychoactive or other medications that cannot be appropriately tapered will be excluded from the study.

Eligible participants will be randomized in a 1:1:1:1:1 ratio to receive a single dose of either MM-120 (25, 50, 100, or 200 μg freebase-equivalent) or placebo (inactive) in a controlled clinical setting, which will be administered during a single dosing session. Table 1 contains dose rationale. During the Baseline visit, which is 1-5 days prior to dosing, participants will meet with their DSMs for Participant Education and Preparation for their dosing day. During the dosing session, participants will spend 12-hours under continuous observation by two dosing session monitors (DSMs) who do not provide psychotherapy before, during, or after the session. DSMs regularly monitor vital signs to ensure the subject's physical well-being, and provide psychological support if reassurance is needed during the dosing session. A lead DSM is always required to be present and must be a healthcare provider with graduate-level professional training, clinical experience, and an active license to practice independently in the state of the study site location. A study site-designated licensed physician must be available in the event of a physiological or psychiatric emergency for the duration of the dosing session day. Participants will be evaluated at 8, 9, 10, 11 and 12-hours post-dose to determine fitness for release from the clinic, although all subjects must remain at the clinic for a minimum of 12 hours post-dose. Participants will return to the site the following day to meet with both DSMs for a follow-up session, and complete safety and exploratory endpoints. Participants will also meet with their dosing session monitors at 1 and 2 weeks post dose for follow-up visits. Additional follow up visits are completed with study staff at 4, 8, and 12-weeks after the dosing session day.

In order to help maintain the study blind, HAM-A and MADRS ratings are completed by a central rater who is blinded to both study visit and inclusion criteria. Further, site raters completing the CGI-S/I are prohibited from functioning as a DSM or the study site designated physician during dosing session visits. Table 2 contains the schedule of key activities including secondary and exploratory measures for the MMED008 study.

Table 3: Study MMED008 Power by Model

Model	Power (%)
Sigmoid Emax (ED50, hill)	
10, 1	84.5
100, 5	90.7
100, 10	91.0
150, 10	89.9
Emax (D50 = 100)	83.2
Linear	85.0

Figure 1: Candidate Dose-response Curves

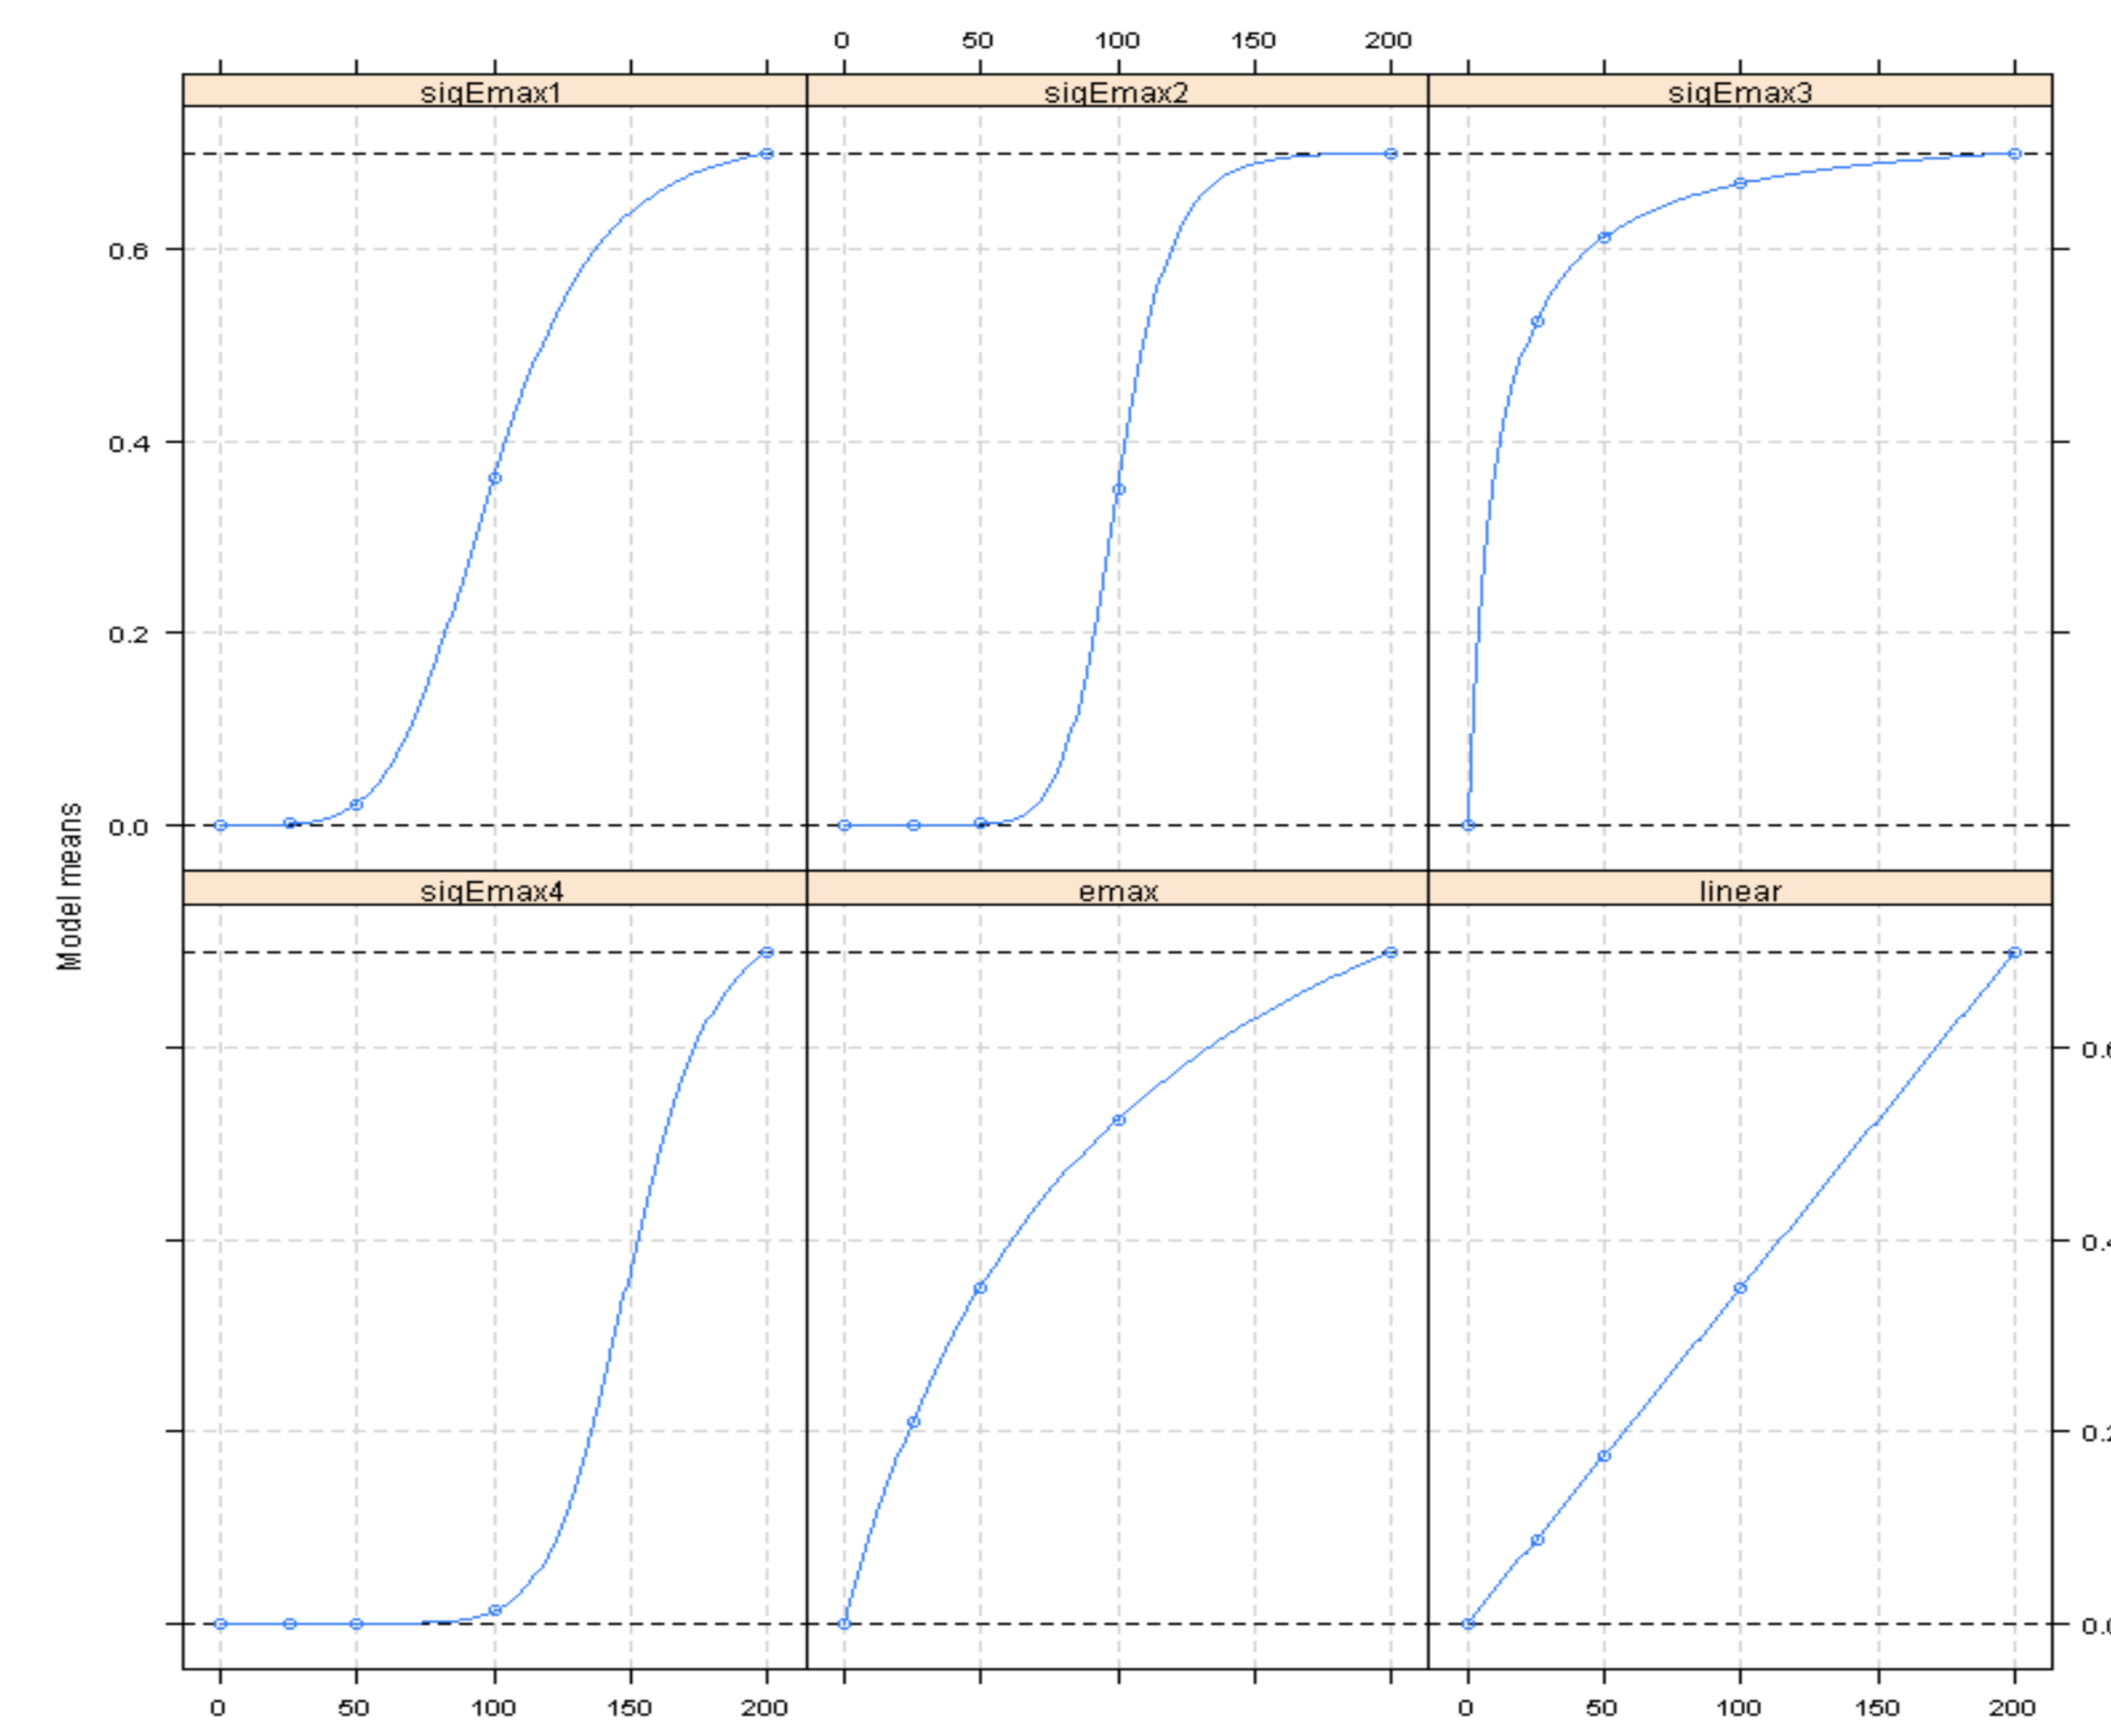
sigEmax1
sigEmax2
sigEmax3
sigEmax4
emax
linear
Model means
0.0
0.2
0.4
0.6
0
50
100
150
200

Results

The primary objective of this 12-week study is to determine the dose-response relationship of four dose levels versus a true inactive placebo and establish the efficacy of MM-120 in GAD, as measured by the change in HAM-A Total Score from Baseline to Week 4. The key secondary measure is change in the HAM-A score from Baseline to Week 8 for the respective arms.

The multiple comparisons procedure methodology (MCP-Mod)[10–11] will be employed to assess the primary objective: to investigate the dose-response relationship for different doses of MM-120 versus placebo in change from Baseline in HAM-A Total Score at Week 4 (Figure 1). A total sample of 180 subjects (36 per dose arm and 36 for the placebo arm) is required to ensure a mean power >87% for an MCP-Mod Analysis rejecting the hypothesis of a constant dose-response curve using the MCP, assuming a null placebo response, a maximum standardized effect of 0.6 within the doses' range, and a common standard deviation within the dose arms, if a study-wise one-sided type-1 error rate < 0.05 is required. The analysis assumes 4 doses of active study drug (25, 50, 100, and 200 μg freebase-equivalent) and placebo. The R package Dose Finding (https://CRAN.R-project.org/package=DoseFinding) has been used to estimate the sample size and the corresponding power. The power for each model is estimated as shown in Table 3.

Secondary efficacy endpoints include measures of co-morbid depression (MADRS), quality of life, and functional outcome measures (Table 2). Exploratory endpoints include evaluation of drug effect, mystical experience, quantification of altered states of consciousness, and a treatment unblinding questionnaire, which occur the day following the dosing session day.

Conclusion

In the first randomized placebo controlled trial of LSD in modern history, the MMED008 study design incorporates a novel dose-finding paradigm, leveraging FDA guidance on developing psychedelic molecules and the gold standard in RCT methodology.

References

1. Carhart-Harris RL, Kaelen M, Whalley MG, Bolstridge M, Feilding A, Nutt DJ. LSD enhances suggestibility in healthy volunteers. Psychopharmacology (Berl). 2015 Feb;232(4):785-94.
2. Gasser P, Holstein D, Michel Y, Doblin R, Yazar-Klosinski B, Passie T, Brenneisen R. Safety and efficacy of lysergic acid diethylamide-assisted psychotherapy for anxiety associated with life-threatening diseases. J Nerv Ment Dis. 2014;202(7):513-20
3. Holze, Friederike, et al. Lysergic acid diethylamide–assisted therapy in patients with anxiety with and without a life-threatening illness: a randomized, double-blind, placebo-controlled phase II study. Biological Psychiatry 93.3 (2023): 215-223
4. Holze, Friederike, et al. Acute dose-dependent effects of lysergic acid diethylamide in a double-blind placebo-controlled study in healthy subjects. Neuropsychopharmacology 46.3 (2021): 537-544.
5. Holze, F., Caluori, T.V., Vizeli, P. et al. Safety pharmacology of acute LSD administration in healthy subjects. Psychopharmacology 239, 1893–1905 (2022). https://doi.org/10.1007/s00213-021-05978-6
6. Psychedelic Drugs: Considerations for Clinical Investigations Guidance for Industry DRAFT GUIDANCE, U.S. Department of Health and Human Services Food and Drug Administration Center for Drug Evaluation and Research (CDER), June 2023 Clinical/Medical
7. Mitchell, J.M., Ot'alora G., M., van der Kolk, B. et al. MDMA-assisted therapy for moderate to severe PTSD: a randomized, placebo-controlled phase 3 trial. Nat Med (2023). https://doi.org/10.1038/s41591-023-02565-4
8. Goodwin, Guy M., et al. "Single-dose psilocybin for a treatment-resistant episode of major depression." New England Journal of Medicine 387.18 (2022): 1637-1648.
9. Raison CL, Sanacora G, Woolley J, et al. Single-Dose Psilocybin Treatment for Major Depressive Disorder: A Randomized Clinical Trial. JAMA. 2023;330(9):843–853. doi:10.1001/jama.2023.14530
10. Bretz, Frank, José C. Pinheiro, and Michael Branson. "Combining multiple comparisons and modeling techniques in dose-response studies." Biometrics 61.3 (2005): 738-748.
11. Pinheiro, José, et al. "Model-based dose finding under model uncertainty using general parametric models." Statistics in medicine 33.10 (2014): 1646-1661.

MindMed