Use of a Pharmacogenomic Biomarker to Retarget a Failed Clinical Trial with Liafensine in Depression

Alphs LD, Luo W, Lu X, Alexander R, Sun H, Lu H, Wang G

Methodological Question Being Addressed

Can a DGMTM (Denovo Genomic Marker) genome wide scan platform designed to discover novel pharmacogenomic biomarkers be used to better target an optimal study population for drugs that have previously failed to demonstrate a significant treatment effect?

Introduction

Variable clinical responses to medications likely reflect in part underlying genetic polymorphisms that differentially affect drug metabolism and/or phenotypic expression of the disease. Despite this generally accepted understanding, to date these polymorphisms have not been characterized and applied to clinical trials of CNS diseases. Absent this knowledge, study populations included in Phase 2 and 3 clinical trials represent heterogeneous mixtures of persons with multiple genetic polymorphisms some of whom may not be responsive to the study treatment or may be particularly sensitive to treatment side effects. This heterogeneity increases the variance in treatment response and diminishes the ability to detect significant clinical effects in the population that has been studied.

Selecting study populations with specific genetic profiles may allow the selection of persons into clinical trials who have a greater likelihood of positive treatment response and permit exclusion of persons who are particularly vulnerable to problematic adverse drug reactions. Once the product becomes available for clinical use, these same pharmacogenomic biomarkers would help clinicians apply a personalized medicine approach to persons most likely to have a positive treatment effect.

Drugs With Negative Results May Succeed in the Right Patients

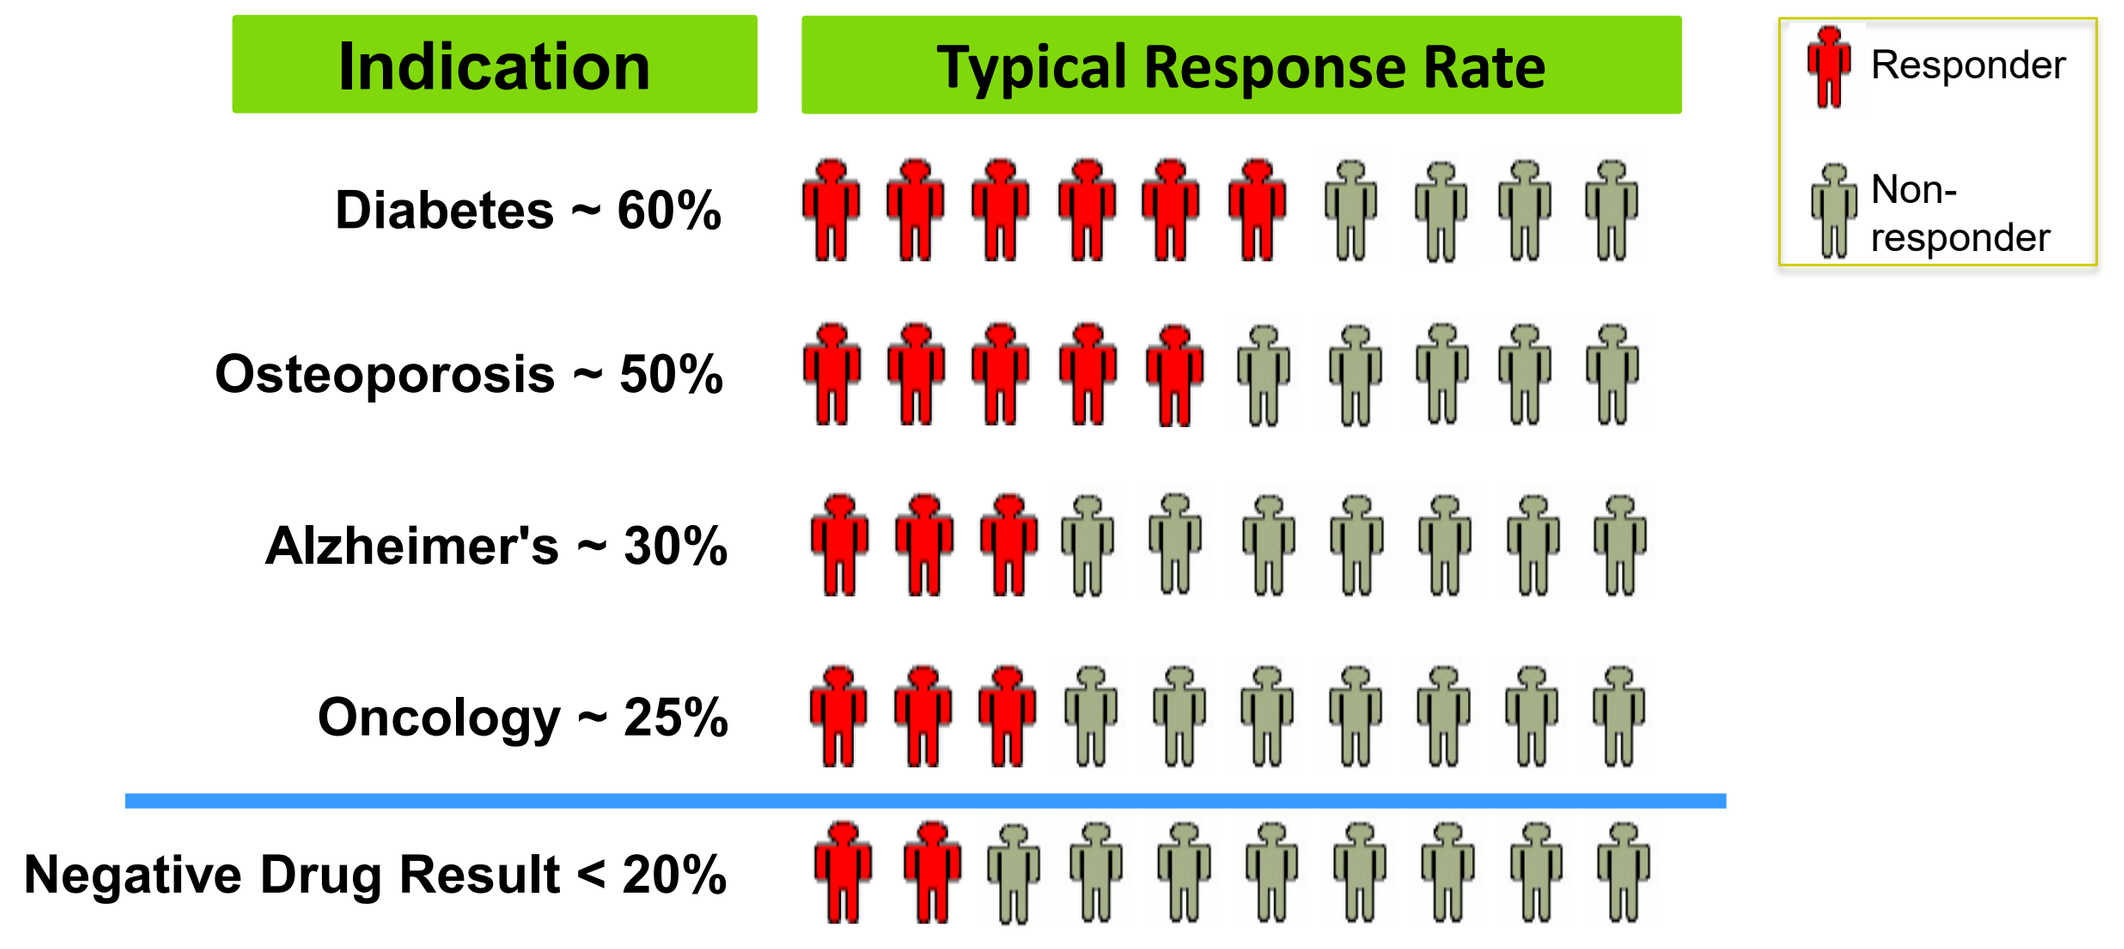
Indication
Typical Response Rate
Responder
Non-responder
Diabetes ~ 60%
Osteoporosis ~ 50%
Alzheimer's ~ 30%
Oncology ~ 25%
Negative Drug Result < 20%

- **Even successful drugs only work on a subset of patients**
- **If responders to a negative drug result can be prospectively identified via a biomarker, it can lead to approval in that specific subset**

Denovo Biopharma

Methods

- DB104 (liafensine) is a potent, selective reuptake inhibitor of three monoamines, serotonin (5-HT), norepinephrine (NE), and dopamine (DA).
- Compound was previously evaluated as monotherapy for treatment-resistant depression (TRD) in two large Phase 2 trials
- SAFETY: DB104 was well tolerated, without evidence of dose-dependent discontinuations due to adverse events.
- EFFICACY: Response to treatment was not different compared to continued treatment
- Program was discontinued.
- Using a subset of archived samples preserved from two DB104 Phase 2 studies, a genome wide scan was completed to identify genetic biomarkers that correlated with good response to liafensine
- A p-value 10-7 was required to control for multiple comparisons that might result in a Type 1 error.

Biomarker Discovery Methodology

Transforming Underprioritized Drugs into Successful Personalized Medicines

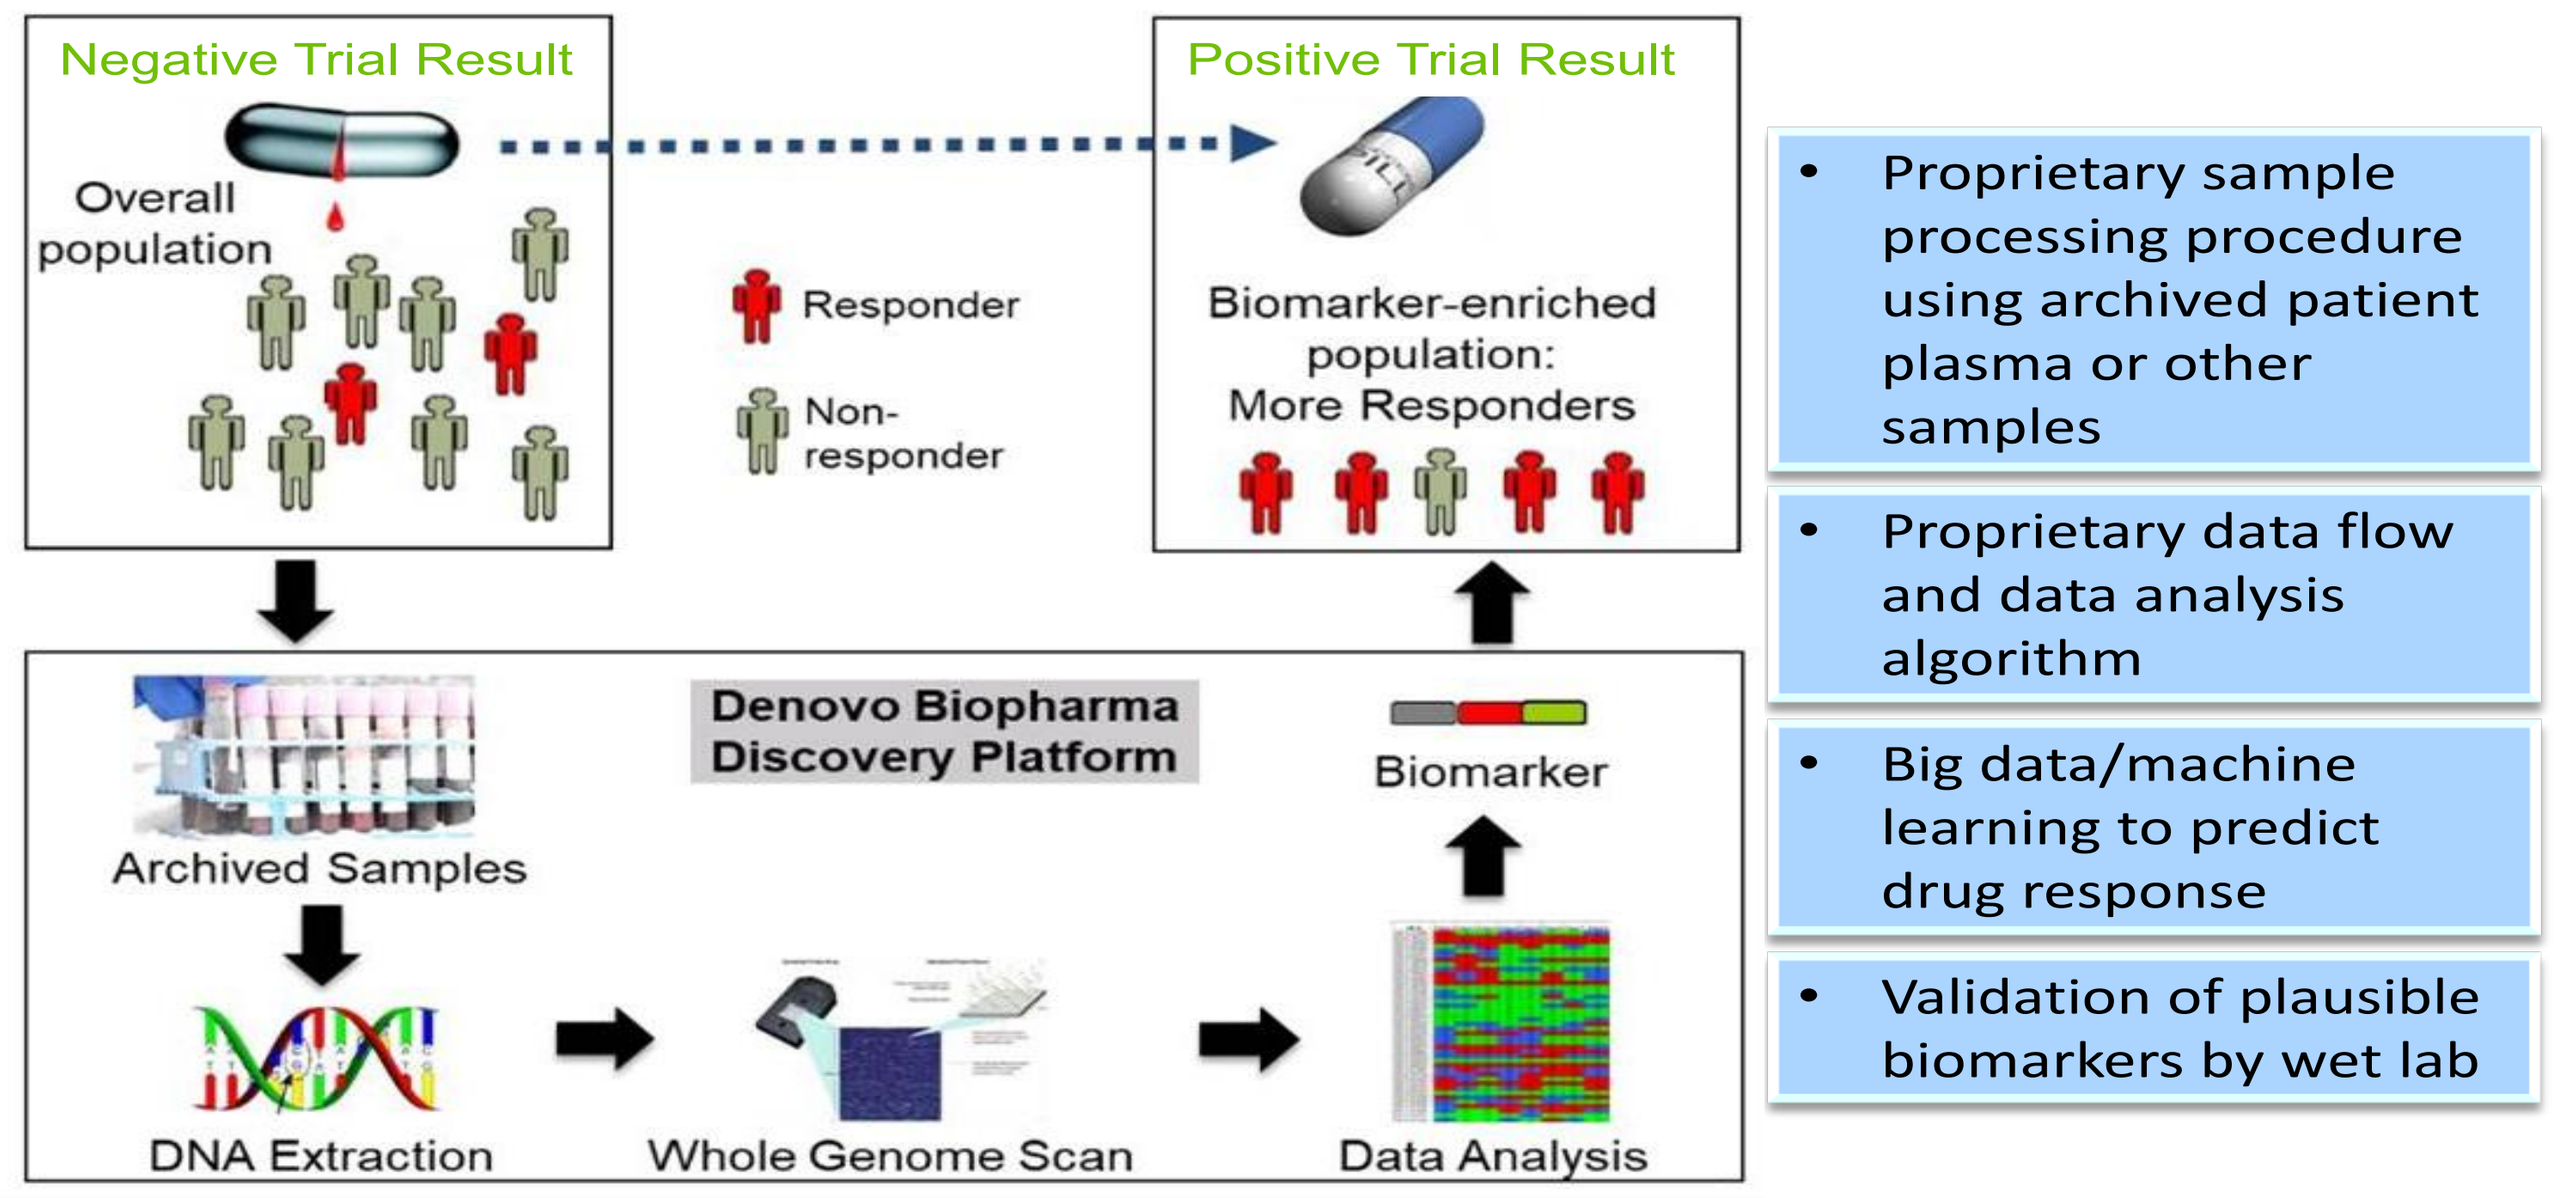
Negative Trial Result
Overall population
Responder
Non-responder
Positive Trial Result
Biomarker-enriched population: More Responders
Archived Samples
Denovo Biopharma Discovery Platform
Biomarker
DNA Extraction
Whole Genome Scan
Data Analysis
Proprietary sample processing procedure using archived patient plasma or other samples
Proprietary data flow and data analysis algorithm
Big data/machine learning to predict drug response
Validation of plausible biomarkers by wet lab

RESULTS

- The genome wide scan identified a genetic biomarker (DGM4 GG) that correlated with good response to liafensine
- Post hoc analyses of prior clinical trial data showed
 - Subjects not carrying the biomarker [DGM AA and DGM AG] did not differentiate from placebo when treated with liafensine
 - Subjects carrying the biomarker [DGM GG] differentiate from placebo as early as Week 3 when treated with liafensine
 - Results were replicated using remaining validation samples.
 - The biomarker does not increase response to duloxetine or escitalopram

Biomarker DGM4 Discovery Set: MADRS Score Change by DGM4 Genotypes

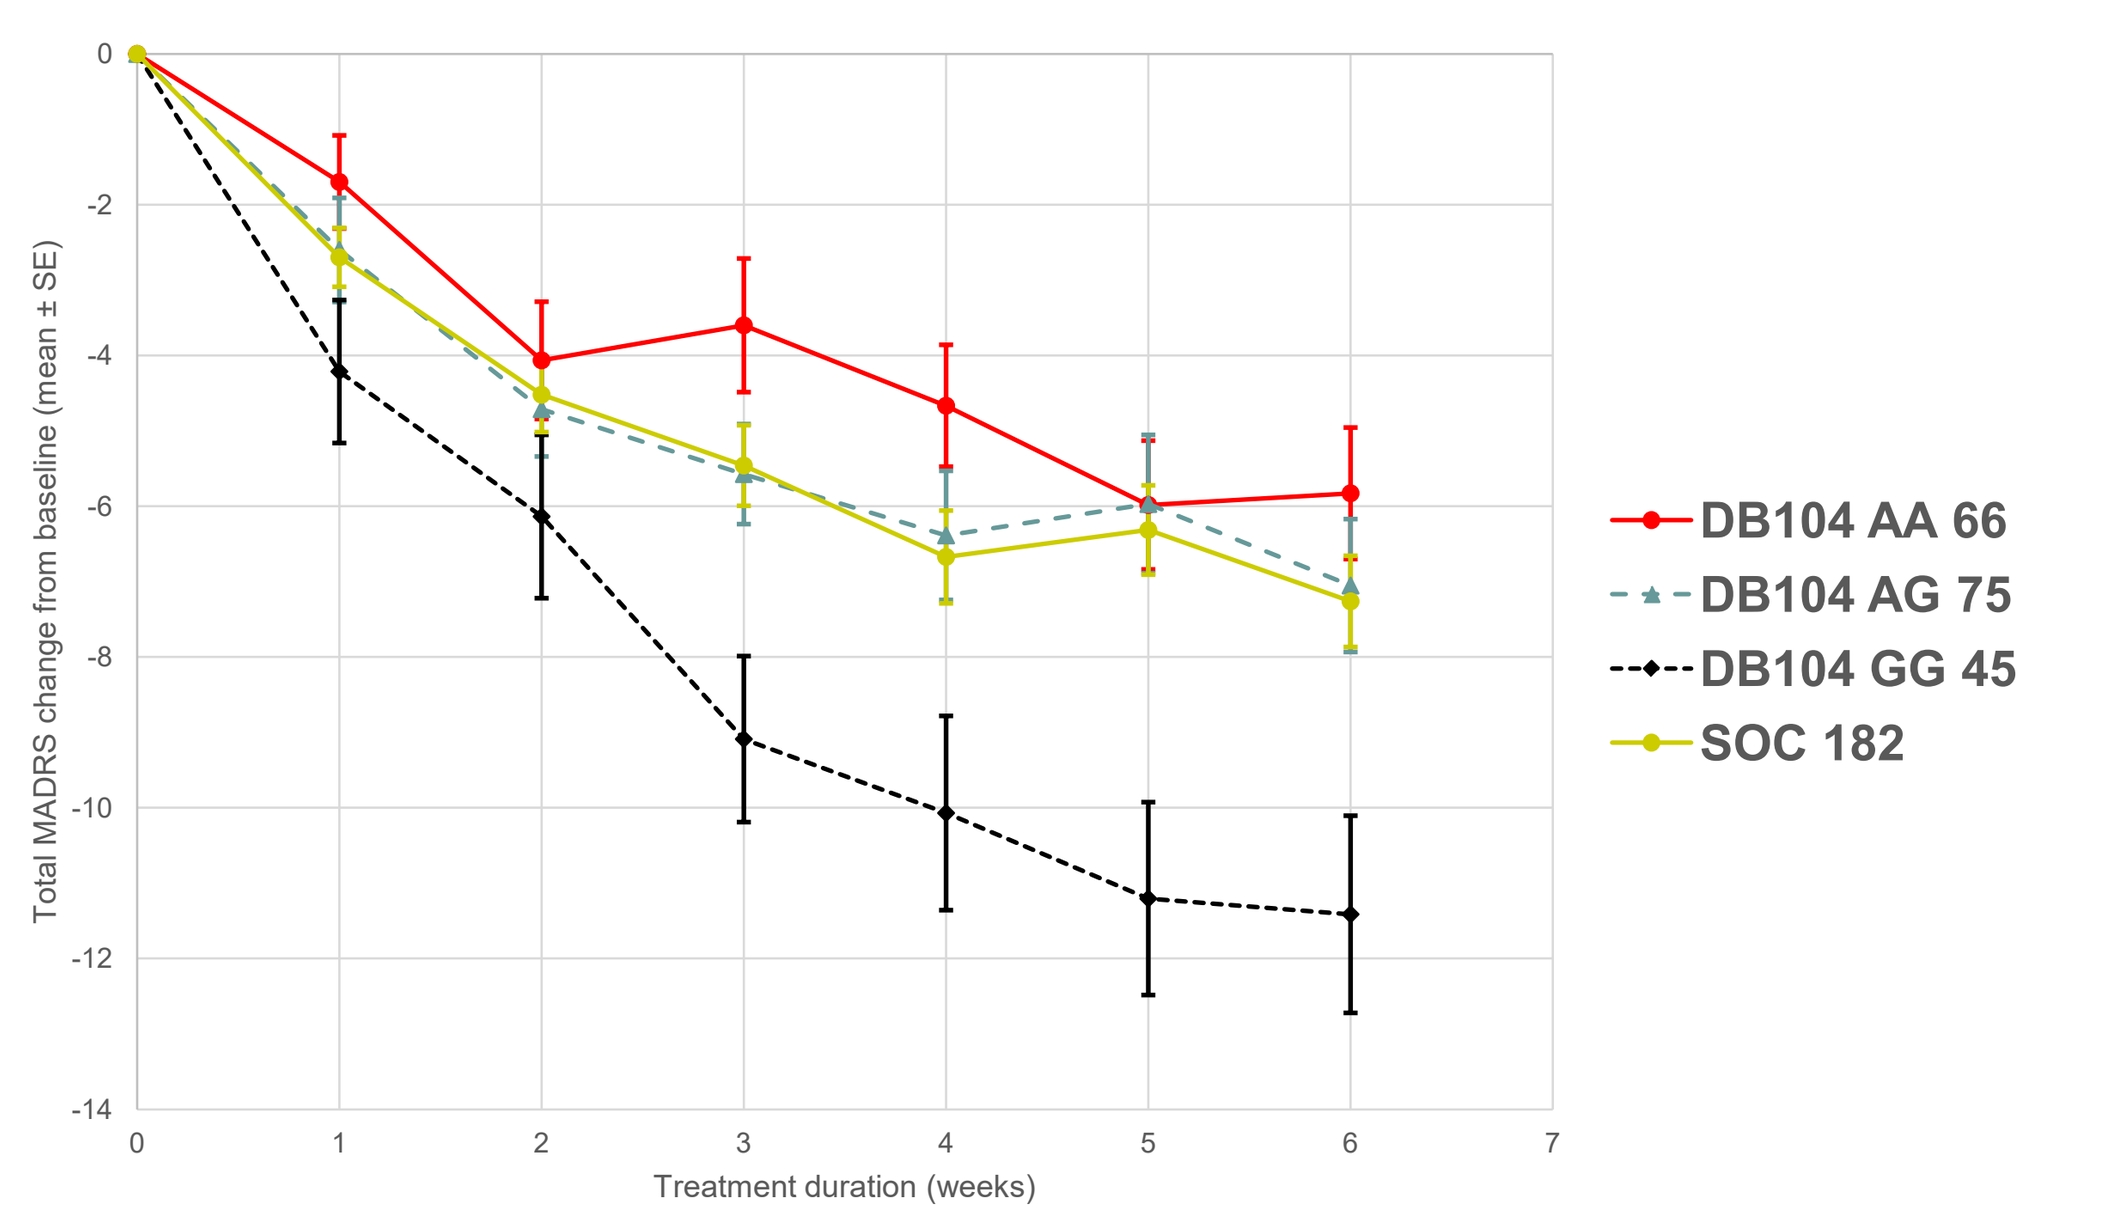
Total MADRS change from baseline (mean ± SE)
Treatment duration (weeks)
DB104 AA 66
DB104 AG 75
DB104 GG 45
SOC 182

Biomarker DGM4 Validation Set: MADRS Score Change by DGM4 Genotypes

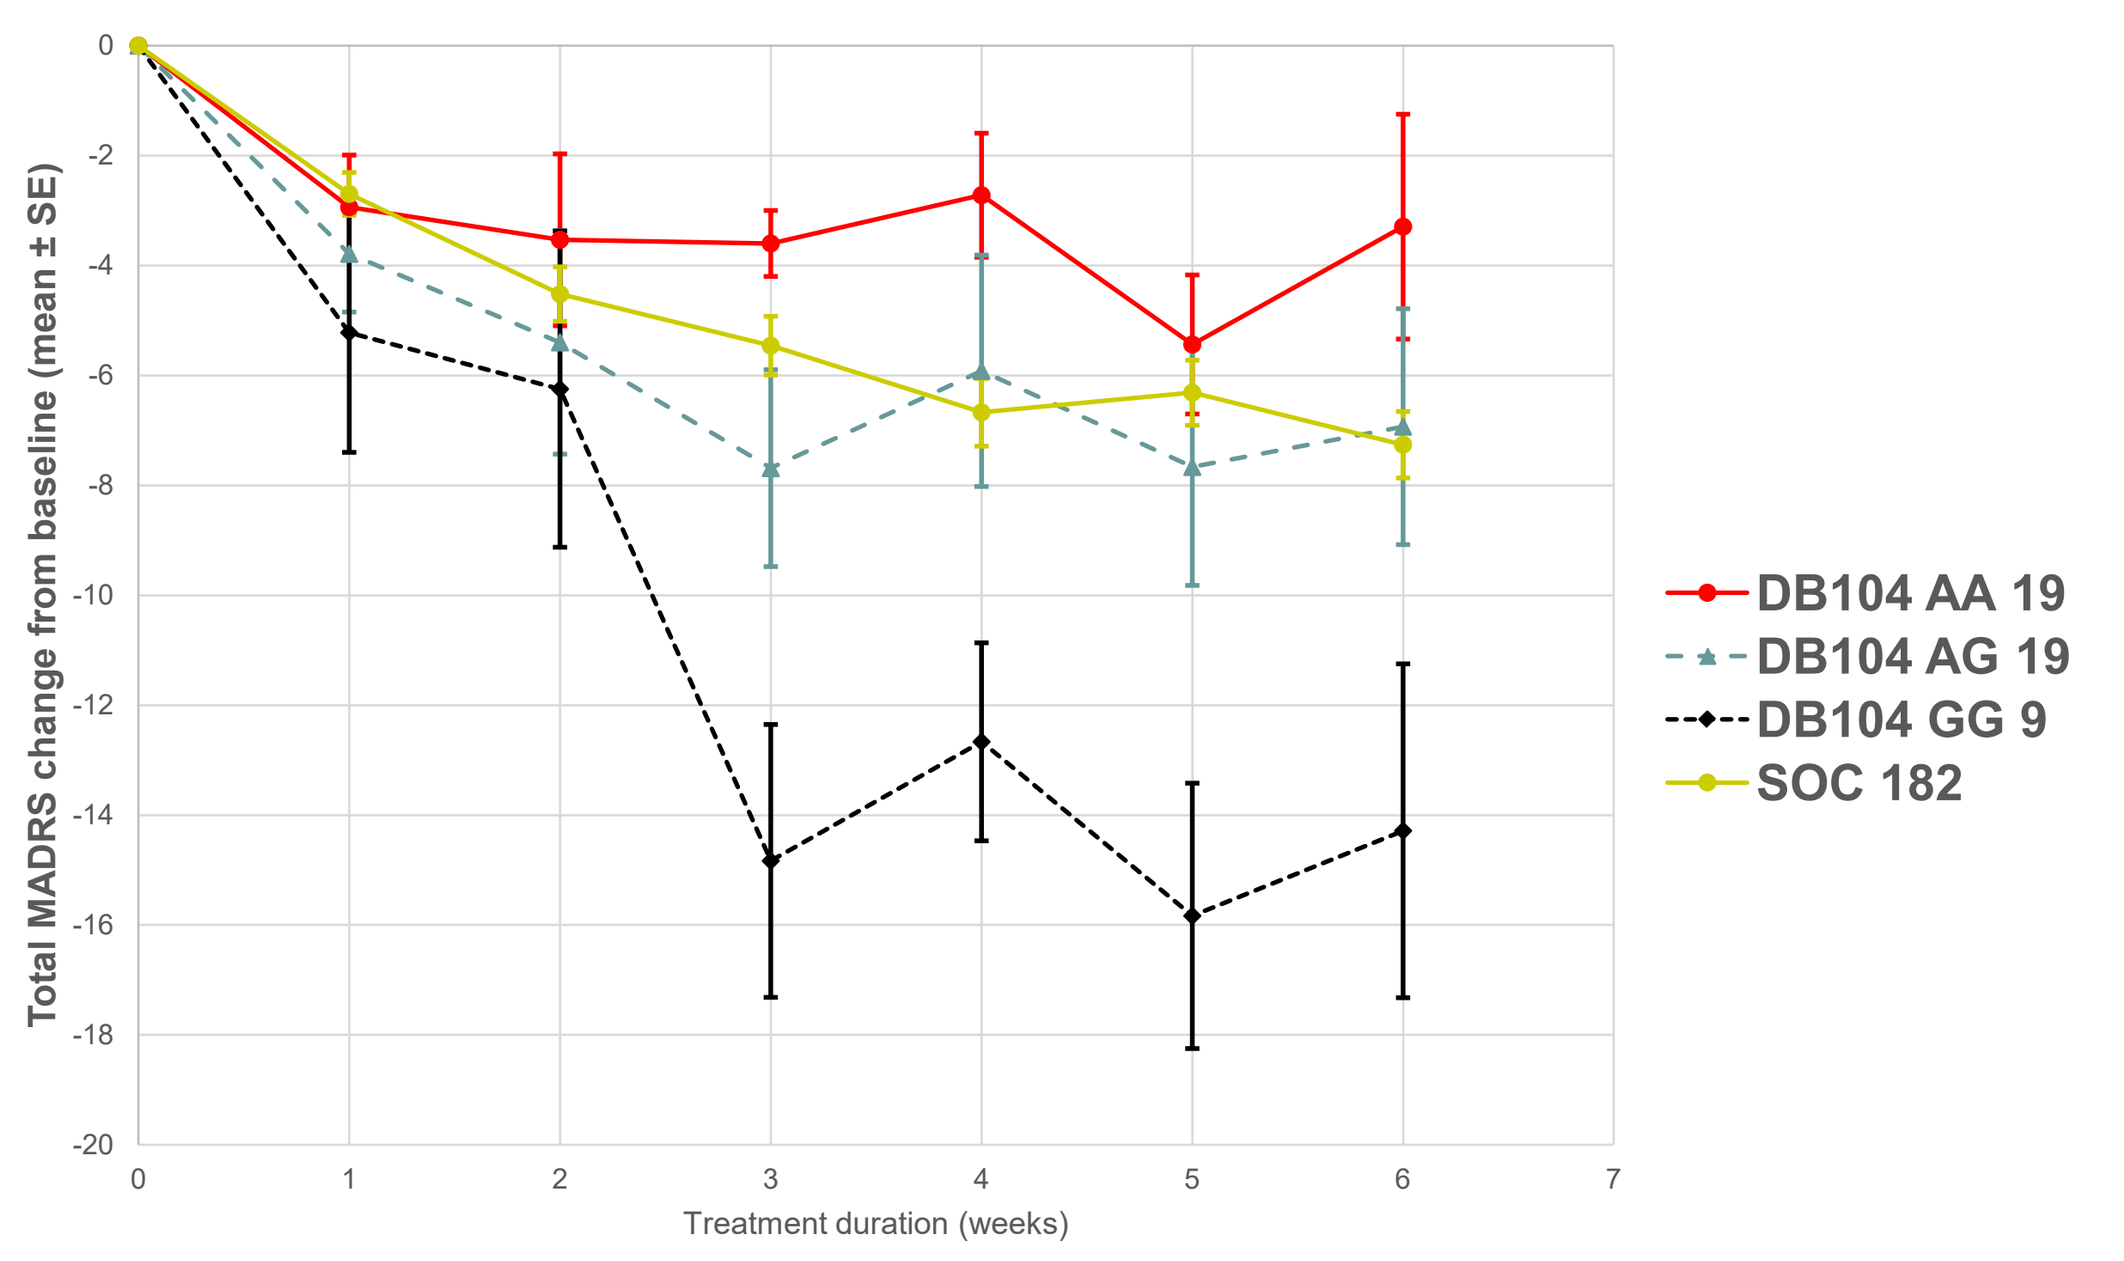
Total MADRS change from baseline (mean ± SE)
Treatment duration (weeks)
DB104 AA 19
DB104 AG 19
DB104 GG 9
SOC 182

MADRS Score Change by DGM4 Genotypes in Control Arm (SOC: Duloxetine/Escitalopram)

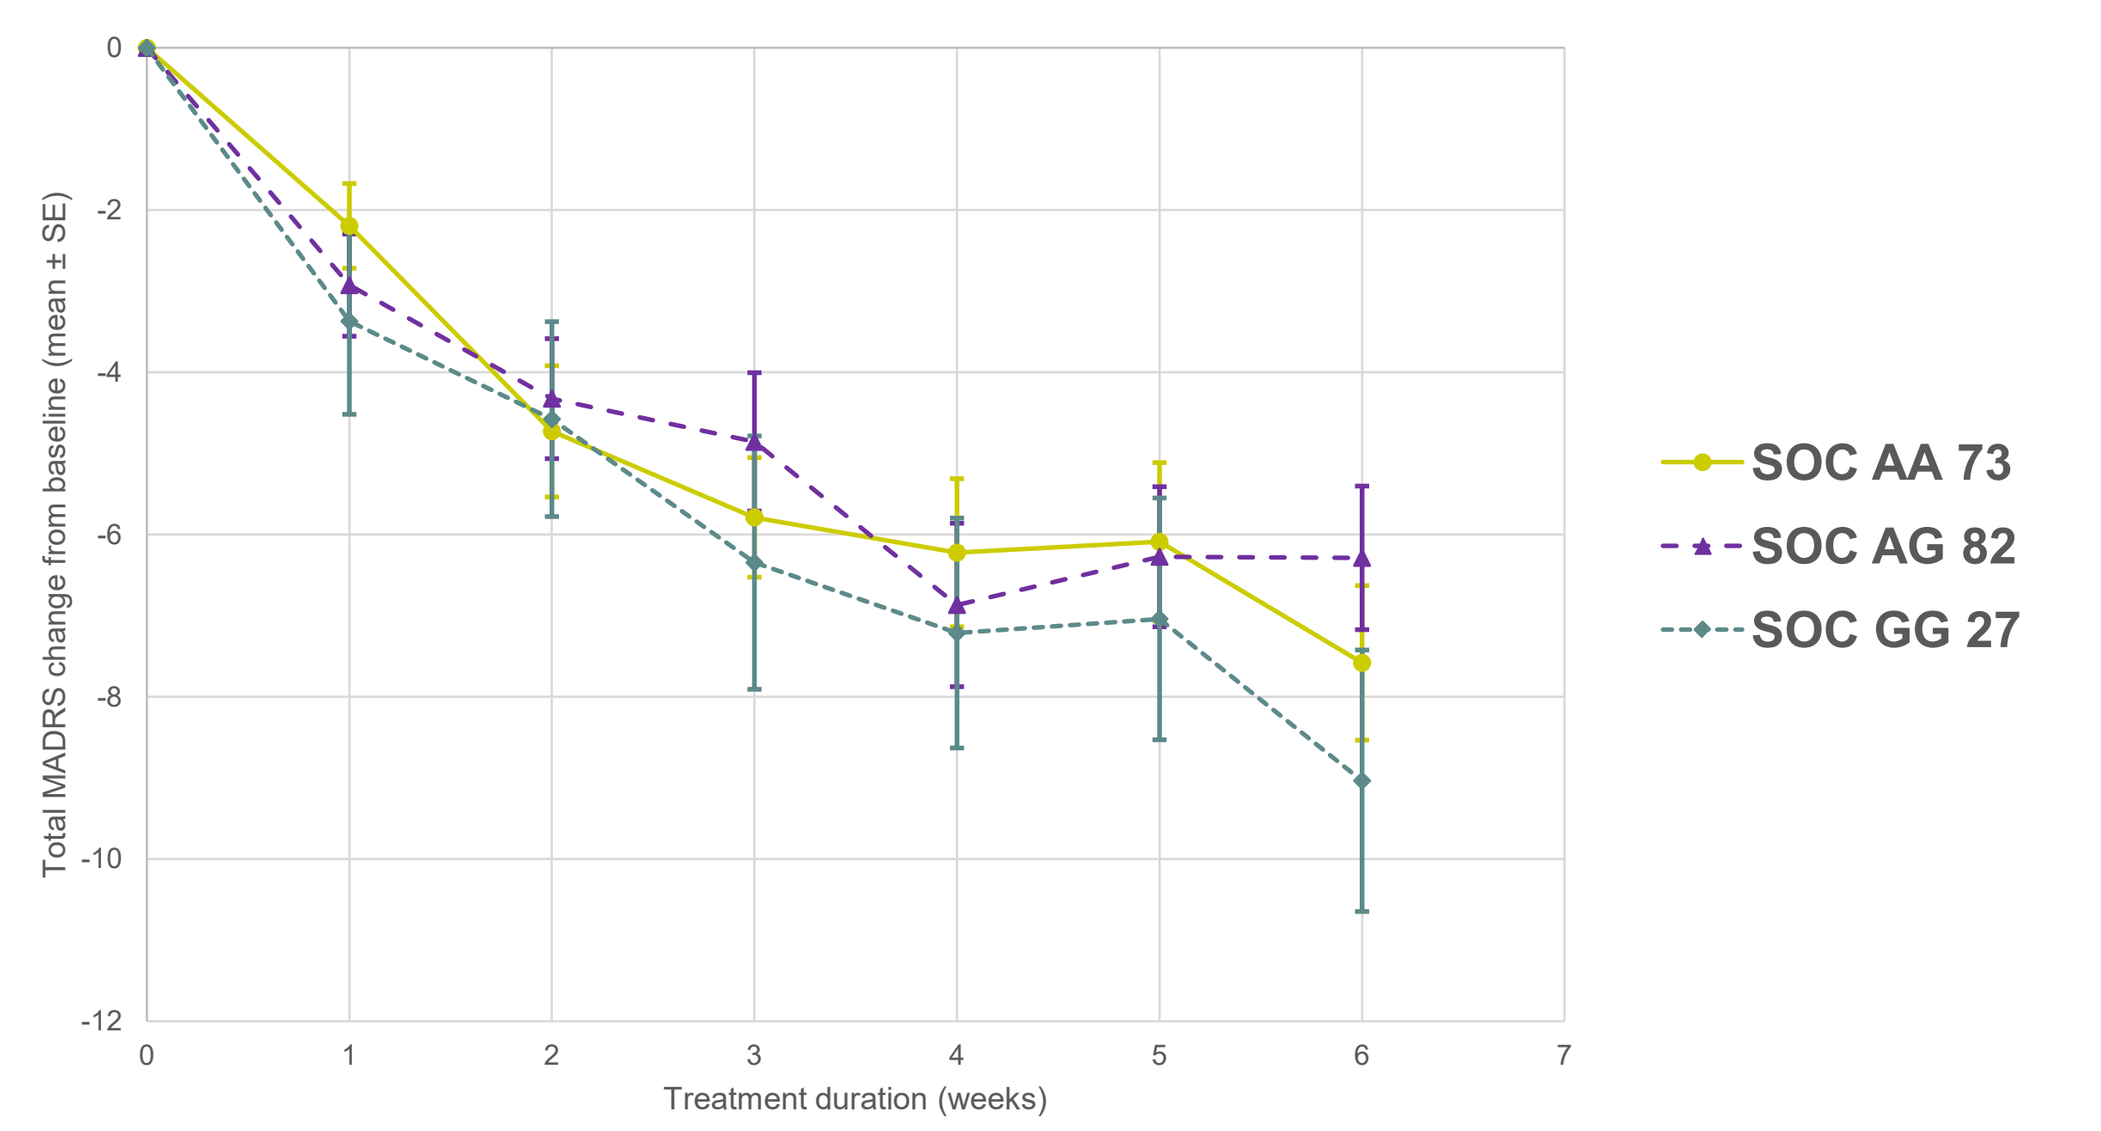
Total MADRS change from baseline (mean ± SE)
Treatment duration (weeks)
SOC AA 73
SOC AG 82
SOC GG 27

MADRS Score Change in All Patients by DGM4 Genotypes

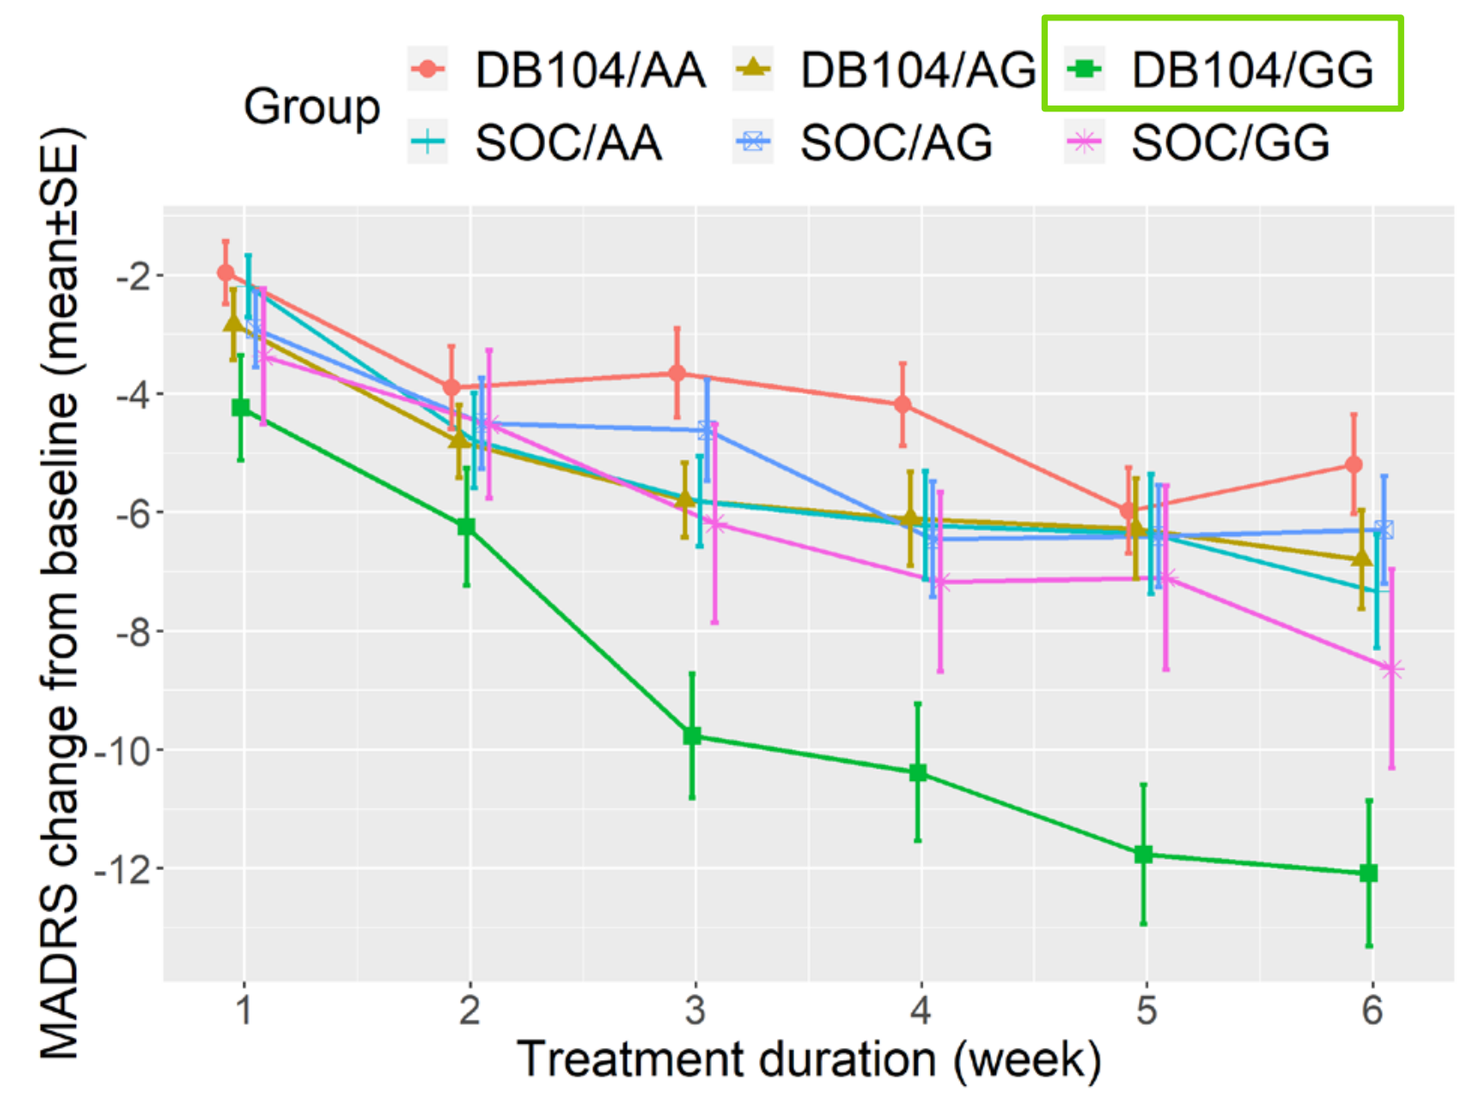
Group
DB104/AA
DB104/AG
DB104/GG
SOC/AA
SOC/AG
SOC/GG
MADRS change from baseline (mean±SE)
Treatment duration (week)

SOC (standard of care): Duloxetine/Escitalopram

Drug/Genotype	Patients (n)
DB104/AA	85
DB104/AG	94
DB104/GG	54
SOC/AA	73
SOC/AG	82
SOC/GG	27
Total	415

Discussion

- The planned study approach represents the one of the first times genomic biomarkers will have been used to personalize treatment to subsets of persons with depression who are most likely to be responsive.
- Given the life-long nature of depression and the poor outcomes that many experience with existing treatments, identifying target populations using genomic biomarkers which are likely responsive to a medication for treatment-resistant depressed persons would be an important public health advance in personalized medicine.
- This biomarker will be used to select the study population in future clinical trials with liafensine for TRD.

Conclusions

- The current work has identified a specific polymorphism (SNP) that may permit detection of individuals likely to show clinical benefit to liafensine.
- Lessons learned from this trial can inform drug development and help build towards better study designs that are more likely to have successful outcomes.

Disclosure
All authors report potential conflicts as they are employees of Denovo Biopharma or have received funding from Denovo Biopharma