Variability of Cognitive Performance on the MATRICS Consensus Cognitive Battery (MCCB): Adding a Different Perspective

David Williamson,[1] Keith Nuechterlein,[2] Todd Tishler,[2] Joseph Ventura,[2] Benjamin Ellingson,[2] Ibrahim Turkoz,[3] Richard Keefe,[4] Amy O'Donnell,[1] Larry Alphs[1,a]

[1]Janssen Scientific Affairs, LLC, Titusville, NJ, USA; [2]University of California, Los Angeles, Los Angeles, CA, USA; [3]Janssen Research and Development, LLC, Titusville, NJ, USA; [4]Duke University, Durham, NC, USA

[a]Affiliation at time of study; Current affiliation: Newron Pharmaceuticals, LLC, Morristown, NJ, USA

INTRODUCTION

- Cognitive impairment is a well-established feature of schizophrenia.[1] The manner in which impairment is quantified has implications for the understanding of patterns of cognitive functioning, the evolution of these patterns over time, and the extent to which they may vary as a function of patient or treatment characteristics
- The MATRICS Consensus Cognitive Battery (MCCB) and the cognitive batteries that are based upon the MCCB have served for more than a decade as the standard by which cognitive impairment is measured in clinical trials. Its 10 tests contribute to a composite score, with mean composite scores typically used to characterize degree of impairment[2]
- Dispersion of test scores across the subtests of a battery within a single administration has also proved to be a useful lens through which to view cognitive data. Increased dispersion is associated with impairment in traumatic brain injury,[3] mild cognitive impairment,[4] dementia,[5] and age-related cognitive decline.[6] Dispersion also has proven utility in the assessment of cognitive functioning in patients with schizophrenia[7,8]; however, an analysis of dispersion among MCCB scores has not been reported in the schizophrenia literature, despite the ubiquity of the MCCB in clinical trials examining cognition in schizophrenia
- The recently completed Disease Recovery Evaluation and Modification (DREaM: NCT02431702) study was conducted to examine whether a long-acting injectable antipsychotic (paliperidone palmitate [PP]), available in monthly and every-3-months formulations, can slow disease progression and possibly modify the course of schizophrenia compared with oral antipsychotics (OAPs) in patients with recent-onset schizophrenia or schizophreniform disorder
 - Here, we present the results of a post hoc analysis examining dispersion of performance on the Neurocognitive Composite of the MCCB in DREaM

OBJECTIVE

- To determine whether dispersion of performance on the tests composing the Neurocognitive Composite of the MCCB provides additional information than can be obtained by examination of the Neurocognitive Composite in isolation

METHODS

DREaM Study Design

- DREaM was a prospective, matched-control, double-randomized, open-label, flexible-dose, multicenter study (**Figure 1**)
- Patients with a diagnosis of schizophrenia or schizophreniform disorder (based on the Diagnostic and Statistical Manual of Mental Disorders, 5th edition [DSM-5]) and first psychotic episode within 2 years were eligible for enrollment

Figure 1. DREaM Study Design

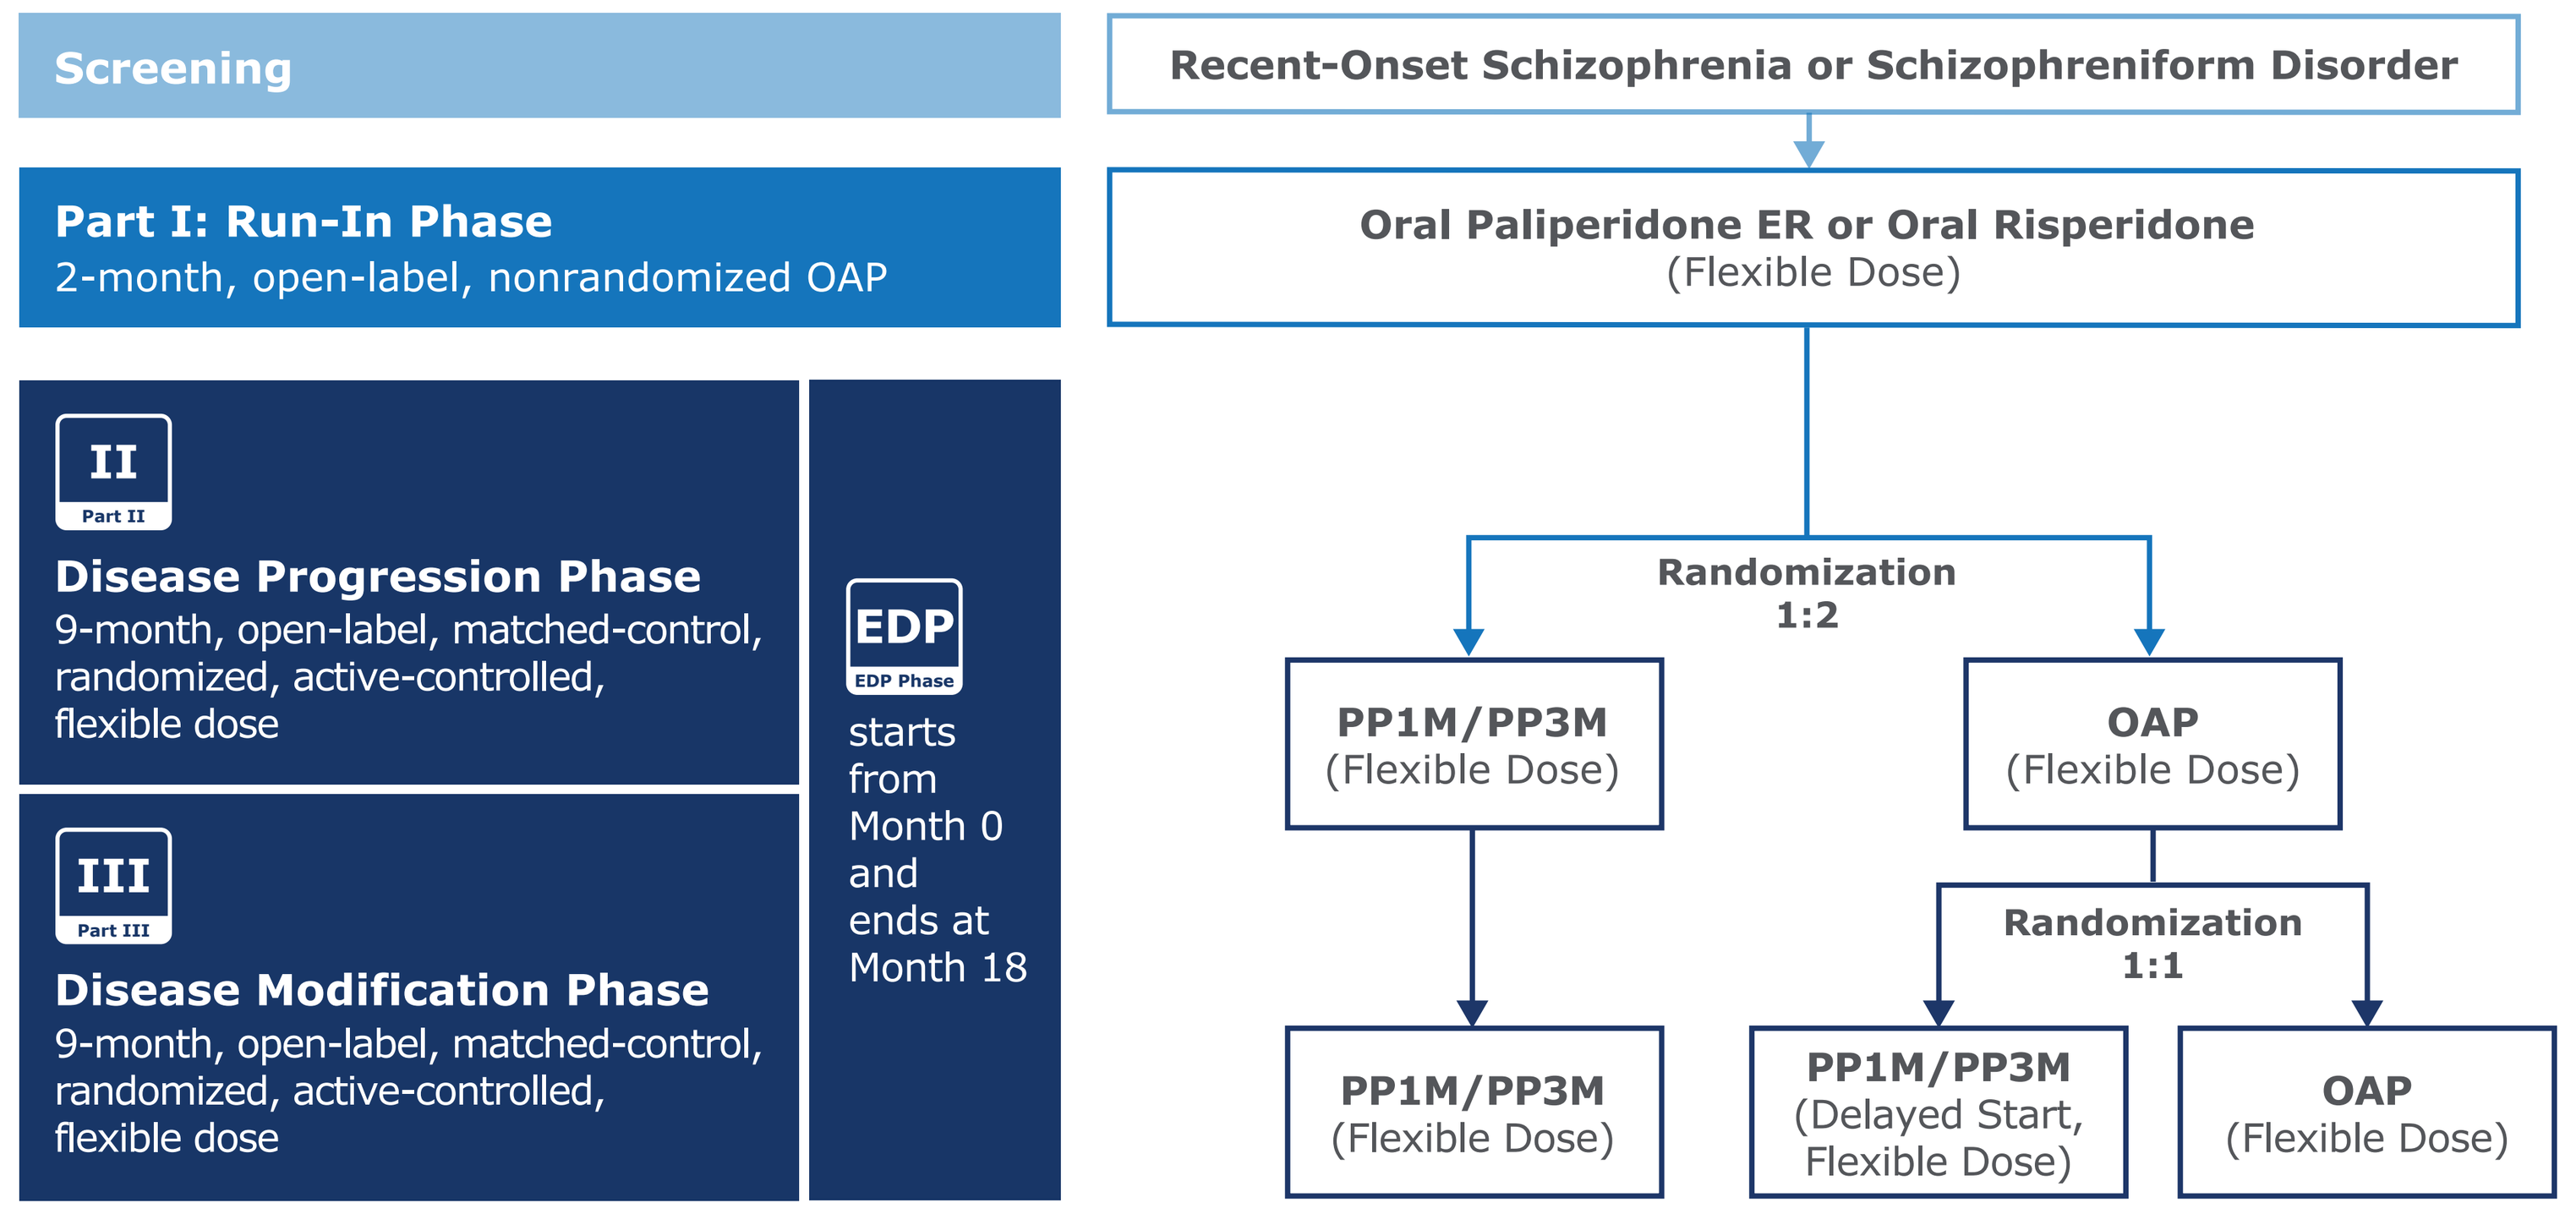
Screening
Recent-Onset Schizophrenia or Schizophreniform Disorder
Part I: Run-In Phase
2-month, open-label, nonrandomized OAP
Oral Paliperidone ER or Oral Risperidone
(Flexible Dose)
II
Part II
Disease Progression Phase
9-month, open-label, matched-control, randomized, active-controlled, flexible dose
EDP
EDP Phase
starts from Month 0 and ends at Month 18
III
Part III
Disease Modification Phase
9-month, open-label, matched-control, randomized, active-controlled, flexible dose
Randomization 1:2
PP1M/PP3M (Flexible Dose)
OAP (Flexible Dose)
Randomization 1:1
PP1M/PP3M (Flexible Dose)
PP1M/PP3M (Delayed Start, Flexible Dose)
OAP (Flexible Dose)

ER, extended release; EDP, extended disease progression; OAP, oral antipsychotic; PP, paliperidone palmitate; PP1M, paliperidone palmitate once monthly; PP3M, paliperidone palmitate once every 3 months.
In Parts II and III: 9-month PP treatment was defined as paliperidone palmitate once monthly (PP1M) for a minimum of 4 months (5 injections) followed by paliperidone palmitate once every 3 months (PP3M) treatment. 5 months/2 injections does not apply to the continued PP arm in Part III.

Statistical Methods

- Measure
 - Cognition was assessed using the Neurocognitive Composite of the MCCB. This composite, often used in clinical trials, represents the average performance relative to community norms, expressed in T-score units, across all MCCB subtests, excluding the measure of social cognition (the Managing Emotions branch of the Mayer–Salovey–Caruso Emotional Intelligence Test)
- Analyses
 - Time points for analysis were at the beginning of randomization (Part II baseline), the end of Part II (Part II end point/Part III baseline), and the end of Part III
 - Dispersion for each participant, by visit, was calculated as the variability of subtest scores relative to mean performance (expressed in T-score units)
 - Data were analyzed with a repeated measures mixed effects model, with treatment and visit as fixed effects and a treatment-by-visit interaction. The correlation of the repeated measures within participants was modeled with an unstructured covariance structure
 - Because primary analyses were not powered to detect differences in dispersion, this post hoc analysis examined contrasts to identify potential differential temporal patterns within groups

RESULTS

Sample Description

- The numbers of patients with MCCB data at baseline of Part II (day 57), day 260 of Part II, and day 260 of Part III, respectively, were 49, 49, and 39 (PP group), and 63, 59, and 44 (OAP group)
- Patient demographics and psychiatric histories at Part II baseline (day 57) are summarized in **Table 1** and **Table 2**

Table 1. Patient Demographics at Part II Baseline[a]

	PP/PP (n = 49)	OAP/OAP (n = 63)
Age, mean (SD), years	23.7 (4.65)	23.0 (4.30)
Male, n (%)	41 (83.7)	47 (74.6)
Race, n (%)		
White	24 (49.0)	23 (36.5)
Black or African American	17 (34.7)	25 (39.7)
Other	8 (16.3)	15 (23.8)
Education, n (%)		
High school or less (≤12 years)	29 (59.2)	39 (61.9)
>12 years	20 (40.8)	24 (38.1)

OAP, oral antipsychotic; PP, paliperidone palmitate; SD, standard deviation.
[a]No significant differences in baseline demographics were observed between treatment groups.

Table 2. Psychiatric History at Part II Baseline[a]

	PP/PP (n = 49)	OAP/OAP (n = 63)
Duration of illness, n (%)		
<6 months	27 (55.1)	40 (63.5)
6-12 months	8 (16.3)	7 (11.1)
>12 months	14 (28.6)	16 (25.4)
CGI-S score, mean (SD)	3.3 (1.09)	3.2 (1.15)
No. of previous psychiatric hospitalizations, mean (SD)	1.1 (1.30)	1.2 (1.12)
MCCB Neurocognitive Composite score, mean (SD)	30.6 (12.14)	30.1 (14.58)

CGI-S, Clinical Global Impression-Severity scale; MCCB, MATRICS Consensus Cognitive Battery; OAP, oral antipsychotic; PP, paliperidone palmitate; SD, standard deviation.
[a]No significant differences in baseline psychiatric histories were observed between treatment groups.

MCCB Results

- Neurocognitive Composite
 - Analyses revealed a significant effect of visit ($P < 0.05$) but no treatment-by-visit interaction ($P = 0.548$) or effect of treatment group ($P = 0.425$)
 - Within-group analyses comparing the baseline and end point values of Parts II and III, respectively, showed that both groups performed similarly on the Neurocognitive Composite of the MCCB (**Figure 2**)
 - Both groups improved significantly during Part II (PP: $P < 0.05$, OAP: $P < 0.001$) and remained stable during Part III (PP: $P = 0.821$, OAP: $P = 0.375$)

Figure 2. MCCB Neurocognitive Composite score by visit and group

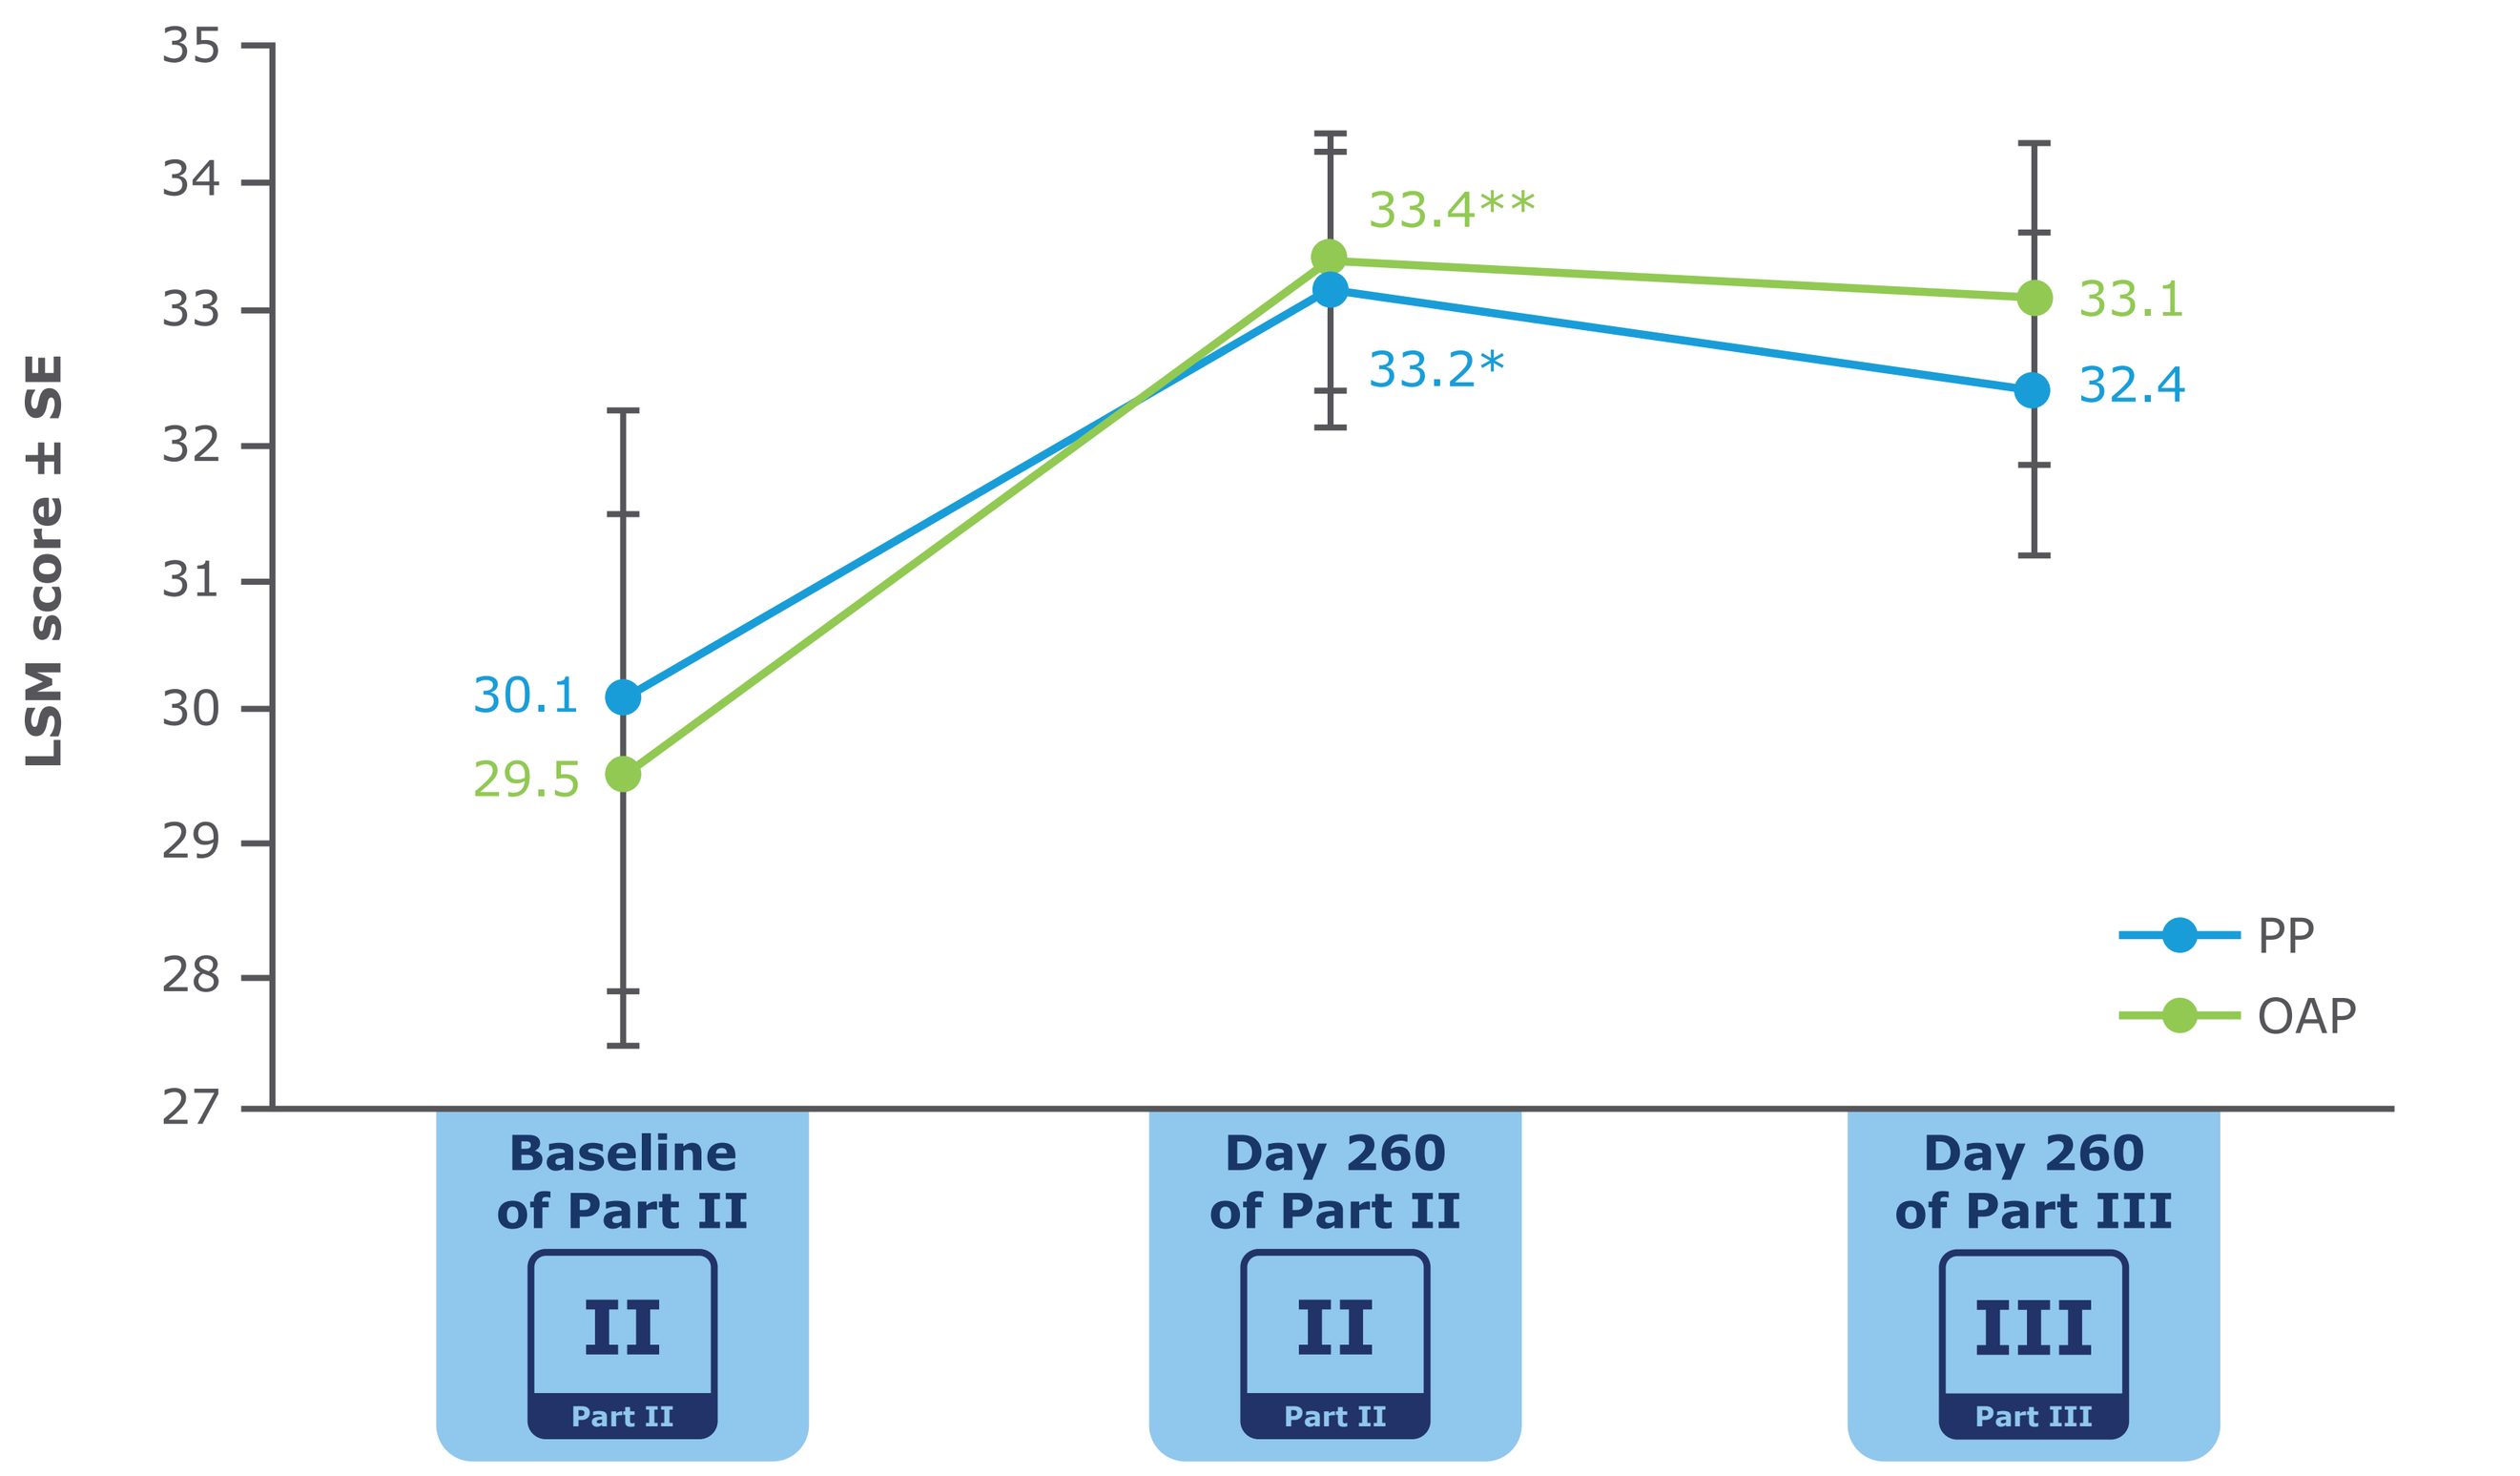
LSM score ± SE
35
34
33
32
31
30
29
28
27
30.1
29.5
33.4**
33.2*
33.1
32.4
PP
OAP
Baseline of Part II
Day 260 of Part II
Day 260 of Part III

LSM, least squares mean; MCCB, MATRICS Consensus Cognitive Battery; OAP, oral antipsychotic; PP, paliperidone palmitate; SE, standard error.
*$P < 0.05$ for Day 260 (Part II) versus baseline; **$P < 0.001$ for Day 260 (Part II) versus baseline.

- Dispersion among Neurocognitive Composite subtest performances
 - In contrast, within-group analyses of dispersion provided a different, nonredundant perspective (**Figure 3**)
 - In Part II, a nonsignificant improvement (ie, decline) in dispersion was observed in both groups. In Part III, dispersion remained stable in the PP group, whereas in the OAP group it became significantly more variable ($P < 0.01$)

Figure 3. Dispersion between performances on MCCB Neurocognitive Composite component subtests by visit and group

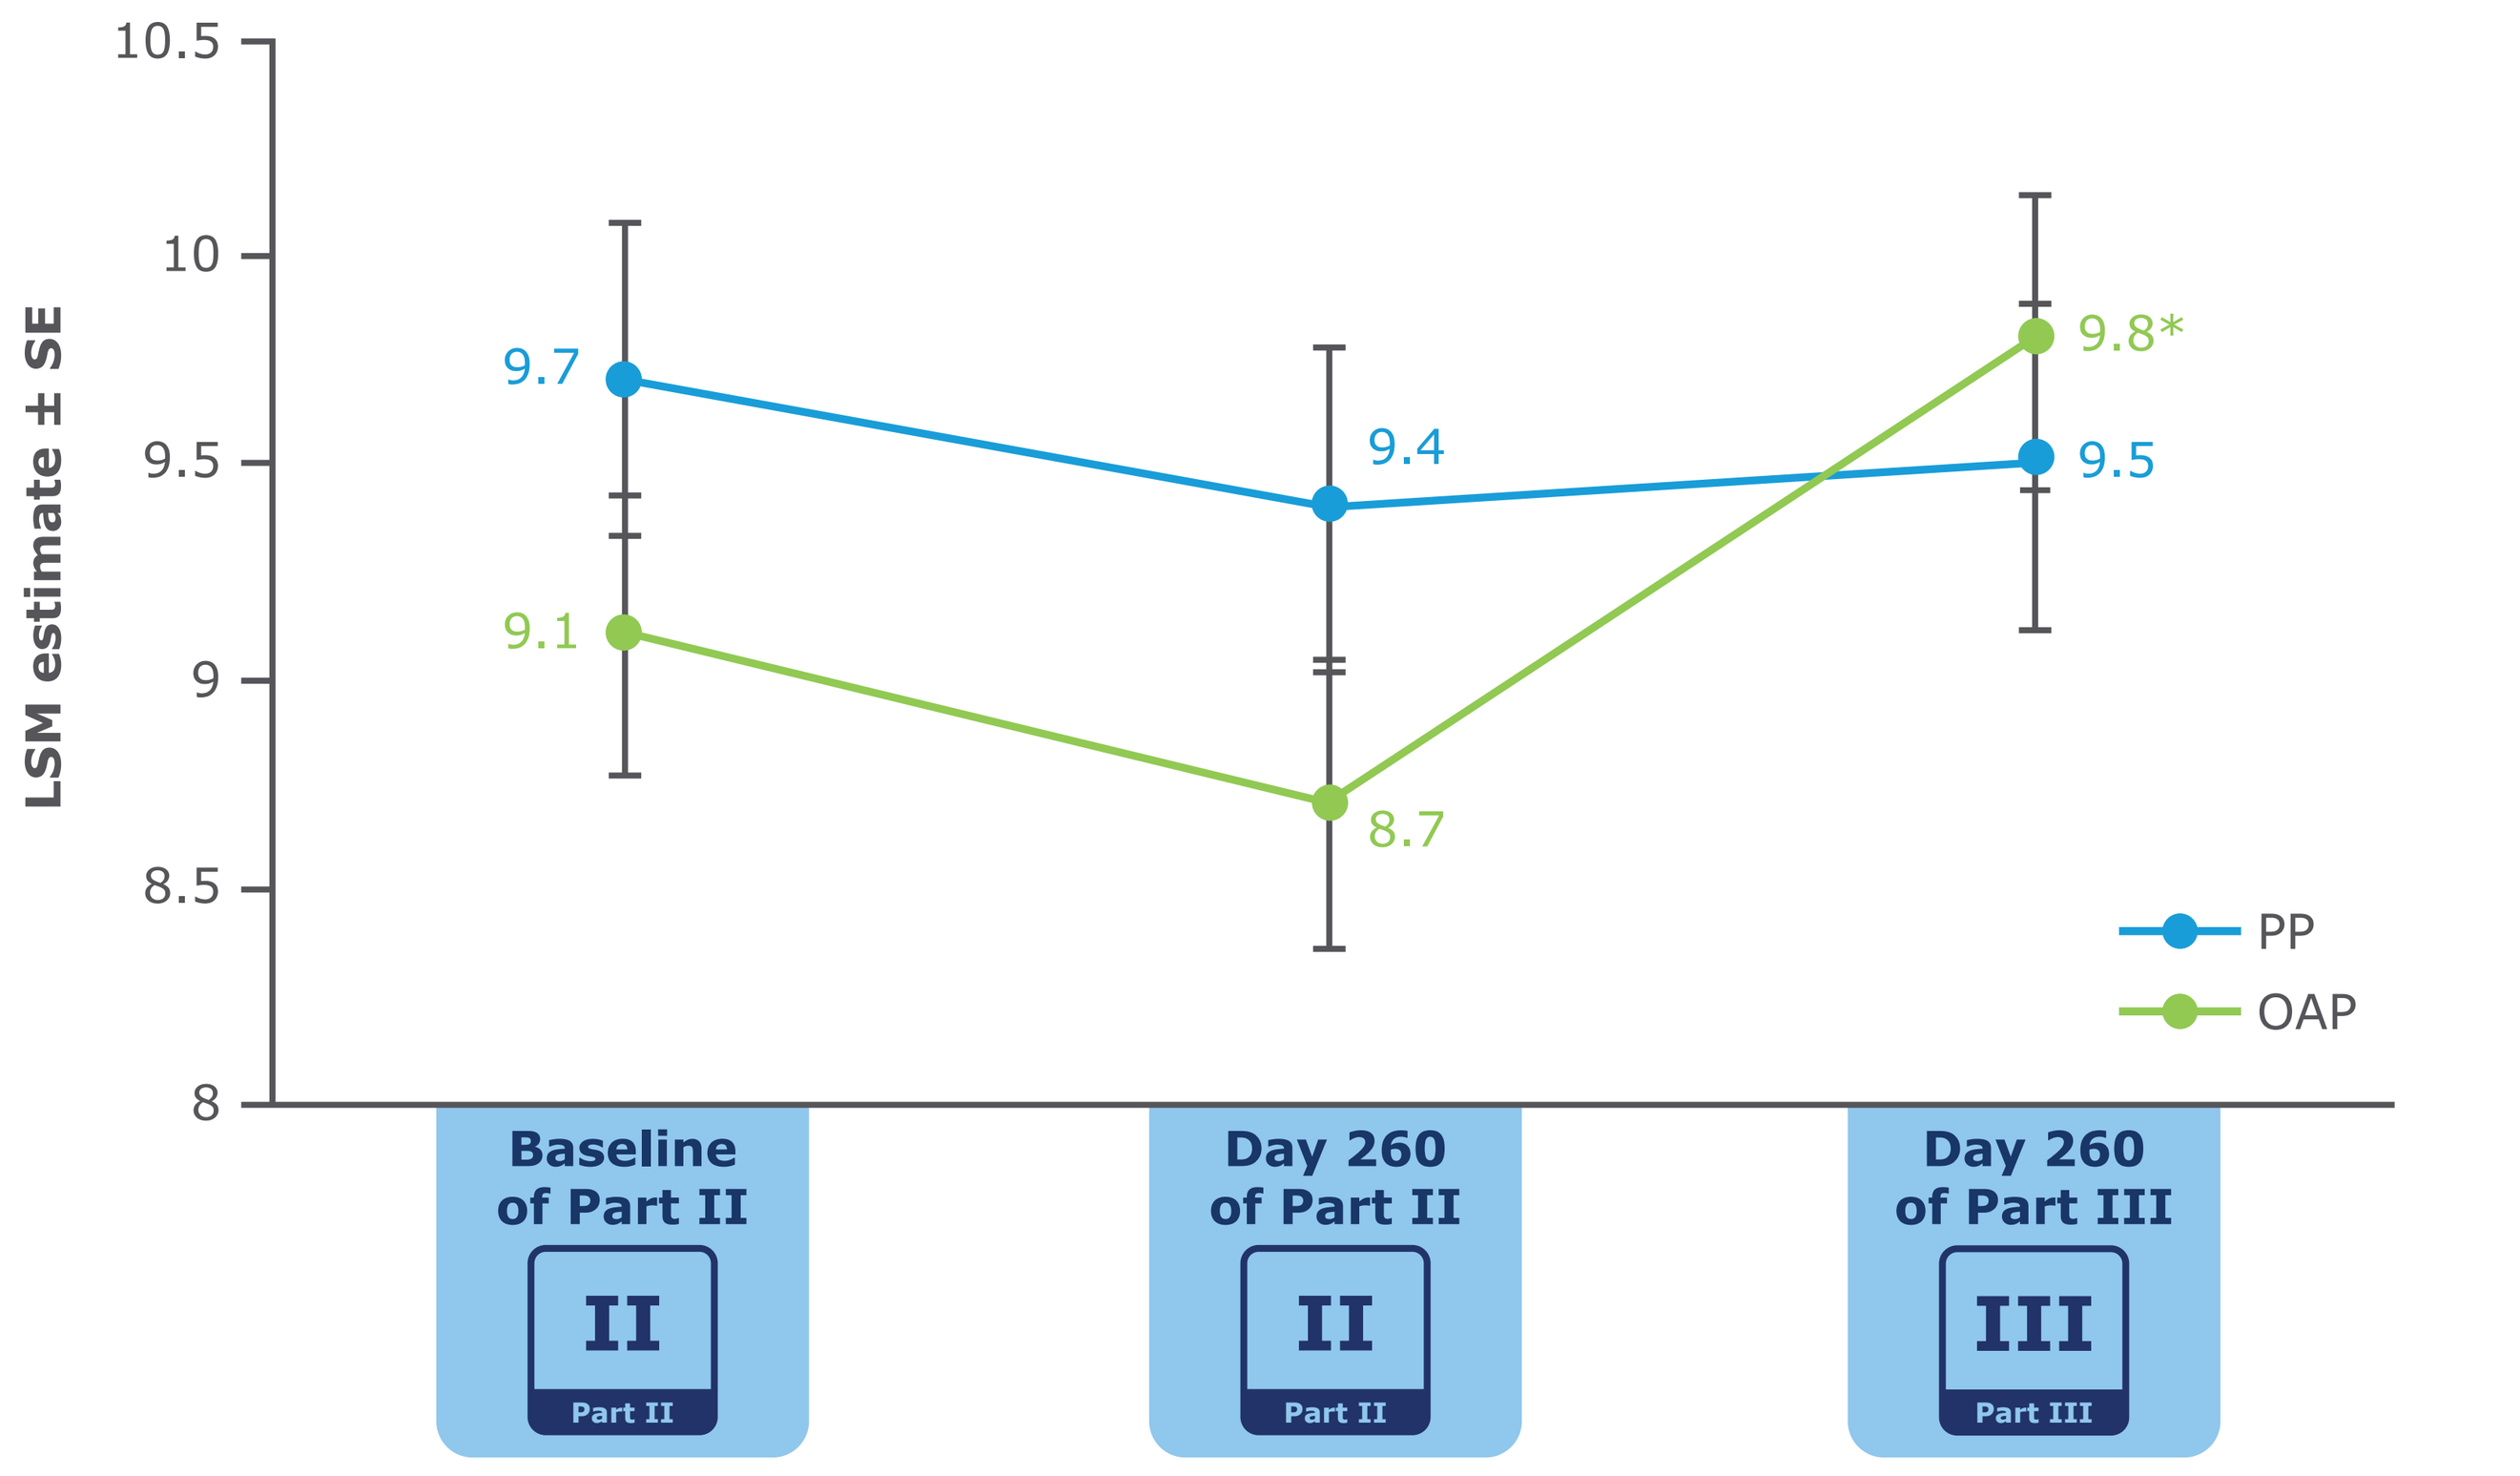
LSM estimate ± SE
10.5
10
9.5
9
8.5
8
9.7
9.1
9.4
8.7
9.8*
9.5
PP
OAP
Baseline of Part II
Day 260 of Part II
Day 260 of Part III

LSM, least squares mean; MCCB, MATRICS Consensus Cognitive Battery; OAP, oral antipsychotic; PP, paliperidone palmitate; SE, standard error.
*$P < 0.01$ for Day 260 (Part II) versus Day 260 (Part III).

DISCUSSION

- Similar to the pattern observed in in other populations,[9,10] intra-individual dispersion of MCCB subtest scores provided information that differed from that evident in composite scores based on the same battery
- Whereas the Neurocognitive Composite scores varied little between the groups in terms of magnitude or pattern over time, the dispersion of scores suggested that both groups improved modestly during the first 9 months of treatment after oral run-in, but these gains were maintained more effectively in the PP group than in the OAP group during the ensuing 9 months
- Given the relatively small sample sizes, the relatively small absolute magnitude of the differences, and the post hoc nature of these analyses, replication in a larger sample would be beneficial. The clinical meaningfulness of these differences merits further investigation

References

1. Keefe R et al. *Handb Exp Pharmacol*. 2012;213:11-37.
2. Nuechterlein K et al. *Am J Psych*. 2008;165:203-213.
3. Hill BD et al. *Arch Clin Neuropsychol*. 2013;28:640-648.
4. Halliday DWR et al. *J Intell*. 2018;6:12.
5. Reckess GZ et al. *Neuropsychology*. 2014;28:254-260.
6. De Felice S, Holland CA. *Front Psychol*. 2018;9:1973.
7. Cole VT et al. *Schizophr Res.* 2011;129:91-93.
8. Reichenberg A et al. *Schizophr Res*. 2006;85:49-57.
9. MacDonald SWS, Stawski RS. In: Diehl M et al, eds. *Handbook of Intraindividual Variability Across the Life Span*. Routledge/Taylor & Francis Group; 2015: 231-257.
10. Ram N et al. *Psychol Aging*. 2005;20:623-633.

Disclosures

Financial Support

The authors thank Susanna Bae, PharmD, and Madeline Pfau, PhD (ApotheCom, New York, NY) for editorial and writing services, which were funded by Janssen Scientific Affairs, LLC.

Conflicts of interest

DW, IT, and AO are employees of Janssen Pharmaceuticals and hold stock in Johnson & Johnson, Inc. LA is a former employee of Janssen Pharmaceuticals and holds stock in Johnson & Johnson, Inc. KN has received research grant support from Janssen Scientific Affairs, LLC, and has served as a consultant to Astellas, Genentech, Janssen, Medincell, Otsuka, Takeda, and Teva. TT has no conflicts of interest to disclose. JV has received research grants from Posit Science and Genentech, Inc., and has been a consultant to Boehringer-Ingelheim and Posit Science, Inc. BE has received research grant support from Siemens, Janssen, VBL, the National Brain Tumor Society, and the American Cancer Society, and has served as a consultant to Image Analysis Group (IAG), Onconceutics, Inc., BeiGene, Tocagen, and the Global Coalition for Adaptive Research (GCAR); BE has also attended an advisory board or served as a paid consultant to Medicenna, MedQIA, LLC, NeoSoma, Agios Pharmaceuticals, Siemens, Imaging Endpoints, Kazia/Novogen, NW Pharmaceuticals, and the NIH/NCI Cancer Imaging Steering Committee. RK owns VeraSci, a for-profit company that provides comprehensive services to more than 100 business entities, most of which are pharmaceutical companies, including Janssen.

An electronic version of the poster can be viewed by scanning the QR code.

The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way. These are electronic reproductions of posters presented at The International Society of CNS Clinical Trials and Methodology (ISCTM) 2020 Autumn Virtual Conference; September 21-25, 2020. All copyrights remain those of the copyright holder. This page will not be available after October 25, 2020.

https://bit.ly/2EAzT0X
(This URL is case sensitive.)

Supported by Janssen Scientific Affairs, LLC

Presented at the International Society of CNS Clinical Trials and Methodology (ISCTM) 2020 Autumn Virtual Conference; September 21-25, 2020