ADVANCE: Adherence to Antipsychotic and Adjunctive Pimavanserin in Patients With Negative Symptoms of Schizophrenia

Brandon Abbs,[1] Dragana Bugarski-Kirola,[2] Mona Darwish,[1] I-Yuan (Cathy) Liu,[3] Srdjan Stankovic[3]

[1]ACADIA Pharmaceuticals Inc., Princeton, NJ, USA; [2]ACADIA Pharmaceuticals GmbH, Lucerne, Switzerland; [3]ACADIA Pharmaceuticals Inc., San Diego, CA, USA

METHODOLOGICAL QUESTION ADDRESSED

- Could checking medication adherence at screening potentially improve adherence in a clinical trial and improve clinical trial quality and outcomes?

BACKGROUND

- Treatment nonadherence is common in patients with psychotic disorders and is associated with higher costs of care and worse clinical outcomes[1]
- Treatment nonadherence is problematic for patient care in both real-world and clinical trial settings[1,2]
- In a clinical trial, nonadherence and failure to achieve therapeutic levels of the study treatment can complicate the interpretation of the results[2] and contribute to the failed development of a potentially effective treatment
- High adherence in a clinical trial can complement positive efficacy and safety results, providing additional evidence that treatment improved patient care without treatment-limiting side effects
- Therefore, it is important to test for adherence when determining eligibility for a trial, measure adherence during the trial, and review the adherence results after trial completion, regardless of study outcome
- The randomized, double-blind, placebo-controlled, phase 2 ADVANCE study (NCT02970305) evaluated the efficacy and safety of pimavanserin, a serotonin (5-hydroxytryptamine) 5-HT_{2A} receptor antagonist/inverse agonist, as adjunctive treatment for the negative symptoms of schizophrenia[3]
 - The ADVANCE study demonstrated significant improvement in negative symptoms of schizophrenia with adjunctive pimavanserin; pimavanserin met the primary efficacy endpoint of superiority to placebo in the change in the Negative Symptom Assessment-16 (NSA-16) total score from baseline to Week 26[3]
- Here we present treatment adherence (defined as a measurable drug blood level) for patients in the ADVANCE study and key elements of the study design that may have contributed to treatment adherence

METHODS

- ADVANCE (NCT02970305) was a phase 2, 26-week, randomized, double-blind, placebo-controlled study in outpatients with schizophrenia and predominant negative symptoms while on treatment with an antipsychotic medication (Figure 1)

Figure 1. ADVANCE study design

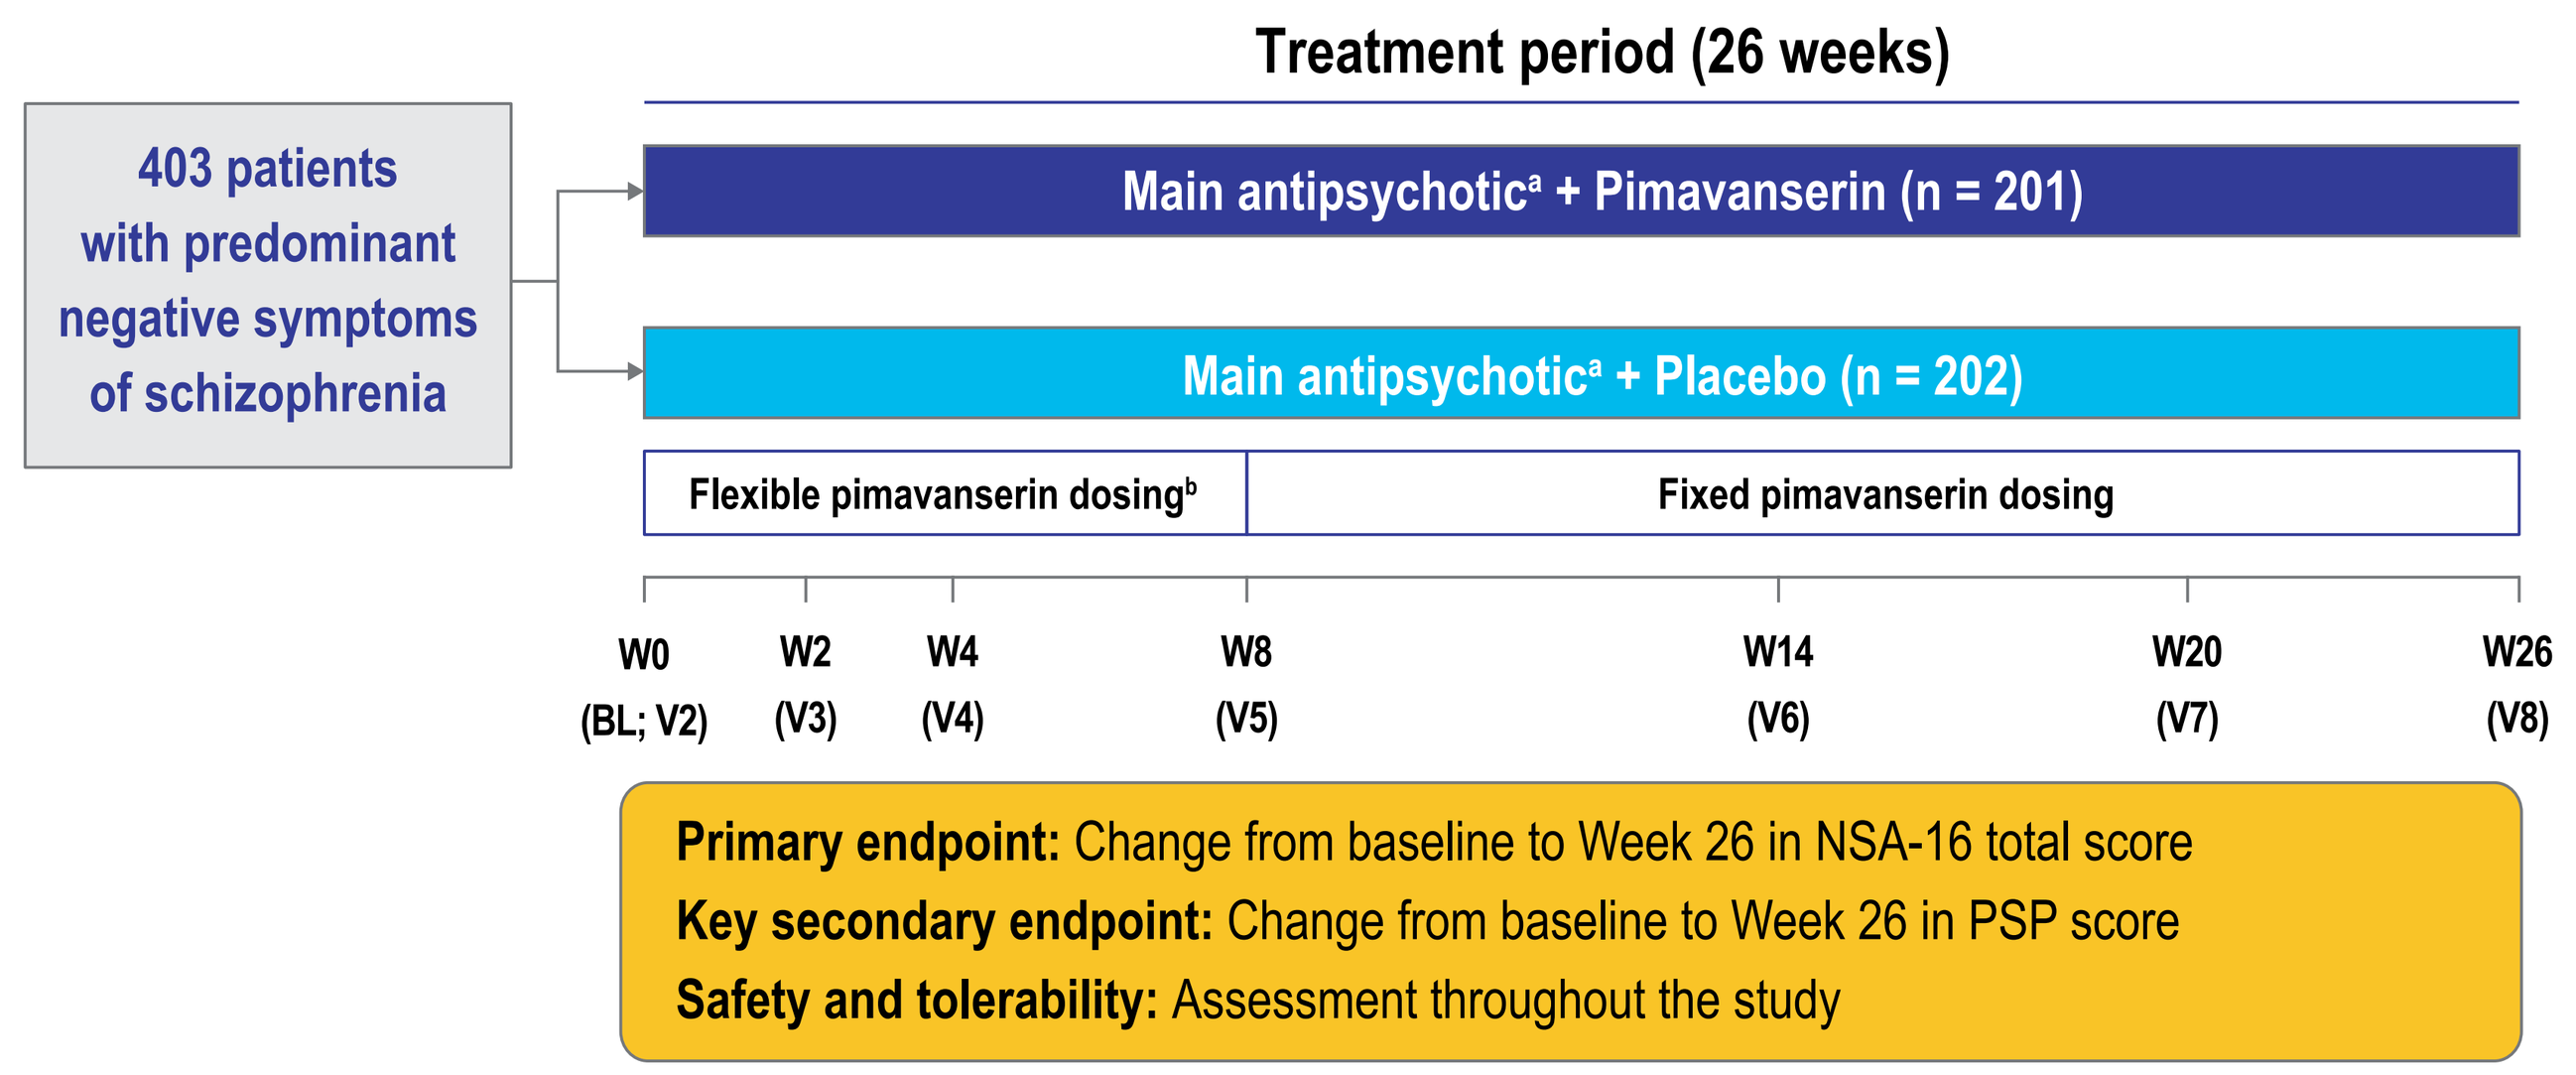
Treatment period (26 weeks)
403 patients with predominant negative symptoms of schizophrenia
Main antipsychotic[a] + Pimavanserin (n = 201)
Main antipsychotic[a] + Placebo (n = 202)
Flexible pimavanserin dosing[b]
Fixed pimavanserin dosing
W0 (BL; V2)
W2 (V3)
W4 (V4)
W8 (V5)
W14 (V6)
W20 (V7)
W26 (V8)
Primary endpoint: Change from baseline to Week 26 in NSA-16 total score
Key secondary endpoint: Change from baseline to Week 26 in PSP score
Safety and tolerability: Assessment throughout the study

[a]One of the following antipsychotics: aripiprazole (oral or long-acting injection), asenapine, brexpiprazole, cariprazine, lurasidone, olanzapine, and risperidone (oral or long-acting injection); [b]Starting dose of pimavanserin or placebo was 20 mg daily and could be adjusted up to 34 mg or down to 10 mg daily after 2 weeks based on investigator discretion
BL, baseline; NSA-16, Negative Symptom Assessement-16; PSP, Personal and Social Performance Scale; V, visit; W, week

- The study included several eligibility and testing requirements to demonstrate adequate treatment and adherence to the main antipsychotic before entry into the trial
 - Inclusion criteria
 - Documented treatment (eg, copy of prescription) for ≥8 weeks before screening with an adequate dose of an antipsychotic (within the dose range recommended according to the local prescribing information) and continuing on the same dose throughout the screening period
 - No dose change within 4 weeks before screening if oral antipsychotic and within 16 weeks before screening if long-acting injectable
 - Reliable caregiver to help monitor and promote treatment adherence
 - Screening procedures
 - Laboratory testing of blood sample to confirm the presence or absence of the main antipsychotic
 - Patients with confirmed absence of the main antipsychotic were excluded
 - Structured telemedicine interview of the patient by an independent clinician to confirm that the patient has schizophrenia with predominant negative symptoms while on adequate treatment with an antipsychotic
 - Comprehensive review of patient eligibility by the sponsor (DBK and BA) and site management personnel
 - Testing during the study
 - Blood samples were collected at baseline and Weeks 2, 8, 14, and 26 (or early exit) for pharmacokinetic assessments of the main antipsychotic and adjunctive pimavanserin
- A post hoc analysis used descriptive statistics to examine the proportion of patients who were adherent to the main antipsychotic at screening and to the main antipsychotic and pimavanserin during the study
- For this analysis, treatment adherence to pimavanserin and the main antipsychotic was defined as a blood sample test result above the lower limit of quantification for pimavanserin and the main antipsychotic, respectively

RESULTS

- Overall, 608 patients were screened with 36 rescreens, for a total of 644 screenings
- Failure to confirm the main antipsychotic in the screening blood sample was a common reason for screening failure (n = 32, Table 1), accounting for 13.3% of all screen failures
 - Rescreen was permitted exceptionally when a temporary condition may have contributed to nonadherence. Only 1 patient was randomized after an initial screen failure for nonadherence. This patient was switched to a different generic version of the main antipsychotic after no blood levels were detected while on the previous generic
- Other common reasons for screen failure included patients not meeting study inclusion criteria for presence of predominantly negative symptoms of schizophrenia and withdrawal of consent (Table 1)
 - Withdrawal of consent frequently resulted from the rigorous screening process required for the study

Table 1. Most common reasons for screening failure

	Screening failures[a] (n = 217)
Inclusion/exclusion criteria[b]	
Did not meet inclusion criteria	86
Sum of scores on the 8 PANSS Marder positive factor items of ≤22, with no more than 2 of the P1, P3, and P6 items (delusions, hallucinatory behavior, and suspiciousness/persecution) having score of 4 and none having score of ≥5 at both screening and baseline	32
Sum of scores on the 7 PANSS Marder negative factor items of ≥20 at screening and baseline, with score of ≥4 on at least 3 of the items or score of ≥5 on at least 2 of the items	25
Treatment with an adequate dose (within recommended dose range) of the main antipsychotic for ≥8 weeks before screening and stable throughout screening	9
Met exclusion criteria	135
Laboratory testing confirmed absence of main antipsychotic	32
Patient was judged to be inappropriate for the study[c]	20
Uncontrolled diabetes mellitus or requirement for insulin treatment	14
Urine drug screen at screening or baseline shows presence of prohibited substance of potential abuse	12
Condition present that would interfere with the patient's ability to comply with the study instructions, or that might confound the study results or put the patient at undue risk[d]	12
Withdrawal of consent[b]	21
Other[b]	3

[a]A patient could fail screening more than once; [b]For each screen failure, 1 category of reason (inclusion/exclusion criteria, withdrawal of consent, or other) is checked; however, for the inclusion/exclusion criteria category, there may have been multiple criteria not met; [c]Assessment by the investigator or medical monitor; [d]Assessment by the investigator
PANSS, Positive and Negative Syndrome Scale

- Patient demographics (Table 2) and disease characteristics (Table 3) were similar in the pimavanserin and placebo groups

Table 2. Baseline patient demographics

	Pimavanserin (n = 201)	Placebo (n = 202)
Age at screening, mean (SD), years	37.7 (9.4)	36.7 (9.2)
Sex – male, n (%)	131 (65.2)	137 (67.8)
Race – white, n (%)	187 (93.0)	186 (92.1)
Region, n (%)		
Europe	177 (88.1)	177 (87.6)
North America	24 (11.9)	25 (12.4)

SD, standard deviation

Table 3. Baseline disease characteristics

	Pimavanserin (n = 201)	Placebo (n = 202)
Main antipsychotic, n (%)		
Aripiprazole	53 (26.4)	52 (25.7)
Aripiprazole LA1	10 (5.0)	17 (8.4)
Lurasidone	2 (1.0)	2 (1.0)
Olanzapine	50 (24.9)	62 (30.7)
Risperidone	69 (34.3)	52 (25.7)
Risperidone LA1	17 (8.5)	17 (8.4)
Duration of main antipsychotic use, mean (SD), months	28.7 (39.6)	24.8 (38.7)
NSA-16 total score, mean (SD)	61.7 (8.5)	60.9 (8.7)
PANSS positive scale score, mean (SD)	13.1 (3.3)	13.7 (3.1)
PANSS negative scale score, mean (SD)	27.5 (3.7)	27.5 (3.5)
CGI-SCH-S score, mean (SD)	4.6 (0.6)	4.7 (0.6)
PSP score, mean (SD)	47.3 (11.7)	46.7 (10.8)
DAI-10 total score, mean (SD)	5.7 (3.1)	5.7 (3.3)

CGI-SCH-S, Clinical Global Impression of Schizophrenia – Severity Scale; DAI-10, 10-item Drug Attitude Inventory; NSA-16, 16-item Negative Symptom Assessment; PANSS, Positive and Negative Syndrome Scale; PSP, Personal and Social Performance scale; SD, standard deviation

- At baseline, 95.0% (383/403) of patients were determined to be adherent to their main antipsychotic based on measurable blood sample drug levels (Table 4); this is a substantial improvement over the adherence rate of 88.0% that would have been observed if patients with confirmed nonadherence at screening were enrolled (assuming they remained nonadherent at baseline)
- High adherence rates (>90%) for both the main antipsychotic and adjunctive pimavanserin were observed at Weeks 2, 8, 14, and 26/early exit (Table 4)
- Of the measured pimavanserin concentrations postbaseline, 91.7% were at or above the lower limit of the 90% prediction interval of the model-predicted median expected level
- At study exit (Week 26 or early exit), 91.8% (369/402) of patients were considered adherent to their main antipsychotic and 95.0% (190/200) of patients were considered adherent to pimavanserin (Table 4)

Table 4. Adherence to main antipsychotic and pimavanserin by study visit

	Pimavanserin (n = 201)	Placebo (n = 202)
Patients with detectable levels of main antipsychotic, n/N (%)		
Baseline	193/201 (96.0)	190/202 (94.1)
Week 2	186/192 (96.9)	179/192 (93.2)
Week 8	176/184 (95.7)	171/185 (92.4)
Week 14	162/177 (91.5)	166/179 (92.7)
Week 26/early exit[a]	186/200 (93.0)	183/202 (90.6)
Patients with detectable levels of pimavanserin, n/N (%)		
Week 2	186/191 (97.4)	NA
Week 8	183/184 (99.5)	NA
Week 14	173/177 (97.7)	NA
Week 26/early exit[a]	190/200 (95.0)	NA

[a]Data from patients at last study visit (Week 26 or early exit visit)
NA, not applicable; n/N, number of patients with measurable treatment levels/total number of patients with blood samples taken

- Rates of overall adherence during the study (with measurable drug levels at all 4 timepoints during treatment) were >82% for the main antipsychotic treatment in both treatment groups and 92% for adjunctive pimavanserin (Figure 2)

Figure 2. Adherence to main antipsychotic and adjunctive pimavanserin during study treatment

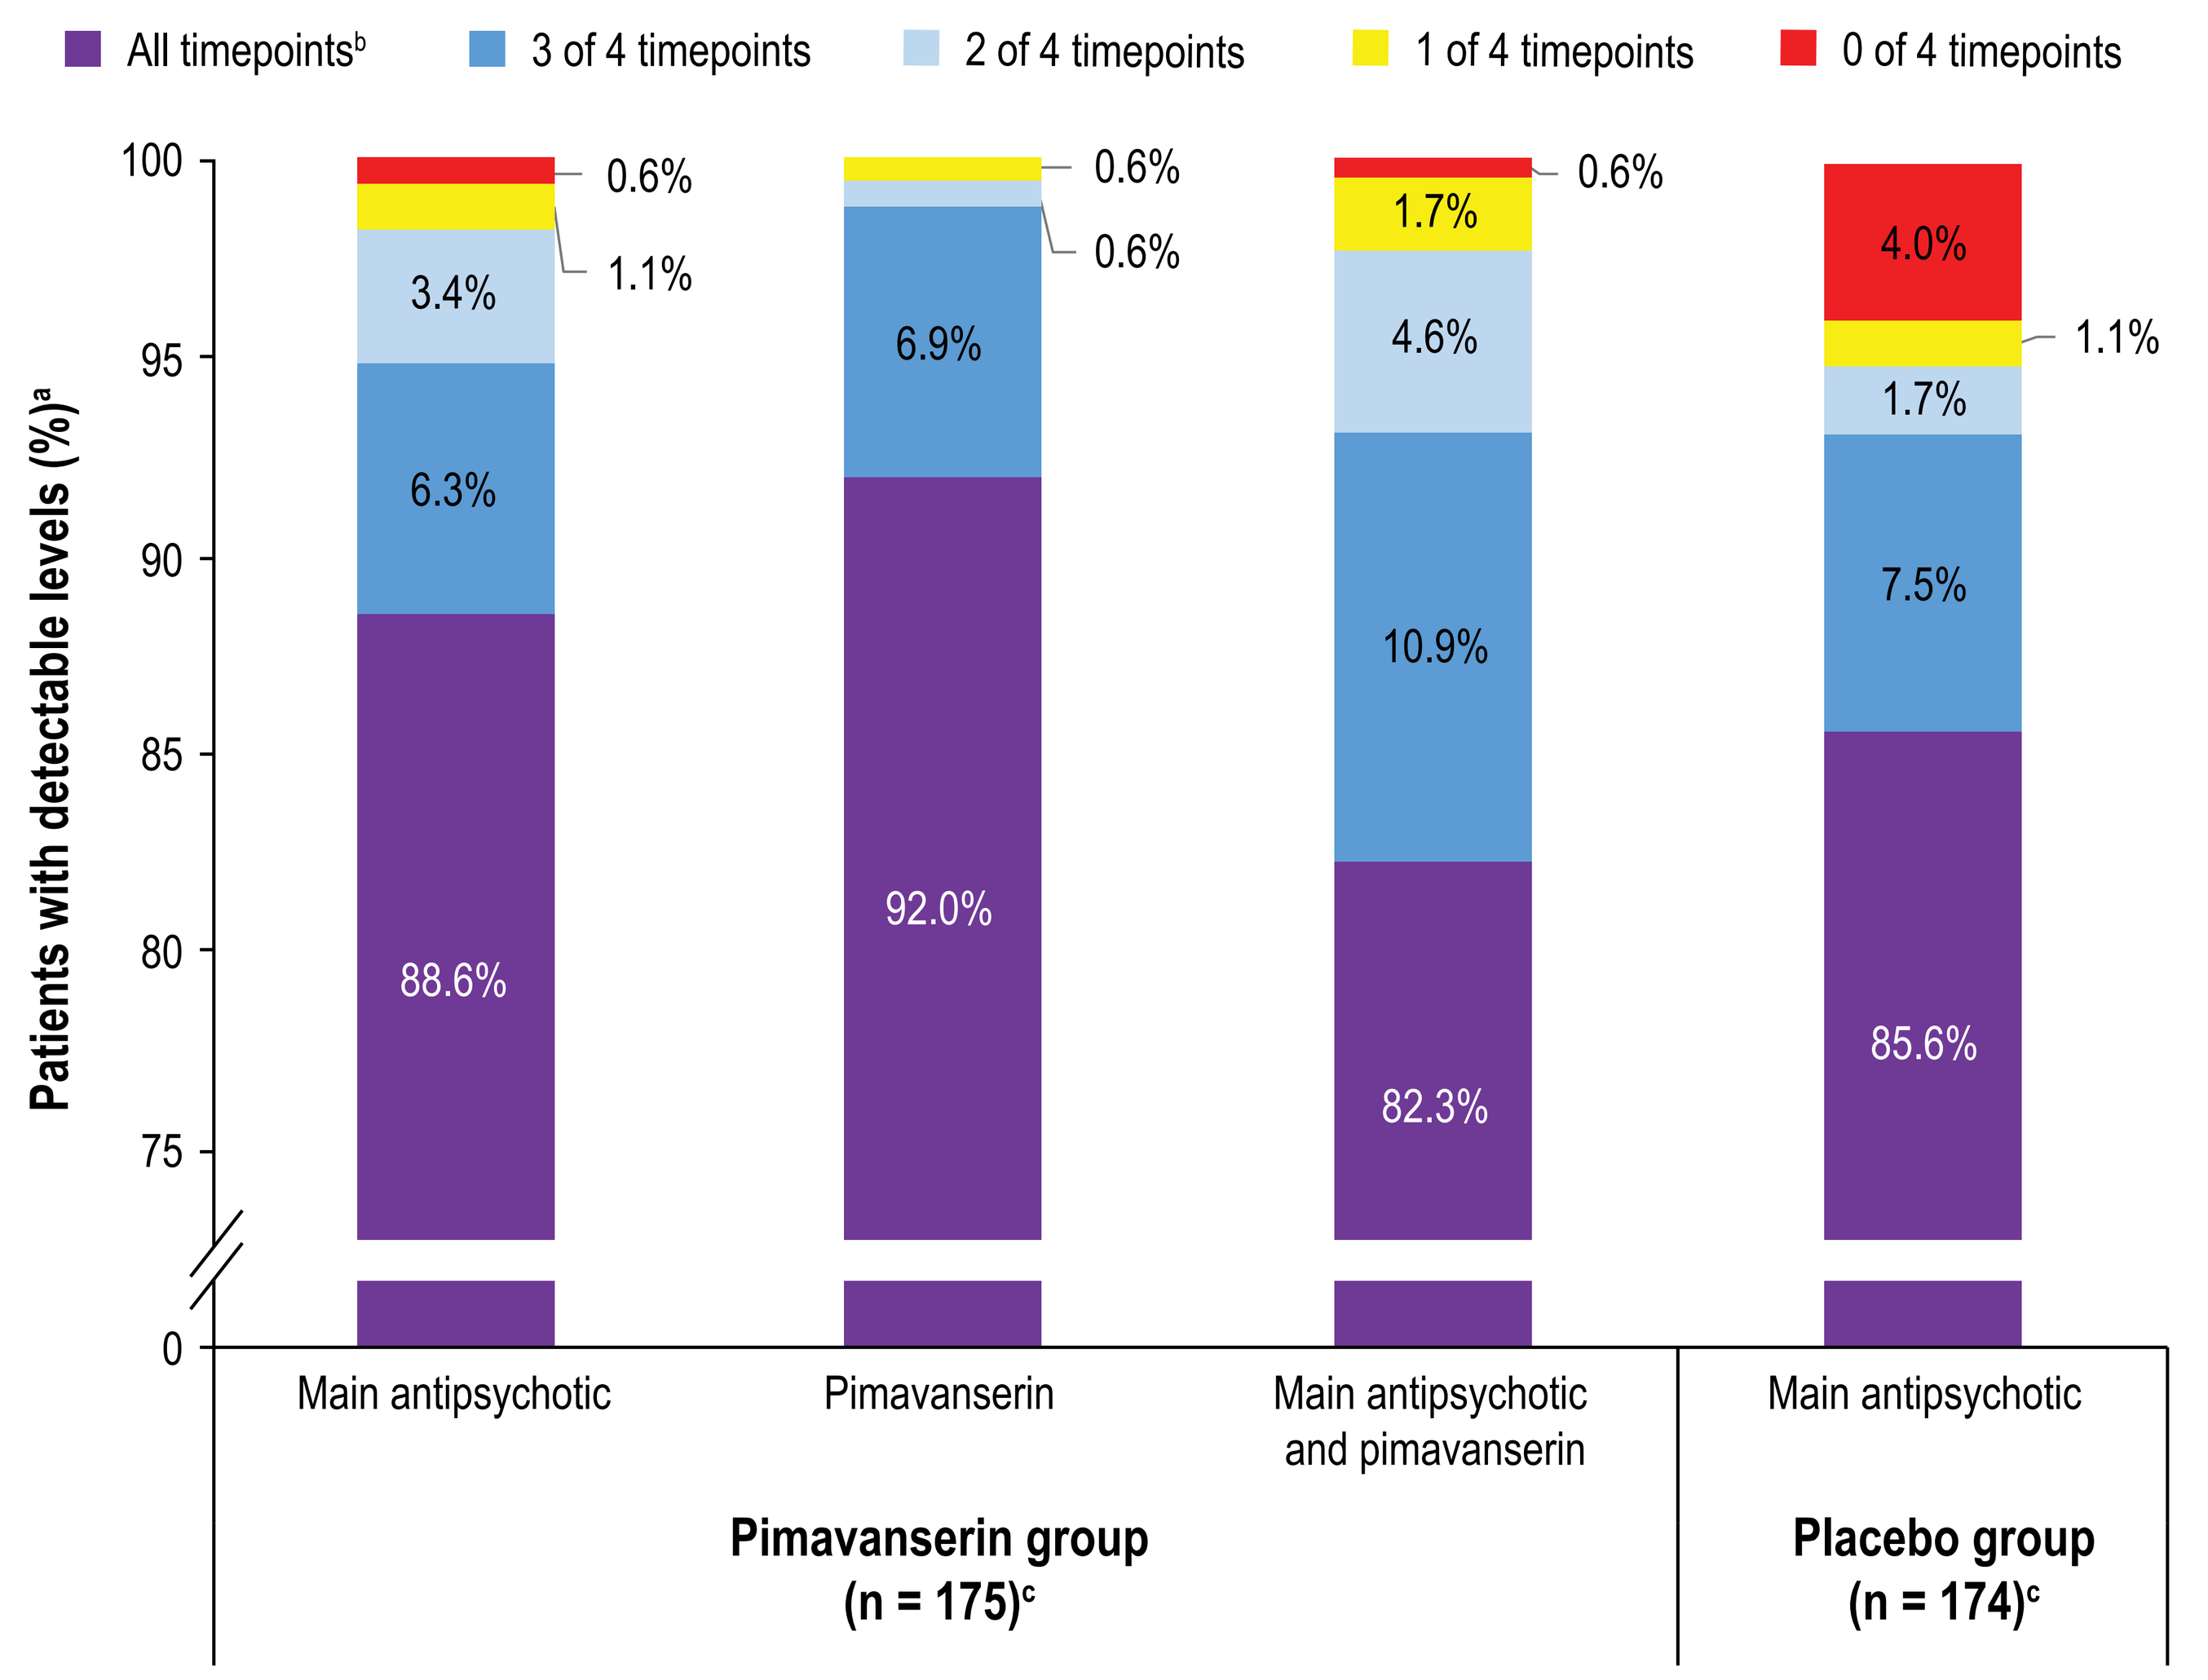
All timepoints[b]
3 of 4 timepoints
2 of 4 timepoints
1 of 4 timepoints
0 of 4 timepoints
Patients with detectable levels (%)[a]
100
95
90
85
80
75
0
88.6%
6.3%
3.4%
1.1%
0.6%
92.0%
6.9%
0.6%
0.6%
82.3%
10.9%
4.6%
1.7%
0.6%
85.6%
7.5%
1.7%
1.1%
4.0%
Main antipsychotic
Pimavanserin
Main antipsychotic and pimavanserin
Main antipsychotic
Pimavanserin group (n = 175)[c]
Placebo group (n = 174)[c]

[a]Drug levels assessed by analyzing blood samples taken during study visits; [b]Samples were collected at a total of 4 postbaseline timepoints; [c]Includes only patients for whom all 4 postbaseline samples were analyzable

CONCLUSIONS

- Patients in the ADVANCE study achieved a high level of treatment adherence (measured by blood levels) to both the main antipsychotic and adjunctive pimavanserin throughout the 26-week study
- These adherence results complement the positive efficacy and safety results previously reported for the ADVANCE study[3]
- Use of adjunctive pimavanserin treatment had no apparent effect on adherence to the main antipsychotic
- High adherence likely resulted from:
 - Rigorous screening procedures employed in ADVANCE, including testing for measurable levels of the main antipsychotic
 - Frequent testing for measurable levels of the main antipsychotic and pimavanserin in blood samples
 - The tolerability of the combination
- Furthermore, most patients in ADVANCE were enrolled in Europe, where treatment paradigms may promote adherence by including long-term relationships with treatment teams, continuity of care support from family, and the potential to lose disability benefits if regular assessments are missed
- The results highlight the importance of clinical review and oversight during screening for supporting patient adherence in clinical trials of antipsychotics
- Confirming medication adherence at screening could potentially improve adherence in a clinical trial and improve both the clinical trial quality and outcomes

REFERENCES

1. Kane JM, et al. *World Psychiatry*. 2013;12(3):216-226.
2. Howes OD, et al. *Am J Psychiatry*. 2017;174(3):216-229.
3. Bugarski-Kirola D, et al. Presentation at the American Society of Clinical Psychopharmacology (ASCP) virtual meeting, May 29-30, 2020.

ACKNOWLEDGMENTS

This study was supported by ACADIA Pharmaceuticals Inc. (San Diego, CA, USA). Medical writing support was provided by Kate Ivins, PhD, of Evidence Scientific Solutions, Inc. and funded by ACADIA Pharmaceuticals Inc.

DISCLOSURES

BA is a former employee of and a stockholder in ACADIA Pharmaceuticals Inc. **DB-K, MD, I-YL,** and **SS** are employees of and stockholders in ACADIA Pharmaceuticals Inc.

To receive a copy of this poster, scan QR code via barcode reader application.
By requesting this content, you agree to receive a one-time communication using automated technology. Message and data rates may apply. Links are valid for 30 days after the congress presentation.

Poster A2

Presented at the International Society for CNS Clinical Trials and Methodology (ISCTM) 2020 Autumn Virtual Conference, September 21–25, 2020

ACADIA® Pharmaceuticals