The Design of a Double-blind, Active-controlled, Randomized Study in Treatment-resistant Schizophrenia

Walling D[1], Forray C[2], Hertel P[3], Löf E[3], Kane J[4]

[1]CNS Network, LLC, Long Beach, California, USA; [2]Lundbeck Pharmaceuticals LLC, New Jersey, USA; [3]H Lundbeck A/S, Valby, Denmark; [4]Hofstra Northwell School of Medicine, The Zucker Hillside Hospital, Hempsead, New York, USA

INTRODUCTION AND AIMS

- More than 21 million people worldwide are affected by schizophrenia, and it is estimated that up to a third of these have treatment-resistant schizophrenia (TRS).[1,2]
- TRS is defined as a failure to respond to two trials of antipsychotic agents of adequate dose and duration (Table 1).
- TRS has severe humanistic, clinical, and economic impact on patients, family, caregivers, and society.[5,6]
- At present, clozapine is the only approved treatment for TRS but is associated with serious safety issues.
- There is a need for a new efficacious treatment with a better safety profile based upon clinical trials that demonstrate these effects.
- Guidelines on TRS study design are available from the Treatment Response and Resistance in Psychosis consensus working group (TRRIP)[7], and the European Medicines Agency (EMA)[8]. The study design considerations in these guidelines are also in line with recommendations from the US Food and Drug Administration (FDA).[9,10]
- The proposed study design aimed at operationalizing the guidelines to a phase III clinical study to test the efficacy and safety of a potential new medication for TRS.

METHODS

- The study was designed in line with the TRRIP[7] and EMA[8] guidelines (Table 2 and 3) and in line with recommendations from the US FDA.[9,10]

Table 1. The definition of TRS according to international guidelines[2,3,4]

Guideline	Prior antipsychotic treatment failures	Treatment duration	Failure criteria
APA (2004)[2]	≥2 failures, ≥1 atypical antipsychotic	≥6 weeks	Little or no symptomatic response to a trial of adequate duration and dose (therapeutic range).
NICE (2014)[3]	≥2 sequential failures, ≥1 non-clozapine atypical antipsychotic	4–6 weeks	Illness has not responded adequately, despite established adherence to antipsychotic medication, prescribed at an adequate dose and for the correct duration.
WFSBP (2012)[4]	≥2 failures, ≥2 different chemical classes, ≥1 atypical antipsychotic	2–8 weeks	No significant improvement in psychopathology and/or target symptoms; assured treatment adherence.
Common definition of TRS: No significant improvement in target symptoms after treatment with ≥2 different antipsychotics (at an adequate dose and duration).			

APA=American Psychiatric Association; TRS=treatment-resistant schizophrenia; NICE=National Institute for Health and Care Excellence; WFSBP=World Federation of Societies of Biological Psychiatry

Table 2. Criteria for assessment and definition of TRS according to TRRIP[7]

Disease characteristics	At least moderate disease severity and <20% symptom reduction during a prospective trial or observation of ≥6 weeks.
	At least moderate functional impairment measured using a validated scale (e.g. SOFAS).
	An illness duration of ≥12 weeks.
Treatment-resistance	Information about past treatment response to be gathered from patient/carer reports, staff and case notes, pill counts, and dispensing charts.
	≥2 prior treatments with different antipsychotic drugs, of ≥6 weeks at a therapeutic dosage (equivalent to ≥600 mg of clorpromazine per day); ≥1 prior treatment utilises a long-acting injectable antipsychotic (for ≥4 months).
	Prospective evaluation of treatment using a standardised rating scale (e.g. PANSS, BPRS, SANS, SAPS).
Treatment adherence (minimum requirement for documentation)	≥80% of prescribed doses taken. Adherence should be assessed using at least two sources (pill counts, dispensing chart reviews, and patient/carer report). Antipsychotic plasma levels monitored on at least one occasion.

SOFAS=Social and Occupational Functioning Assessment Scale, PANSS=Positive And Negative Syndrome Scale, BPRS=Brief Psychiatric Rating Scale, SANS=Scale for the Assessment of Negative Symptoms, SAPS=Scale for the Assessment of Positive Symptoms

Table 3. Key elements of a TRS study design from a regulatory perspective

Choice of endpoints	Essentially the same as for other schizophrenia trials.[8]
Identification of patients with TRS	Demonstration of treatment-resistance as a lack of satisfactory improvement to at least two different antipsychotic agents. It is acceptable for one of the these treatment failures to be retrospectively assessed, but the second one needs to be demonstrated prospectively.[8]
	Confirmation of treatment adherence.[8]
Choice of comparator	It is usually essential, in order to demonstrate effectiveness of the new drug in non-responders, to randomize patients to both the failed drug and the test drug.[9] Randomizing non-responder patients to the new drug and placebo would document effectiveness of the new agent but would not show effectiveness in non-responders, because a history of non-response does not predict non-response in a new trial. To show success in non-responders, randomization to the new and failed treatment is critical.[10]
	EMA, Option 1: Demonstration of non-inferiority to a medicinal product that is approved in the EU for the indication treatment-resistant schizophrenia. At present the only such drug is clozapine.[8]
	EMA, Option 2: Demonstration of superiority to an active comparator, which should be a medicinal product with which treatment failure has been documented prospectively.[8]

STUDY DESIGN

- The design of a multinational, double-blind, active-controlled, fixed dose study to evaluate a new treatment for TRS (NCT02717195, Figure 1).
- The number of patients to be included in the study was 964, which was calculated to detect a difference of 5 points in Positive And Negative Syndrome Scale (PANSS) total score.

Figure 1. The study design

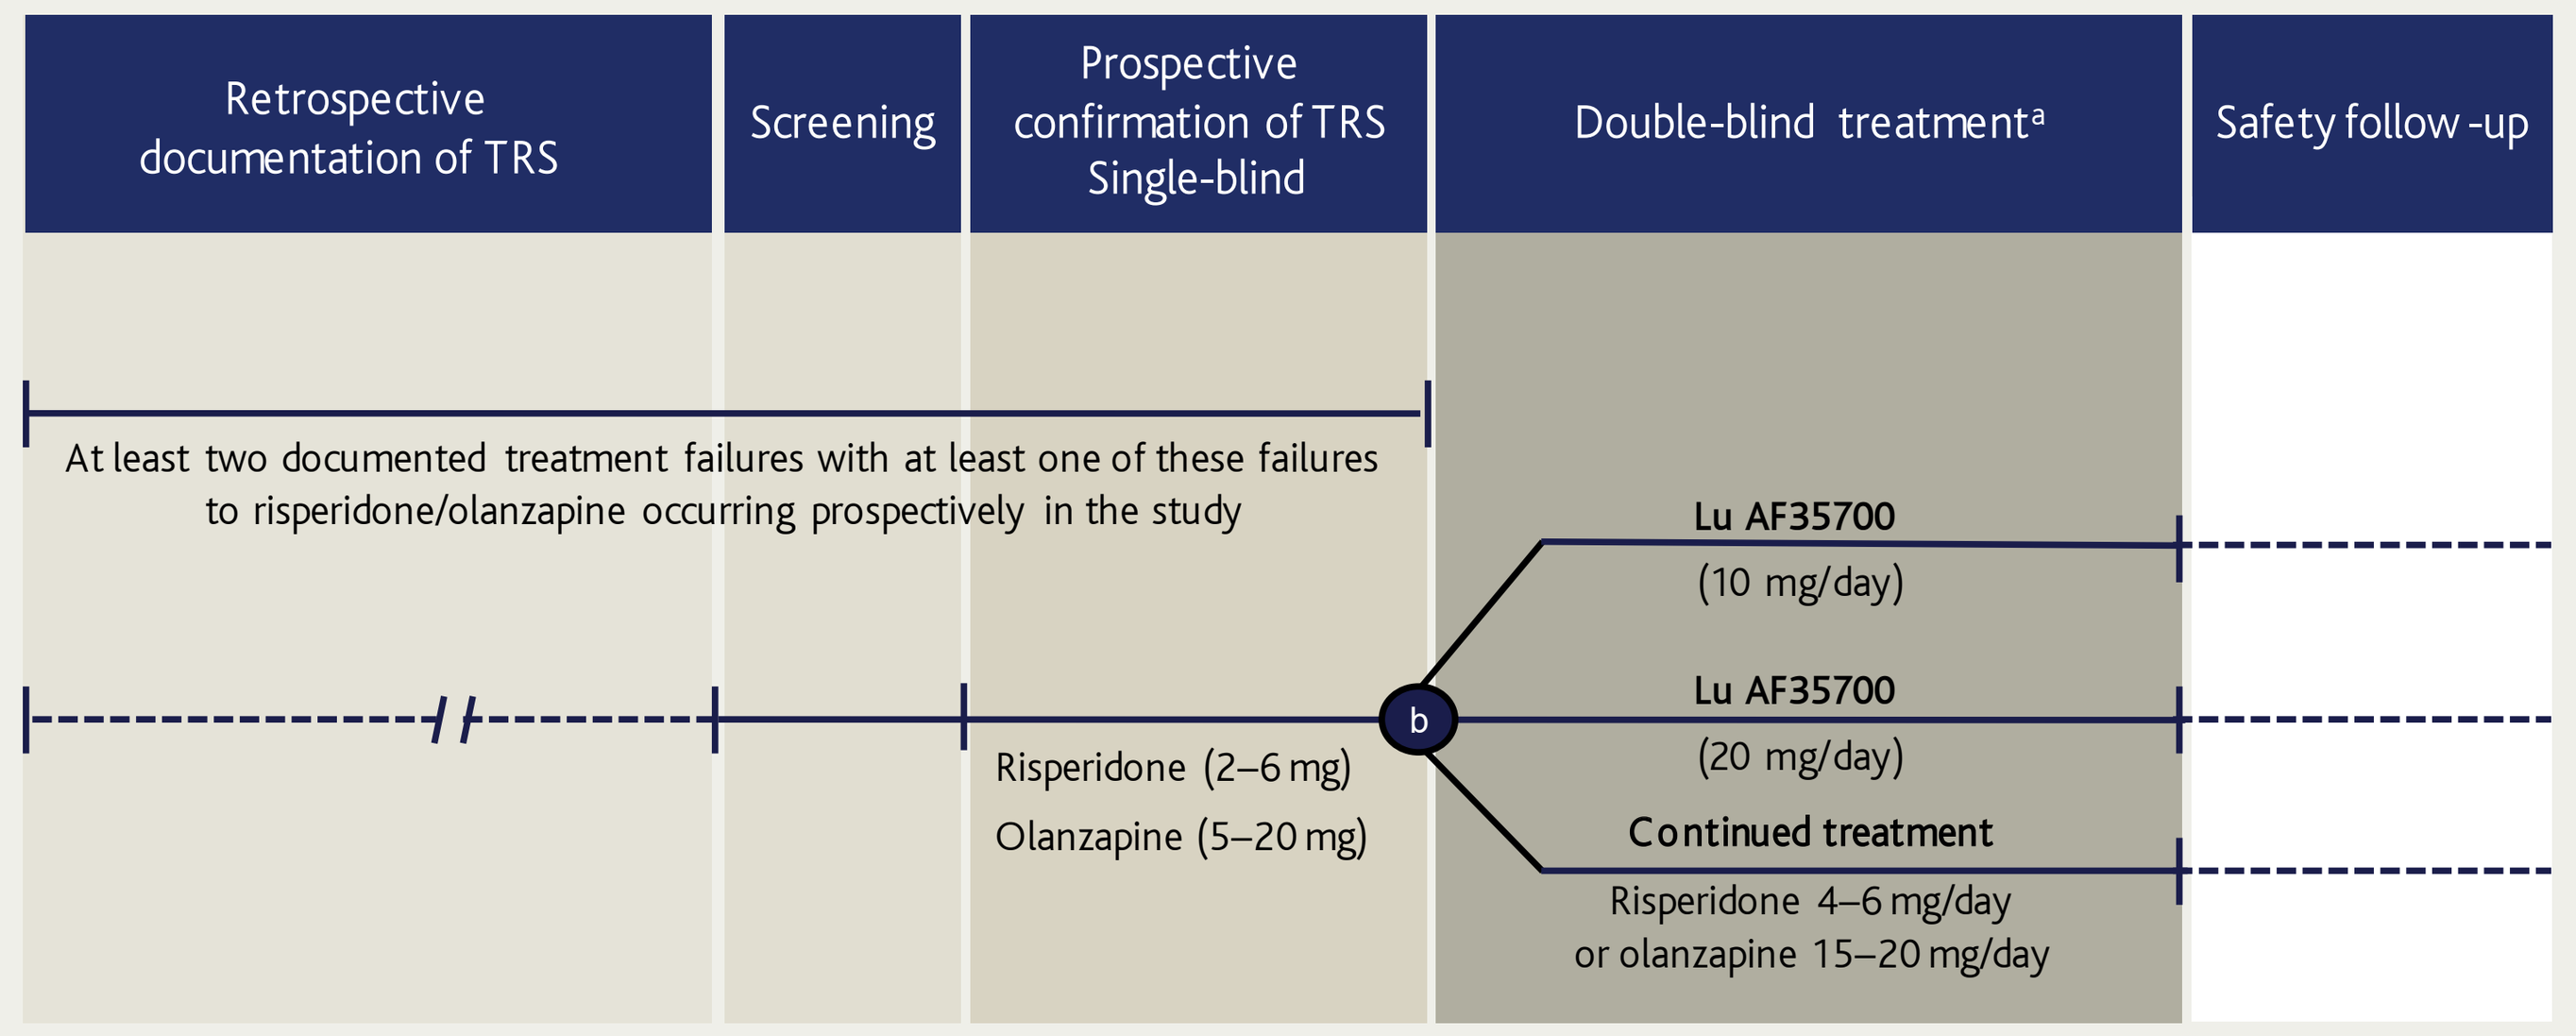

Retrospective documentation of TRS
Screening
Prospective confirmation of TRS Single-blind
Double-blind treatment[a]
Safety follow-up
At least two documented treatment failures with at least one of these failures to risperidone/olanzapine occurring prospectively in the study
Risperidone (2–6 mg)
Olanzapine (5–20 mg)
b
Lu AF35700 (10 mg/day)
Lu AF35700 (20 mg/day)
Continued treatment
Risperidone 4–6 mg/day or olanzapine 15–20 mg/day

a. Responders excluded, non-responders randomised to treatment.
b. Baseline

CONSIDERATIONS AND RATIONALE

- The current recommendations for studies evaluating efficacy in TRS[7,8,9,10] were operationalized in the design of a phase III study, testing the efficacy and safety of the investigational product against the comparative arm.
- The overall study design is based on previous study designs in the TRS population[11], adapted to the more recent definition of treatment-resistance and current treatments (Table 1 and 5).

Table 4. Confirmation of treatment-resistance

Retrospective confirmation	**At least one previously documented treatment failure** The patient has failed to show an adequate response in the level of psychotic symptoms despite at least one treatment trial with an adequate dose of an antipsychotic agent prescribed for an adequate time (at least lasting for 6 weeks) during 2 years prior to screening.
	Ongoing psychotic symptomatology at enrollment The patient has a Clinical Global Impression – Severity scale (CGI-S) score of ≥4, a PANSS total score of ≥80 and a score of ≥4 on at least 2 of the following PANSS items: • P2 - Conceptual disorganization • P3 - Hallucinatory behaviour • P6 - Suspiciousness/persecution • G9 - Unusual thought content
Prospective confirmation	**Lack of clinically relevant improvement** The specific inclusion criteria for the double-blind treatment period was blinded to the investigators to further reduce possible sources of bias in the assessment of efficacy in TRS patients.
	Adequate dose and duration of treatment with antipsychotics Patients were treated with risperidone (4-6 mg/day), or if currently under treatment with risperidone, with olanzapine (15-20 mg/day).
	Assured treatment adherence The plasma level of risperidone or olanzapine was measured prior randomization.

- The key operational questions were how to identify patients with TRS, choice of endpoints, and choice of comparator.
- The criteria for identification of patients with TRS were set in alignment with current recommendations:
 - Patients with schizophrenia of a moderate to severe symptomatology were identified based on PANSS and Clinical Global Impression – Severity scale (CGI-S), in alignment with the TRRIP guideline.
 - Patients were identified as having TRS using both a retrospective and a prospective trial of antipsychotic treatment as described in Table 4, in alignment with the TRRIP and EMA guidelines.
 - Treatment adherence was documented as described in Table 4, in addition to pill count, which is in alignment with the TRRIP and EMA guidelines.
 - Patients who adhered to treatment in the prospective confirmation phase and did not show treatment response were defined as having TRS.

Table 5. Reasons for choosing risperidone and olanzapine as comparators

Olanzapine and risperidone are both second-generation antipsychotics with well-established efficacy and safety profiles which will enable a clear differentiation between responders and non-responders. Risperidone and olanzapine are thus appropriate comparators for the investigational product.
Mimics clinical practice. TRS patients often stay on non-clozapine treatment in spite of a limited symptom improvement, due to the serious safety risks of clozapine treatment.
Improvement of symptoms as a consequence of being in a clinical study setting can be taken into account.
Given the limited treatment options for this patient population, it is considered ethically justifiable to keep patients on a treatment with which treatment failure has been documented.
Blinding and recruitment is operationally more feasible for these comparators than for clozapine as a comparator.

- The choice of endpoints was based on common practice for schizophrenia studies, in alignment with the EMA guideline:
 - The primary endpoint was symptoms of schizophrenia as change from baseline to end of treatment in PANSS total score.
 - Other assessments represented symptom and functional evaluations, including CGI-S and Personal and Social Performance scale (PSP).
- The choice of comparator was in alignment with current recommendations:
 - Based on the EMA guideline[8], as a choice of comparator it was an option to either use clozapine or let the eligible patients stay on the treatment with which they did not show improvement in the prospective confirmation period. Based on the reasons listed in Table 5, risperidone/olanzapine was the choice of comparator.
 - Eligible patients with TRS were randomized to either stay on the treatment used in the prospective confirmation period (risperidone or olanzapine) or switch to the investigational product in the double-blind treatment period, in alignment with the EMA[8] guideline and recommendations from the US FDA.[9,10]

CONCLUSIONS

- When combining the available recommendations, the presented study design for assessing monotherapy of TRS is a feasible methodology.
- New in this study is the operationalization of TRS based on the TRRIP consensus guideline.

References

1. World Health Organization (WHO). Fact Sheet. Schizophrenia. 2016. Accessed August 2018.
2. Lehman AF. Practice guideline for the treatment of patients with schizophrenia, second edition. Am J Psychiatry 2004;161 (2 Suppl):1-56.
3. National Institute for Health and Care Excellence (NICE). nice.org.uk/guidance/cg178. Accessed August 2018.
4. Hasan A et al. World Federation of Societies of Biological Psychiatry (WFSBP) Guidelines for Biological Treatment of Schizophrenia, part 1: update 2012 on the acute treatment of schizophrenia and the management of treatment resistance. World J Biol Psychiatry 2012;13:318–378.
5. Kennedy JL et al. The social and economic burden of treatment-resistant schizophrenia: a systematic literature review. Int Clini Psychopharmacol 2014; 29: 63-76.
6. Bartolomeis A et al. Differential cognitive performances between schizophrenic responders and non-responders to antipsychotics: Correlation with the course of the illness, psychopathology, attitude to treatment and antipsychotic doses. Psychiatry Res 2013; 210: 387-395.
7. Howes OD et al. Treatment-Resistant Schizophrenia: Treatment Response and Resistance in Psychosis (TRRIP) Working Group Consensus Guideline on Diagnosis and Terminology. Am J Psychiatry 2017; 174: 216–229.
8. European Medicines Agency (EMA). Committee for Medicinal Products for Human Use (CHMP). Guideline on clinical investigation of medicinal products, including depot preparations in the treatment of schizophrenia. CHMP/40072/2010 Rev. 1, 2012.
9. US Food and Drug Association (FDA). Guidance for Industry. Enrichment Strategies for Clinical Trials to Support Approval of Human Drugs and Biological Products. Draft Guidance 2012.
10. Temple R. A regulator's view of comparative effectiveness research. Clinical Trials 2012; 9: 56–65.
11. Kane J et al. Clozapine for the Treatment-Resistant Schizophrenic. Arch Gen Psychiatry 1988; 45: 789-796.

Disclosures

Elin Löf and Peter Hertel are employees of H. Lundbeck A/S. Carlos Forray is an employee of Lundbeck Pharmaceuticals LLC.

Dr. Kane has been a consultant for or received honoraria from Alkermes, Eli Lilly, EnVivo Pharmaceuticals (Forum), Forest (Allergan), Genentech, H. Lundbeck. Intracellular Therapies, Janssen Pharmaceutica, Johnson and Johnson, Merck, Neurocrine, Otsuka, Pierre Fabre, Reviva, Roche, Sunovion, Takeda and Teva. Dr. Kane has received grant support from Otsuka, Lundbeck and Janssen. Dr. Kane has participated in Advisory Boards for Alkermes, Intracellular Therapies, Lundbeck, Neurocrine, Otsuka, Pierre Fabre, Takeda, Teva. Dr. Kane is a Shareholder in Vanguard Research Group and LB Pharmaceuticals, Inc.

David P. Walling, Ph.D., has received fees as a consultant from Otsuka and Janssen, and has received grants or research support from Novartis, J&J PRD, Sunovion, Janssen, Pfizer, Abbvie, Alkermes, Allergan, Takeda, Otsuka, Noven, Zogenix, Omeros, CoMentis, IntraCellular, Lupin, Avanir, Lundbeck, Roche, Boehringer Ingelheim, and Acadia.

Sponsorship

This poster and the work done to design the study described in this poster was funded by H. Lundbeck A/S.

Presented at the ISCTM 2018 Autumn Conference, 15-16 October 2018The Ritz-Carlton, Marina del Rey, CA USA