Self-Report on the SIBAT: Sensitivity to Rapid Change in Thinking Related to Suicide

David Williamson,[1] Carla Canuso,[2] Dong-Jing Fu,[2] Yun Zhang,[2] Rosanne Lane,[2] Larry Alphs[1]

[1]Janssen Scientific Affairs, LLC, Titusville, NJ; [2]Janssen Research and Development, LLC, Titusville, NJ

INTRODUCTION

- Several scales are available to assess suicidal ideation. However, none were explicitly designed to be sensitive to the rapid changes in suicidal ideation that may be observed when a patient is admitted to an acute psychiatric care setting after exhibiting suicidal thoughts or behaviors.[1-3] As this epoch has been targeted by recent treatments, appropriate measurement tools must be developed and validated
- The Suicide Ideation and Behavior Assessment Tool (SIBAT) combines clinician-report, patient-report, and patient-performance components, some of which are expected to change rapidly and some of which are not
- This analysis examines the sensitivity of the My Current Thinking (MCT) module to changes in suicidal thinking at 4 hours and 24 hours after initiation of standard-of-care (SoC) and SoC plus esketamine treatment

FEATURES OF THE SIBAT

- Modules of the SIBAT includes both patient- and clinician-reported information relevant to suicide risk assessment
- The SIBAT separates the measurement of constructs prone to rapid change (eg, suicidal ideation) from those that change more slowly (eg, history of suicidal behavior). The SIBAT also permits quantification of dynamic constructs and decreases patient burden so that the assessment of stable constructs is not needlessly repeated
- The SIBAT captures the clinician's assessment of severity of suicidality, imminent and long-term suicide risk, and his/her plan to manage the patient under real-world treatment conditions
- The SIBAT is being developed to meet FDA regulatory standards for use in clinical trials that are useful for both efficacy and safety assessments
- The SIBAT is a valuable clinical tool for tracking patient response to therapeutic interventions

STRUCTURE OF THE SIBAT

- The SIBAT includes 8 modules that capture information on demographics, known suicide risk factors, history of suicidal behavior, and severity of suicidal ideation (**Figure 1**)
 - The modules are organized into separate units according to category and susceptibility to change
 - The modular format allows different question sets to be administered independently at different times
 - Compared with most scales, the SIBAT assesses a broader range of severity for each module item, thus potentially allowing it to be more sensitive to changes in suicidal ideation and behavior (SIB)
 - Patients with a history of SIB review the items and their wording
 - During revisions of the provisional versions of the SIBAT, modules were added and item wordings were refined
- The patient-reported modules (Modules 1–5) are completed as assigned for a given rating session
- Clinician-reported modules (Modules 6-8):
 - The clinician reviews the patient-reported modules (Modules 1–5)
 - Following the clinician's review, the clinician conducts a brief, semistructured interview of the patient (Module 6)
 - The clinician's review and interview provide the basis for completing the Clinical Global Impression of Severity of Suicidality (revised) (CGI-SS-R) scale, the Clinical Global Impression of Imminent Suicidality (CGI-SR-I) scale, and the Clinical Global Impression of Long-Term Suicide Risk (CGI-SR-LT) scale (Module 7), and for making decisions about optimal clinical management (Module 8)

Figure 1. Summary of SIBAT Modules

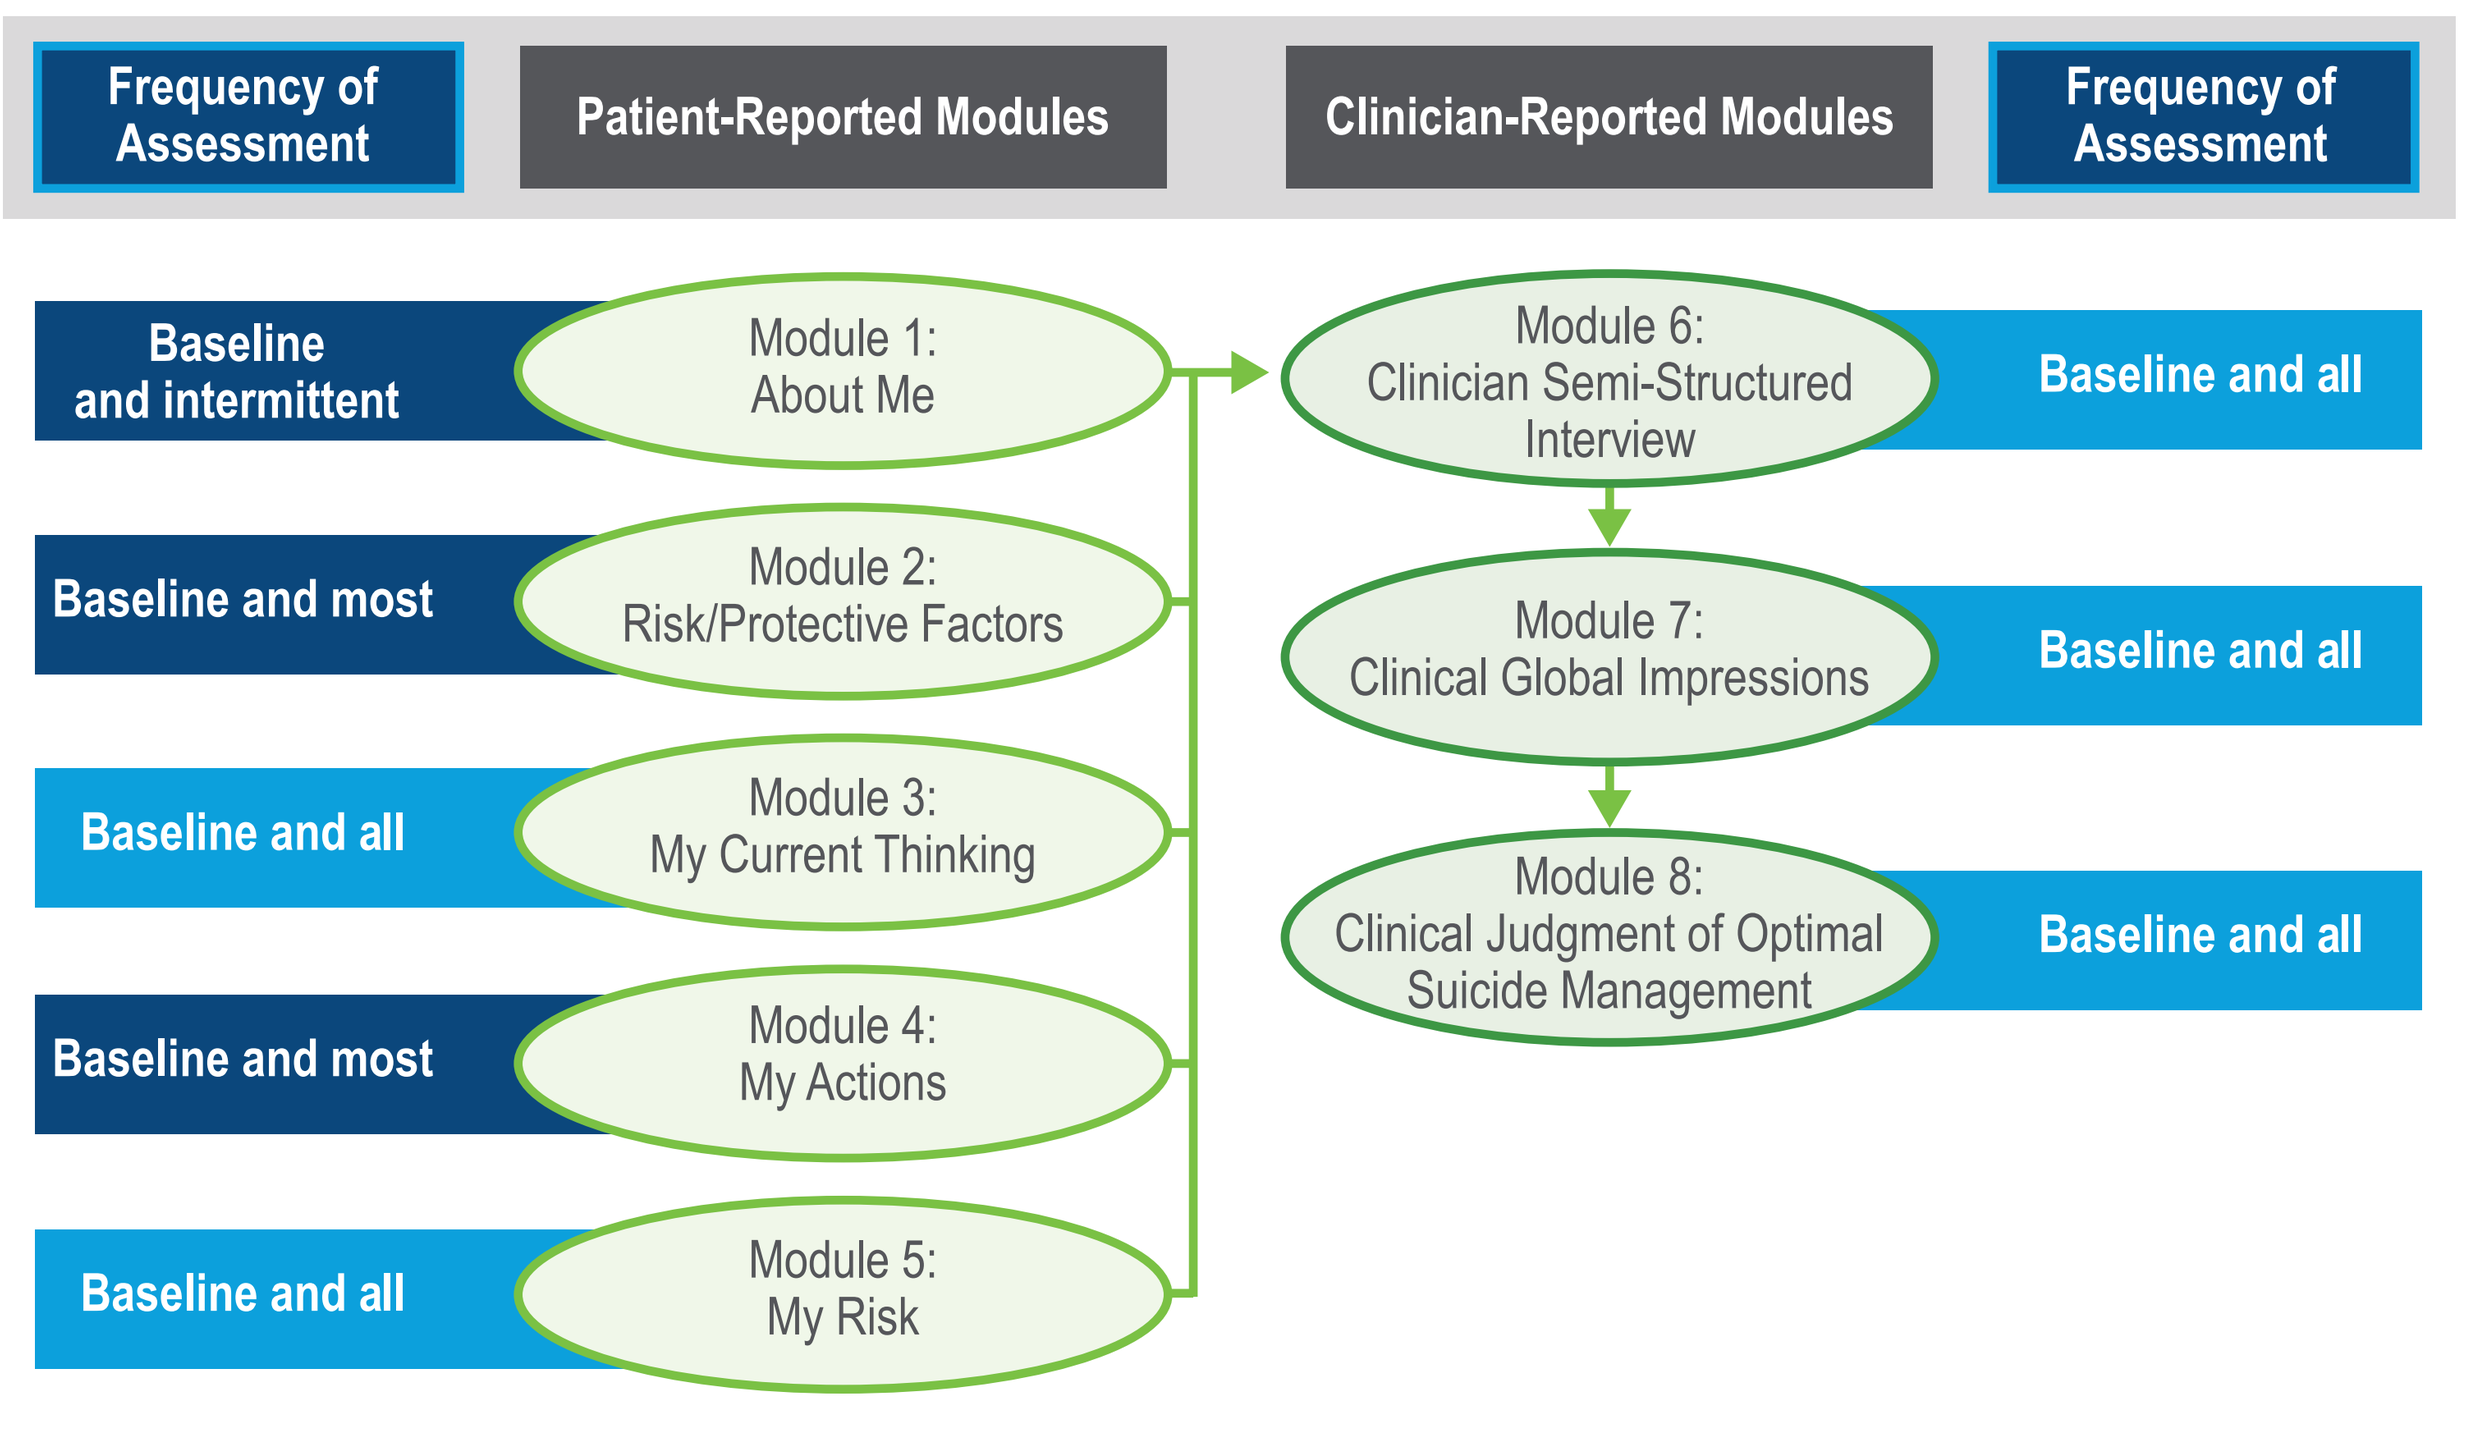
Frequency of Assessment
Patient-Reported Modules
Clinician-Reported Modules
Frequency of Assessment
Baseline and intermittent
Module 1: About Me
Baseline and most
Module 2: Risk/Protective Factors
Baseline and all
Module 3: My Current Thinking
Baseline and most
Module 4: My Actions
Baseline and all
Module 5: My Risk
Module 6: Clinician Semi-Structured Interview
Baseline and all
Module 7: Clinical Global Impressions
Baseline and all
Module 8: Clinical Judgment of Optimal Suicide Management
Baseline and all

METHODS

- These findings are based on data from a study that examined the efficacy and safety of intranasal (IN) esketamine in patients with major depressive disorder (MDD) at imminent risk of suicide[4]

Evaluating Sensitivity to Change

- Patients were treated with IN esketamine plus SoC or placebo plus SoC and were followed up for 25 days with double-blind assessments on the SIBAT (**Figure 2**)

Figure 2. Study design.

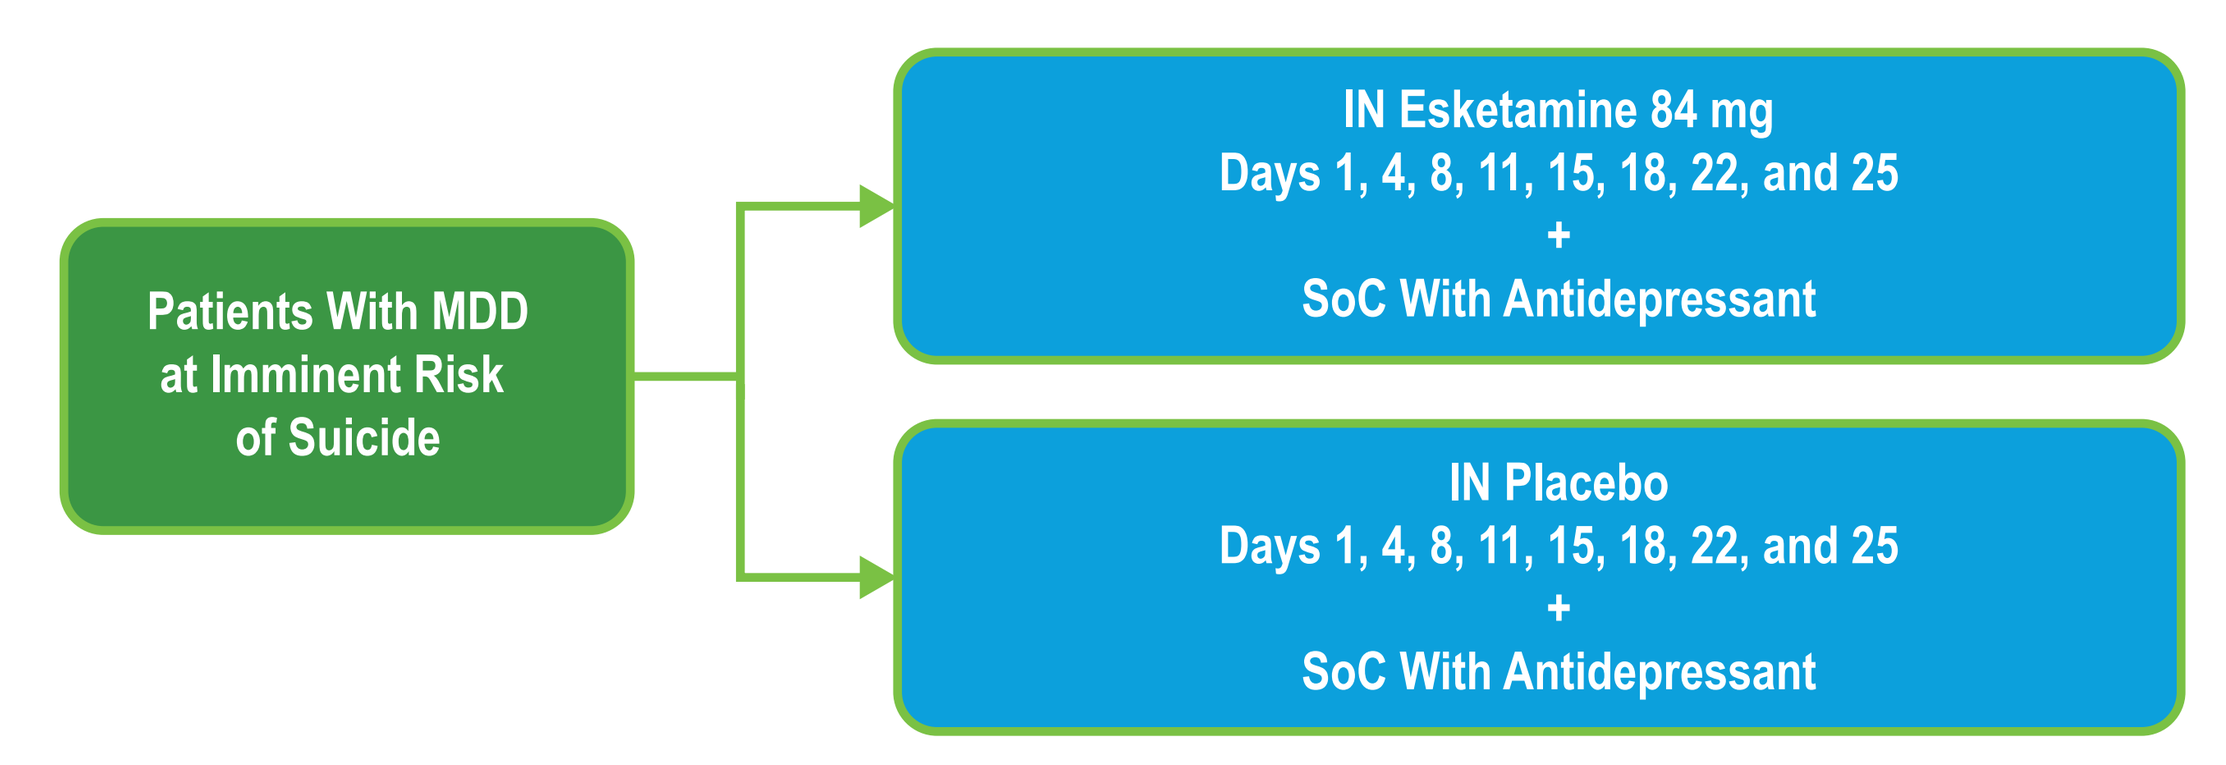
Patients With MDD at Imminent Risk of Suicide
IN Esketamine 84 mg
Days 1, 4, 8, 11, 15, 18, 22, and 25
+
SoC With Antidepressant
IN Placebo
Days 1, 4, 8, 11, 15, 18, 22, and 25
+
SoC With Antidepressant

Administration and Scoring

- Data were collected electronically to ensure an efficient, secure process
- Scores were obtained from patient self-reporting (Modules 1–6) and from a semistructured interview by an experienced, well-trained clinician (Module 7)
- The scoring of Modules 8, 9, and 10 was based on the clinician's judgment, using his or her knowledge of suicide risk factors and information from the patient

BASELINE DEMOGRAPHICS

- The demographics of patients assessed on the SIBAT following treatment with IN esketamine or placebo were comparable (**Table 1**)

Table 1. Baseline Demographics

	Placebo + SoC n = 31	IN Esketamine 84 mg + SoC n = 35
Age, years, mean (SD)	36.0 (12.8)	35.7 (13.4)
Age category, years, n (%)		
18–34	18 (58.1)	18 (51.4)
35–54	10 (32.3)	12 (34.3)
55–64	3 (9.7)	5 (14.3)
Sex, n (%)		
Male	10 (32.3)	13 (37.1)
Race, n (%)		
White	15 (48.4)	20 (57.1)
Black/African American	13 (41.9)	12 (34.3)
Asian	0	1 (2.9)
Multiple	1 (3.2)	0
Other	2 (6.5)	0
Not reported	0	2 (5.7)

Assessment

- The SIBAT's MCT module is a patient-report component of the SIBAT that assesses 31 suicide-related thoughts and beliefs, each scored on a 5-point Likert scale. Twenty-one of these items are written such that a higher score indicates more suicidality or beliefs associated with greater expected suicide risk. Ten of these items are written in the opposite manner
- The MCT total score is the sum of all items, with these latter 10 items reverse-scored (multiplied by −1)
- Subjects completed this module at baseline prior to treatment and again at 4 hours and 24 hours after the initiation of treatment

Statistical Methods

- A mixed-effects model using repeated measures was performed on change in MCT total score from baseline to 4 hours and from baseline to 24 hours
- The model included baseline MCT total score as a covariate, and included visit, treatment (esketamine or placebo), SoC treatment (antidepressant monotherapy or antidepressant plus augmentation), site, and visit-by-treatment interaction as fixed effects, as well as a random-subject effect
- An autoregressive (1) covariance structure was used

RESULTS

- The MCT total score had acceptable internal consistency (Cronbach's alpha = 0.9284)

Overall Sensitivity to Change (Figure 3)

Figure 3. Least-squares mean changes from baseline in MCT total score for entire sample (n = 66).

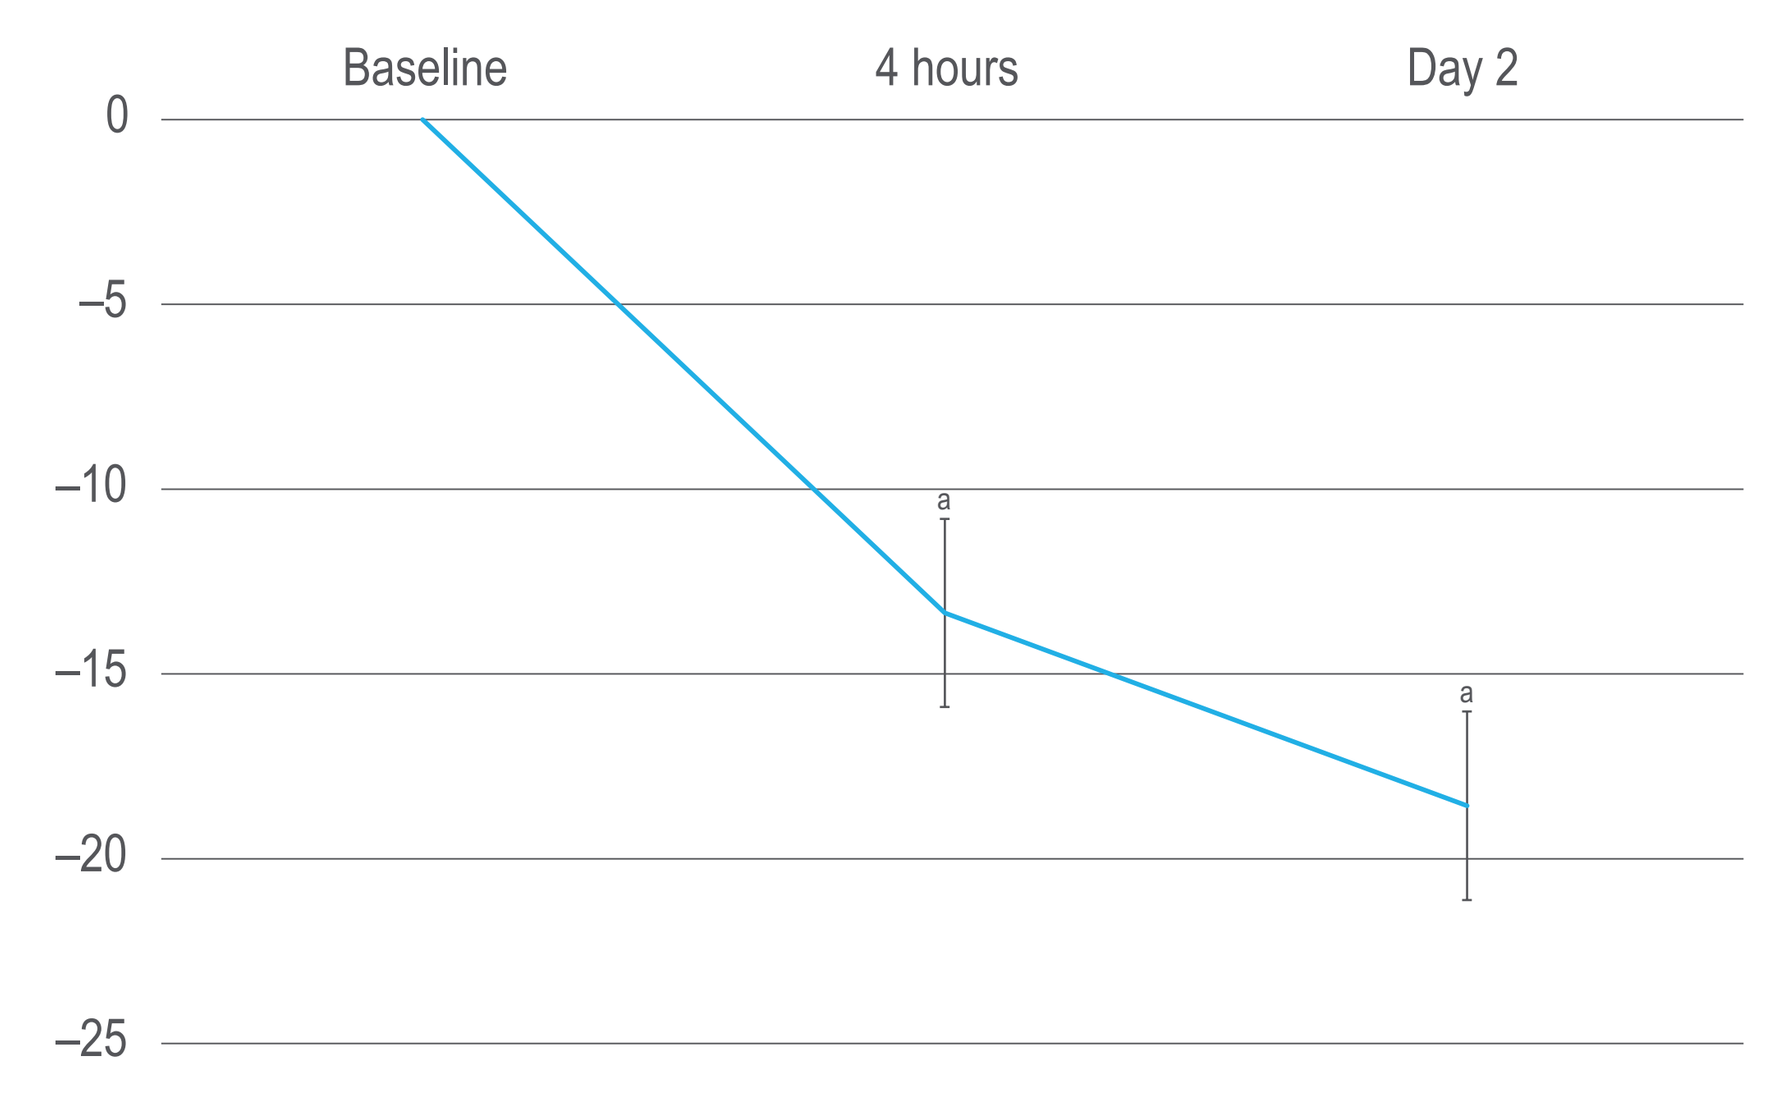
Baseline
4 hours
Day 2
0
–5
–10
–15
–20
–25
a
a

Changes from baseline were analyzed using a mixed model for repeated measures (MMRM). Error bars indicate the standard error of the least-squares mean (LSM).
[a]$P<0.0001$ indicates significant differences between MCT total scores at baseline and at 4 hours and 24 hours, respectively.

Detection of Treatment Differences (Figure 4)

Figure 4. LSM changes from baseline in MCT total score by treatment group.

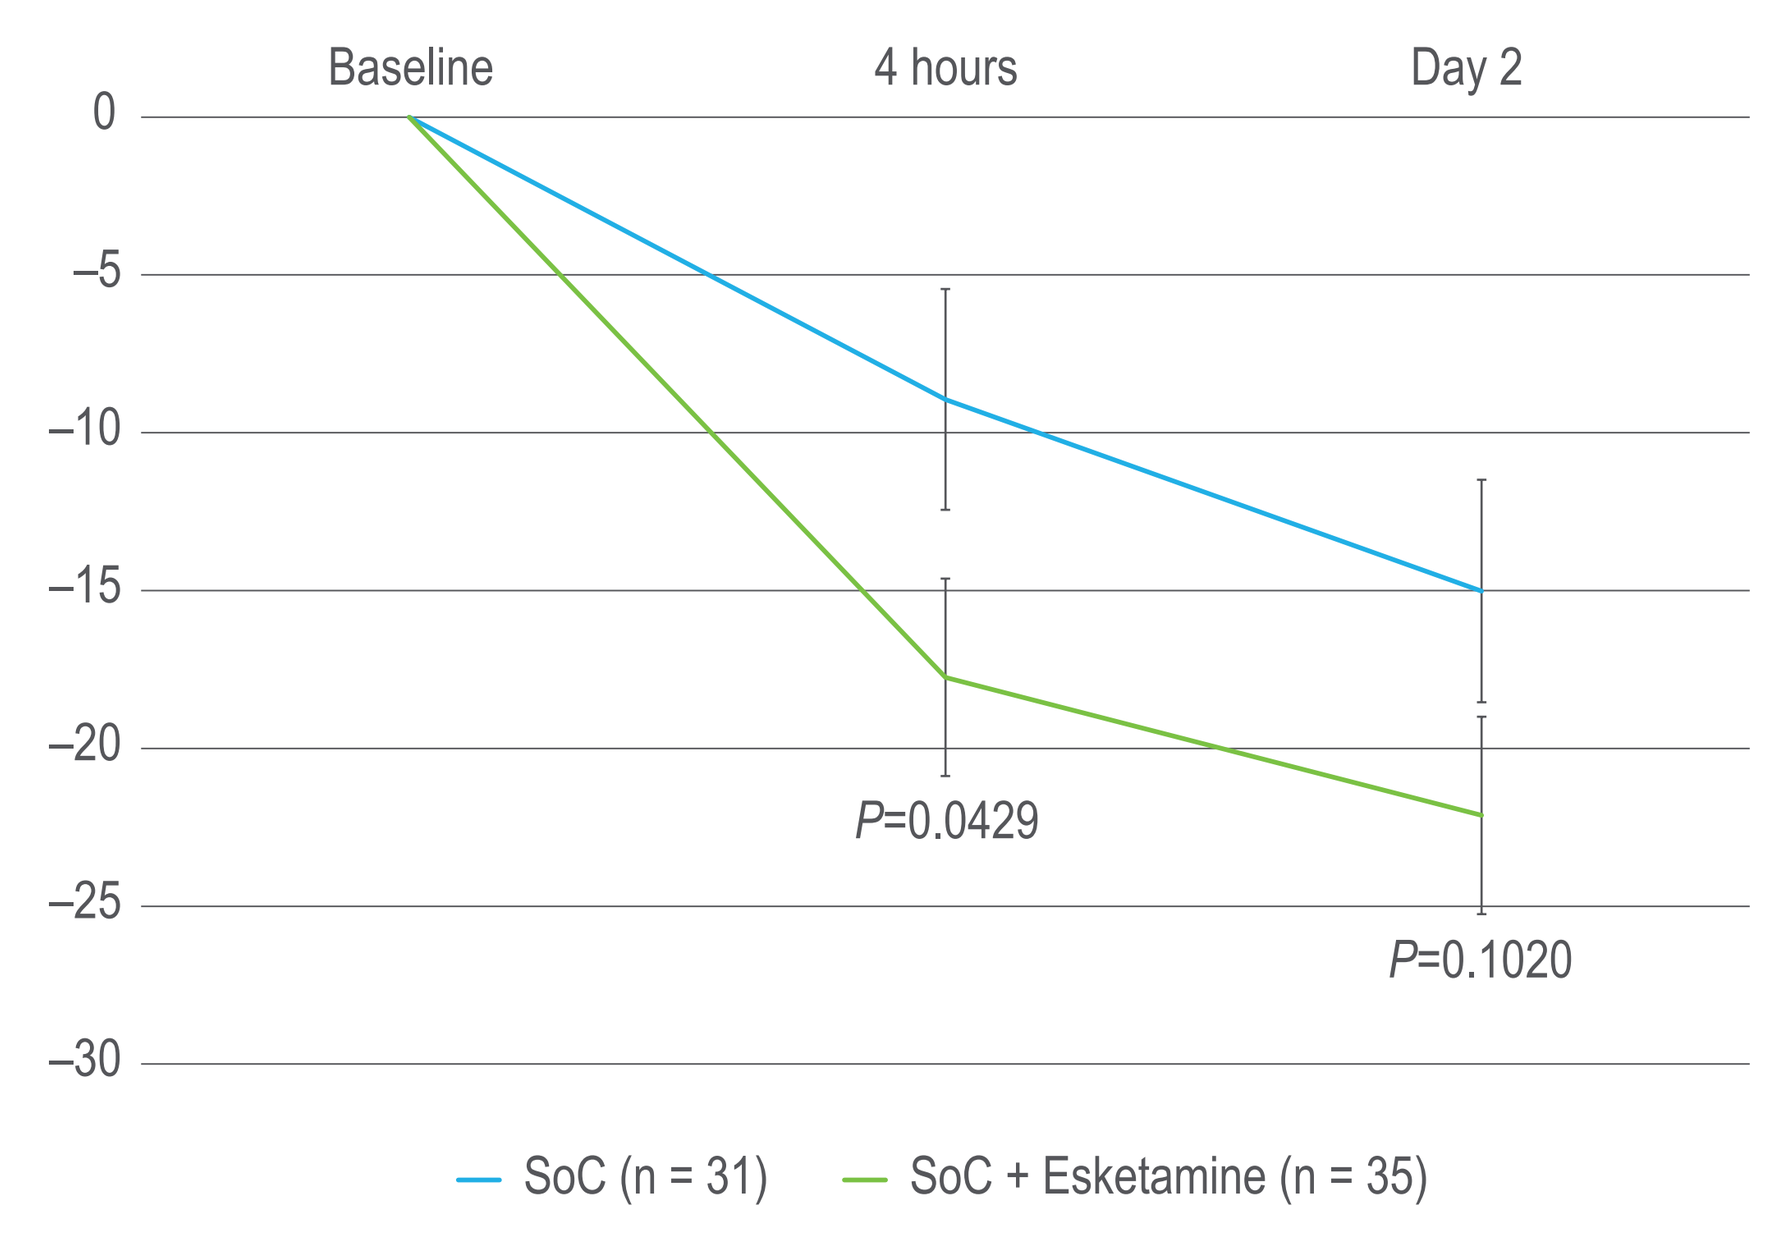
Baseline
4 hours
Day 2
0
–5
–10
–15
–20
–25
–30
P=0.0429
P=0.1020
SoC (n = 31)
SoC + Esketamine (n = 35)

Changes from baseline were analyzed using an MMRM. Error bars indicate the standard error of the LSM. *P* values indicate differences in short-term changes in thoughts associated with suicidality between treatment groups IN esketamine plus SoC and placebo plus SoC. Statistical significance for this proof-of-concept study was based on a two-sided 0.20 significance level.

DISCUSSION

- The SIBAT is a novel instrument for efficiently and comprehensively identifying and monitoring SIB in patients at risk for suicide
- The total score derived from the SIBAT's patient-reported MCT module proved quite sensitive to changes about thoughts related to suicidality as soon as 4 hours and 24 hours after initiation of treatment
- All mean changes were in the direction of less severe suicide-related thinking

CONCLUSIONS

- **The current analysis supports the use of the SIBAT as an efficacy and safety instrument to measure patient- and clinician-based assessment of imminent suicide risk**
- **The MCT total score performed well as a self-report measure in the context of measuring rapid change, suggesting promise for the approach of segregating issues with the potential to change acutely (eg, current suicidal ideation) from those less likely to change (eg, history of suicide attempts)**
- **We anticipate that the SIBAT will be a valuable tool for assessing change—particularly rapid change—in patients at risk for suicide**

REFERENCES

1. Meltzer HY et al. *Arch Gen Psychiatry*. 2003;60:82-91.
2. Lindenmayer JP et al. *Schizophr Res*. 2003;63:161-170.
3. Posner K et al. *Am J Psychiatry*. 2011;168:1266-1277.
4. Canuso CM et al. Poster presented at: Society of Biological Psychiatry 71st Annual Meeting; May 12–14, 2016; Atlanta, GA.

Acknowledgments

The authors thank the SIBAT Consortium members:

- Larry Alphs and David Williamson (Janssen Scientific Affairs, LLC)
- Steve Ascher, Carla Canuso, Erin Lee, Lian Mao, Bruce Simonson, and Jaskaran Singh (Janssen Research and Development, LLC)
- Emily Bruer, Roberta May, and Richard C. Shelton (University of Alabama at Birmingham)
- Phillip Chappell and Michelle Stewart (Pfizer)
- Bill Coryell (University of Iowa)
- Chris Gray (Medical Outcome Systems)
- John Greist (Health Technology Systems)
- Magdalena Harrington (Shire Pharmaceuticals)
- Jean-Pierre Lindenmayer (New York University)
- Cheryl McCullumsmith (University of Cincinnati)
- David Miller (Bracket)
- Jim Mundt (State of Wisconsin)
- Dennis Revicki (Evidera)
- David Sheehan (University of South Florida)
- Paul Wicks (Patients Like Me)
- Frank Wiegand (Janssen Scientific Affairs, LLC)

Writing and editorial support was provided by Matt Grzywacz, PhD, and Lynn Brown, PhD, of ApotheCom (Yardley, PA). This support was provided by Janssen Scientific Affairs, LLC.

Disclosures

D. Williamson and L. Alphs are employees of Janssen Scientific Affairs, LLC, and are Johnson & Johnson stockholders. C. Canuso, D.-J. Fu, Y. Zhang, and R. Lane are employees of Janssen Research and Development, LLC, and are Johnson & Johnson stockholders.

An electronic version of the poster can be viewed by scanning the QR code. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way. These are electronic reproductions of posters presented at the International Society for CNS Clinical Trials and Methodology (ISCTM) 2016 Autumn Conference; September 26–27, 2016; Philadelphia, PA. All copyrights remain those of the copyright holder. This page will not be available after October 27, 2016. http://bit.ly/2bQ2oXO (This URL is case sensitive.)

Supported by Janssen Scientific Affairs, LLC

Presented at International Society for CNS Clinical Trials and Methodology Autumn Conference; September 26–27, 2016; Philadelphia, PA