Using PANSS Score Profiles to Predict Early Termination in a Study on Acute Exacerbation of Schizophrenia

Mark Opler[1,2], Jonathan Lam[1], Jennifer Lord-Bessen[1], Elizabeth Hanson[3], Atul Mahableshwarkar[3], Xinxin Dong[3], Maggie McCue[3], Tom Macek[3]

[1]ProPhase LLC, New York, NY, [2]New York University, School of Medicine, New York, NY, [3]Takeda Development Center Americas, Inc., Deerfield, IL

Background

Early termination in clinical trials, especially in those involving schizophrenia, is a significant concern, with some studies showing early discontinuation rates of over 50% (Rabinowitz and Davidov, 2008). Missing data due to early dropouts can potentially compromise the results of a trial. While this area has been identified as an issue in antipsychotic clinical trials, there is little research on understanding the factors that lead to early termination (Mocks et al., 2002). One strategy that may be useful is to identify subjects who present with atypical symptom profiles. For instance, one would expect a patient who presents with high levels of hallucinatory behavior (represented by P3) to also show a correspondingly high level for conceptual disorganization (represented by P2). A patient who instead shows high scores on P3 but low scores on P2 is somewhat atypical. Atypical symptom profiles may represent subjects who are inappropriate for the clinical trial.

A recent Phase 2 study was completed in the US, evaluating the efficacy, safety and tolerability of treatment for 6 weeks with TAK-063 compared with placebo in subjects with acutely exacerbated schizophrenia. TAK-063 is a potent and selective inhibitor of the phosphodiesterase 10A (PDE10A) enzyme that is expressed primarily in the striatal medium spiny neurons (MSN) of the basal ganglia complex (Coskran et al., 2006), which receives extensive cortical (glutamatergic), thalamic, and nigral (dopaminergic) input. In phase 1 studies, TAK-063 has been shown to be safe and well tolerated at single doses up to 1000 mg in healthy subjects and following multiple dosing once daily (QD) for 7 days up to 100 mg in subjects with stable schizophrenia. A retrospective analysis of preliminary blinded data from this Phase 2 clinical trial presented an opportunity to investigate what strategies might be effective in identifying early terminators.

Objectives

Analyses were aimed to determine how different atypical PANSS score profiles (identified using algorithms) were able to predict early termination.

Methods

- Inpatient adults with a diagnosis of schizophrenia (N=153) receiving randomly-assigned treatment (including placebo) and identified as experiencing an acute exacerbation within the last 60 days were evaluated with the PANSS by raters.
- Twenty-one algorithms were examined at both the screening and baseline visits.
- First sensitivity and specificity analyses were run to broadly examine each algorithm's individual usefulness at identifying early terminators.
- Second, a CART analysis (specifically the use of recursive partitioning and regression trees) was run in order to isolate which (if any) algorithm combinations were most effective at identifying early terminators. Those item relationships that were flagged by the CART analysis as showing an ability to correctly identify early terminators were then further examined using logistic regression.
- Two approaches using logistic regression are compared in the receiver operating characteristic (ROC) curves shown. An ROC curve plots sensitivity versus specificity in order to demonstrate the cost/benefit analysis of different models. The better a model separates the early terminators from the completers, the more area under the curve (AUC).
- All analyses were run using R.

Results

- Forty-nine subjects (32.0%) were identified as early terminators from the study (see Table 1 for demographic information).
- Sensitivity and specificity analyses on the algorithms revealed a wide range of sensitivity and specificity values (Sensitivity: 0 to .94; Specificity: 0 to .99; see Table 2-6).
- Two algorithms had the most useful trade-off between sensitivity and specificity, one involving negative symptoms (labeled NEG: N1 [Blunted Affect] and N5 [Difficulty in Abstract Thinking]), and one involving two positive symptoms (labeled POS1: P2 [Conceptual Disorganization] and P1 [Delusions]).
- Use of recursive partitioning and regression trees also identified these two algorithms for early terminators (see Figure 1).
- Finally, logistic regression analyses revealed a significant interaction effect between these two algorithms (z=2.1, p < .03). The ROC curves show the use of NEG only (red) in a logistic regression (AUC = .55) while the blue curve shows the use of both POS1 and NEG as predictors (AUC = .61; see Figure 2).

Table 1. Demographic Characteristics of Sample

Mean Age (SD)	42.2 (10.5)
Mean PANSS Score at Baseline (SD)	98.2 (10.4)
Sex	29 (19.0%) Female 124 (81.0%) Male
Race	
Black/African-American	103 (67.3%)
White	46 (30.1%)
Other	4 (2.6%)
Hispanic	17 (11.1%)

Table 2. Contingency Table for ANX Algorithm

		Early Termination: **No**	Early Termination: **Yes**
ANX	**No**	100	49
	Yes	4	0

Table 3. Contingency Table for POS1 Algorithm

		Early Termination: **No**	Early Termination: **Yes**
POS1	**No**	66	32
	Yes	38	17

Table 4. Contingency Table for POS2 Algorithm

		Early Termination: **No**	Early Termination: **Yes**
POS2	**No**	13	8
	Yes	91	41

Table 5. Contingency Table for NEG Algorithm

		Early Termination: **No**	Early Termination: **Yes**
NEG	**No**	67	37
	Yes	37	12

Figure 1. CART Analysis of Algorithms

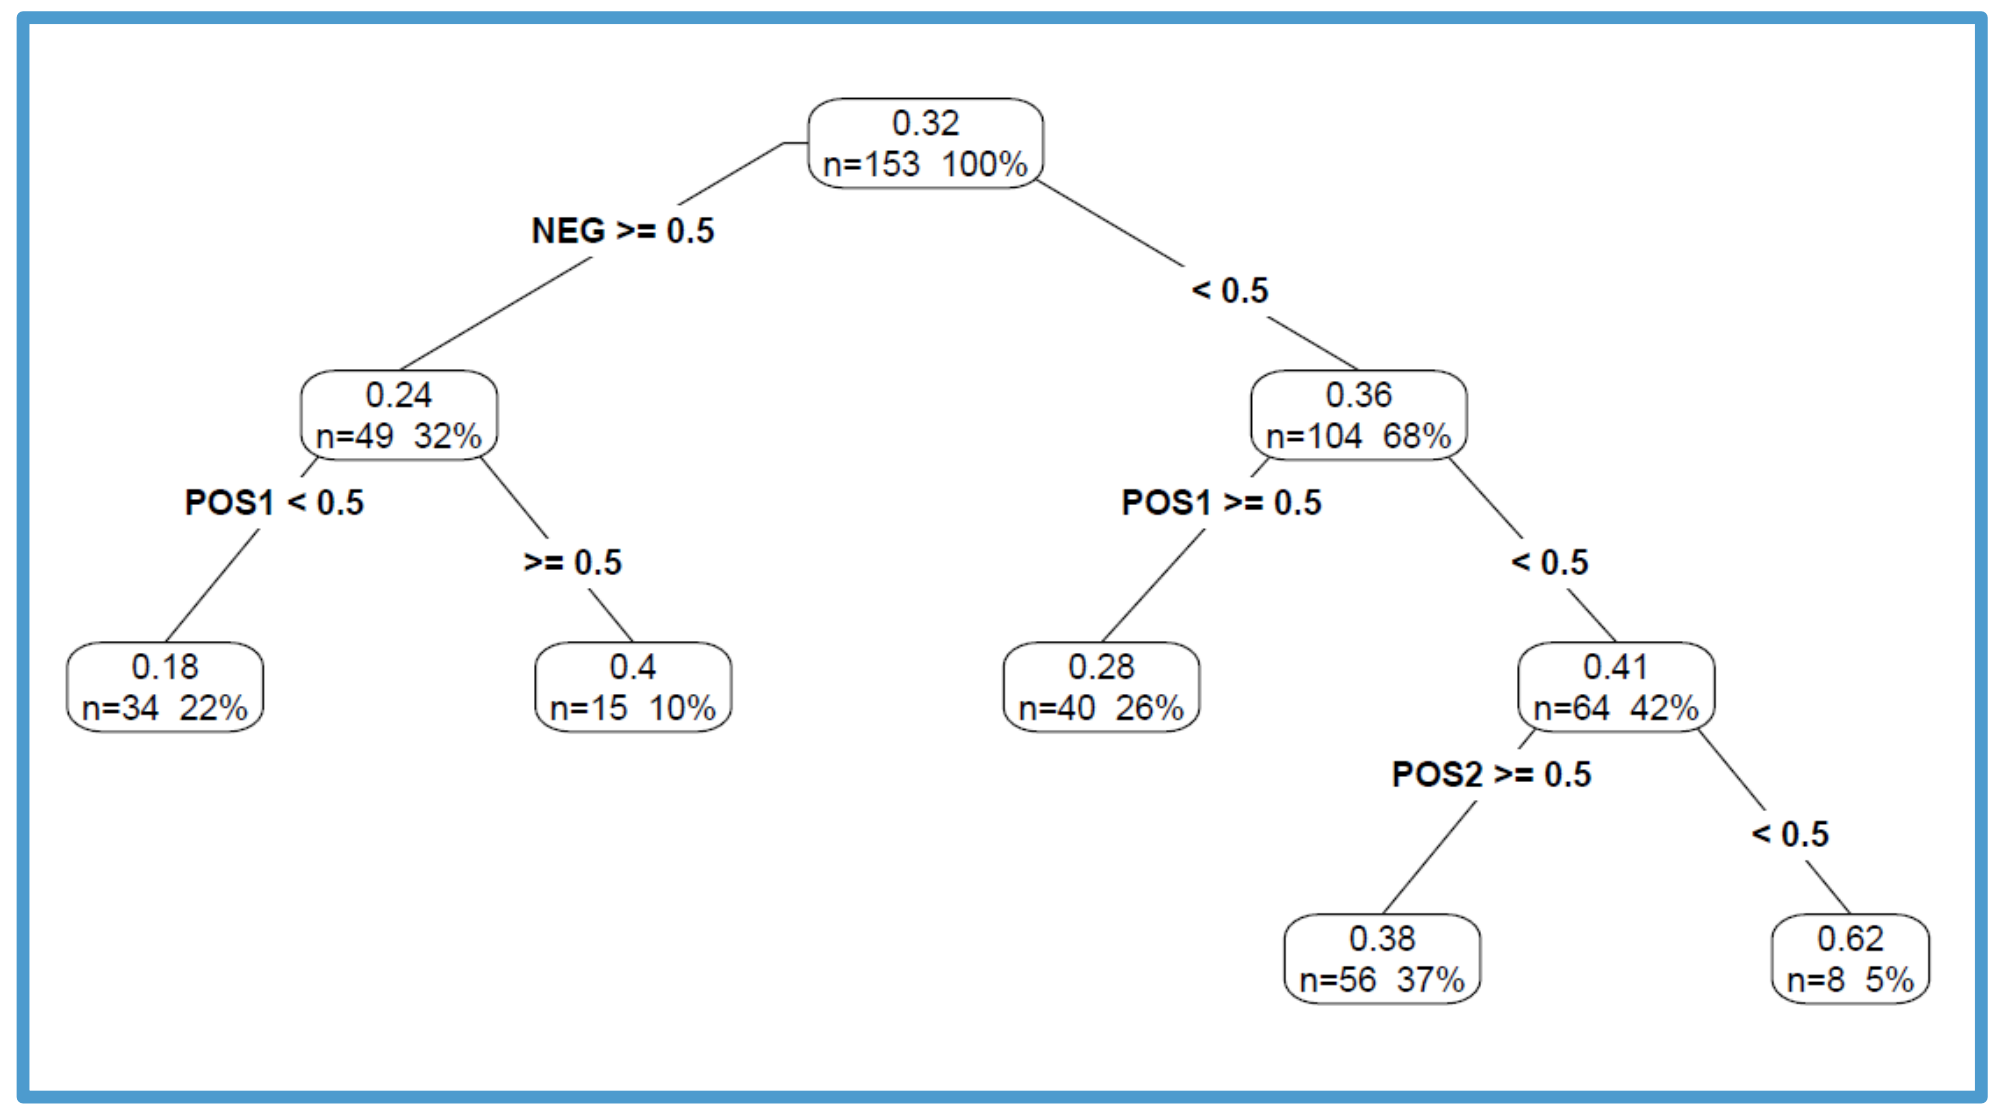
0.32
n=153 100%
NEG >= 0.5
< 0.5
0.24
n=49 32%
0.36
n=104 68%
POS1 < 0.5
>= 0.5
POS1 >= 0.5
< 0.5
0.18
n=34 22%
0.4
n=15 10%
0.28
n=40 26%
0.41
n=64 42%
POS2 >= 0.5
< 0.5
0.38
n=56 37%
0.62
n=8 5%

Figure 2. ROC curve showing use of NEG, as well as NEG and POS1

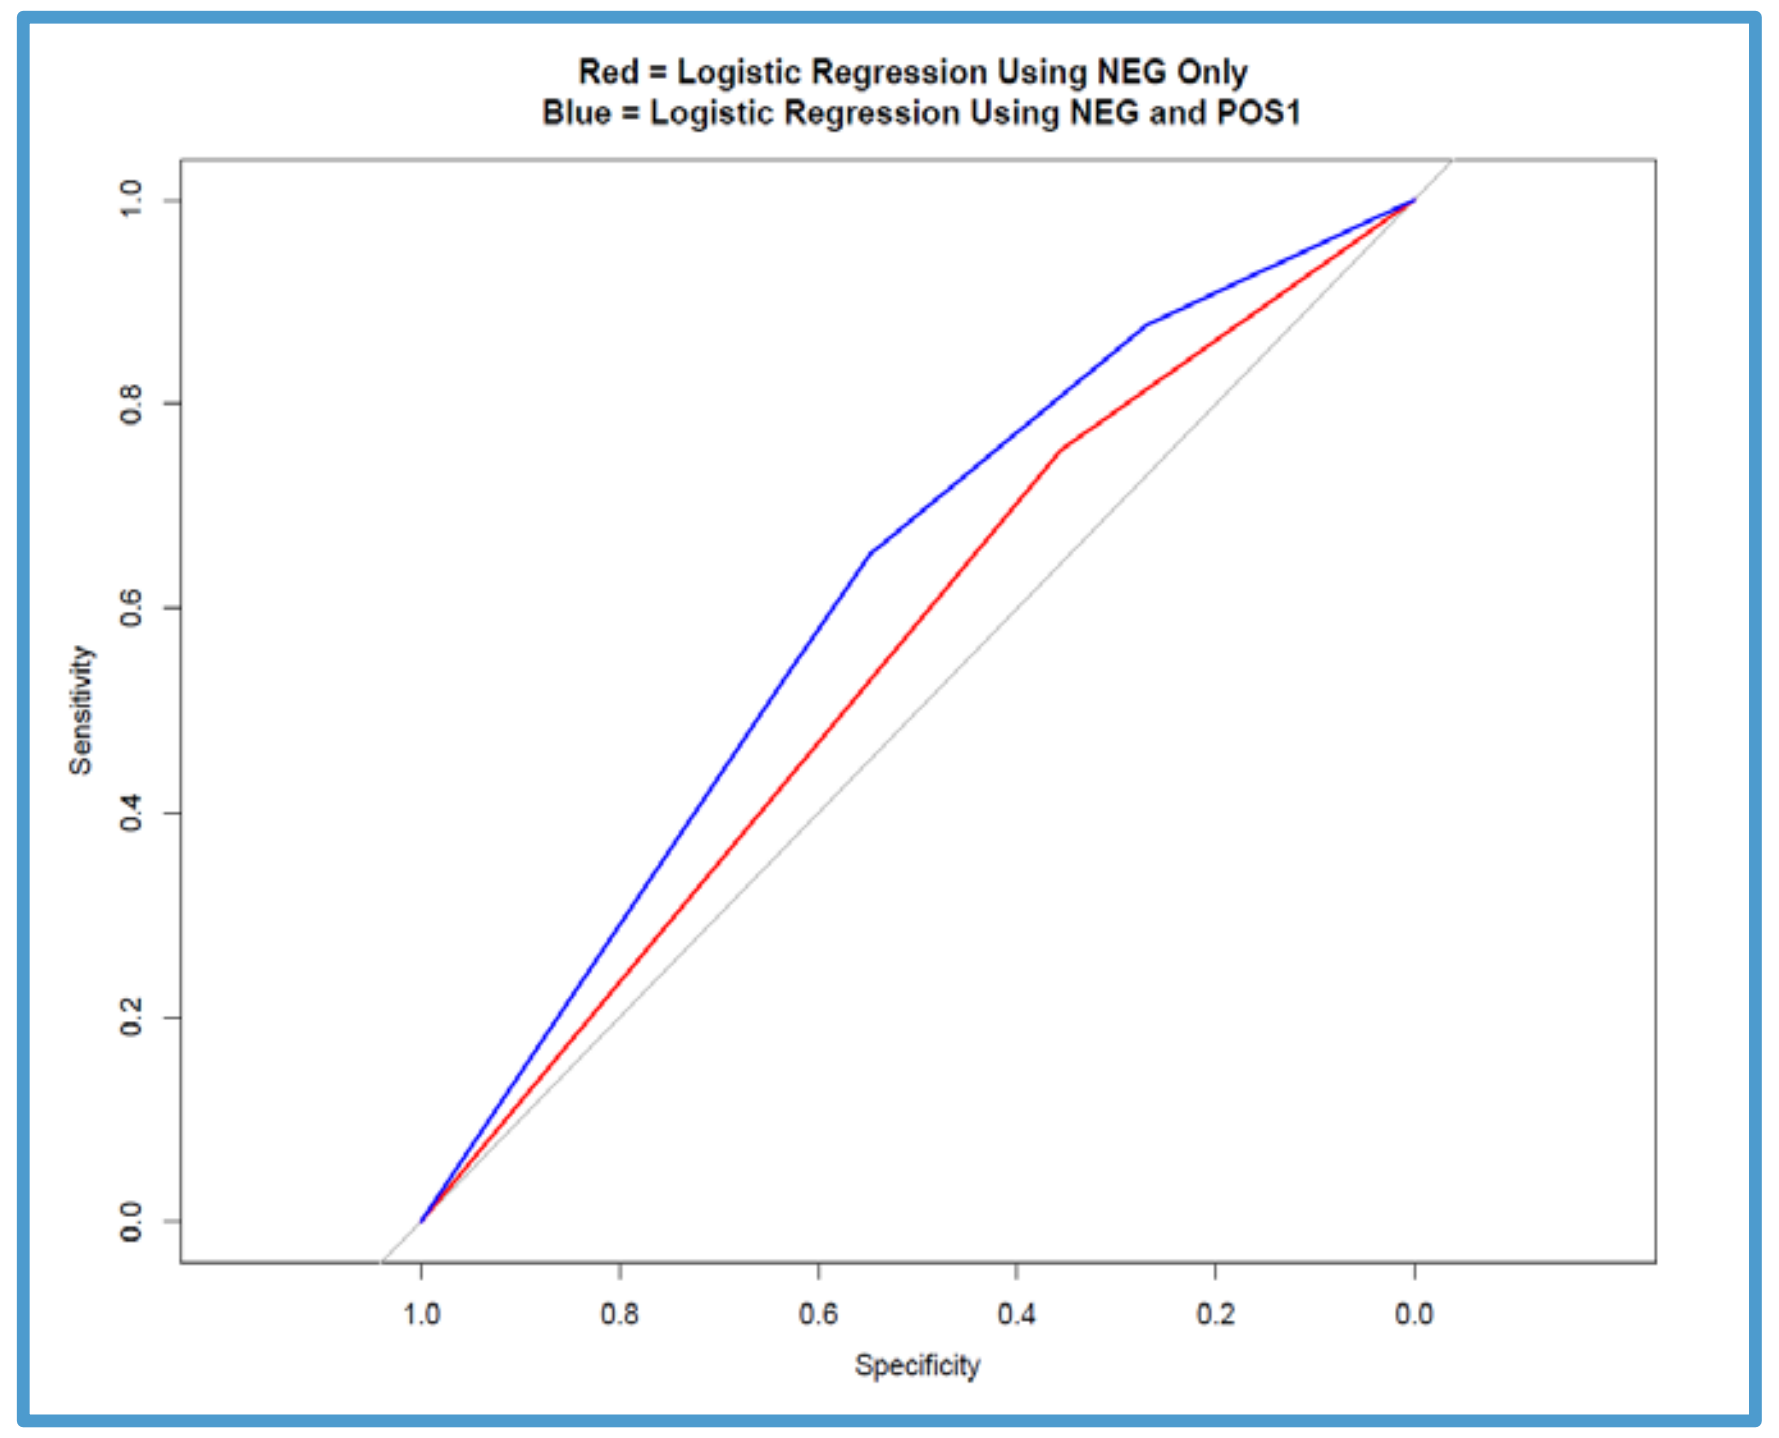
Red = Logistic Regression Using NEG Only
Blue = Logistic Regression Using NEG and POS1
Sensitivity
Specificity
0.0
0.2
0.4
0.6
0.8
1.0

Table 6. Selection of Algorithms and Sensitivity/Specificity Analyses

Algorithm Label	Relevant PANSS items	Sensitivity	Specificity	PPV	NPV
ANX	G2 (Anxiety), G4(Tension)	.00	.96	.00	.67
POS1	P2 (Conceptual Disorganization), P1 (Delusions)	.35	.63	.31	.67
POS2	P3 (Hallucinatory Behavior), G15 (Preoccupation)	.94	.07	.32	.70
NEG	N1 (Blunted Affect), N5 (Difficulty in Abstract Thinking)	.24	.64	.24	.64

Conclusions

- These data provide preliminary evidence that atypical score profiles on the PANSS can be useful as method of detecting patients who are early terminators.
- First, dependent on the identification strategy desired, different algorithms may be particularly helpful. For instance, one strategy might be to focus on identifying all early terminators regardless of how many false positives are also identified. Using this strategy, an algorithm that maximizes sensitivity such as POS2 (Sensitivity= .94) might appear desirable. However, more realistically, a strategy such as this would lead to a waste of resources as it also identifies a large number of patients who do not early terminate (Specificity= .07). It is more likely that one would want to use an algorithm that maximizes sensitivity and specificity.
- Our results showed that individually, NEG and POS1, showed a reasonable trade-off between sensitivity and specificity. Each algorithm by itself was able to correctly predict around 25%-35% of those who early terminated.
- In addition, CART and logistic regression analyses were able to show that the combination of these two algorithms may also be useful in further refining identification of subjects who early terminate.
- Use of algorithms such as these may be helpful in first identifying individuals who are likely to early terminate; these individuals may require further screening to determine their appropriateness for the clinical trial.
- Future analyses should examine these algorithms with data from other samples of patients from schizophrenia clinical trials.

References

Coskran TM, Morton D, Menniti FS, Adamowicz WO, Kleiman RJ, Ryan AM, et al (2006). Immunohistochemical localization of phosphodiesterase 10A in multiple mammalian species. *J Histochem Cytochem* **54**:1205-1213.

Mocks J, Kohler W, Scott M, Maurer J, Budde M, Givens S (2002). Dealing with selective dropout in clinical trials. *Pharm Stat* **1**: 119-130.

Rabinowitz J, Davidov O (2008). The association of dropout and outcome in trials of antipsychotic medication and its implications for dealing with missing data. *Schizophrenia Bulletin* **34**: 286-291.

Acknowledgment
Sponsored by Takeda Development Center Americas, Inc., Deerfield, IL.
Presented during The International Society for CNS Clinical Trials and Methodology Annual Meeting (ISCTM), September 26-27, 2016, Philadelphia, PA.
Study 2002: NCT02477020

Disclosures
Mark Opler, Jonathan Lam, and Jennifer Lord-Bessen are employees of ProPhase LLC, New York, NY, and consultants to Takeda Development Center Americas, Inc., Deerfield, IL.

Elizabeth Hanson, Atul Mahableshwarkar, Xinxin Dong, Maggie McCue, and Thomas A. Macek are employees of Takeda Development Center Americas, Inc., Deerfield, IL.