NONPARAMETRIC METHODS FOR ANALYZING COGNITIVE TEST DATA WHERE MISSING VALUES MAY REFLECT COGNITIVE IMPAIRMENT: ANOTHER LOOK AT PEARL III

Judith Jaeger[1]; Cynthia Siu[2]; Michael Tocco[3]; Robert Goldman[3]; Antony Loebel[3]

[1] Albert Einstein College of Medicine, NY and CognitionMetrics, LLC; [2] COS & Associates Ltd., Central, Hong Kong; [3] Sunovion Pharmaceuticals Inc., Fort Lee, NJ

ABSTRACT

INTRODUCTION: Results from a 6-week, double-blind, placebo-controlled study demonstrated that patients treated with lurasidone 160 mg/day performed significantly better at study endpoint on a measure of cognitive function (computerized Cogstate Schizophrenia Battery (CSB) than patients receiving placebo (p=0.038) or quetiapine XR 600 mg/day (p=0.018). Patients treated with lurasidone 80 mg/day did not significantly differ from placebo or quetiapine XR groups. (Harvey et al, 2013). However, this finding was only observed in a subset (n=267) of patients in the intent to treat (ITT) population (n=482) whose cognitive testing was determined to be evaluable at both baseline and Week 6 based on prespecified criteria. This has raised concerns that the finding might not be generalizable to the full sample. While a subsequent study introduced adaptations to the CSB that have improved its acceptability in acutely ill patients (Citrome et al, 2016), there is evidence in the present study to suggest that greater impairment in the non-evaluable sample may have contributed to their difficulty in completing the tests. For example subjects with evaluable compared to non-evaluable cognition scores had significantly lower mean PANSS total scores at baseline (p<0.001) (Harvey et al, 2013).

METHODS: In order to address the issue of incomplete data from the non-evaluable sample, an analysis based on the full ITT sample was conducted using rank ANCOVA, in which non-evaluable scores at baseline and/or week-6, missing cognitive testing scores and/or early dropouts were assigned the lowest possible rank score (=0). Tied ranks were resolved by standard averaging methods. Accordingly, a positive change in rank reflects improvement in position relative to the whole sample, whereas a negative change in rank reflects a worsening of relative position. This method allowed for missing or partial data from all patients to inform the analysis, and assumes that missing data reflects the lowest performance level.

RESULTS: The results from the full ITT data sample were comparable to previous analyses using the evaluable subgroups. Patients treated with lurasidone 160 mg/day improved in rank between baseline and 6 weeks by 10.2 rank points, whereas those on placebo experienced an average drop in rank of -16.8 points, a difference which was statistically significant (p<0.05). Patients in the lurasidone 80 mg/day group improved by 6 rank points (p=0.08) and the quetiapine XR 600 mg/day group did not differ from placebo.

CONCLUSION: In this post-hoc analysis, a nonparametric approach was employed to permit the inclusion of missing cognitive data from patients who initially failed the evaluability criteria. Using this technique, results from the full patient ITT sample, including those with significant cognitive impairment at the time of testing due to severe psychiatric illness, were shown to be consistent with a subsample of evaluable patients. Informed by this experience, the (computerized) CSB was modified to permit rest periods if needed, as is routine with paper and pencil cognitive testing, the test sequence in the was altered. Using this modified presentation of the identical tests, Citrome et al (2016) achieved 100% evaluable data on the CSB in an acute schizophrenia sample.

INTRODUCTION

Results from a 6-week, double-blind, placebo-controlled study demonstrated that patients treated with lurasidone 160 mg/day performed significantly better at study endpoint on a measure of cognitive function (computerized Cogstate Schizophrenia Battery (CSB)) than patients receiving placebo (p=0.038) or quetiapine XR 600 mg/day (p=0.018). Patients treated with lurasidone 80 mg/day did not significantly differ from placebo or quetiapine XR groups. (Harvey et al, 2013).

However, this finding was only observed in a subset (n=267) of patients in the intent to treat (ITT) population (n=482), whose cognitive testing was determined to be evaluable at both baseline and Week 6 based on prespecified criteria. This has raised concerns that the finding might not be generalizable to the full sample.

There is evidence in the present study to suggest that greater impairment in the non-evaluable sample may have contributed to their difficulty in completing the tests (Harvey et al, 2013).

Furthermore, a recent study showed that new adaptations to the Cogstate Schizophrenia Battery have improved its acceptability in acutely ill patients (Citrome et al, 2016).

OBJECTIVE

The objective of this post-hoc analysis was to investigate the utility of applying rank ANCOVA (Koch et al., 1990) to account for missing/incomplete cognitive data in a short-term phase 3 study.

Sponsored by Sunovion Pharmaceuticals Inc.

International Society for CNS Clinical Trials and Methodology (ISCTM)

Autumn Conference | September 26-27, 2016 | Philadelphia, PA

METHODS

- Hospitalized patients who met DSM-IV criteria for schizophrenia were randomized to 6-weeks of double-blind treatment with fixed doses of lurasidone 80 mg/d (N=125, LUR80), lurasidone 160 mg/d (N=121, LUR160), quetiapine XR 600mg/d (N=120, QXR), or placebo (N=122), administered once-daily in the evening with a meal or within 30 minutes after eating.
- Primary study outcome was the mean change in PANSS total score for lurasidone fixed-dose groups vs. placebo.

Figure 1. Study Design

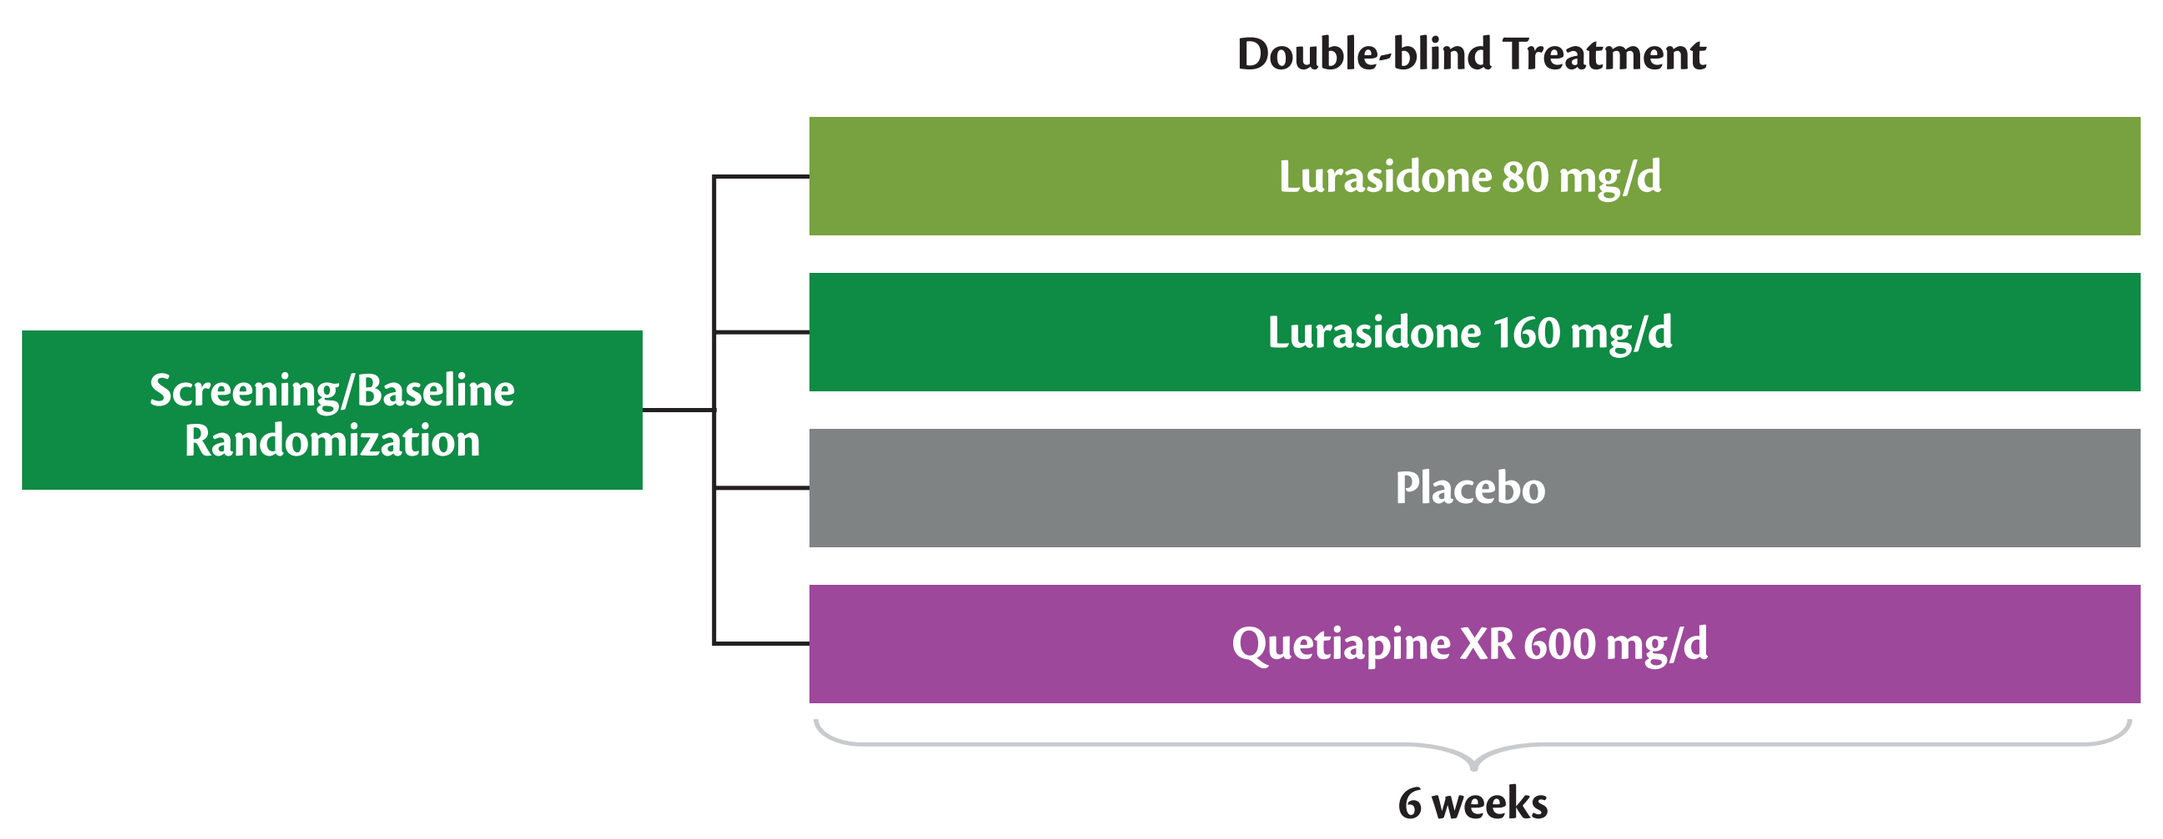
Double-blind Treatment
Screening/Baseline Randomization
Lurasidone 80 mg/d
Lurasidone 160 mg/d
Placebo
Quetiapine XR 600 mg/d
6 weeks

Titration: Lurasidone 80 mg/d arm – 80 mg/d from Day 1 on (no titration)
Lurasidone 160 mg/d arm – 120 mg/d for Days 1-2; then 160 mg/d from Day 3 on
Quetiapine XR 600 mg/d arm – 300 mg/d for Days 1-2; then 600 mg/d from Day 3 on

Participants

- Subjects had a PANSS total score ≥80 at screening and baseline, with a score ≥4 (moderate) on 2 or more of the following PANSS items: delusions, conceptual disorganization, hallucinations, and unusual thought content.
- Subjects had a score ≥4 on the CGI-S at screening and baseline.

Cogstate Test Scores: Data Evaluability Validation

- All Cogstate data were reviewed for completion and performance based on pre-specified criteria and Cogstate internal standard operating procedures. "Performance check" failures indicate when performance is sufficiently poor to suggest that the subject is not performing in accordance with test instructions, and hence that that the test score may not reflect the cognitive operation the test was designed to assess.
- Cogstate task completion were defined as follows:
 – Detection, Identification, One Card Learning, One Back, and SECT: Response given for at least 75% of trials
 – ISLT: All three rounds of the task were completed.
 – GMLT: All five rounds were completed (28 correct moves on the last round).
 – GMR: 28 correct moves were made.
- Cogstate performance check criteria were set as follows:
 – Detection Task: Accuracy of performance on the Detection Task >75%
 – Identification Task: Accuracy of performance on the Identification Task >75%
 – One Back Learning Task: Accuracy greater than 50%
 – One Card Learning Task: Accuracy greater than 50%
 – SECT: Accuracy greater than 25%

Statistical Methods

An analysis based on the full ITT sample was conducted using rank ANCOVA, in which non-evaluable scores at baseline and/or week-6, missing cognitive testing scores and/or early dropouts were assigned the lowest possible rank score (=0). Tied ranks were resolved by standard averaging methods.

Accordingly, a positive change in rank reflects improvement in position relative to the whole sample, whereas a negative change in rank reflects a worsening of relative position. This method allowed for missing or partial data from all patients to inform the analysis, and assumes that missing data reflects the lowest performance level.

RESULTS

Table 1. Demographics and Baseline Characteristics

	LUR160 N=121	LUR80 N=125	PBO N=120	QXR N=116
Age, Mean (SD)	37.9 (11.3)	36.2 (10.9)	37.3 (10.8)	37.3 (10.3)
Gender, n (%)				
Male	82 (68%)	96 (77%)	76 (63%)	75 (65%)
Female	39 (32%)	29 (23%)	44 (37%)	41 (35%)
Race, n (%)				
White	63 (52%)	75 (60%)	68 (57%)	69 (59%)
Black	29 (24%)	22 (18%)	24 (20%)	19 (16%)
Asian/Other	29 (24%)	28 (22%)	27 (23%)	28 (25%)
PANSS Total Score, Mean (SD)	97.5 (11.8)	97.7 (9.7)	96.6 (10.2)	97.7 (10.2)

RESULTS

Figure 2. Changes in Rank Score from Baseline to Week 6 in Cognitive Composite Score ITT Sample(N=482)

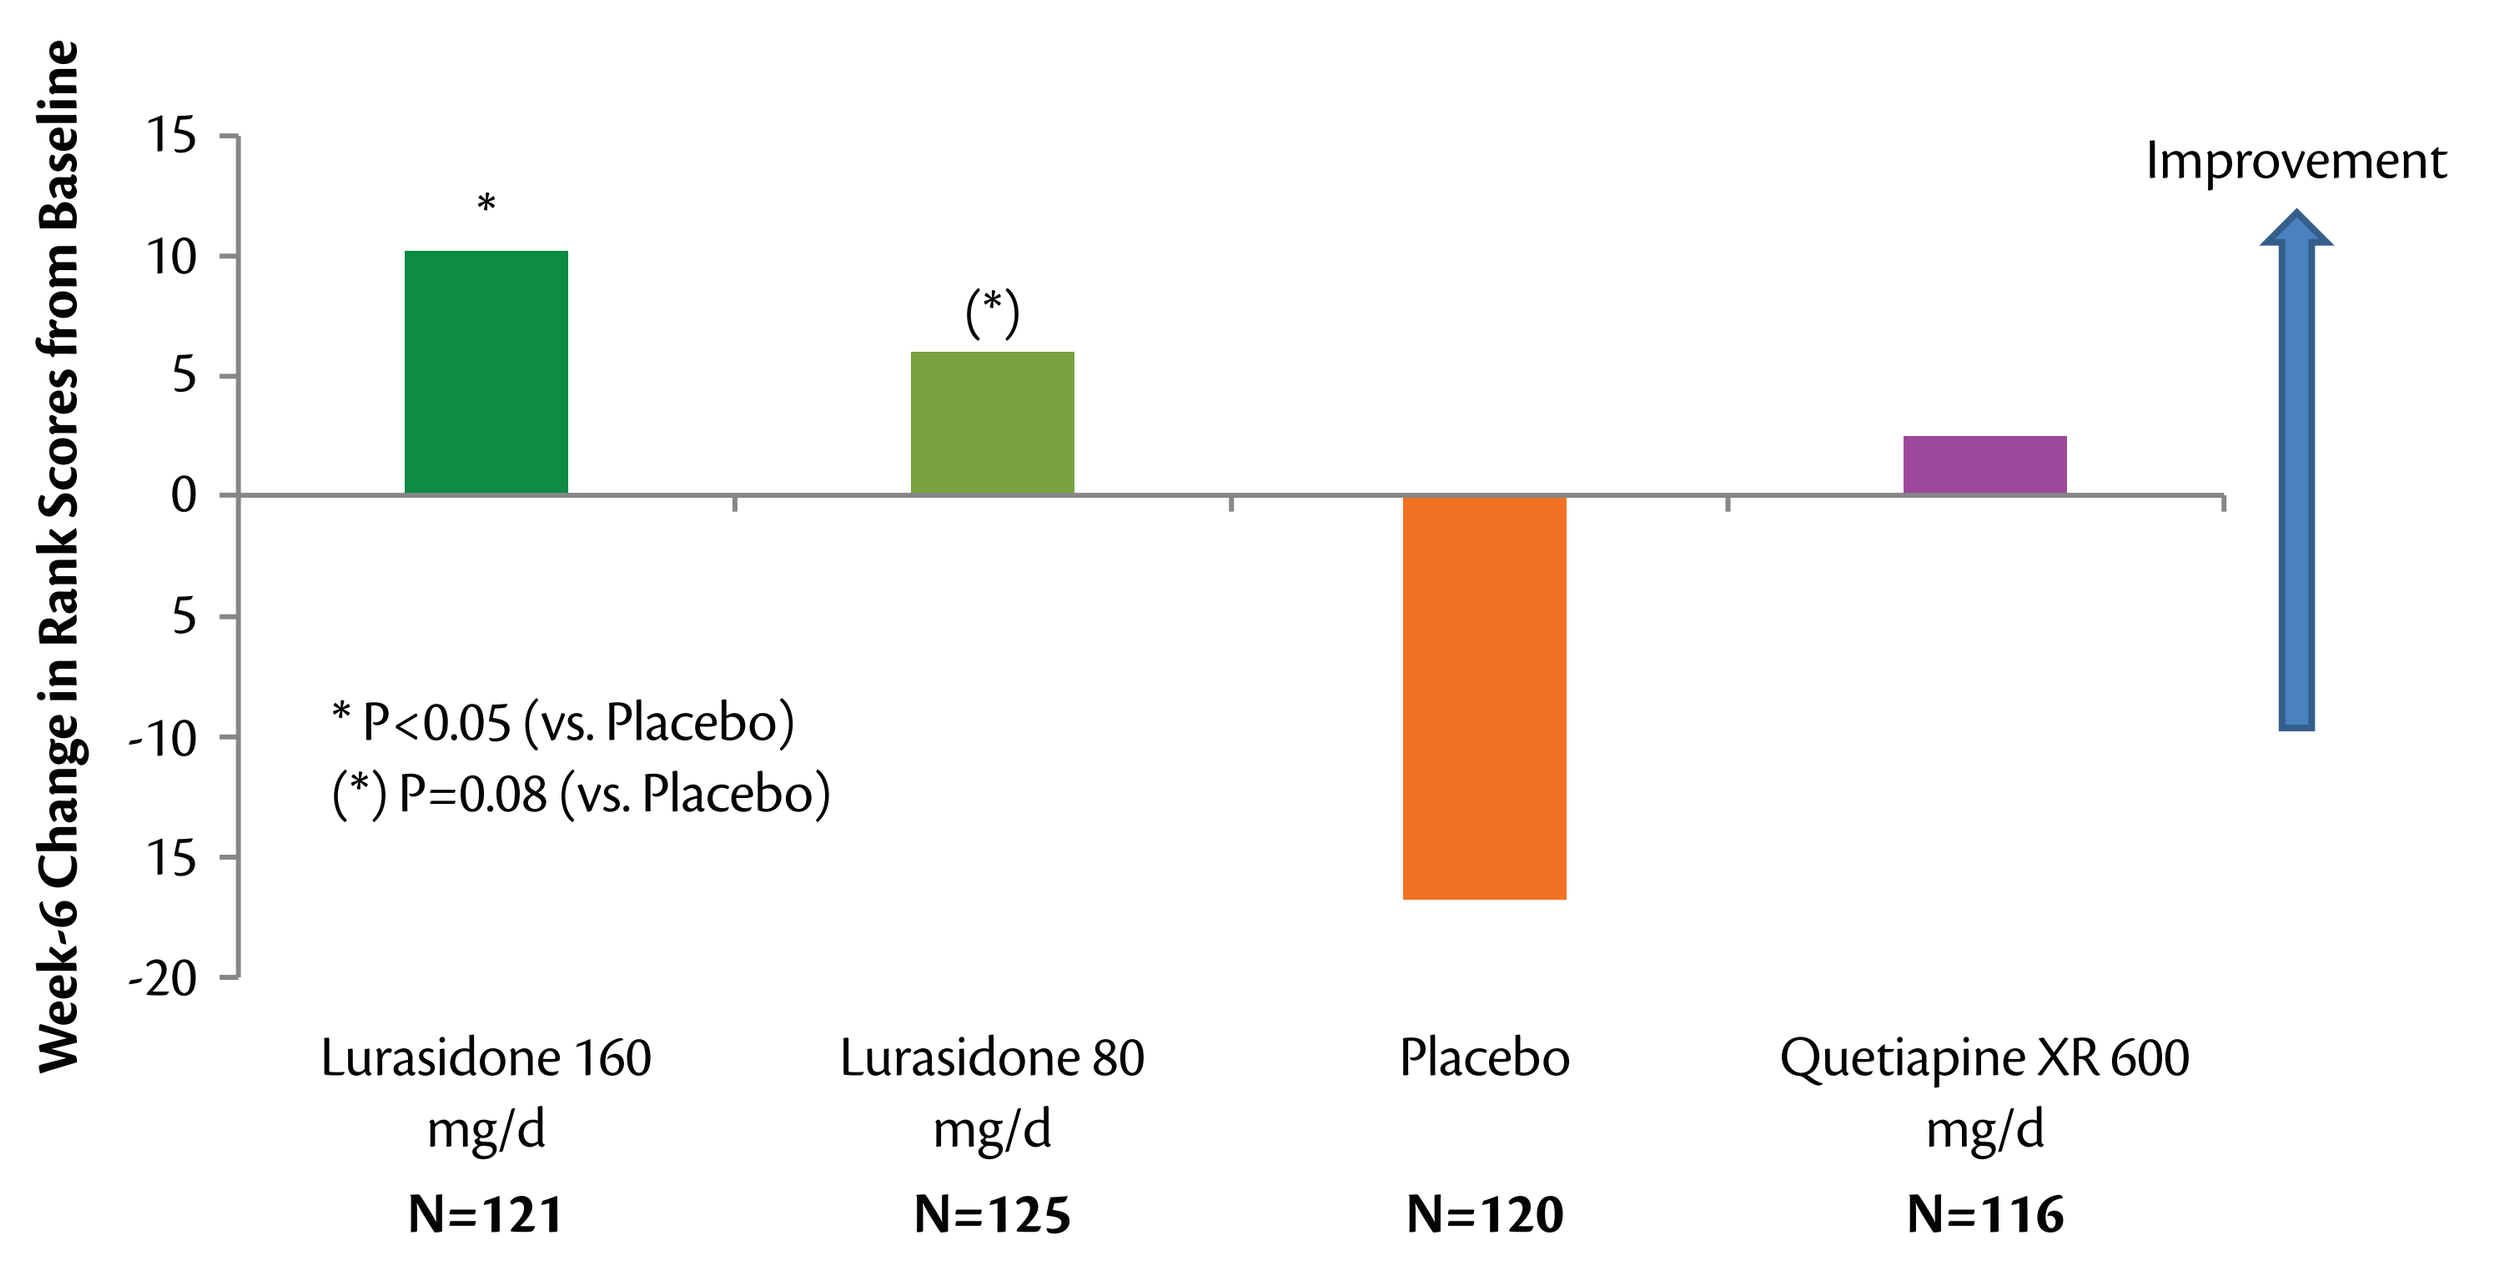
Week-6 Change in Rank Scores from Baseline
15
10
5
0
5
-10
15
-20
*
(*)
Improvement
* P<0.05 (vs. Placebo)
(*) P=0.08 (vs. Placebo)
Lurasidone 160 mg/d
Lurasidone 80 mg/d
Placebo
Quetiapine XR 600 mg/d
N=121
N=125
N=120
N=116

Rank Score = 0 (non-evaluable composite score or missing or early dropout).

Figure 3. Rate of Improvement (Rank Score Change from Baseline > 0) at Week 6: ITT Sample (N=482)

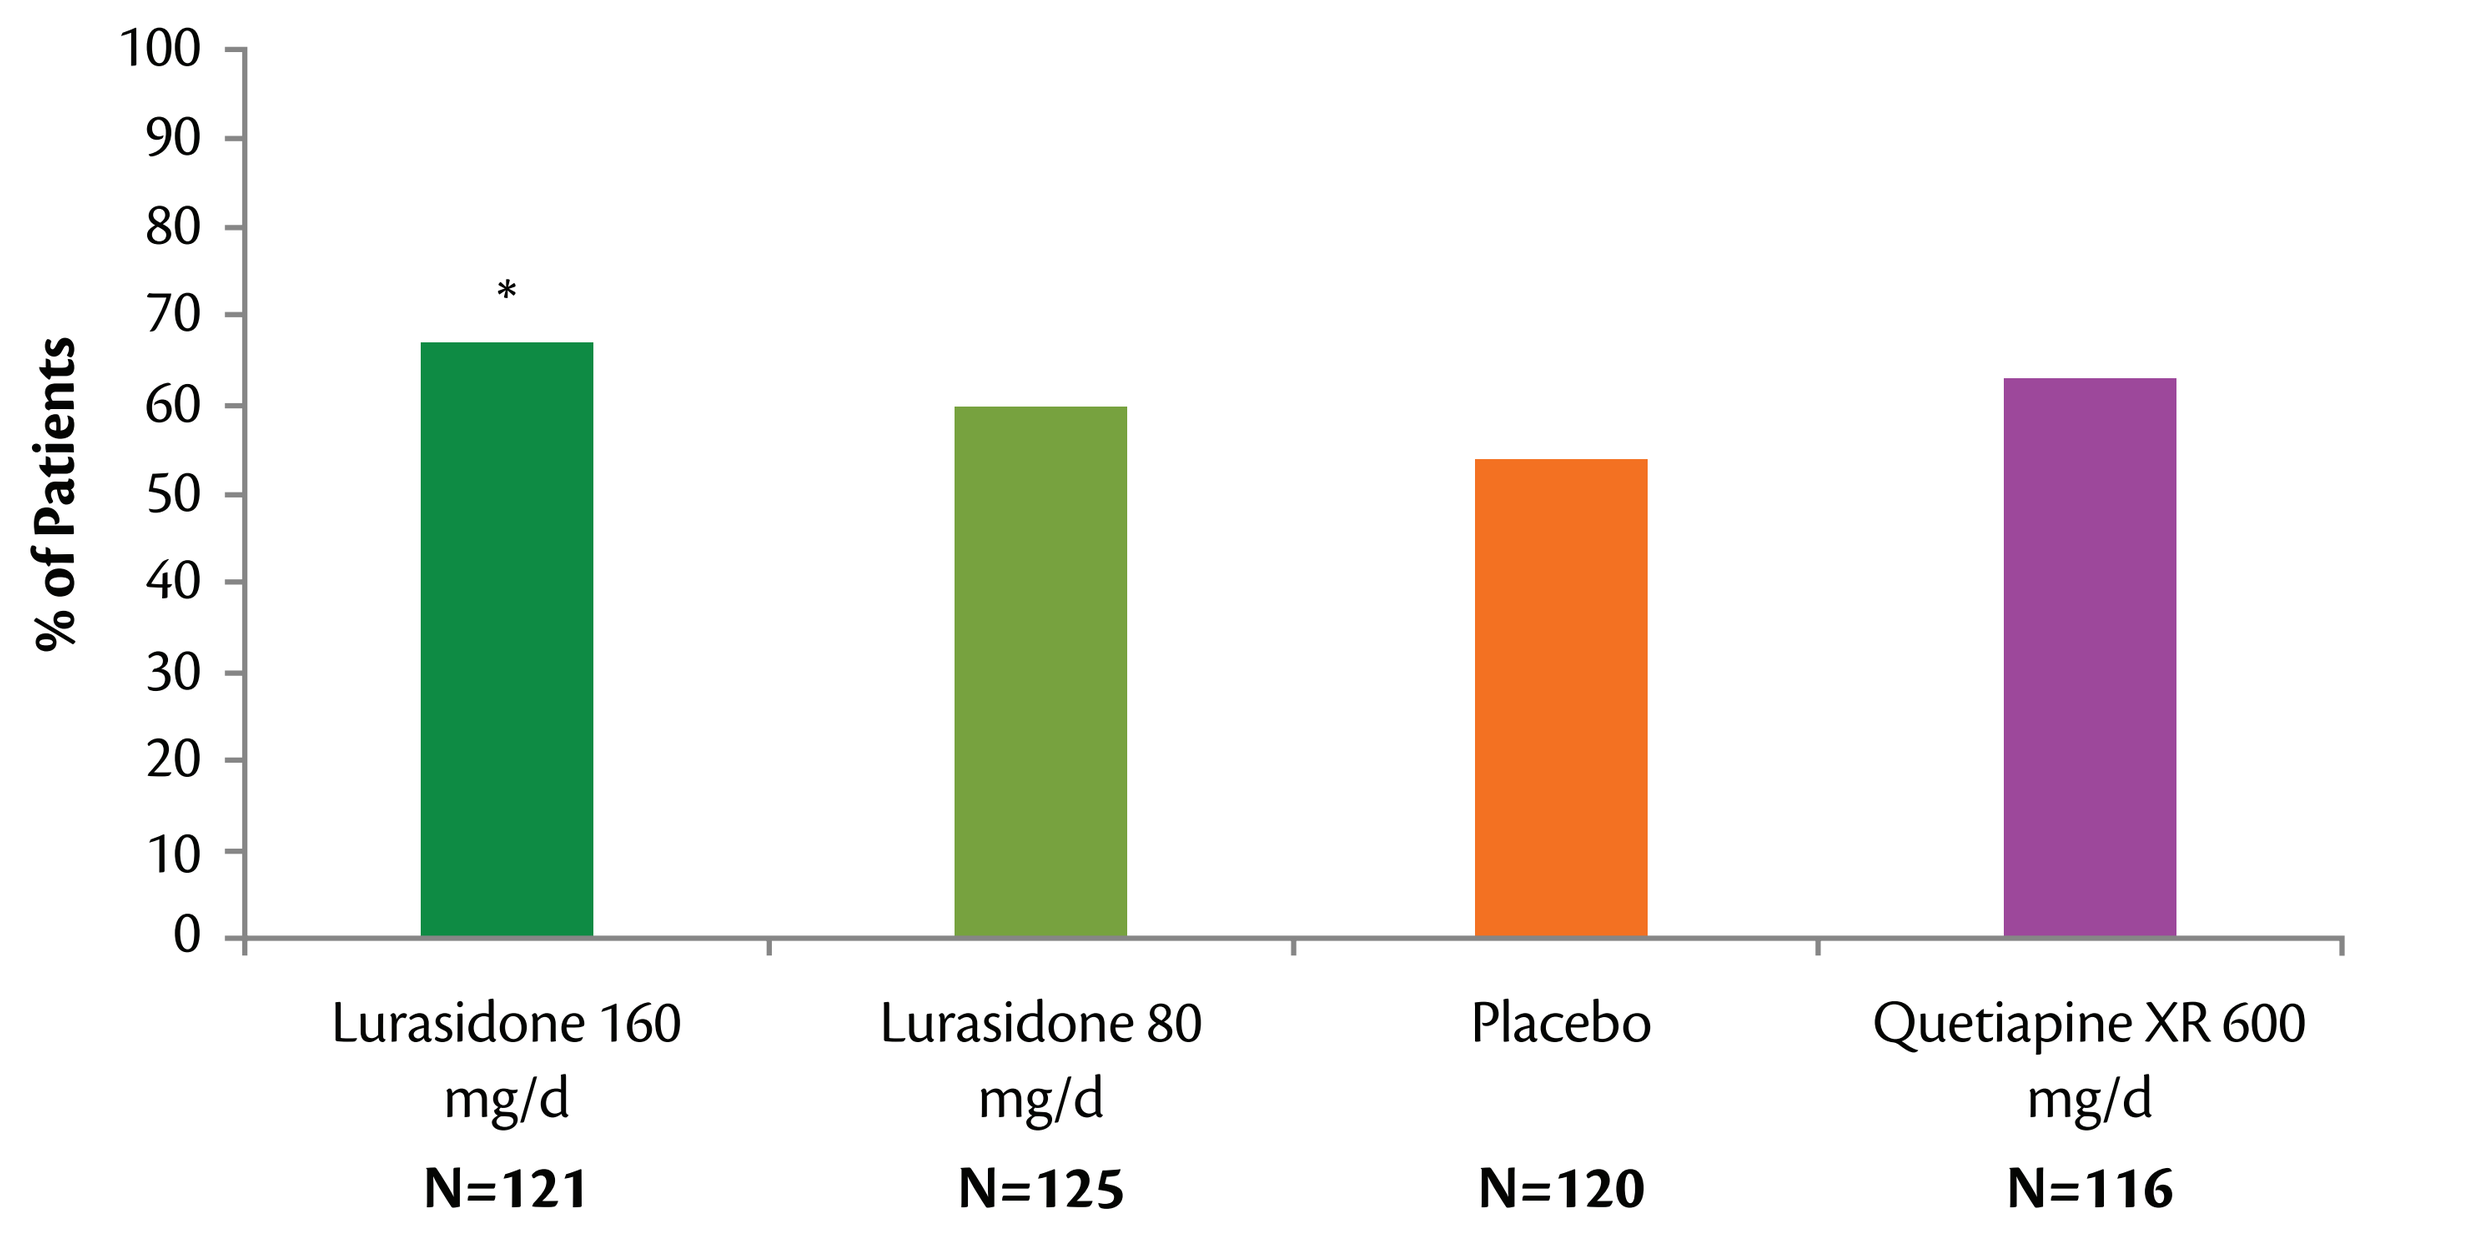
% of Patients
100
90
80
70
60
50
40
30
20
10
0
*
Lurasidone 160 mg/d
Lurasidone 80 mg/d
Placebo
Quetiapine XR 600 mg/d
N=121
N=125
N=120
N=116

Improvement = 1 if shift from baseline to week 6 in Rank score > 0.
* P<0.05 (vs. Placebo)

Figure 4. Changes in Rank Score from Baseline to Week 6 in Groton Maze Learning Test Score: ITT Sample (N=482)

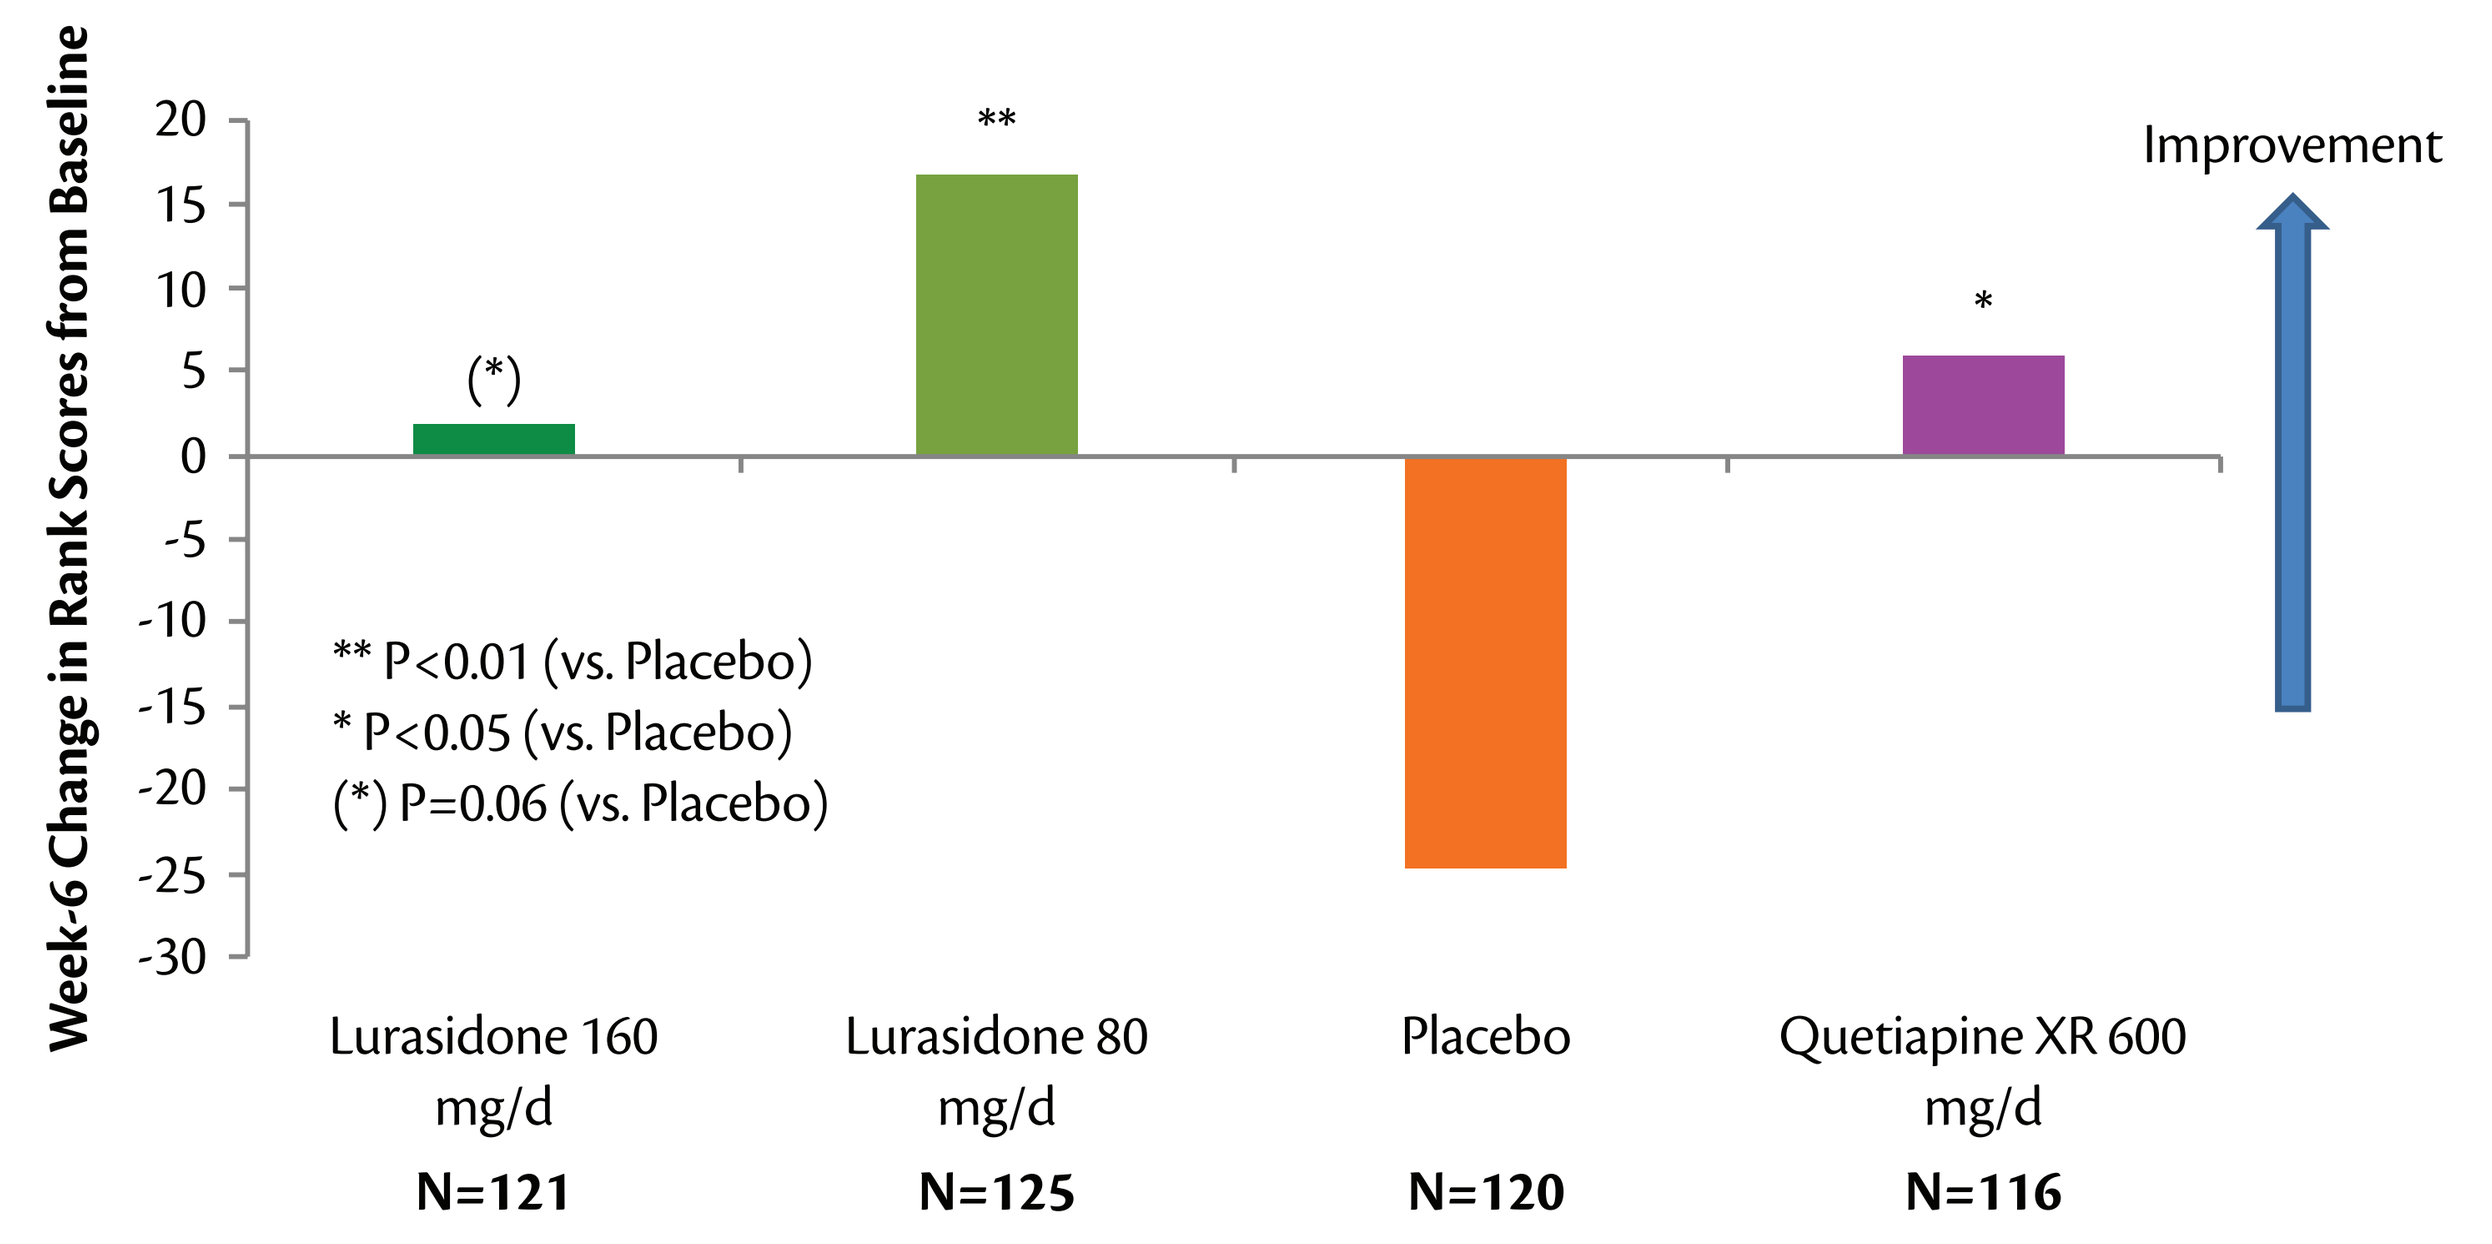
Week-6 Change in Rank Scores from Baseline
20
15
10
5
0
-5
-10
-15
-20
-25
-30
(*)
**
*
Improvement
** P<0.01 (vs. Placebo)
* P<0.05 (vs. Placebo)
(*) P=0.06 (vs. Placebo)
Lurasidone 160 mg/d
Lurasidone 80 mg/d
Placebo
Quetiapine XR 600 mg/d
N=121
N=125
N=120
N=116

Rank Score = 0 (non-evaluable composite score or missing or early dropout).

Figure 5. Groton Maze Learning Test Error Taxonomy: Executive vs. Non-Executive Learning Errors (N=429)

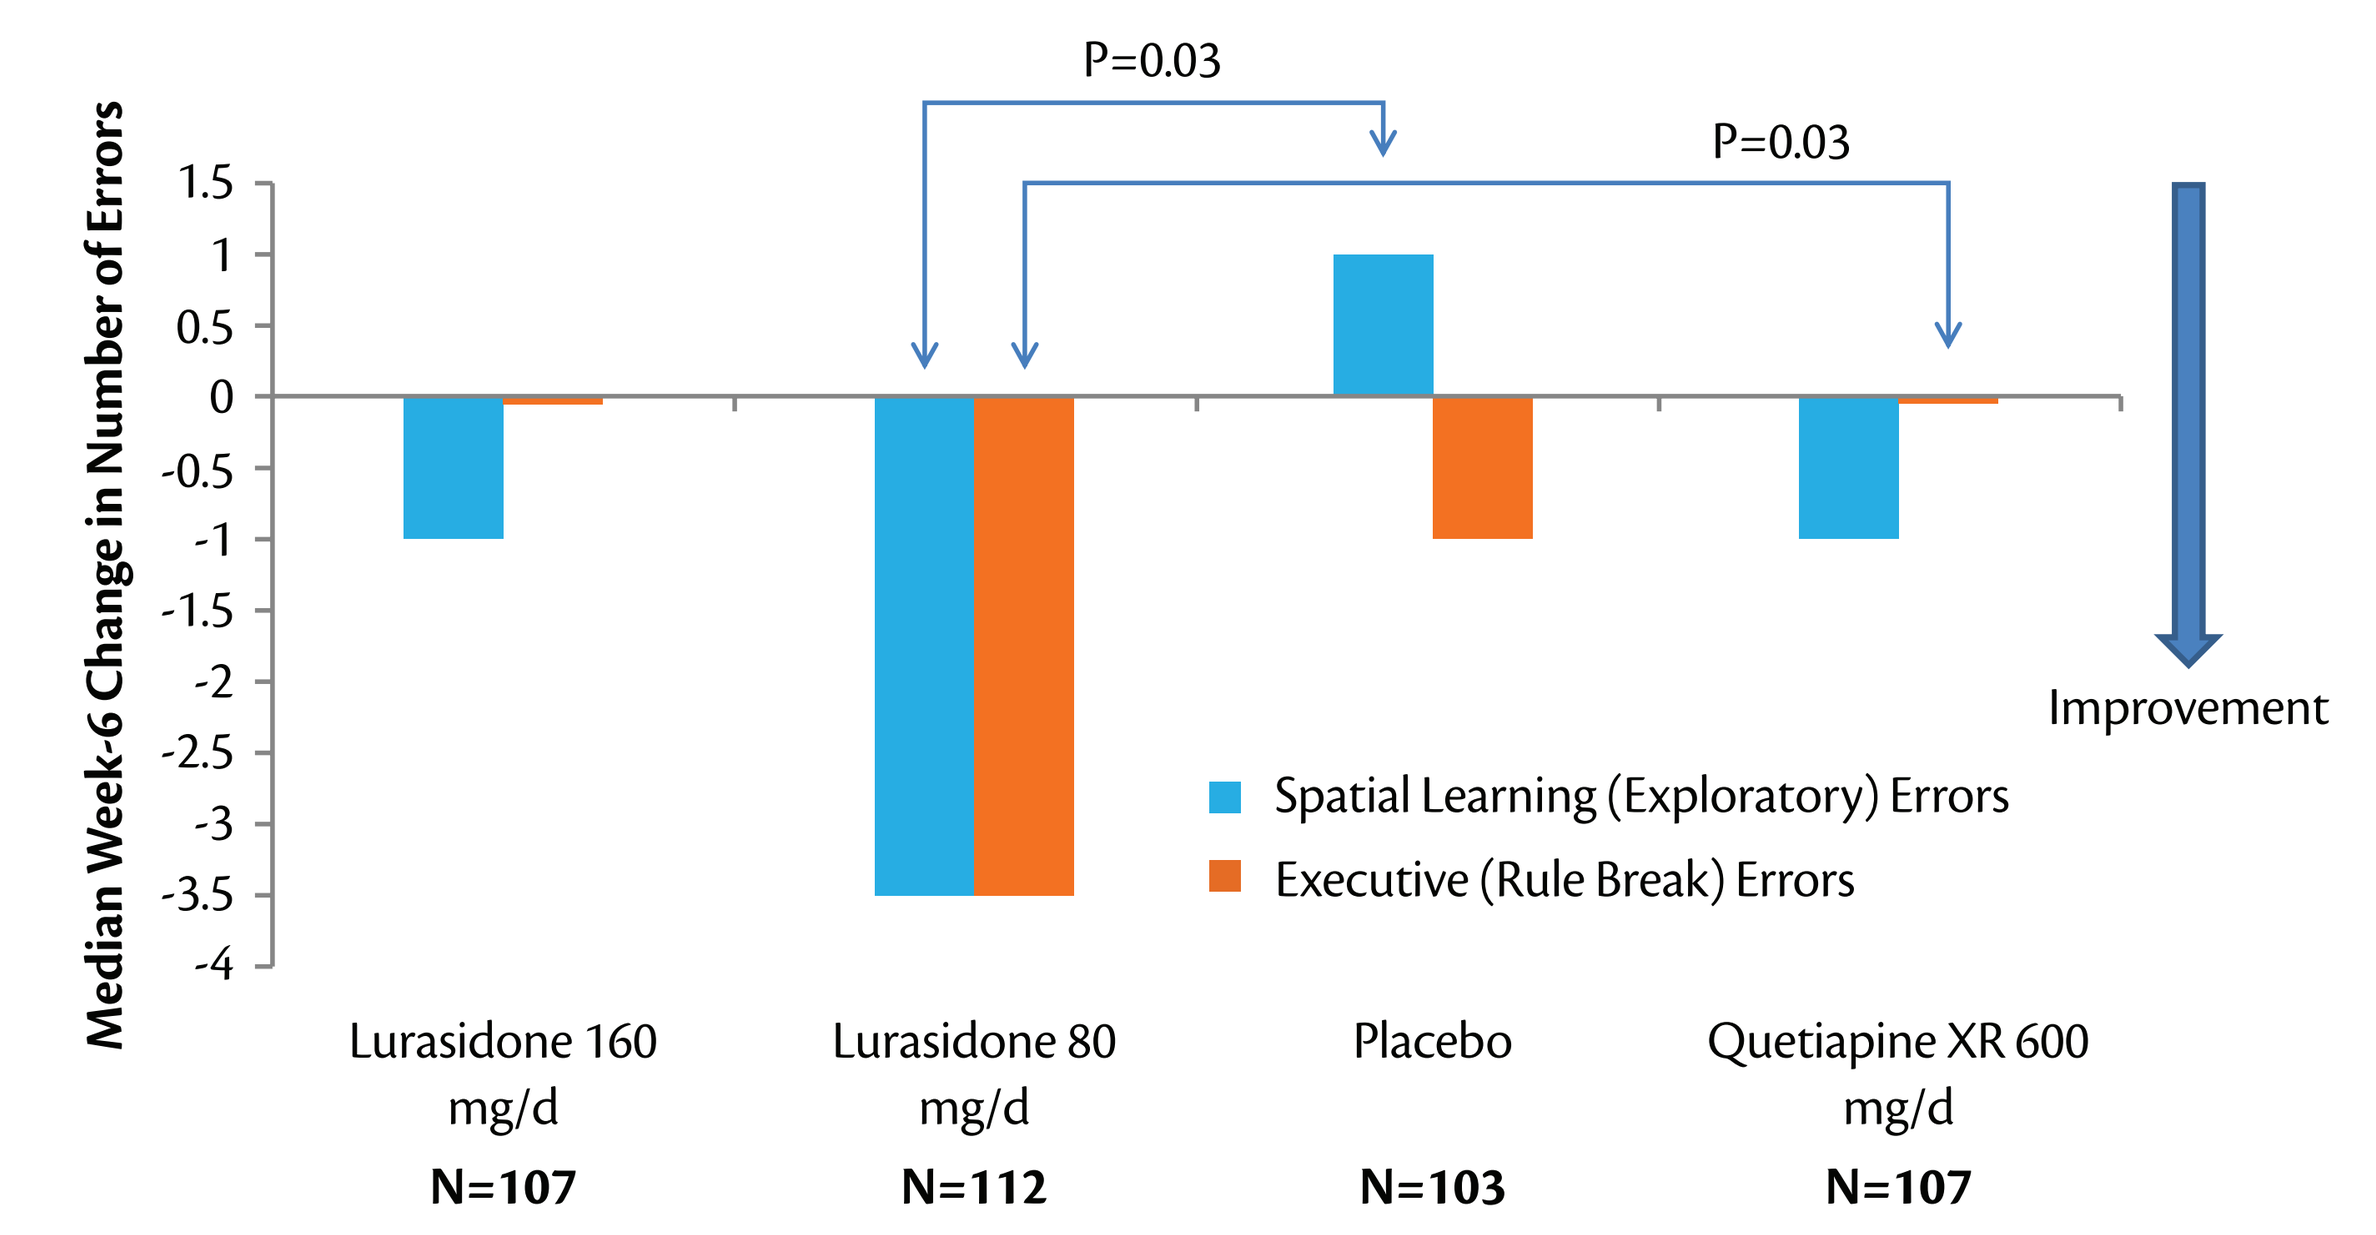
P=0.03
P=0.03
Median Week-6 Change in Number of Errors
1.5
1
0.5
0
-0.5
-1
-1.5
-2
-2.5
-3
-3.5
-4
Improvement
Spatial Learning (Exploratory) Errors
Executive (Rule Break) Errors
Lurasidone 160 mg/d
Lurasidone 80 mg/d
Placebo
Quetiapine XR 600 mg/d
N=107
N=112
N=103
N=107

Figure 6. Distribution of Evaluable Domain Tasks (Evaluable Analysis Sample)

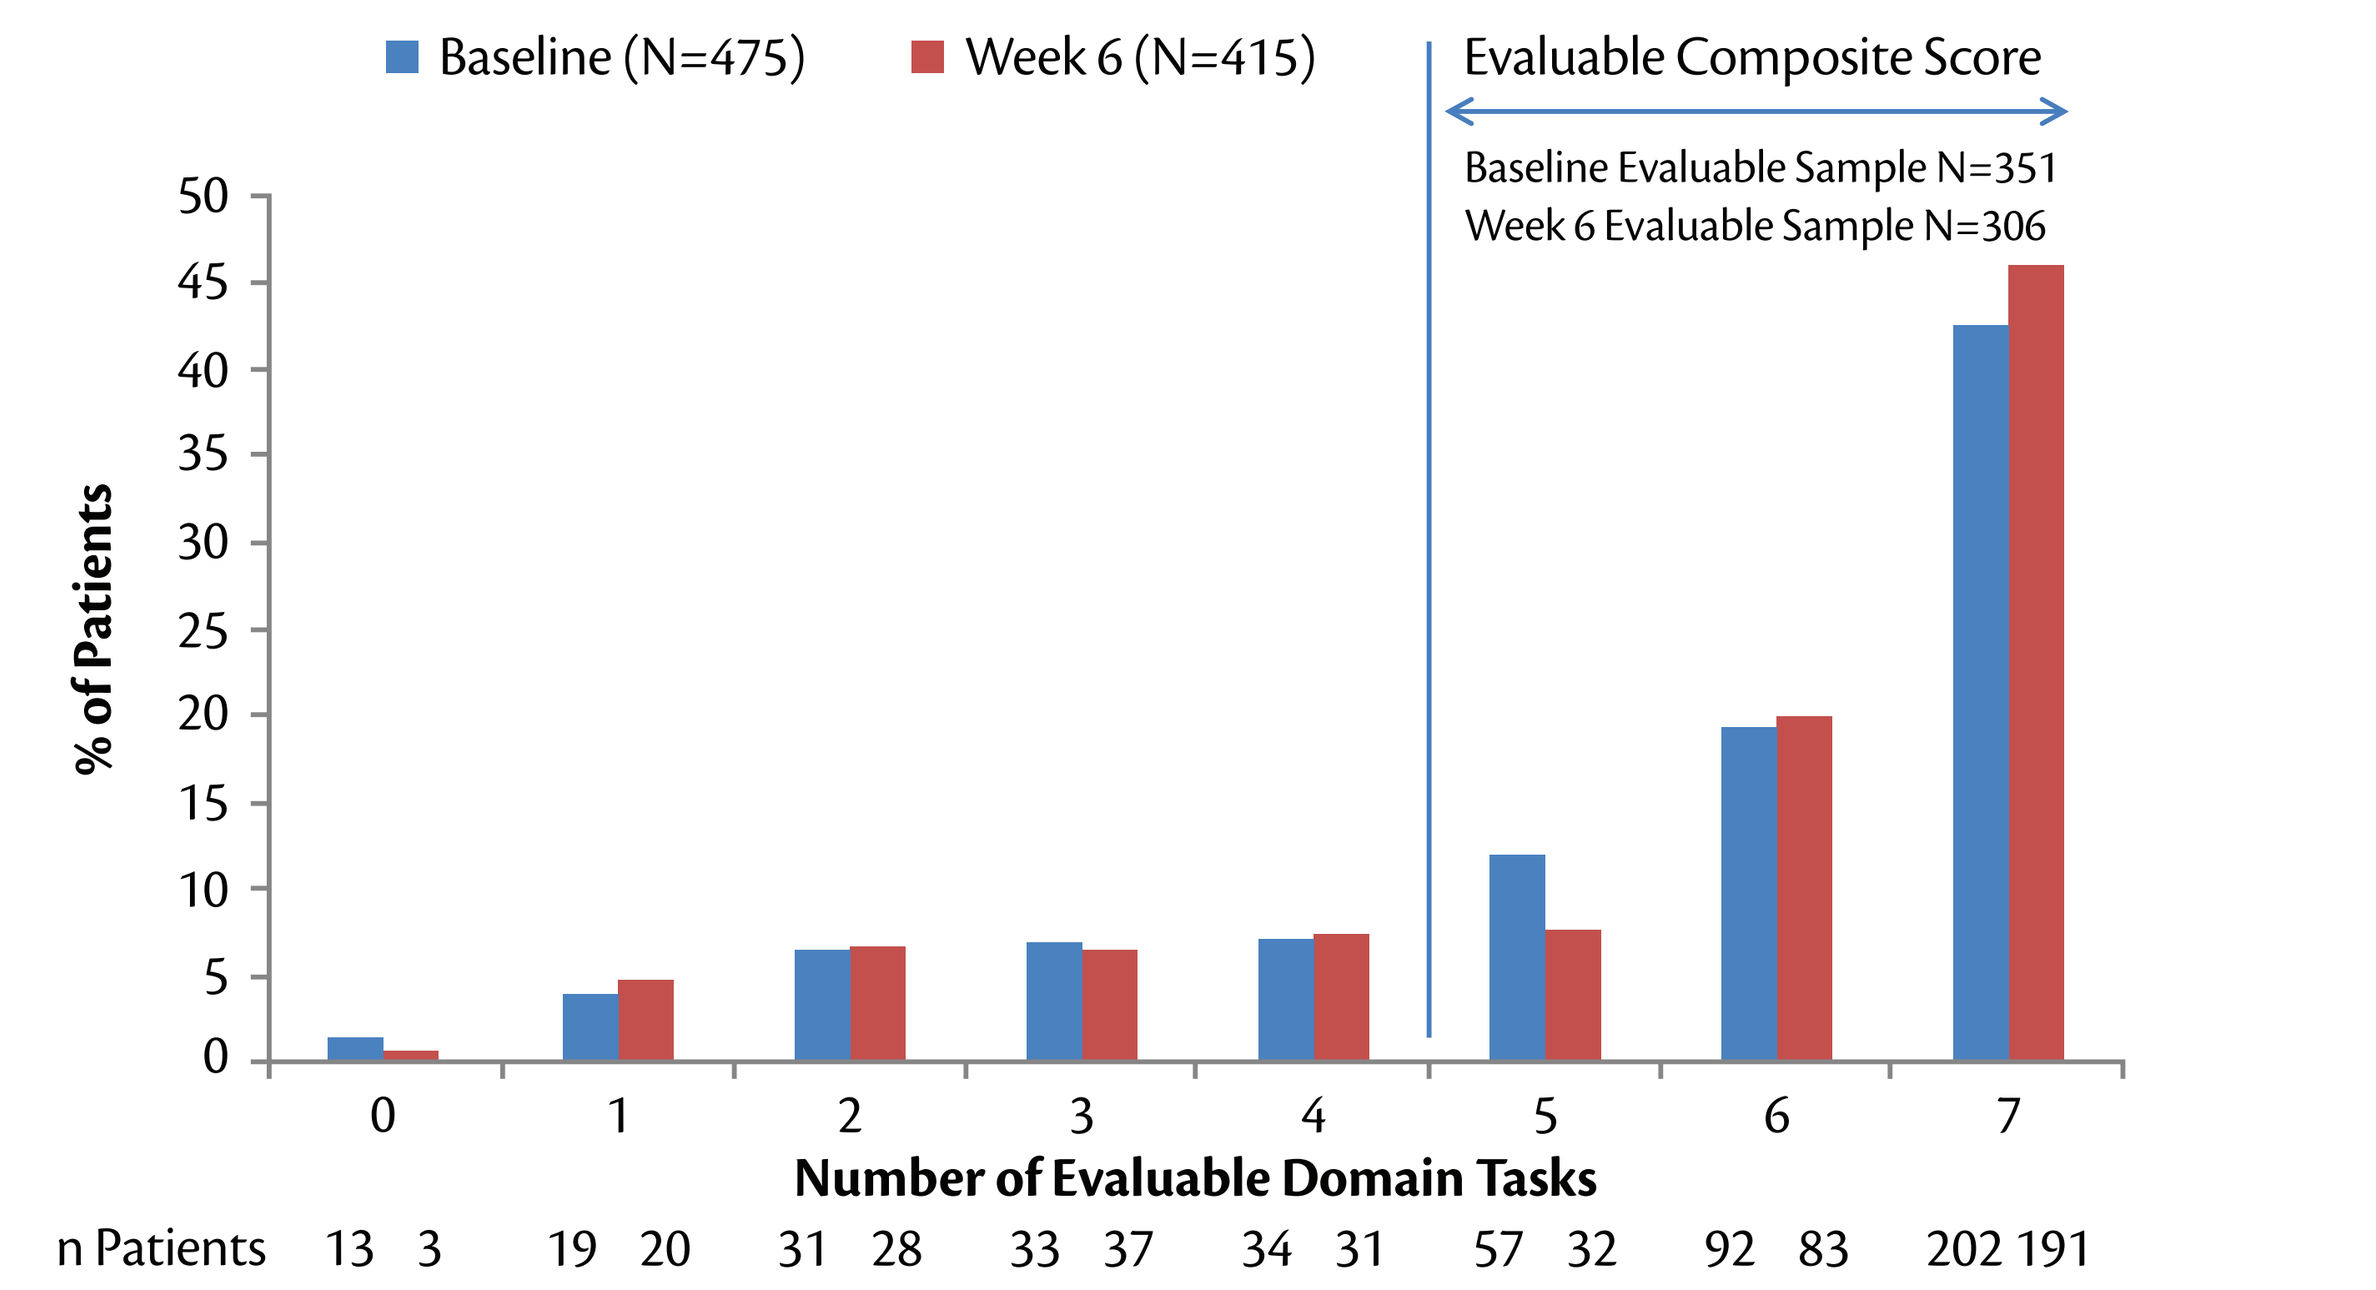
Baseline (N=475)
Week 6 (N=415)
Evaluable Composite Score
Baseline Evaluable Sample N=351
Week 6 Evaluable Sample N=306
% of Patients
50
45
40
35
30
25
20
15
10
5
0
0
1
2
3
4
5
6
7
Number of Evaluable Domain Tasks
n Patients 13 3 19 20 31 28 33 37 34 31 57 32 92 83 202 191

DISCUSSION

In this double-blind, placebo- and active-controlled trial of subjects with an acute exacerbation of schizophrenia, patients treated with lurasidone 160 mg/d demonstrated improved cognition compared to placebo in the full ITT sample, while both executive (rule break) and spatial learning (exploratory) errors were significantly improved in the lurasidone 80 mg/d group compared to the quetiapine XR and the placebo groups, respectively.

In this post-hoc analysis, a nonparametric approach was employed to permit the inclusion of patients with significant cognitive impairment at the time of testing due to severe psychiatric illness. Consistent results were observed when compared to a subsample of evaluable patients as reported in previous study (Harvey et al., 2013).

Informed by this experience, the computerized Cogstate Schizophrenia Battery was modified to permit rest periods if needed, as is routine with paper and pencil cognitive testing, and the test sequence in the was altered. Using this modified presentation of the identical tests, Citrome et al (2016) achieved 100% evaluable data on the Cogstate Schizophrenia Battery in an acute schizophrenia sample.

REFERENCES

1. Koch, G. G., Carr, G. J., Amara, I. A., Stokes, M. E., and Uryniak T. J. (1990), "Categorical Data Analysis," in Berry, D. A. ed., Statistical Methodology in Pharmaceutical Sciences, New York: Marcel Dekker, 291–475.
2. Citrome L, Ota A, Nagamizu K, Perry P, Weiller E, Baker RA. HYPERLINK "http://www.ncbi.nlm.nih.gov/pubmed/26963842"The effect of brexpiprazole (OPC-34712) and aripiprazole in adult patients with acute schizophrenia: results from a randomized, exploratory study. Int Clin Psychopharmacol. 2016 Jul;31(4):192-201. doi: 10.1097/YIC.0000000000000123.
3. Harvey PD, Siu CO, Hsu J, Cucchiaro J, Maruff P, Loebel A. HYPERLINK "http://www.ncbi.nlm.nih.gov/pubmed/24035633"Effect of lurasidone on neurocognitive performance in patients with schizophrenia: a short-term placebo- and active-controlled study followed by a 6-month double-blind extension. Eur Neuropsychopharmacol. 2013 Nov;23(11):1373-82. doi: 10.1016/j.euroneuro.2013.08.003. Epub 2013 Aug 27.

DISCLOSURES

Dr. Jaeger provided scientific consulting to Axsome, Boeringer-Ingelheim, Biogen, Eisai, Lundbeck, Pfizer, Sunovion, Teva and Takeda. While previously employed by Cogstate, she currently has no financial interest in Cogstate. Dr. Siu consults to Sunovion and Pfizer. Drs. Tocco, Goldman and Loebel are employees of Sunovion.