Digital Twin Simulation for Longevity Trials: A Multi-Domain Monte Carlo Modeling Framework for CNS and Aging Outcomes

Lakhan SE, Rice JS

Boricua Bio, San Juan, PR, USA

Methodological Issue Being Addressed

Simulation based trial design offers a way to study longevity interventions without requiring multi year or multi decade follow up. Aging is multidimensional and varies across individuals, which introduces feasibility and power challenges for CNS relevant outcomes such as cognitive decline, sleep deterioration, and loss of physiologic resilience. Digital twins allow investigators to generate individualized computational trajectories and compare counterfactual outcomes for treated and untreated virtual participants. This improves endpoint selection, effect size estimation, and sample size planning before conducting a physical trial.

Introduction

Longevity interventions target interacting biologic domains such as metabolism, vascular aging, sleep, activity, cognition, and inflammatory burden. Conventional trials are limited by slow outcome emergence and high variability. Digital twins provide a computational approach to model aging trajectories using baseline characteristics, clinical history, and digital biomarkers. By simulating treatment effects across large virtual cohorts, investigators can anticipate signal detection windows, refine primary and secondary outcomes, and identify high yield subgroups for future CNS oriented trials. This submission describes a digital twin framework designed to support the planning of simulation based longevity studies.

Figure 1. Digital Twin Simulation Architecture

Integrated framework for longitudinal longevity trials. (Left) Multi-domain data ingestion (CNS, physiology, sleep) to create patient-specific digital blueprints. (Center) Monte Carlo engine (10k iterations) generating parallel counterfactual trajectories for treated vs. untreated twins. (Right) Strategic outputs including power projections and bio-signal sensitivity to optimize trial design and reduce sample size.

Methodology

A multi domain digital twin model was developed using published epidemiologic and aging data sets including NHANES, UK Biobank, and ADNI. Each twin incorporated demographic variables, metabolic markers, blood pressure, sleep continuity, heart rate variability, activity regularity, mood indices, and cognitive screening scores. Probabilistic transition matrices estimated year over year changes in risk for cognitive decline, functional loss, and accelerated biological aging. Interventional inputs were drawn from published literature on metabolic modulators, rapalog pathways, behavioral programs, and digital therapeutic components. Monte Carlo simulation followed 10,000 virtual participants for up to 10 simulated years. Outcomes included biological age trajectories, incident mild cognitive impairment, composite resilience indices, and expected treatment effect sizes. Sensitivity analyses examined the impact of digital biomarkers on early signal detection and sample size estimates.

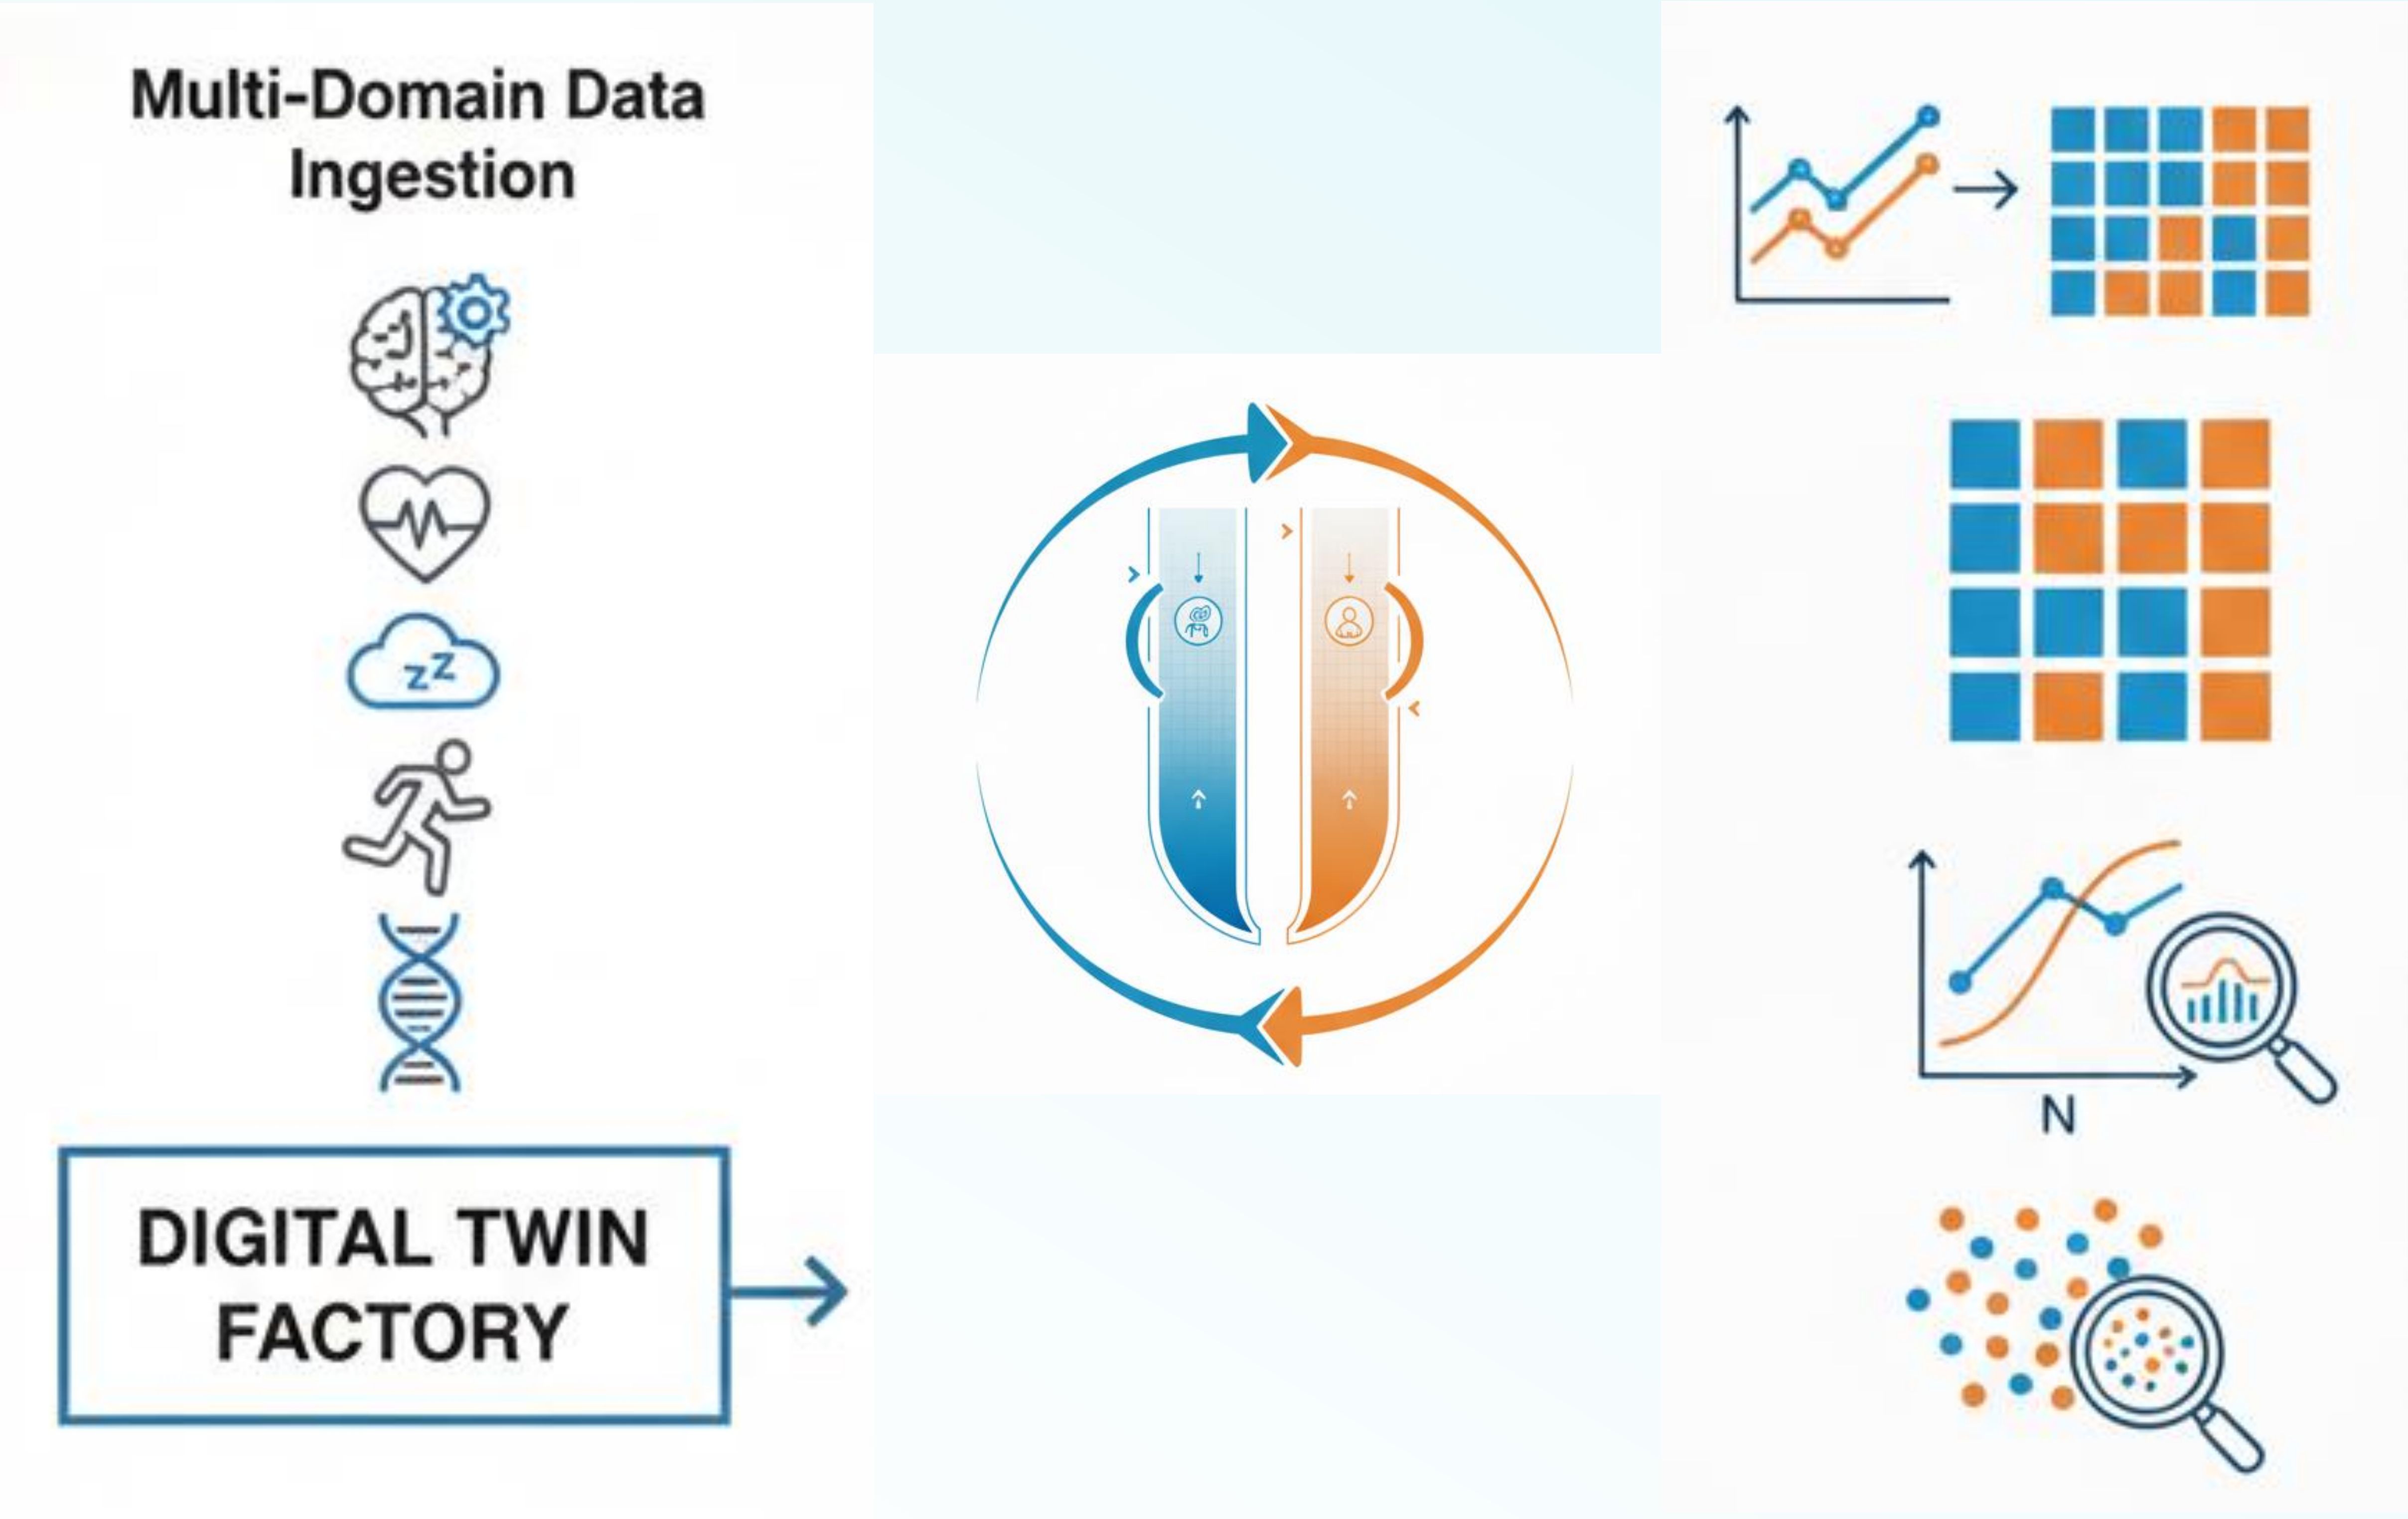
Multi-Domain Data Ingestion
DIGITAL TWIN FACTORY
N

Results

Simulations showed that multimodal interventions produced larger projected effects than single domain approaches. Biological age deceleration was detectable within 12 to 24 simulated months, cognitive decline effects within 3 to 4 years, and mortality related endpoints within approximately 5 years. Including digital biomarkers such as sleep variability and activity regularity improved early signal detection and reduced projected sample sizes by 25 to 40 percent. Counterfactual comparisons between treated and untreated twins identified subgroups with higher responsiveness, particularly those with elevated metabolic or sleep related risk at baseline. Uncertainty narrowed when digital biomarkers were incorporated into the model, suggesting improved stability of effect estimation.

Conclusion

Digital twin simulation is a practical approach for planning longevity trials when traditional designs are limited by long time horizons and high variability. This framework supports feasibility assessment, endpoint prioritization, and effect size estimation for CNS related aging outcomes. Incorporating digital biomarkers strengthens early detection and improves modeling precision. Simulation based approaches may help accelerate the development of interventions intended to influence aging and resilience across multiple physiologic domains.