Evaluating the Relationship Between Esketamine Monotherapy Efficacy and Dissociation in Patients With Treatment-Resistant Depression

Dionne Williams,[1,2*] Dong-Jing Fu,[1] Cynthia Bossie,[1]
Xin Qiu,[1] Abi Bangerter[1] and Carla Canuso[1]

[1]Johnson & Johnson, Titusville, NJ, USA.
[2]Drexel University College of Medicine, Philadelphia, PA, USA.

*Presenting author

Background

- A new generation of antidepressants including esketamine (an approved antidepressant), ketamine and psychedelics (both investigational) have unique side effects that can lead to functional unblinding.[1]
- Previous studies have reported no significant correlation between dissociative effects as measured by the Clinician-Administered Dissociation States Scale (CADSS) and antidepressive response to esketamine used in conjunction with an oral antidepressant.[2]
- The relationship between the antidepressant efficacy and dissociation effects of esketamine monotherapy in patients with treatment-resistant depression (TRD) is yet to be evaluated.

Objectives

- To determine the response rates, the number of treatment-emergent adverse events (TEAE) of dissociation for esketamine and placebo treatment groups and assess whether the efficacy of esketamine is associated with its dissociative effects.

Methods

Study Design

- Data were collected from a randomized, double-blind (DB), placebo-controlled study (NCT04599855) that evaluated the efficacy of 56 mg and 84 mg esketamine nasal spray as a monotherapy versus matching placebo nasal spray in adults with TRD.

Assessments

- Response status at DB endpoint (Day 28): response was defined as ≥50% reduction from baseline in Montgomery and Asberg Depression Rating Scale (MADRS) total score.
- TEAEs of dissociation on dosing days during the DB phase: defined as patient-reported TEAE of dissociation.

Statistical Analysis

- Response rates at DB endpoint were compared in participants with versus without reported TEAE of dissociation using Fisher's exact test.

Results

- Participants in the esketamine treatment groups had higher response rates than those in the placebo group.

Table 1. Response rates based on MADRS total score at the end of DB phase

	Placebo	Esketamine 56 mg	Esketamine 84 mg
Endpoint (DB)			
N	197	85	95
Response (≥50% improvement), n (%)	30 (15.2)	26 (30.6)	27 (28.4)
Non-response (<50% improvement), n (%)	167 (84.8)	59 (69.4)	68 (71.6)

DB, double-blind; MADRS, Montgomery and Asberg Depression Rating Scale.

- Higher proportion of participants in the esketamine treatment groups exhibited dissociation compared to the placebo group.

Table 2. Proportion of participants with TEAE of dissociation by treatment groups

	Placebo	Esketamine 56 mg	Esketamine 84 mg
Dissociation, n (%)	7 (2.8)	23 (21.9)	55 (26.4)

TEAE, treatment-emergent adverse events.

- There is no significant difference in response rates among participants with and without dissociation for the two esketamine treatment groups.

Table 3. Response rates among participants with and without dissociation by treatment groups

	Dissociation	No dissociation
Placebo		
N	7	190
Response, n (%)	3 (42.9)	27 (14.2)
Fisher's Exact test		P=0.073
Esketamine 56 mg		
N	19	66
Response, n (%)	6 (31.6)	20 (30.3)
Fisher's Exact test		P=1.000
Esketamine 84 mg		
N	23	72
Response, n (%)	7 (30.4)	20 (27.8)
Fisher's Exact test		P=0.796

Key Takeaway

These data suggest that dissociation does not account for the efficacy observed in participants receiving esketamine monotherapy.

Discussion and Conclusions

- At the DB endpoint, a two-fold increase in the overall response rates was observed in the esketamine 56 mg and 84 mg (30.6% and 28.4%, respectively) treatment groups versus placebo (15.2%).
- Higher incidence of dissociation was observed for esketamine 56 mg and 84 mg (21.9% and 26.4%, respectively) compared to placebo (2.8%).
- Similar response rates were observed between participants with (31.6% and 30.4%) and without dissociation (30.3% and 27.8%) in the esketamine 56 and 84 mg groups, respectively, at DB endpoint.
- Although there are higher response rates in the placebo group (42.9%) in participants that experienced dissociation than those that did not experience dissociation (14.2%), the interpretation of the results for the placebo group with dissociation are confounded by the low N (N=7).
- These data are consistent with findings from Chen et al. 2022 showing no difference in response rates among TRD patients with and without dissociation.[2]

Limitations

- Dissociation measurement based on CADSS scores, as demonstrated in Chen et al. 2022, was not included in this analysis.[2]
- This post-hoc analysis used dissociation data collected through TEAE reports only, which may have resulted in an underestimation of dissociative effects.

Acknowledgements

Editorial support was provided by Sonali Satam, PhD, and graphic designing support was provided by Sandeep Chavan (both Siro Medical Writing Pvt. Ltd., India).

Funding

This study was funded by Johnson & Johnson.

Disclosures

All authors are employees of Johnson & Johnson.

Novel Pathways in Depression

Presented at the International Society for CNS Clinical Trials and Methodology (ISCTM) 2025 – 21st Annual Scientific Meeting, February 19-21, 2025, Washington DC, USA.

REFERENCES:

1. Ballard, E.D., et al. (2020). *Nature Communications*, 11. https://doi.org/10.1038/s41467-020-20190-4.
2. Chen, G., et al. (2022). *International Journal of Neuropsychopharmacology*, 25(4), 69–279. https://doi.org/10.1093/ijnp/pyab084.