Presented at the 21st Annual ISCTM Scientific Meeting, Feb 19 - 21, 2025, Washington, D.C.

Examining patterns of commonly observed quality issues across global schizophrenia clinical trial research sites

Martina Micaletto, Andrei Iacob, Juliet Brown, Alan Kott, David Daniel
Signant Health

BACKGROUND

Schizophrenia has been a particularly challenging area of therapeutic treatment development. Placebo-drug differences have diminished (Leucht et al. 2019), and hallmarked by complex primary outcome scales such as the PANSS (Opler, Yavorsky and Daniel, 2017) that may be particularly susceptible to confounding factors, such as rater change (Kott, Brannan, Wang and Daniel, 2017). Understanding common scale quality issues in schizophrenia clinical trials can enhance data validity and reliability, supporting confident “go/no go” decisions on novel clinical therapies.

In 2017, an ISCTM working group[1] investigated the frequency of consistency flags for the PANSS across two global databases to identify prevalent patterns (Rabinowitz et al, 2017). The results offered a novel perspective on remediation strategies to improve data quality in clinical trials. We sought to build on this pivotal work by focusing on higher level data quality patterns that could be identified by either statistical and clinical algorithms or CQR conclusions. We also sought to expand the methodology of our analysis to other commonly used scales in schizophrenia clinical trials.

We aimed to identify and characterize the most frequently observed quality issues across global schizophrenia clinical trials, including both prior known “flag” type findings and new categories of operational inconsistencies, participant eligibility concerns, and more powerful variability metrics.

[1]Algorithms/Flags to Identify Clinical Inconsistency in the Use of Rating Scales in CNS RCTs Working Group

METHODOLOGY

We retrospectively analyzed data from SCID-5-CT, PANSS, PSP, CGI-S, and CGI-I obtained from 21 global phase 2/3 schizophrenia clinical trials conducted at 238 research sites between 2020-2024. Analysis was restricted to sites with minimum 5 randomized patients, across Bulgaria, China, Croatia, Czech Republic, Georgia, Hungary, Japan, Moldova, Poland, Romania, Russia, Serbia, Spain, Ukraine and United States.

The analysis measured quality metrics “actual hits” over “possible hits”. That is, visit or scale-level data where a quality finding occurred over visit or scale-level data where it may have occurred but did not. Figures are reported as % calculated by (actual hits/possible hits) x 100.

Table 1 shows quality meta-categories with individual metrics, possible and actual hit totals. Considering the variety and complexity of scales utilized in schizophrenia clinical trials, and the evidence from the ISCTM working group, we hypothesize Category 1 and 2 would have the highest percentage of hits.

DISCUSSION

The presence of subjects with inclusionary concerns signalled by scale ratings or a second clinical opinion suggests third party eligibility reviews and data monitoring would prove beneficial in ensuring appropriate patient ascertainment in upholding rigorous enrollment gatekeeping. Especially considering that around 44% of subjects hits for the Inclusionary concerns were randomized after Tier 2 clinical review.

Literature also shows that operational consistency factors can introduce considerable noise into study data, jeopardising study success. Rater training highlighting the deleterious effects of rater variability, suboptimal administrations practices and offering evidence-based best practices (with tips for complex cases and ratings) has been seen to mitigate such noise (Verma et al 2020).

Contrary to our hypothesis, category 1 and 2 were not the most prominent data quality concerns in this dataset. These still represent important quality indicators that should be carefully considered and mitigated where possible to support optimal signal detection (Kott et al. 2021).

Overall, we mapped out a “bird’s eye view” of the prominent data quality patterns in recent schizophrenia clinical trials. These findings highlight the importance of designing robust clinical trials data monitoring methodologies (such as enhanced eligibility surveillance, robust analytics or co-ratings) to mitigate these issues.

We plan subsequent granular analyses of individual concerns.

TABLE 1: QUALITY META-CATEGORY COMPONENTS AND POSSIBLE VS. ACTUAL HIT METRICS.

The bolded are statistically-derived findings or meta-data. Italic items represent CQR-derived findings. Underlined items represent a combination of both.

Category	Meta-category	Metrics evaluated	Possible hits	Actual hits
1	Administration quality and rating accuracy	• *Administration errors* • *Scoring errors* • **Between-scale item rating discordance** • **Within-scale rating discordance** • **Unexpected values** • *Rater to CQR rating discordance*	495,200	2,0672
2	Data variability	• **High data variability** • **Low data variability** • **Multivariate inliers**	165,527	10,582
3	Operational Inconsistency	• **Interview duration** • **Rater change**	12,488	115,915
4	Participant Eligibility	• Inclusionary concerns (symptom acuity, diagnosis, inclusionary scale ratings)	32,111	3,453

FIGURE 1: % INCIDENCE OF QUALITY META-CATEGORIES

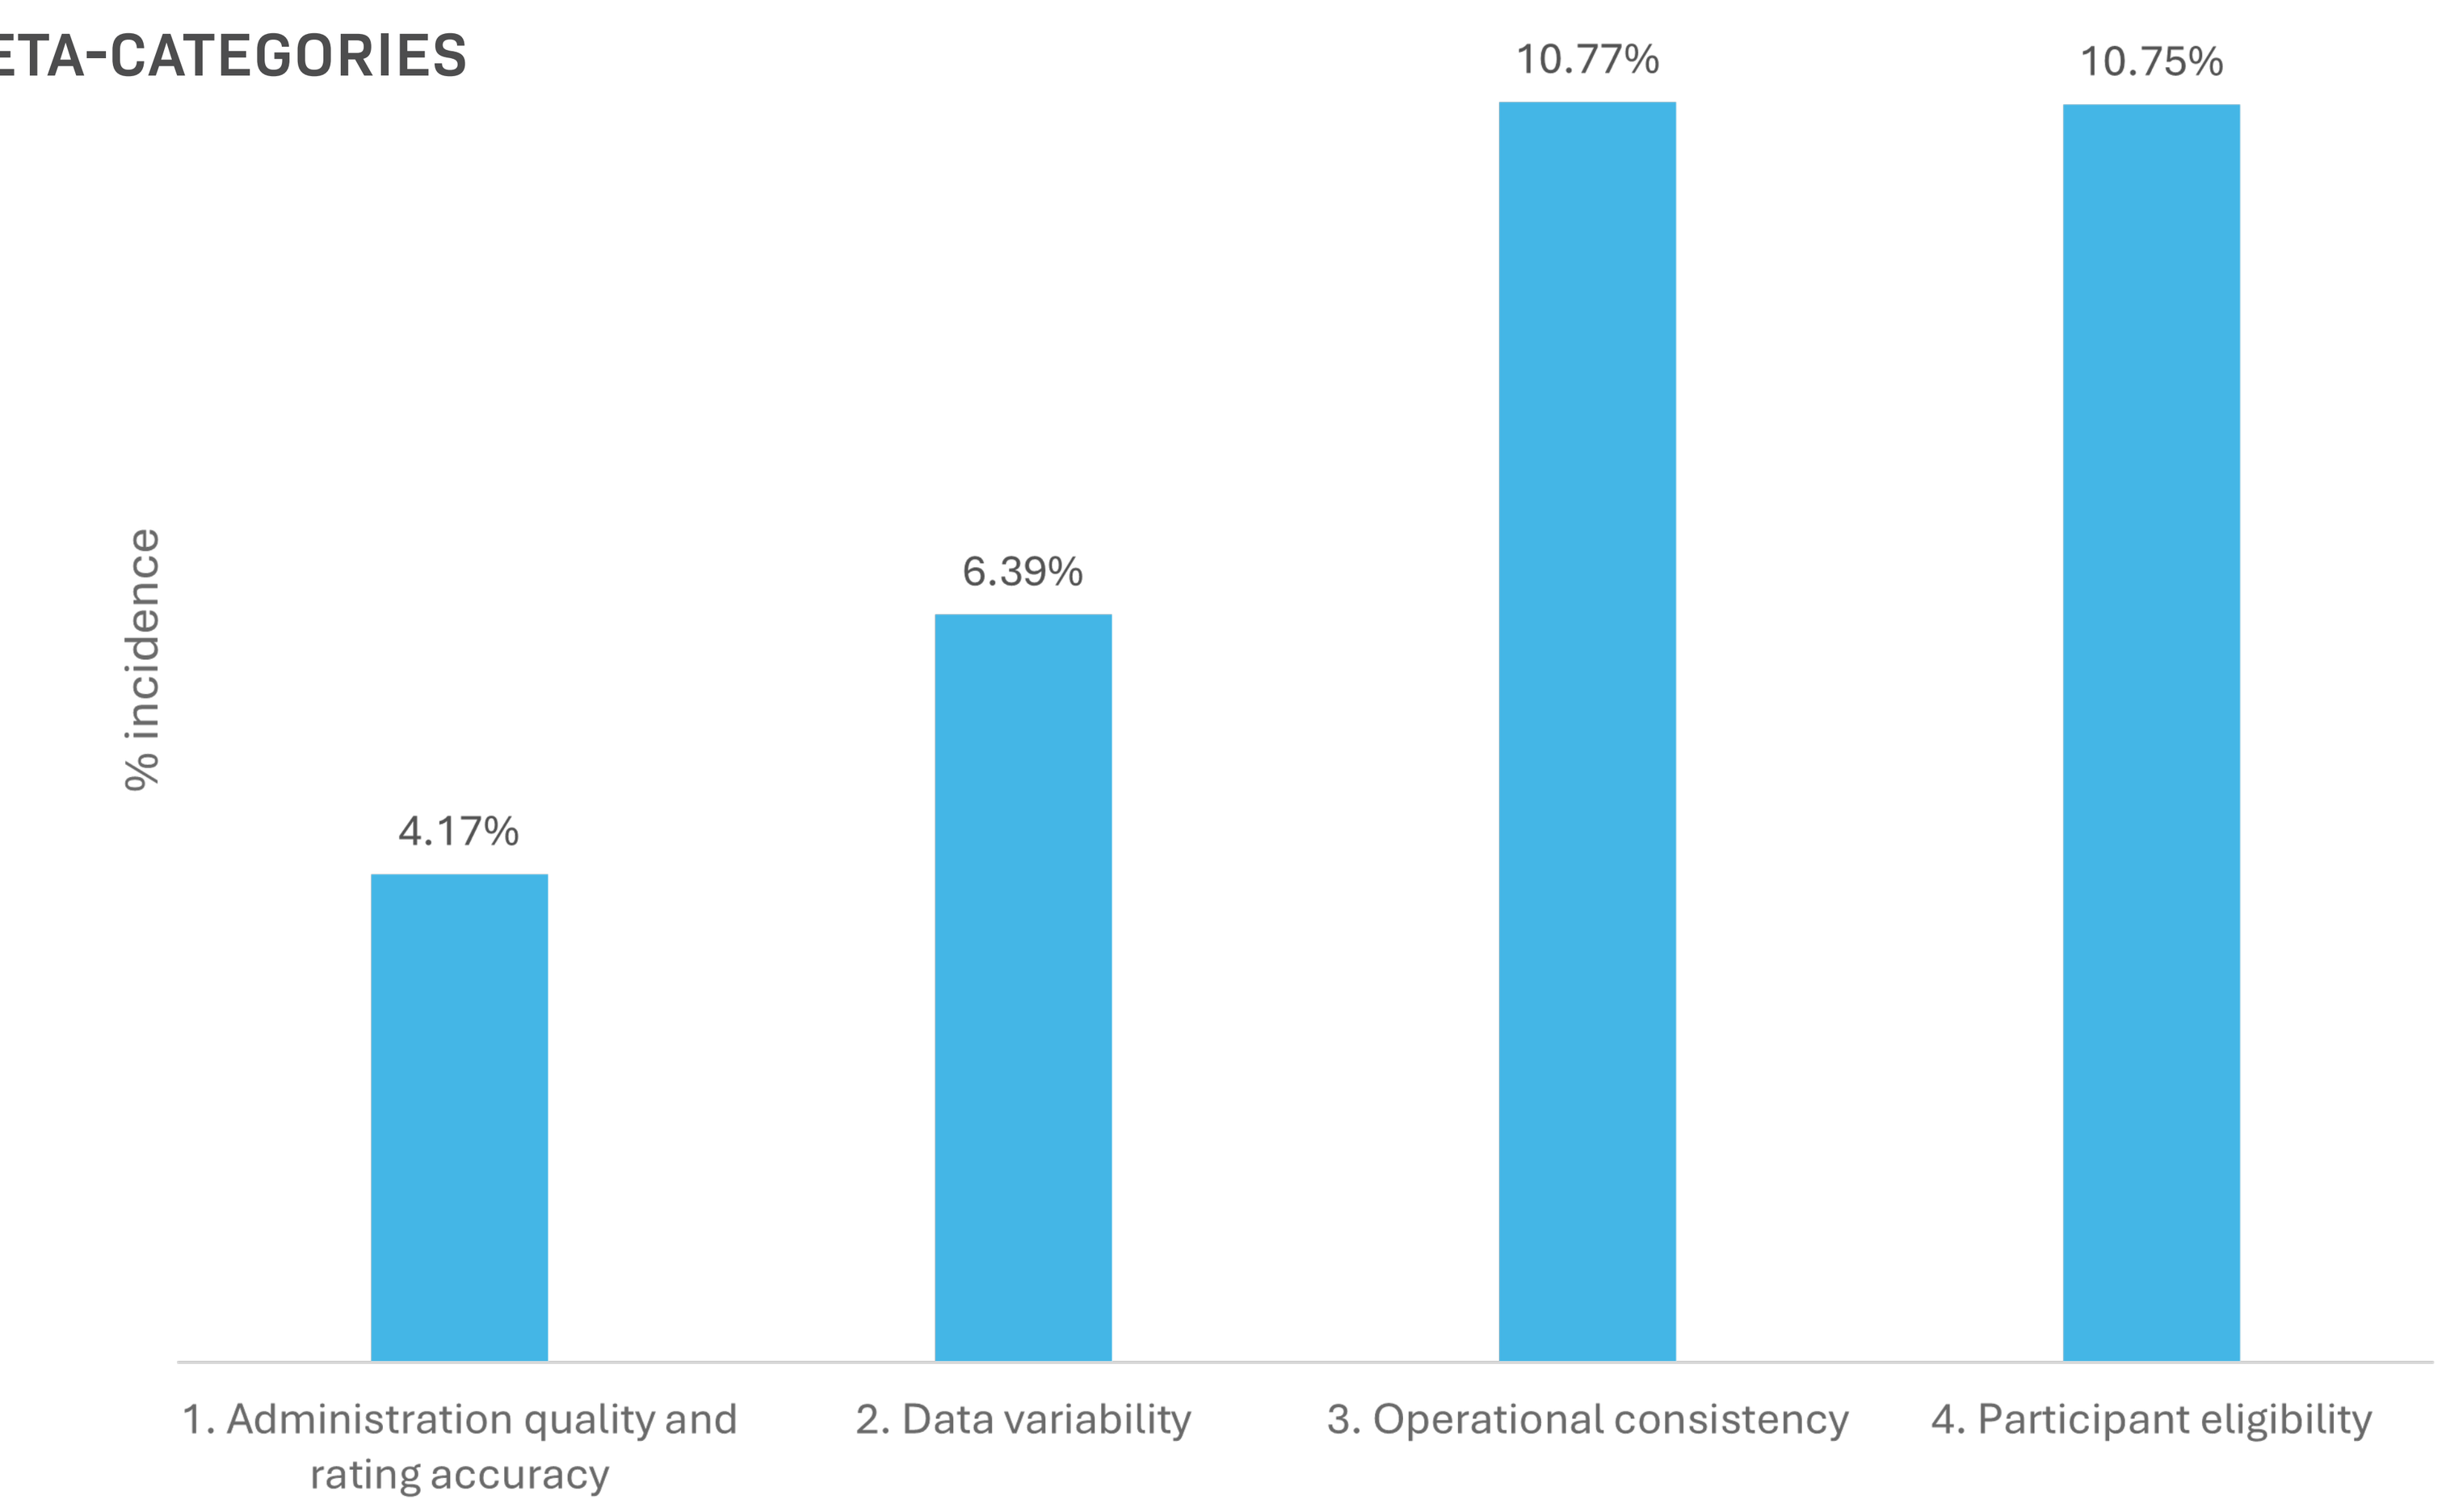
% incidence
4.17%
6.39%
10.77%
10.75%
1. Administration quality and rating accuracy
2. Data variability
3. Operational consistency
4. Participant eligibility

REFERENCES

Kott, A., Brannan, S., Wang, X., & Daniel, D. (2021). The impact of aberrant data variability on drug–placebo separation and drug/placebo response in an acute schizophrenia clinical trial. Schizophrenia Bulletin Open, 2(1), sgab037.

Leucht, S., Chaimani, A., Mavridis, D., Leucht, C., Huhn, M., Helfer, B., ... & Davis, J. M. (2019). Disconnection of drug-response and placebo-response in acute-phase antipsychotic drug trials on schizophrenia? Meta-regression analysis. Neuropsychopharmacology, 44(11), 1955-1966.

Opler, M. G., Yavorsky, C., & Daniel, D. G. (2017). Positive and negative syndrome scale (PANSS) training: challenges, solutions, and future directions. Innovations in clinical neuroscience, 14(11-12), 77.

Rabinowitz, J., Schooler, N. R., Anderson, A., Ayearst, L., Daniel, D., Davidson, M., ... & Zhao, J. (2017). Consistency checks to improve measurement with the Positive and Negative Syndrome Scale (PANSS). Schizophrenia research, 190, 74-76.

Verma P., Micaletto, M., Laird, R., Hong, B., Feaster, T.,Tackett, ZC., Machizawa, S., Zhou, W., Crittenden-Ward, K., Daniel, DG., Kott, A. (2020) Evaluating the Impact of Rater Change on PANSS Score Change: An Item Level Analysis. Presented at the ISCTM 2020 Autumn Virtual Conference, September 21 - 25, 2020.

SIGNANT HEALTH
THE EVIDENCE GENERATION COMPANY™