Presented at the 21st Annual ISCTM Scientific Meeting, Feb 19 - 21, 2025, Washington, D.C.

The impact of PANSS change in screening on week 1 PANSS change in acute schizophrenia clinical trials

Kott, Alan; Wang, Xingmei; Daniel, David Gordon
Signant Health

METHODOLOGICAL QUESTION

Does the PANSS change in the screening period impact week 1 PANSS change in blinded data in acute schizophrenia clinical trials?

INTRODUCTION

Most clinical trials in acute schizophrenia limit the participation of subjects who improve by more than a set cut-off (typically 20%) between screening and baseline.

Anecdotally, clinical trial sponsors are concerned that subjects showing larger change in the screening period may ultimately have larger placebo response and/or diminished placebo-drug separation.

In the current analysis, we assessed the impact of the screening change (PANSS total, factor scores) on the PANSS change at the week 1 visit after randomization.

METHODS

- PANSS data were pulled from 14 double blind, placebo controlled, multicenter, phase 2/3 clinical trials in acute schizophrenia.
- A total of 6,232 subjects were included in the analysis.
- Of these, 5,721 (91.8%) had data for the week 1 post-baseline visit.
- Percent change in the screening period (difference between baseline and screening value) was calculated for the PANSS total and all 5 PANSS factors using the formula subtracting the minimal possible score from the denominator.
- Linear mixed effect models were fitted to the data, with the PANSS percent change at week 1 as the dependent variable and the respective screening change (PANSS total, PANSS factors) and PANSS baseline value included in the model as fixed effects and study as a random effect.
- Linear mixed effect models were used to assess the difference in in the PANSS change in the screening period between the randomized and screen-failed study participants.

RESULTS (FIGURE 1)

- No difference in the PANSS total score change in the screening period was identified between those study participants who randomized and who screen-failed.
- A modest, but statistically significant effect of the PANSS total score change in the screening period on Week 1 PANSS total score change was observed.
- A 10% increase in PANSS improvement in the screening period was associated with an approximately 0.7% decrease in PANSS improvement at week 1 ($p = 0.0014$).
- None of the PANSS factor score changes in the screening period significantly impacted the week 1 PANSS total score change.
- However, a modest, but non-statistically significant effect on the week 1 PANSS total score change was observed for the positive factor score and to a lesser extent the negative and disorganized thought factors.

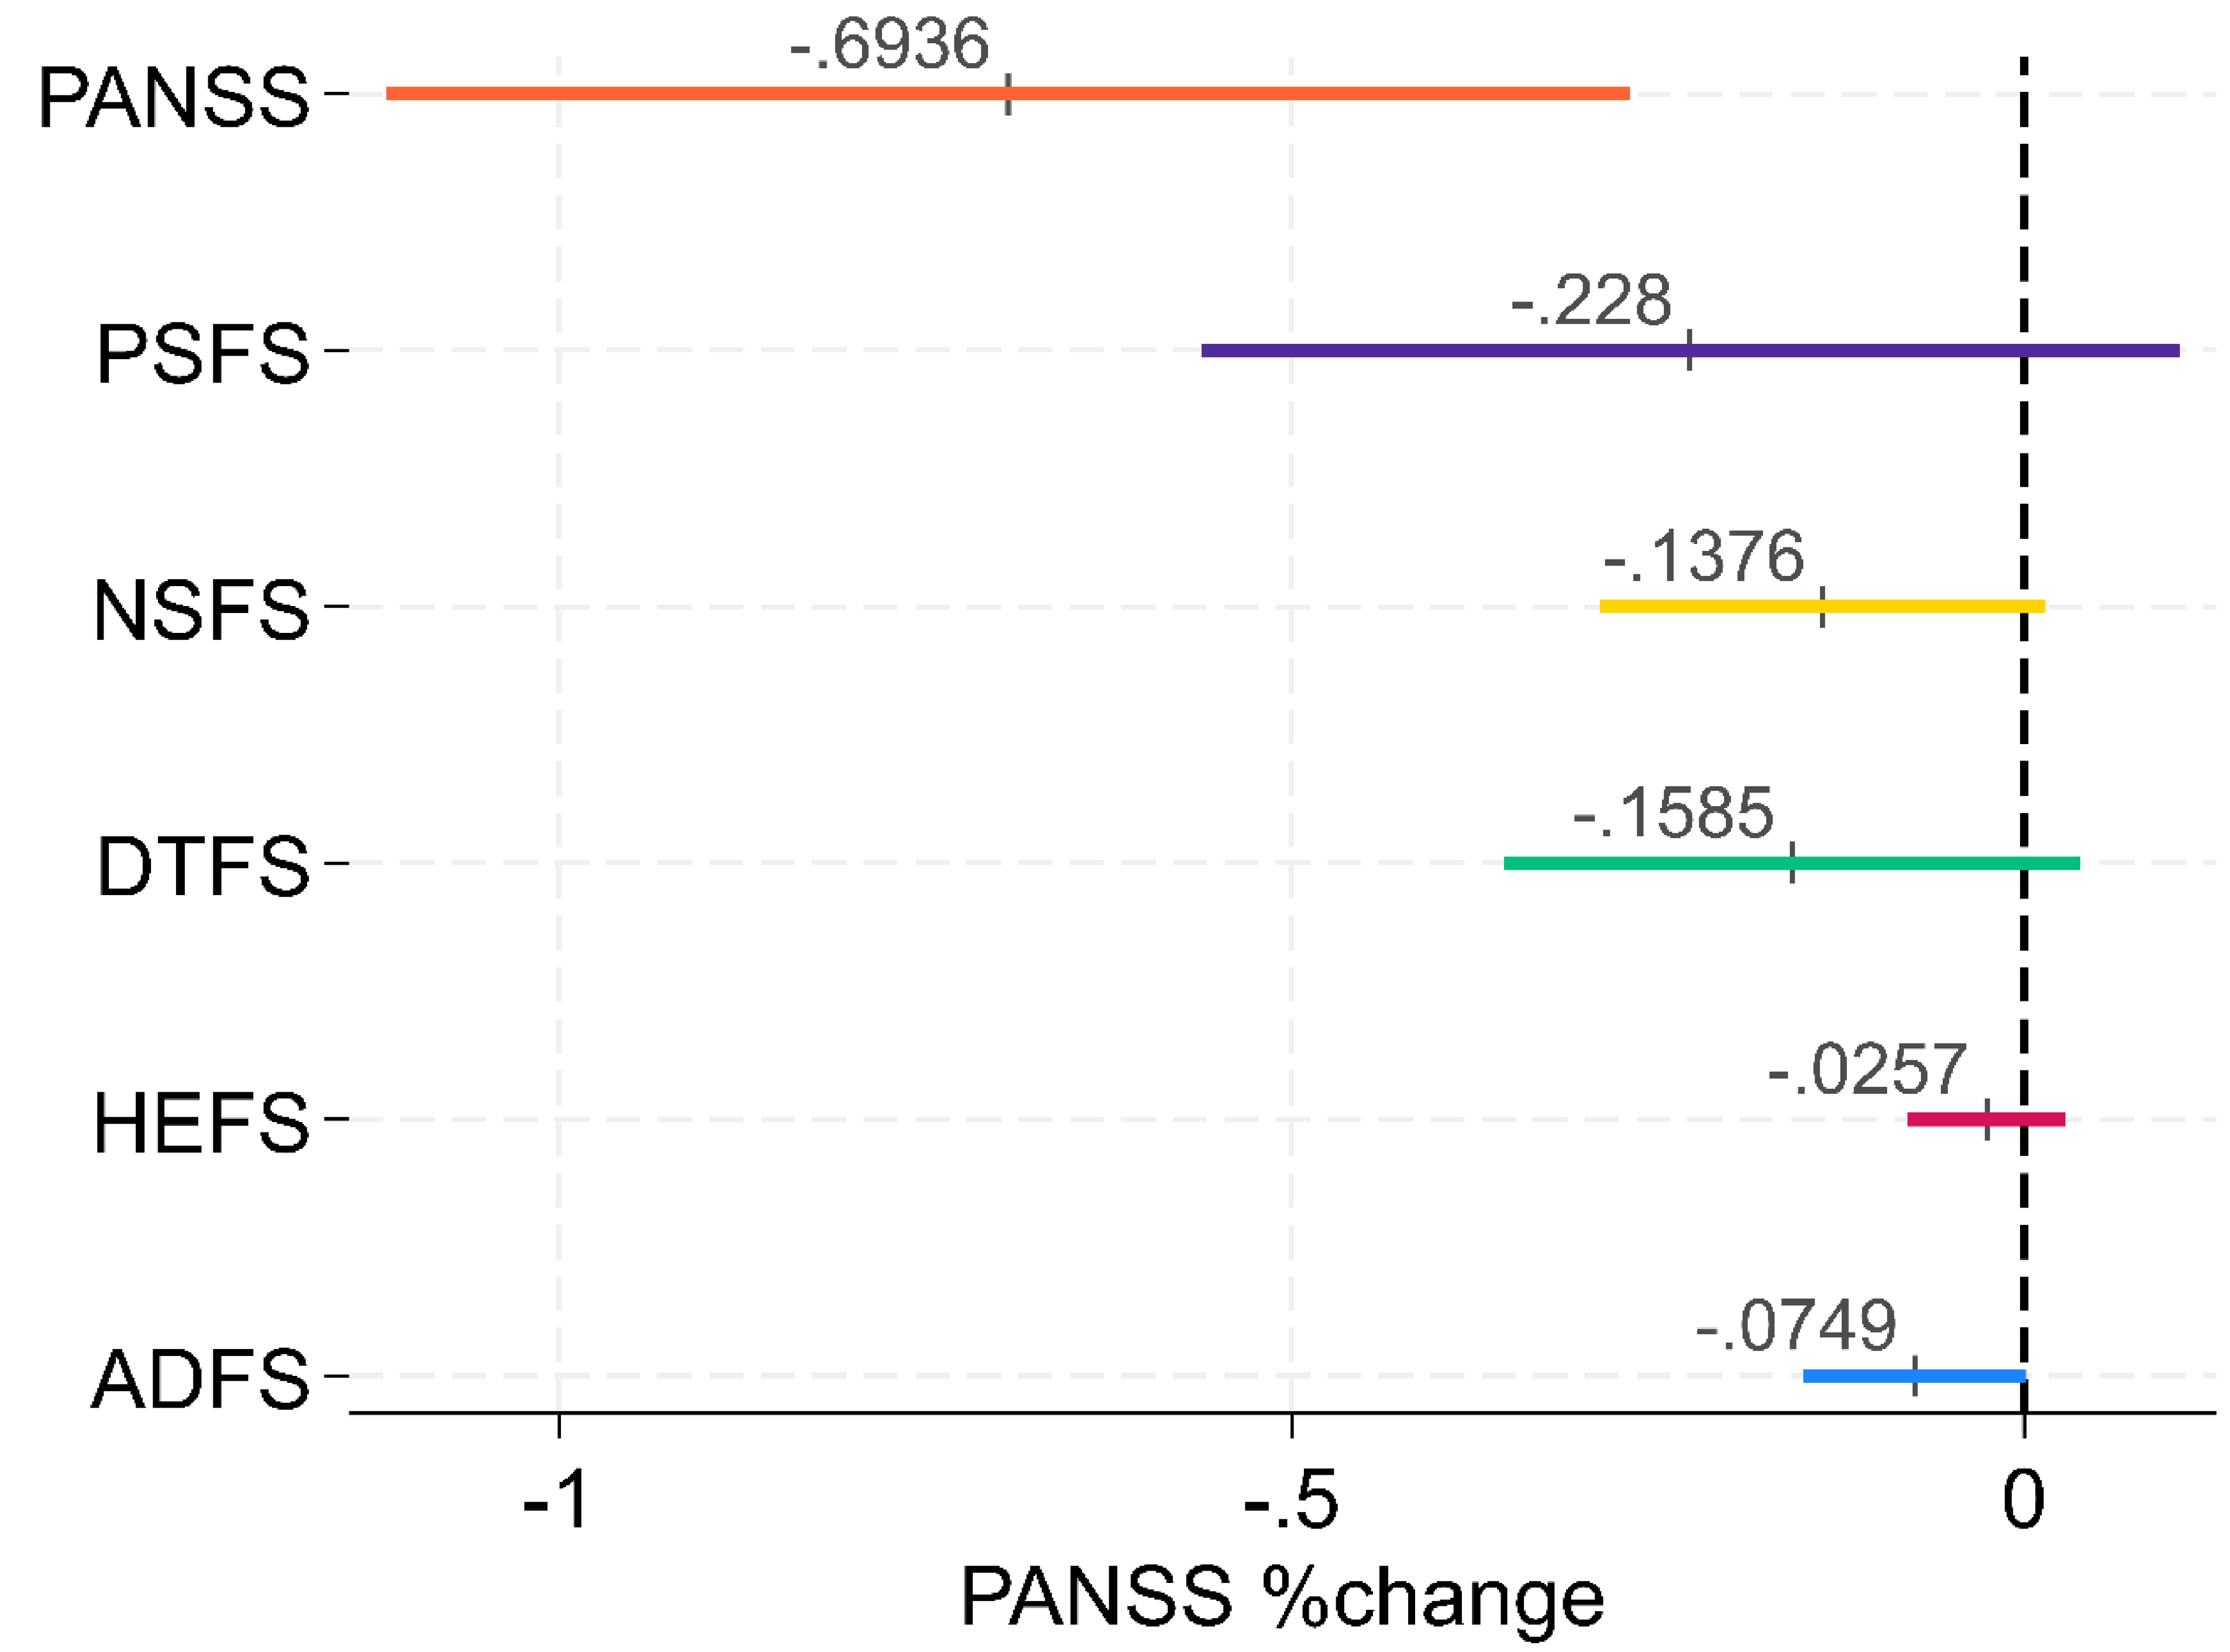
PANSS
PSFS
NSFS
DTFS
HEFS
ADFS
-.6936
-.228
-.1376
-.1585
-.0257
-.0749
-1
-.5
0
PANSS %change

FIGURE 1: IMPACT OF 10% CHANGE BETWEEN SCREENING AND BASELINE ON WEEK 1 PANSS TOTAL CHANGE FROM BASELINE.

PSFS – positive symptoms factor; NSFS – negative symptoms factor; DTFS disorganized thought factor; HEFS – uncontrolled hostility/excitement factor; ADFS – anxiety, depression factor

DISCUSSION

- Our retrospective analysis suggests that the magnitude of symptom change in the screening period has little to no effect on the blinded PANSS change a week after randomization.
- Whether this is true even for improvements beyond 20% is impossible for us to assess because12 of the 14 trials imposed screening change thresholds and the overall sample of subjects improving by more than 20% in the analyzed sample was < 1%.
- We focused on the week 1 visit because the PANSS is assessed at week 1 in most acute schizophrenia clinical trials and its temporal proximity to screening is consistent with potential vulnerability to impact by the screening change.
- Among the limitations in interpreting our results are the retrospective nature of the analysis, the inability to distinguish impact on placebo vs. study drug participants in blinded data and the ceiling created by screening change thresholds.
- More research is needed to confirm the utility of the screening stability requirement implemented in current trials in acute exacerbation of schizophrenia.

KEY FINDING

The magnitude of symptom change in the screening period has little to no clinical effect on the blinded PANSS change a week after randomization.

SIGNANT HEALTH
THE EVIDENCE GENERATION COMPANY™