A novel asynchronous video technology and service to secure protocol compliance in a Phase 2 Major Depressive Disorder trial

Morad Elmi[1], Jorge Aquino[1]; Hedi Kim[1]; Linor Haimson[2]; Yifat Ronen-Protas[2]; Oren Berkowitz[2]; Esther Lukasiewicz-Hagai[2]

Scene Health (Scene)[1], Clexio Biosciences (Sponsor)[2]

Introduction

Medication non-compliance, misuse, abuse, and diversion represent significant challenges in clinical trials, particularly for compounds with high abuse potential or those intended for strict protocol adherence. Poor compliance can lead to unreliable data, and underpowered studies, and may compromise both the safety of participants and the validity of trial outcomes. To mitigate these risks, a technology-enabled adherence review service has been employed. This service confirms that each dose is taken as directed by having subjects video record themselves taking their medication and showing the remaining tablets. This vendor, successfully used in the previous CLEO trial, will again be used in the SOLEO trial to minimize misuse, abuse, and diversion, and to confirm daily medication adherence.

Objective

Sponsor is continuing its use of the technology enabled review service to monitor compliance to trial protocol and to mitigate risk of misuse/abuse/diversion of CLE-100.

Study Design Methodology

- **Participants** (n=90) undergo a 28-day double-blind, placebo-controlled phase followed by a 24-week open-label phase.
- **Adherence** is calculated as the number of confirmed doses divided by the total expected doses; the overall adherence rate is averaged across all participants.
- **Patient Retention Rate** is the percentage of participants completing the treatment phase.
- **Drug Accountability (DA)** is verified by comparing the displayed pill count on video to the expected pill count, allowing for timely corrections. Although DA is not a primary endpoint, both adherence and DA are assessed daily.

Video Directly Observed Therapy (VDOT) combined with Human Review

The SOLEO trial employs a vDOT service to confirm both daily adherence and accurate pill counts. This service captures a short, daily video of each participant ingesting their medication and displaying the remaining tablets to the camera. Each video is then reviewed by a trained adherence reviewer for near real-time confirmation of protocol compliance. Any detected deviations are promptly escalated to the trial site for appropriate intervention. Key elements of this approach include:

- **Smartphone-Based Recording (BYOD):** Patients use their own devices to record a video each day showing themselves taking the dose and displaying remaining pills.
- **Near Real-Time Review:** Within 12 hours, a trained adherence reviewer evaluates each submission, confirming adherence and remaining count.
- **Prompt Escalation:** If deviations from trial protocol are observed (e.g., double-dosing, inaccurate pill counts, or missed submissions), the site is promptly informed for intervention as is appropriate for each patient.
- **Customized Escalation Criteria:** Protocol-specific triggers for further follow-up or corrective action are defined in collaboration with the Sponsor and CRO at implementation.
- **Trial Analytics:** Real-time data provides visibility into the performance of each trial site and identifies patients at risk of not meeting adherence targets.

Expected Outcomes

SOLEO began enrollment in 2024 and is ongoing. We hypothesize participants will demonstrate **similar adherence rates (97.4% - 98.4%)** and **higher patient retention rates (89.2% - 94.2%)** as the previous CLEO study.

Conclusions

Pending results, the asynchronous vDOT Service will directly confirm dosing and obtain accurate pill count in near real-time to mitigate misuse/abuse/diversion. Daily human review of each video submission will allow trial sites to correct patient adherence and pill accountability deviations in a timely fashion. Active adherence monitoring with timely patient interventions will increase patient retention and improve compliance.

Trial Sites

onboard patients. They are notified by vDOT Service team to any protocol deviation that may occur allowing for timely patient specific intervention.

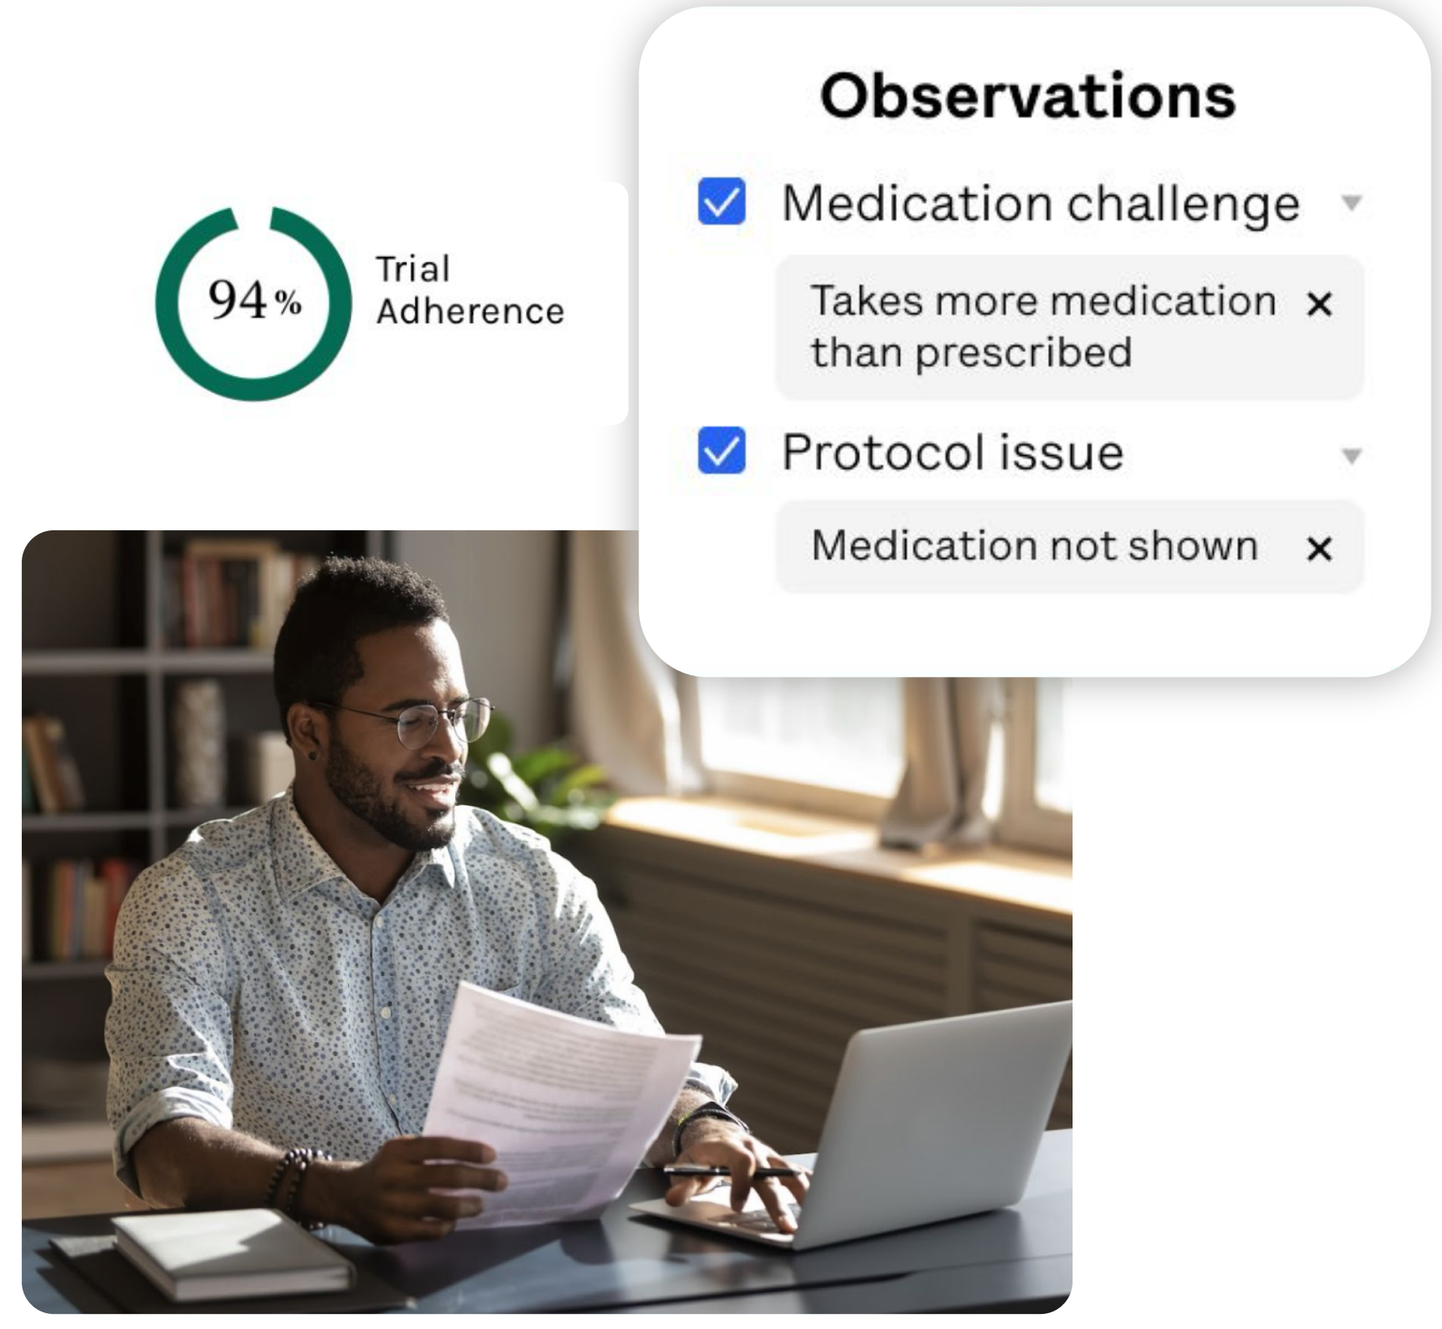
94%
Trial Adherence
Observations
Medication challenge
Takes more medication than prescribed
Protocol issue
Medication not shown

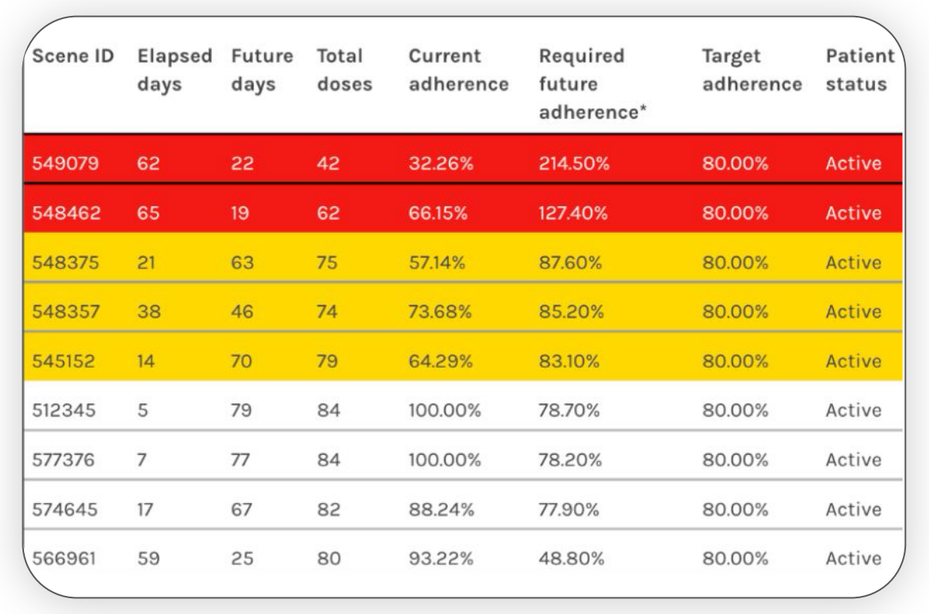

Scene ID	Elapsed days	Future days	Total doses	Current adherence	Required future adherence*	Target adherence	Patient status
549079	62	22	42	32.26%	214.50%	80.00%	Active
548462	65	19	62	66.15%	127.40%	80.00%	Active
548375	21	63	75	57.14%	87.60%	80.00%	Active
548357	38	46	74	73.68%	85.20%	80.00%	Active
545152	14	70	79	64.29%	83.10%	80.00%	Active
512345	5	79	84	100.00%	78.70%	80.00%	Active
577376	7	77	84	100.00%	78.20%	80.00%	Active
574645	17	67	82	88.24%	77.90%	80.00%	Active
566961	59	25	80	93.22%	48.80%	80.00%	Active

Sponsor Dashboards

display real-time adherence trends, trial site performance rates, and flags patients at risk of not meeting adherence targets.

Video DOT Service Review Team

reviews every video submission. Any observed protocol deviation is escalated to the trial site team.

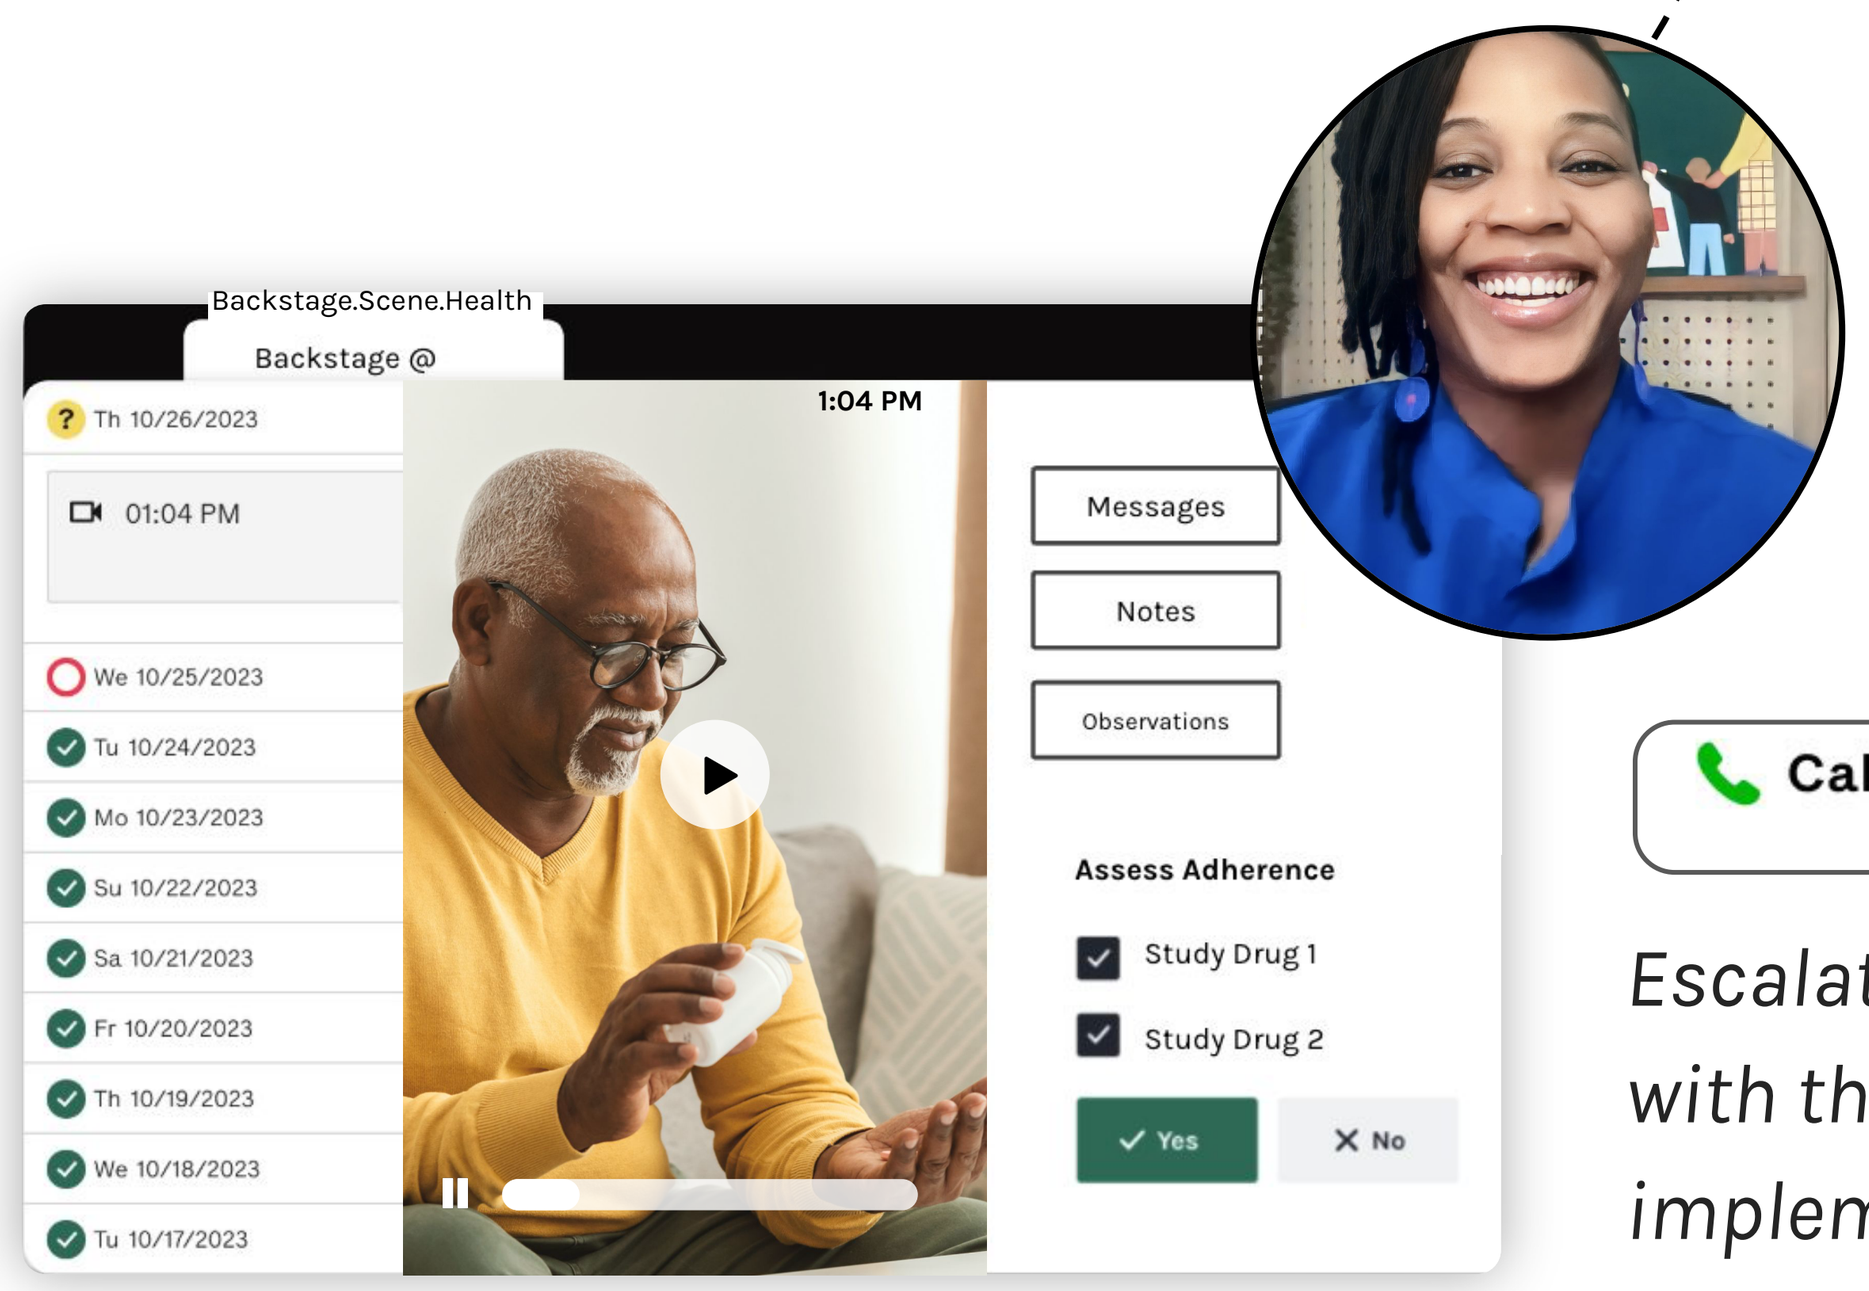
Backstage.Scene.Health
Backstage @
Th 10/26/2023
01:04 PM
We 10/25/2023
Tu 10/24/2023
Mo 10/23/2023
Su 10/22/2023
Sa 10/21/2023
Fr 10/20/2023
Th 10/19/2023
We 10/18/2023
Tu 10/17/2023
1:04 PM
Messages
Notes
Observations
Assess Adherence
Study Drug 1
Study Drug 2
Yes
No

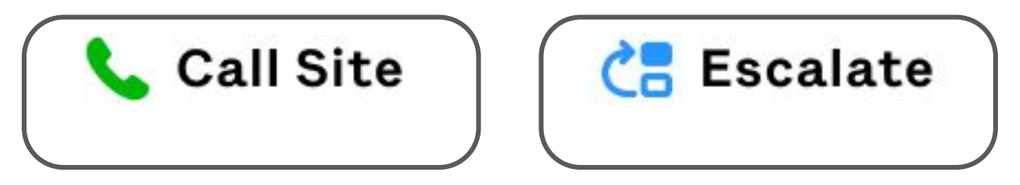
Call Site
Escalate

Escalations designed with the Sponsor during implementation.

Trial Participants

record a short video of themselves (BYOD) taking their daily medication, and show their remaining pills on camera to confirm accurate pill count.

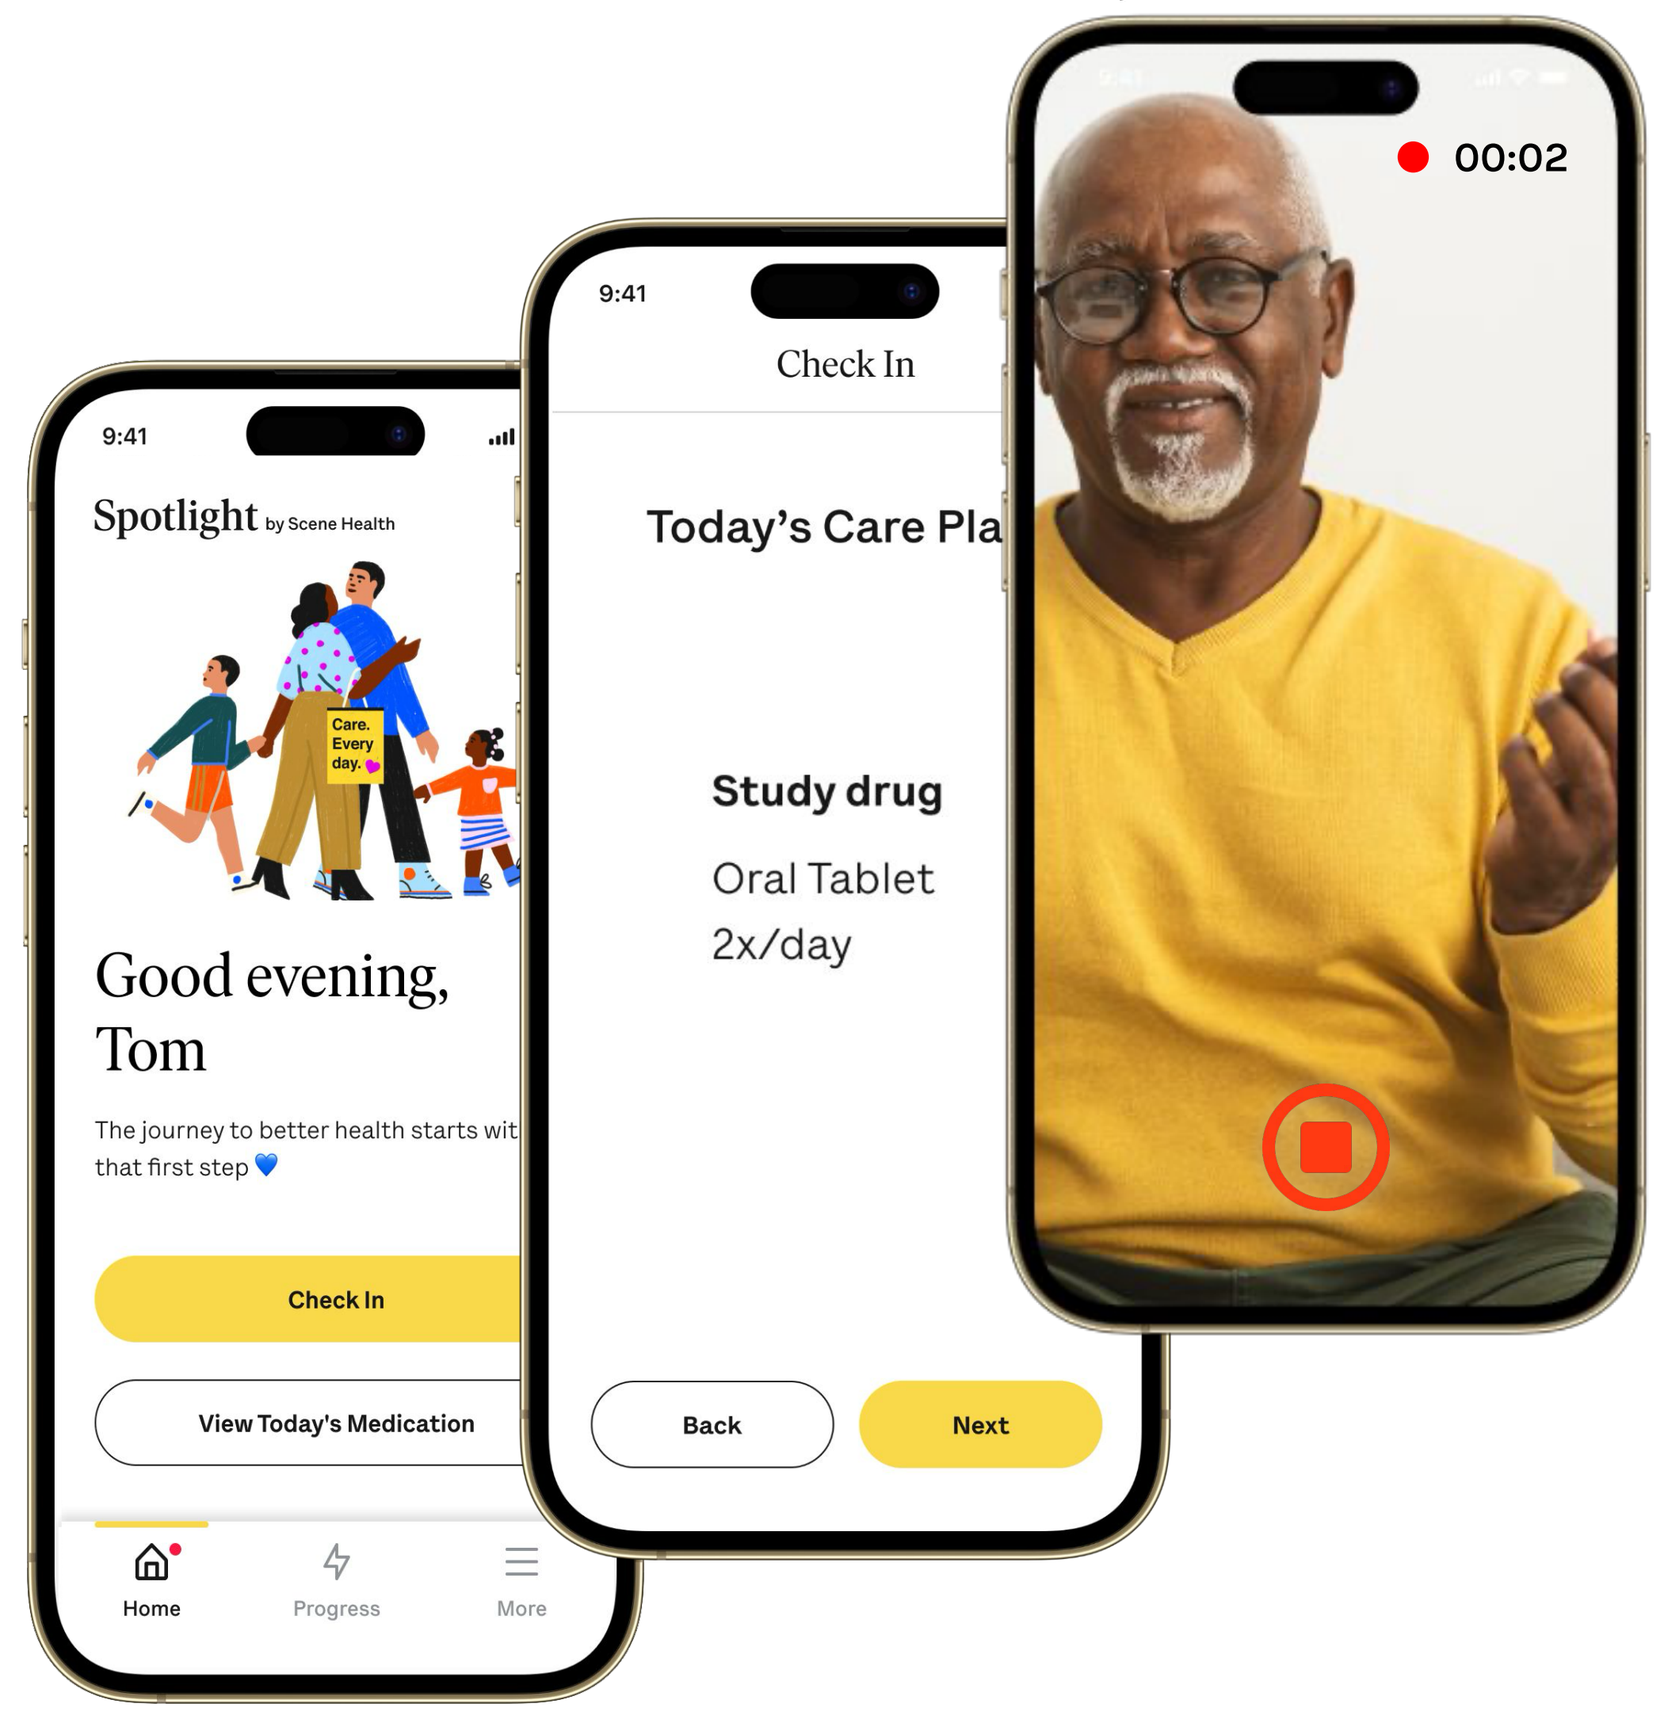
9:41
Spotlight by Scene Health
Good evening, Tom
Check In
View Today's Medication
Home
Progress
More
Check In
Study drug
Oral Tablet
2x/day
Back
Next
00:02

Disclosures: One or more authors report potential conflicts which are described in the program. Morad Elmi and Hedi Kim are employees of the vDOT Service provider, Scene Health.