Presented at the 21st Annual ISCTM Scientific Meeting, Feb 19 - 21, 2025, Washington, D.C.

Comparison of the ten-item pediatric Positive and Negative Syndrome Scale (PANSS-10) to the PANSS-6 in an adult, acutely exacerbated clinical trial population with schizophrenia

Daniel DG[1], Kott A[1], Busner J[1],4, Wang X[1], Langfus J[2], Youngstrom EA[3], Findling R[4]

[1]Signant Health, [2]University of North Carolina at Chapel Hill, [3]University of North Carolina at Chapel Hill; Helping Give Away Psychological Science, 501c3 and Nationwide Children's Hospital and The Ohio State University, Columbus, Ohio, US, [4]Virginia Commonwealth University

METHODOLOGICAL QUESTION

How do the psychometric qualities of the pediatric short Positive and Negative Syndrome Scale (PANSS-10) version compare to the PANSS-6 in an adult, acutely exacerbated clinical trial population with schizophrenia?

INTRODUCTION

The Positive and Negative Syndrome Scale (PANSS) is the most frequently used instrument to measure severity of schizophrenia in clinical research. The scale is long, takes a lot of time to administer, and appears to have many item redundancies. Several attempts were conducted to shorten the instrument to reduce administrative burden while retaining as much of the validity and reliability of the original PANSS as possible. The PANSS-6 (Hieronymous et al, 2021), for example, has shown favorable psychometric qualities and sensitivity to change in antipsychotic clinical trials. Recently, we reported that the 10-item version of the PANSS developed for pediatric population (Findling et al, 2023) shows promising psychometric qualities even in adult populations of acutely exacerbated clinical trial participants with schizophrenia (Daniel et al, 2023). In the current retrospective analysis, we compare psychometric qualities of the PANSS-10 and PANSS-6 in acutely exacerbated schizophrenic patients.

METHODS

Baseline PANSS data (n=8,505) were pooled from 8,505 subjects recruited into 22 acute adult schizophrenia trials. Average item correlations were examined using Spearman correlation coefficients for the PANSS-6 and PANSS-10 separately; the correlation between PANSS-6 total score (or PANSS-10 total score) and the PANSS-30 total score (or CGI-S score) were examined using Pearson or Spearman correlation coefficients. In addition, the difference in the average item scores between each of the short versions and the full version was calculated. Bootstrapping confidence limits were computed using 1000 random samples with replacement for each correlation coefficient.

RESULTS

- The mean inter-item correlation for the PANSS-10 was 0.088 (CI = 0.082, 0.093) and for the PANSS-6 was 0.115 (0.107, 0.124).
- As shown in Figure 1 the correlation of the PANSS-10 with the full PANSS, was r= 0.839 (0.832, 0.845). The correlation of the PANSS6 with the full PANSS was r= 0.681 (0.670, 0.692).
- The correlation between the PANSS-10 and the CGI-S was 0.477 (0.460, 0.494); and the correlation between the PANSS-6 and the CGI-S was 0.425 (0.407, 0.443).
- The mean difference in average item scores between the PANSS-10 and the full PANSS was 0.11 (-0.37, 0.589) and between the PANSS-6 and the full PANSS was 0.6 (-0.21, 1.41).

CONCLUSION

In this post-hoc initial psychometric analysis comparing the PANSS-10 and PANSS-6 at baseline in acutely psychotic patients with schizophrenia, the PANSS-10 demonstrated a modest but statistically significantly stronger relationship to the full 30 item PANSS than did the PANSS-6. The mean inter-item correlations were statistically significantly lower in the PANSS-10 than in the PANSS-6. The relationship with the CGI was similar for both scales. In this analysis, the PANSS -10 displayed relatively favorable psychometric qualities in an adult acutely decompensated population at baseline.

FIGURE 1: THE CORRELATION OF THE PANSS-10 WITH THE FULL PANSS (r=0.839 (0.832, 0.845)).

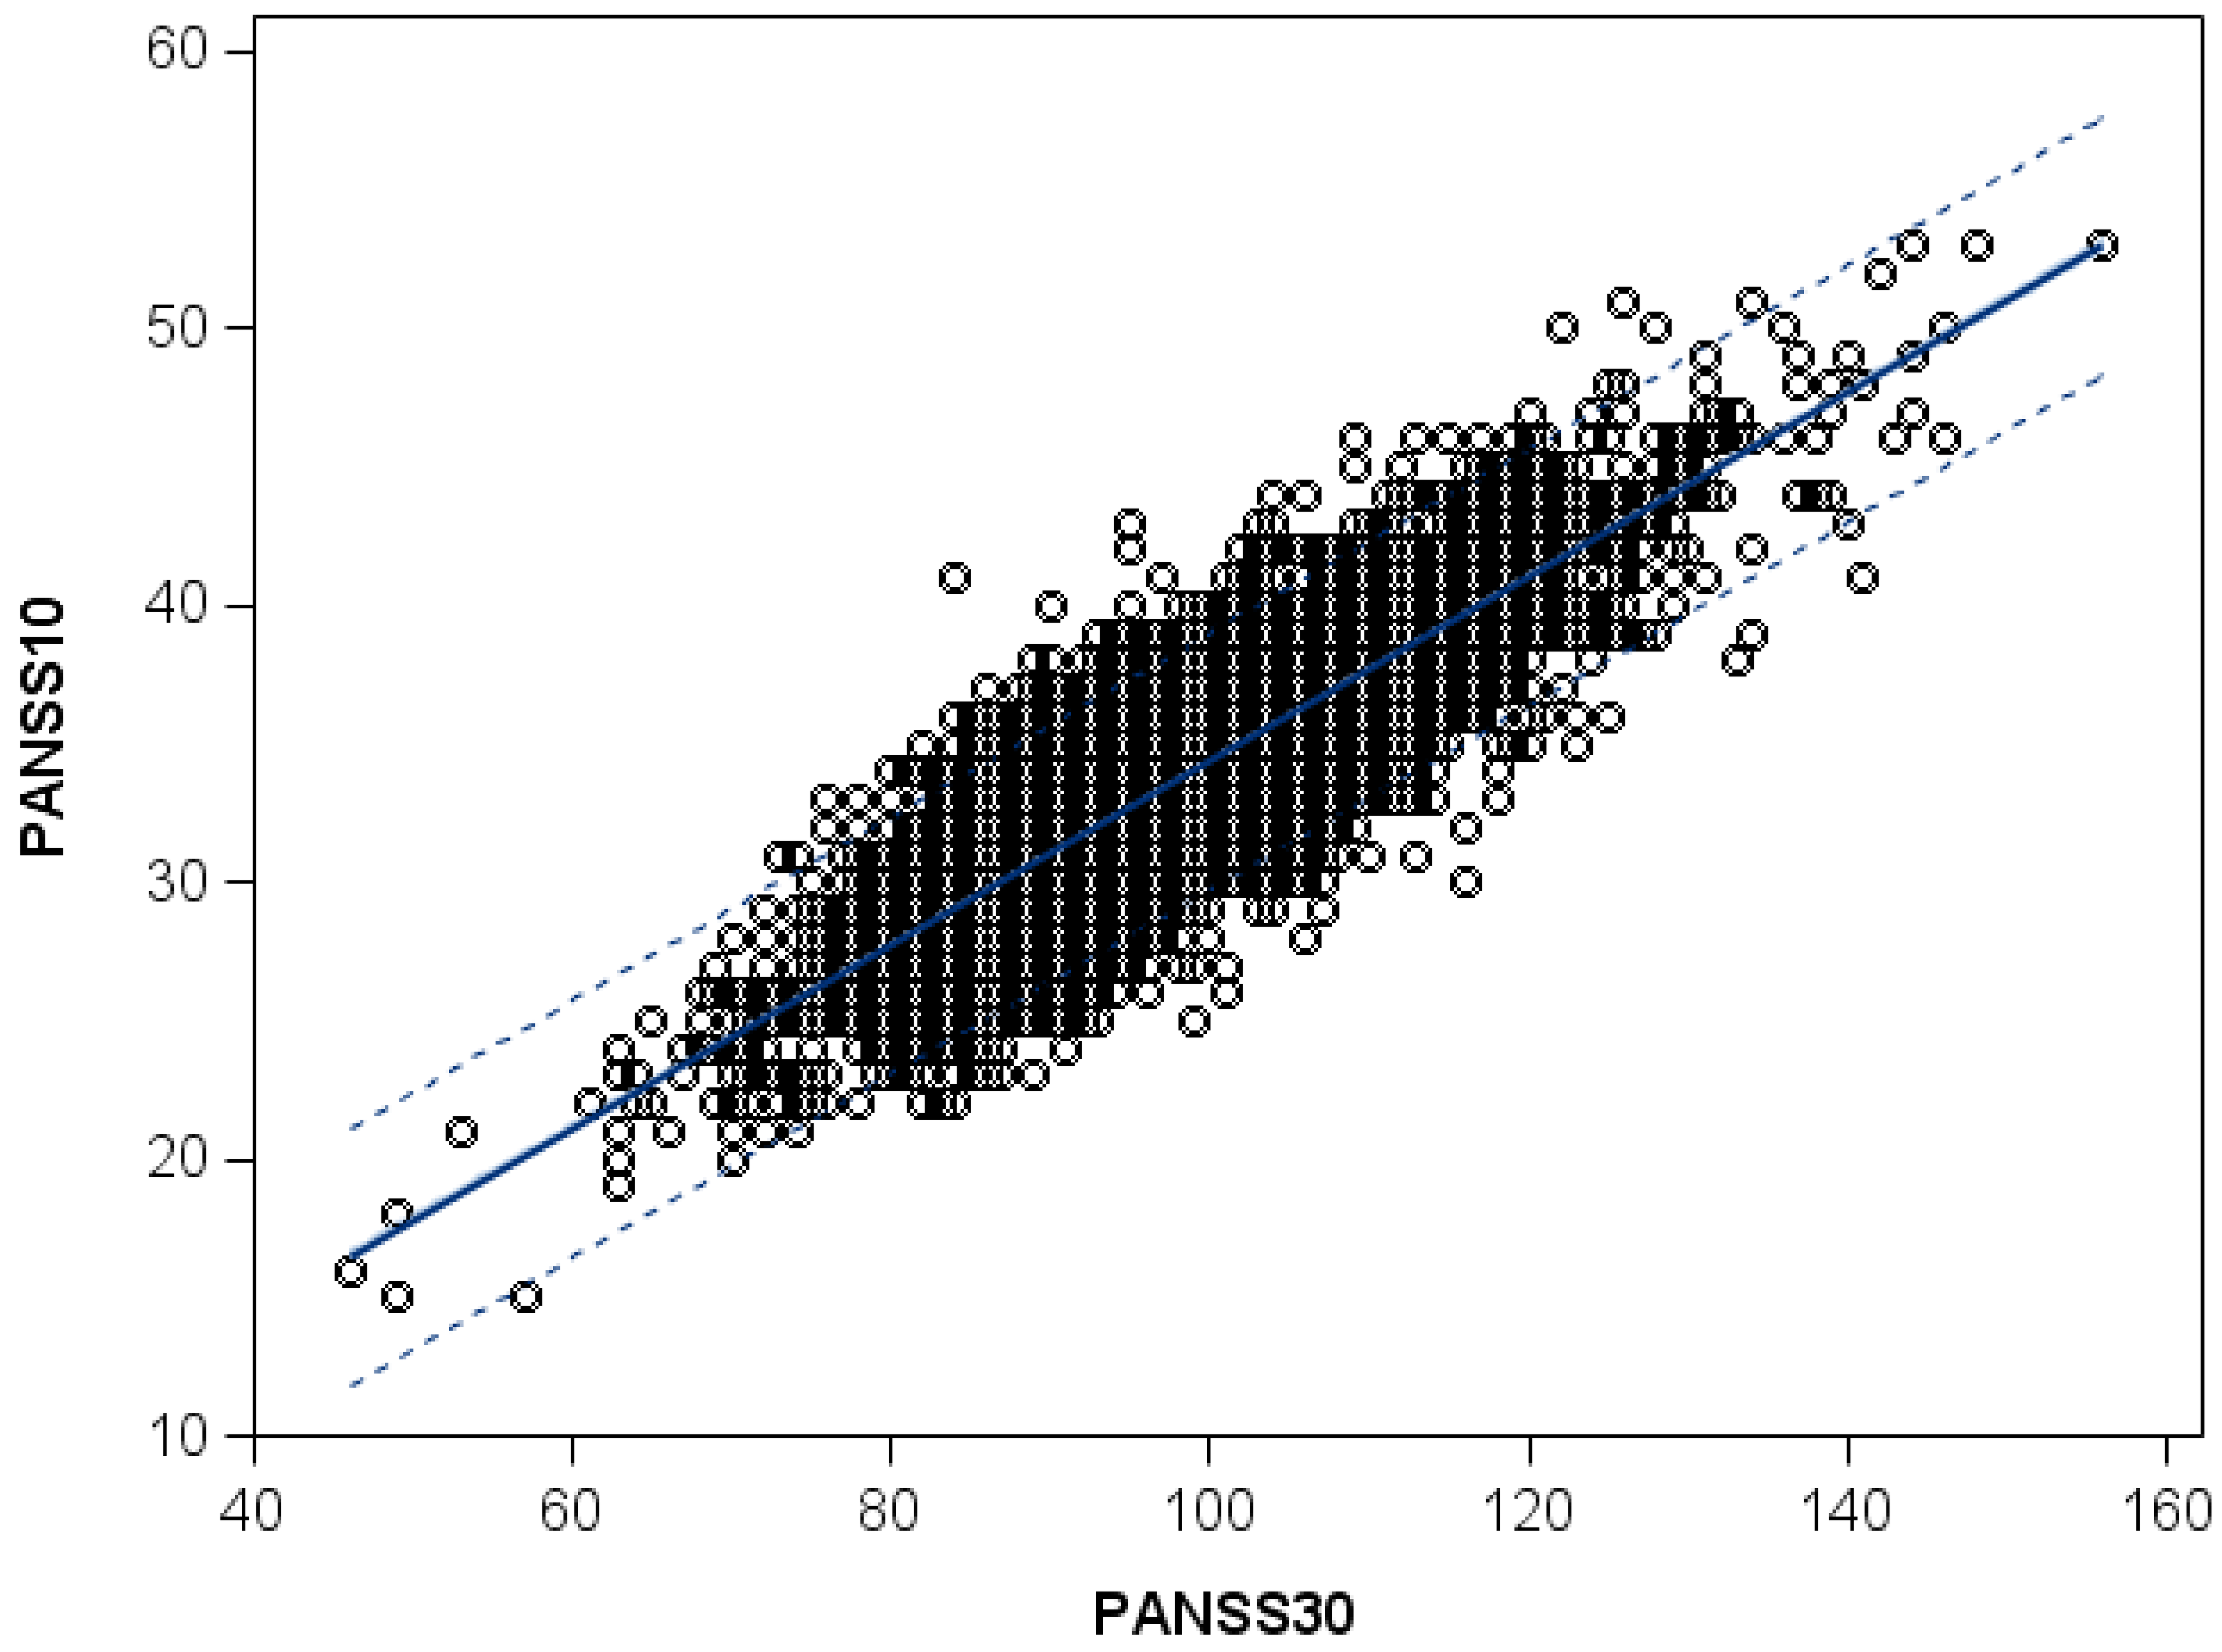
60
50
40
30
20
10
PANSS10
40
60
80
100
120
140
160
PANSS30

REFERENCES

- Daniel DG, Kott A, Busner J, Wang X, Langfus J, Youngstrom EA, Findling R: Initial psychometric evaluation of the pediatric short Positive and Negative Syndrome Scale (PANSS) version in an adult, acutely exacerbated clinical trial population with schizophrenia Innovations in Clinical Neuroscience. Volume 20, No. 10-12, Oct-Dec 2023, Supplement 1. https://icns.mydigitalpublication.com/cns-summit-2023-abstract-supplement.
- Findling RL, Youngstrom EA, McClellan JM, Frazier JA, Sikich L, Daniel DG, Busner J. An Optimized Version of the Positive and Negative Symptoms Scale (PANSS) for Pediatric Trials. J Am Acad Child Adolesc Psychiatry. 2023 Apr;62(4):427-434. doi: 10.1016/j.jaac.2022.07.864. Epub 2022 Dec 13. PMID: 36526163.
- Hieronymus F, Kølbæk P, Correll CU, Østergaard SD. Antipsychotic-placebo separation on the PANSS-6 subscale as compared to the PANSS-30: a pooled participant-level analysis. NPJ Schizophr. 2021 Aug 27;7(1):41. doi: 10.1038/s41537-021-00168-x. PMID: 34453057; PMCID: PMC8397783.

DISCLOSURES

Alan Kott, Xingmei Wang and Joan Busner are employees of Signant Health and may hold stock/equity shares. David Daniel is an Executive Advisor to Signant Health. Eric Youngstrom is Director, Institute for Mental and Behavioral Health research at Nationwide Children's Hospital and Ohio State University, he is the co-founder and Executive Director of Helping Give Away Psychological Science, a 501c3; he has consulted about psychological assessment with Signant Health and received royalties from the American Psychological Association and Guilford Press. Joshua Langfus reports no relevant conflict of interest. In the past 36 months, Dr. Findling has received research support, acted as a consultant and/or has received honoraria from Abbvie, Acadia, Adamas, Afecta, Ajna, Akili, Alkermes, American Academy of Child & Adolescent Psychiatry, American Psychiatric Press, Bioprojet, BioXcel, Corium, Elsevier, Idorsia, Iqvia, Karuna, Lundbeck, Merck, MJH Life Sciences, NIH, Neurim, Novartis, Otsuka, Oxford University Press, PaxMedica, PCORI, Pfizer, Physicians' Postgraduate Press, Radius, Receptor Life Sciences, Sage, Signant Health, Sumitomo Pharma, Sunovion, Supernus Pharmaceuticals, Syneos, Takeda, Tris, and Viatris.

SIGNANT HEALTH
THE EVIDENCE GENERATION COMPANY™