ENRICHMENT BASED ON SPEECH LATENCY ENHANCES TREATMENT EFFECTS IN A PHASE III STUDY OF BRILAROXAZINE.

PRESENTERS: *Alex S Cohen [a,b,c], Brian Kirkpatrick [c,d], Mark Opler [e], Jan A Sedway [e], Kazunori Tatsumi [e], Seema Bhat [f], Laxminarayan Bhat [f]*
AFFILIATIONS: a. Louisiana State University, Department of Psychology, b. Louisiana State University, Center for Computation and Technology, c. Quantic Innovations, Inc, d. Psychiatric Research Institute, University of Arkansas for Medical Sciences, e.WCG Clinical, Inc., f. Reviva Pharmaceuticals Inc

Key Findings

- Automated measurement of speech latency corresponds to clinically rated negative symptoms.
- Excluding the ~40% of individuals with normal speech latencies dramatically increases treatment effect size.
- Speech latency is a promising enrichment tool for CNS clinical trials.

Introduction

Speech latency, a measure of verbal response times, is an objective marker of cognitive, social, and motivational factors that can be assayed directly from psychiatric interviews.

We recently used speech latency to enrich participants for an antidepressant clinical trial (Siegel, Cohen, 2024), resulting in nearly double the drug-placebo effects at half the sample size.

Here we evaluate speech latencies in a clinical trial of schizophrenia.

What is a Speech Latency?

Q: How have you been doing lately?
A: Turn Latency Ok, I don't have much going on, so...

We applied a correction to account for cross-cultural variability.

Methods

Audio recordings from psychiatric interviews in a Phase 3 trial of brilaroxazine were evaluated (k = 2590 recordings for 408 participants from three countries representing eight languages).

Predicted PANSS Marder factor scores were modeled based on turn latency for post-randomization sessions and applied to the screening data for classifying individuals:

- Vocal Biomarker Positive (VBM Pos): slow latency; predicted Marder item average score 4+
- Vocal Biomarker Negative (VBM Neg): Fast latency; predicted Marder item average score 2 or below

Analyses:

We evaluated treatment x time effects for VBM Pos and VBM Neg patients, in:

- PANSS Total scores
- PANSS Marder factor scores
- Clinical Global Impression (CGI)
- Personal and Social Performance Scale (PSP)

The VBM Pos Group shows fast , significant reduction in PANSS Total scores

VBM Positive

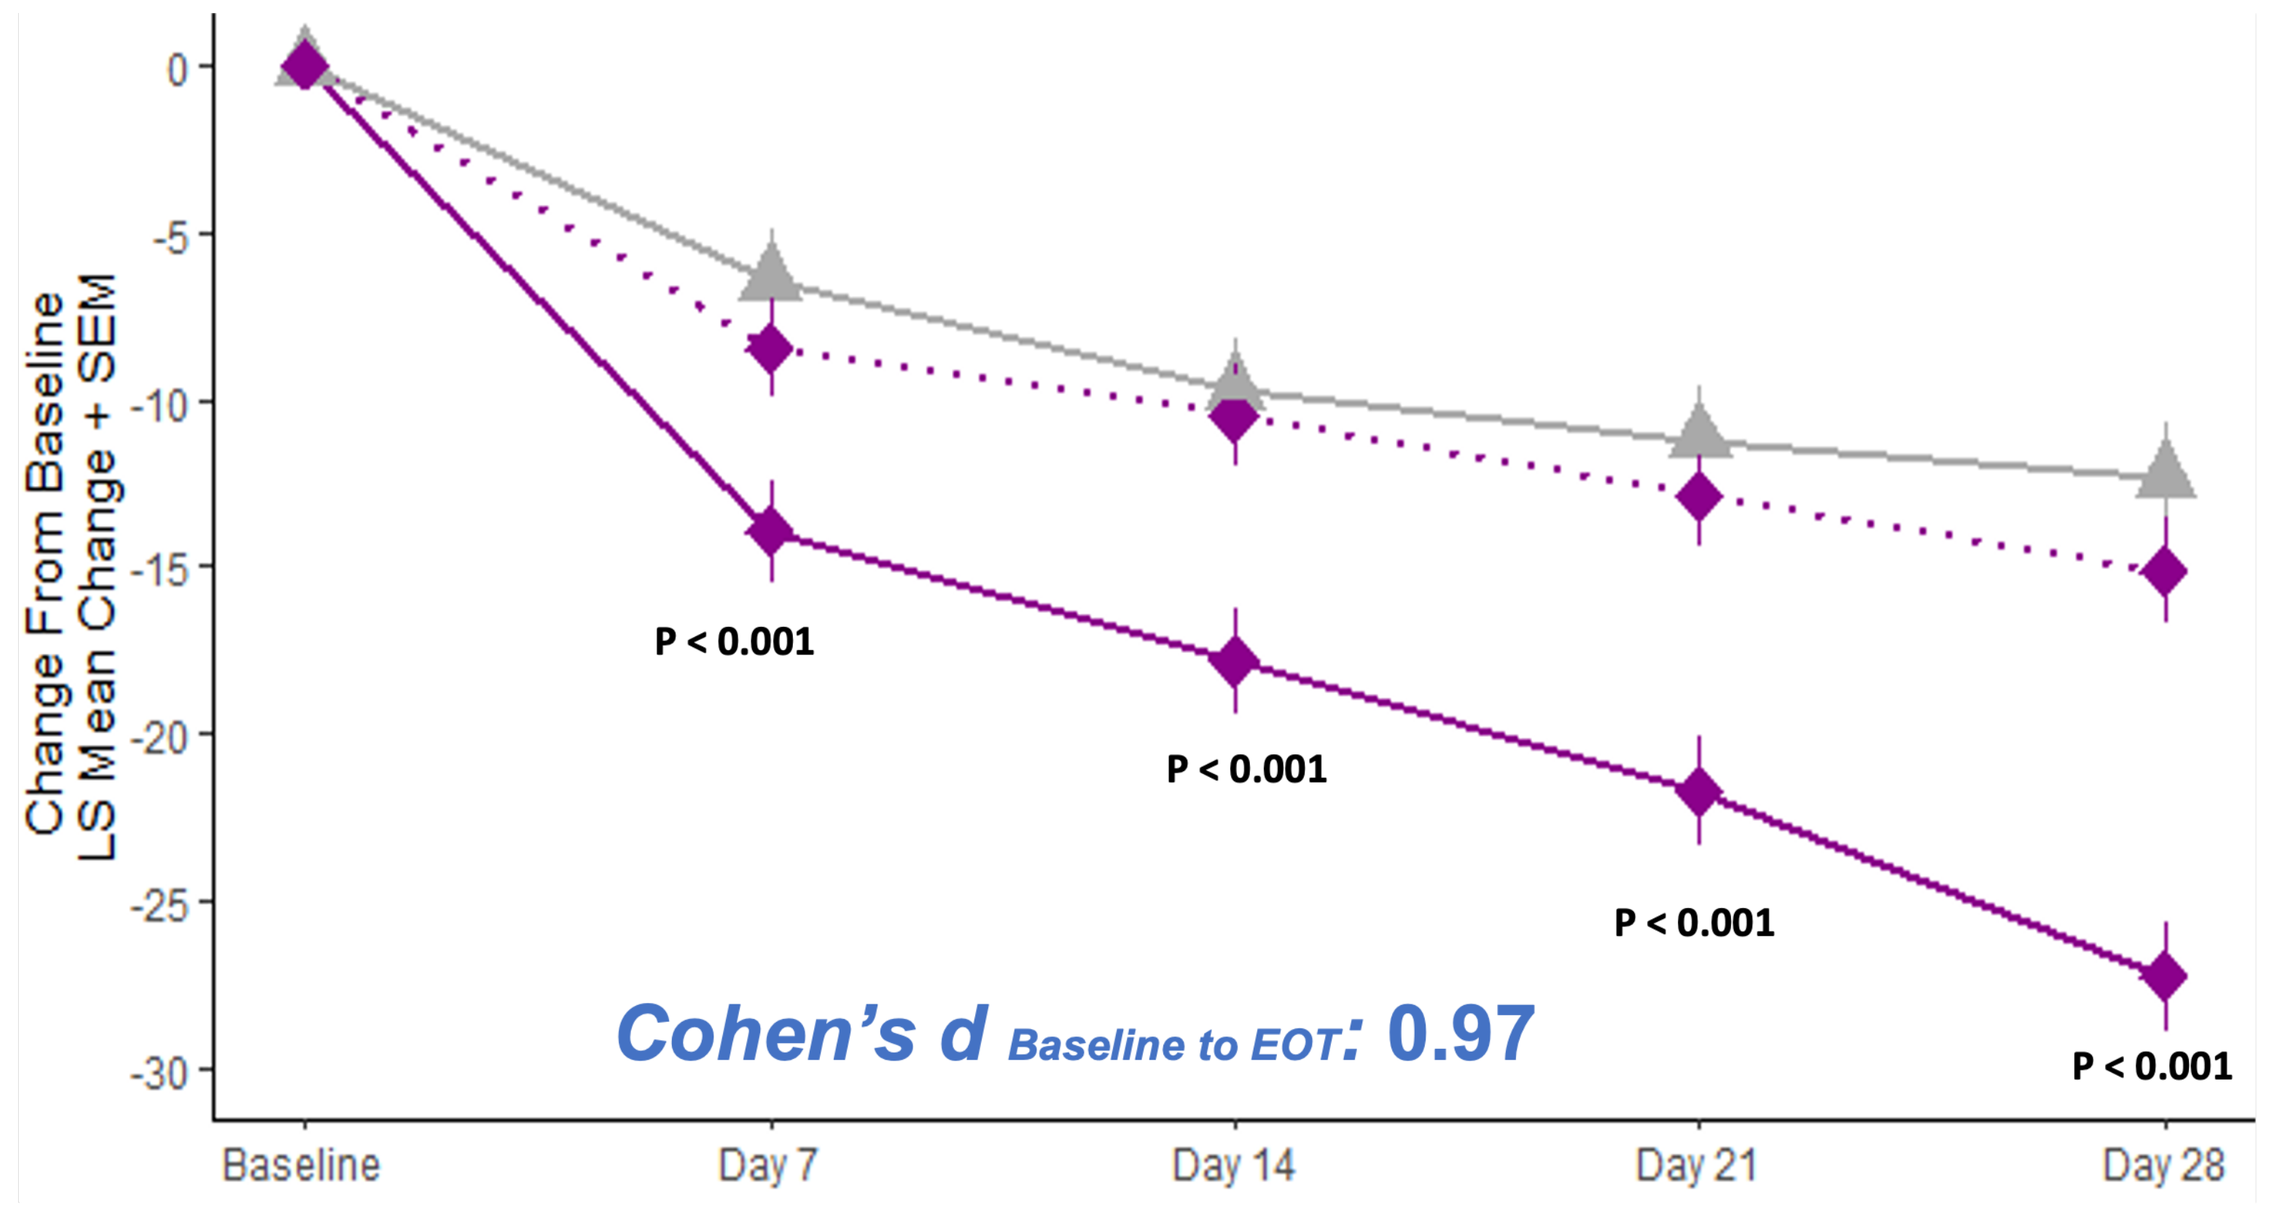
Change From Baseline
LS Mean Change + SEM
P < 0.001
P < 0.001
P < 0.001
P < 0.001
Cohen's d Baseline to EOT: 0.97
Baseline
Day 7
Day 14
Day 21
Day 28

VBM Negative

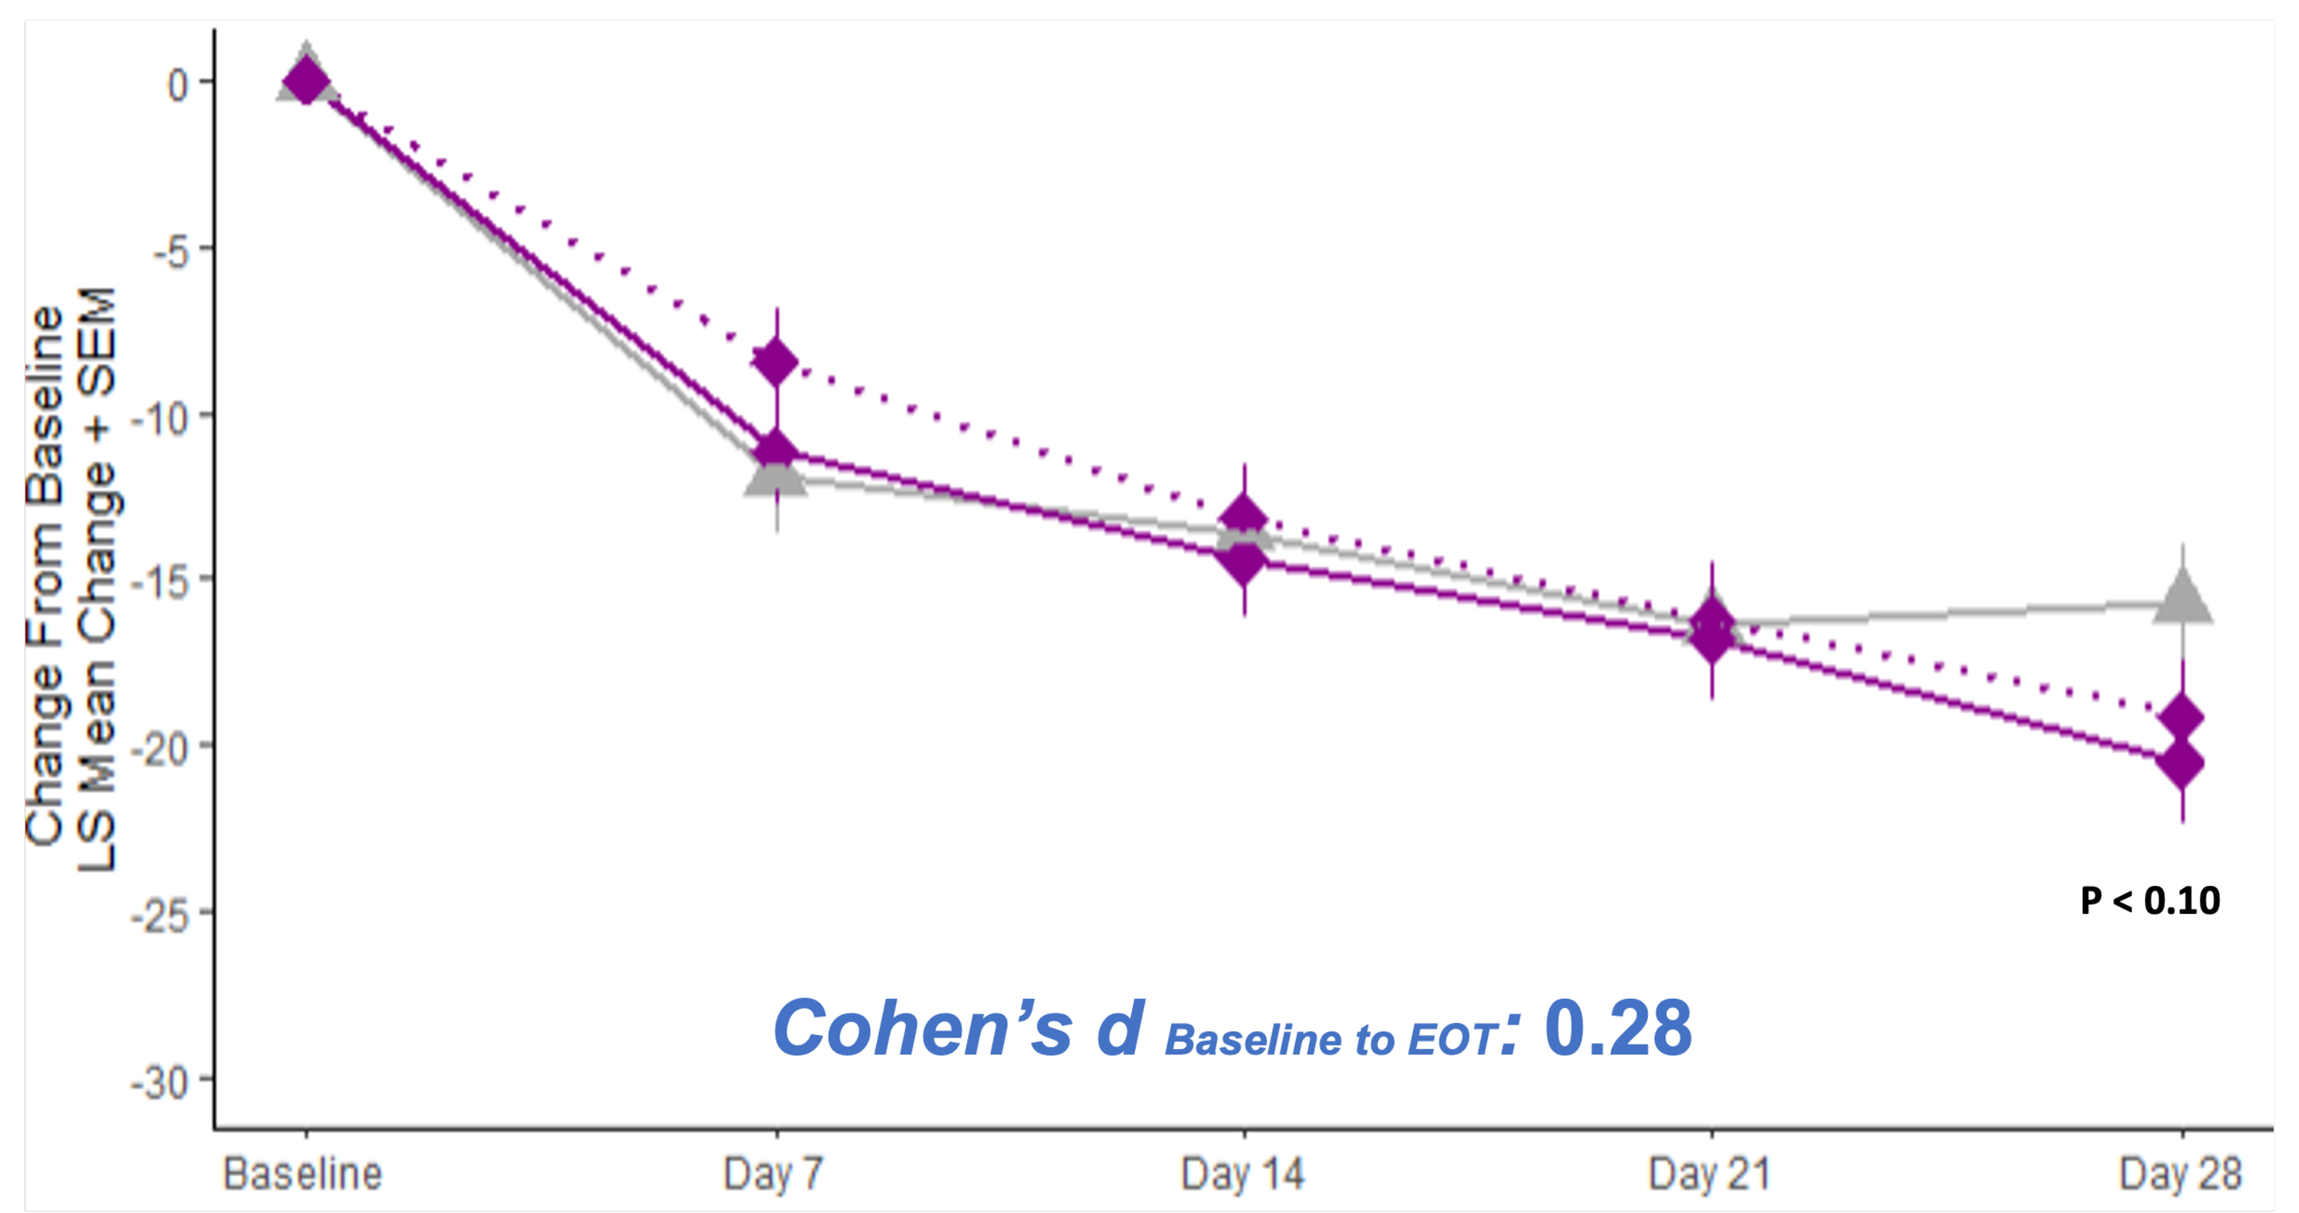
Change From Baseline
LS Mean Change + SEM
P < 0.10
Cohen's d Baseline to EOT: 0.28
Baseline
Day 7
Day 14
Day 21
Day 28

The VBM Pos Group shows fast , significant reduction in Marder Negative Factor scores

VBM Positive

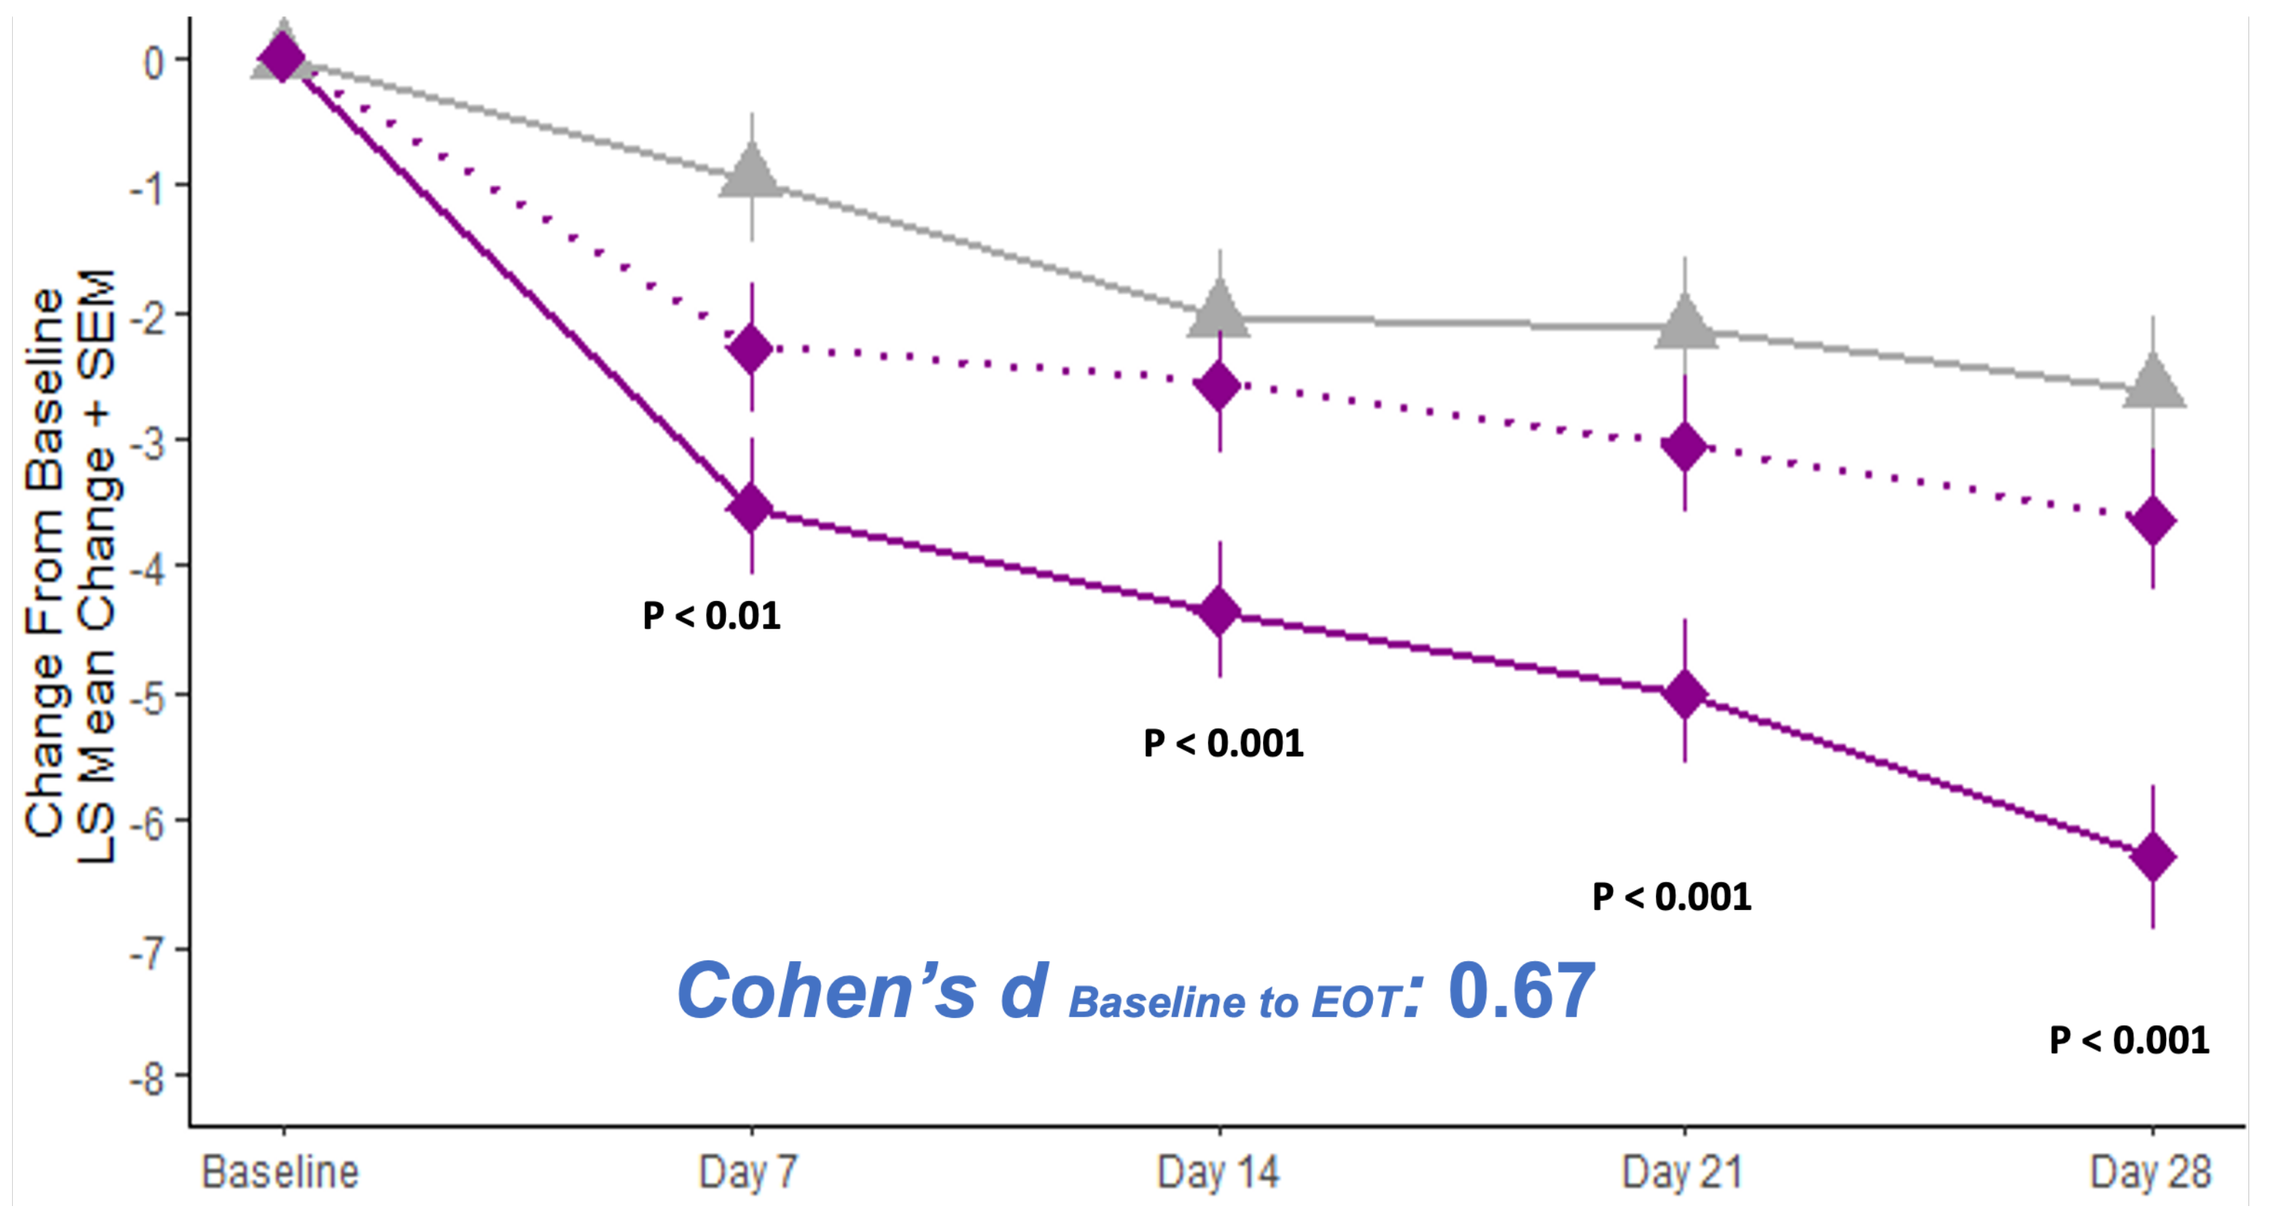
Change From Baseline
LS Mean Change + SEM
P < 0.01
P < 0.001
P < 0.001
P < 0.001
Cohen's d Baseline to EOT: 0.67
Baseline
Day 7
Day 14
Day 21
Day 28

VBM Negative

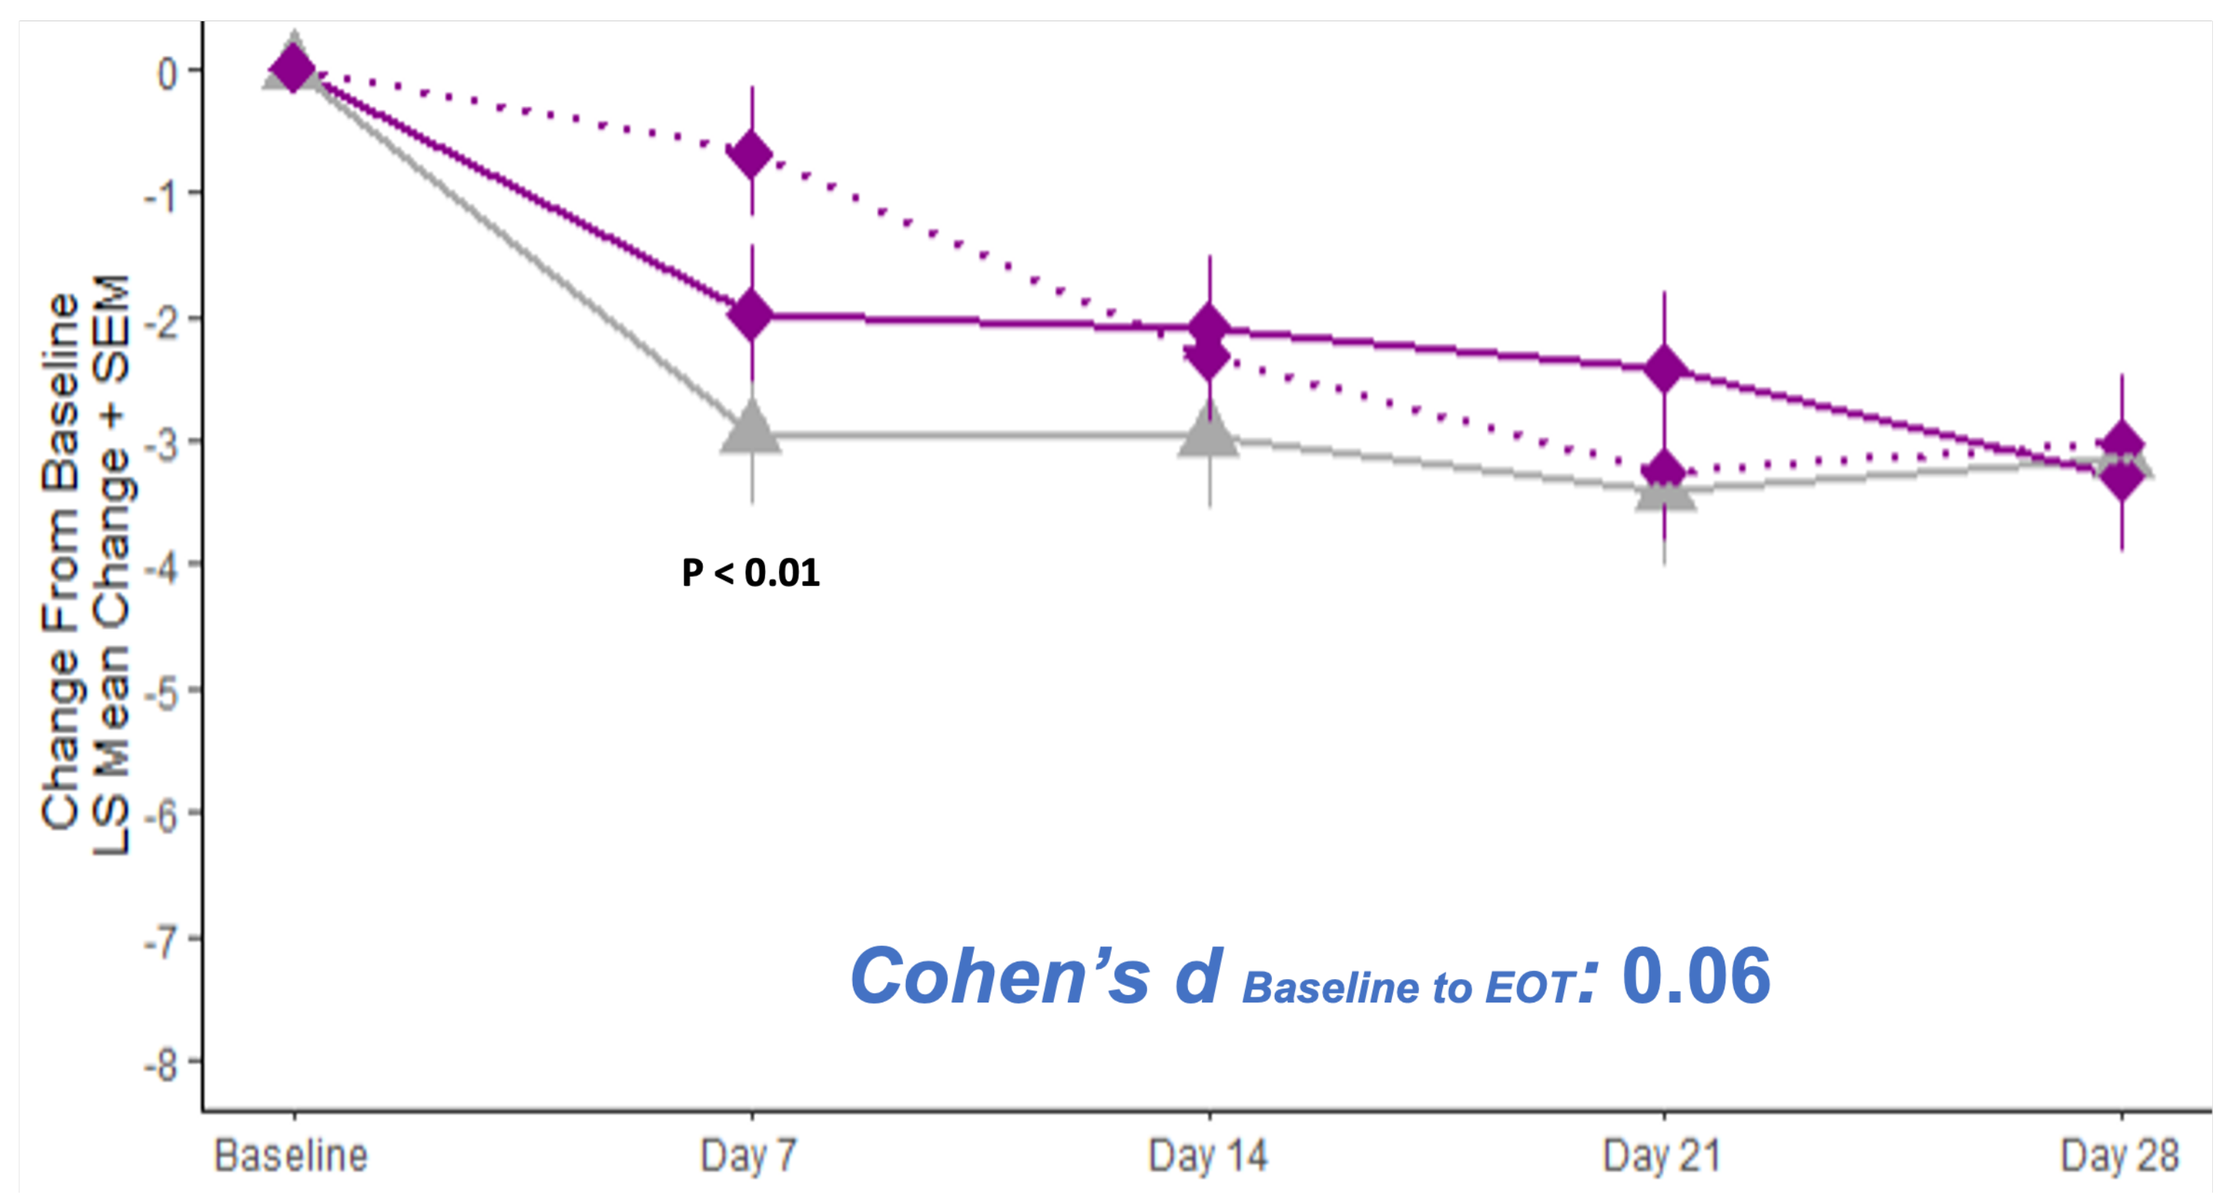
Change From Baseline
LS Mean Change + SEM
P < 0.01
Cohen's d Baseline to EOT: 0.06
Baseline
Day 7
Day 14
Day 21
Day 28

The VBM Pos Group shows similar improvement in PANSS Positive & Disorganization symptoms and improved functioning in CGI and PSP

Clinical Global Impression (CGI)

VBM Positive

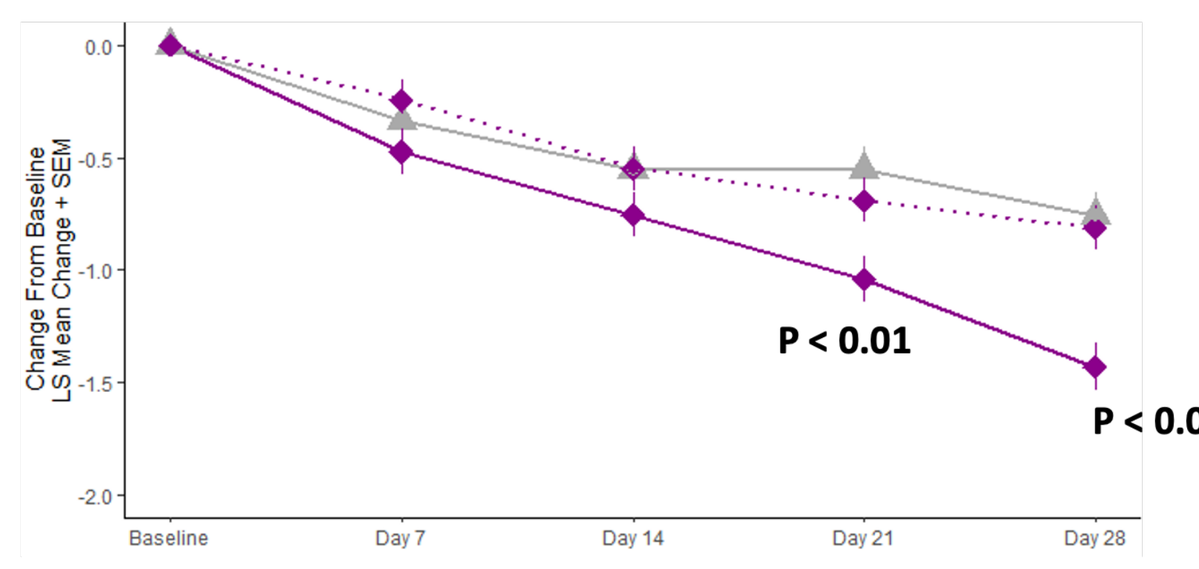
P < 0.01
P < 0.001

VBM Negative

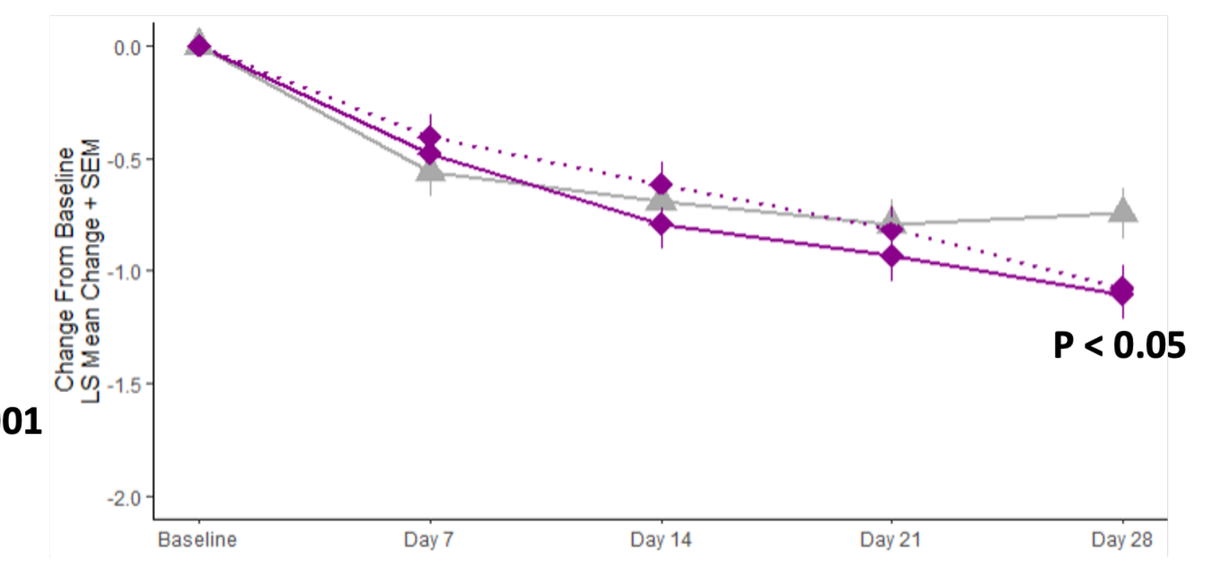
P < 0.05

Personal and Social performance Scale (PSP)

VBM Positive

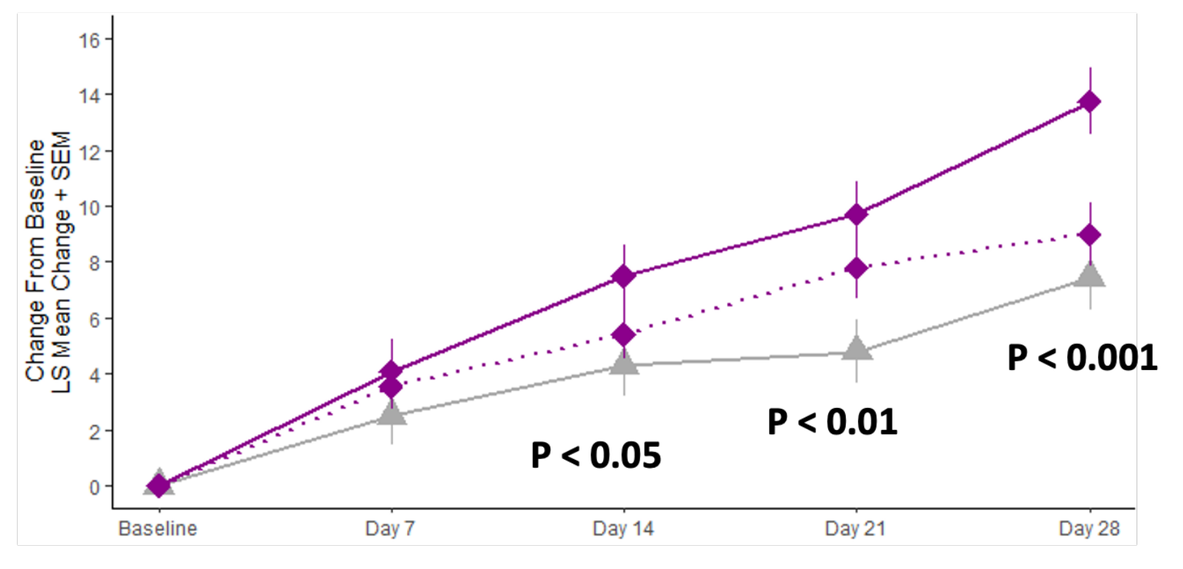
P < 0.05
P < 0.01
P < 0.001

VBM Negative

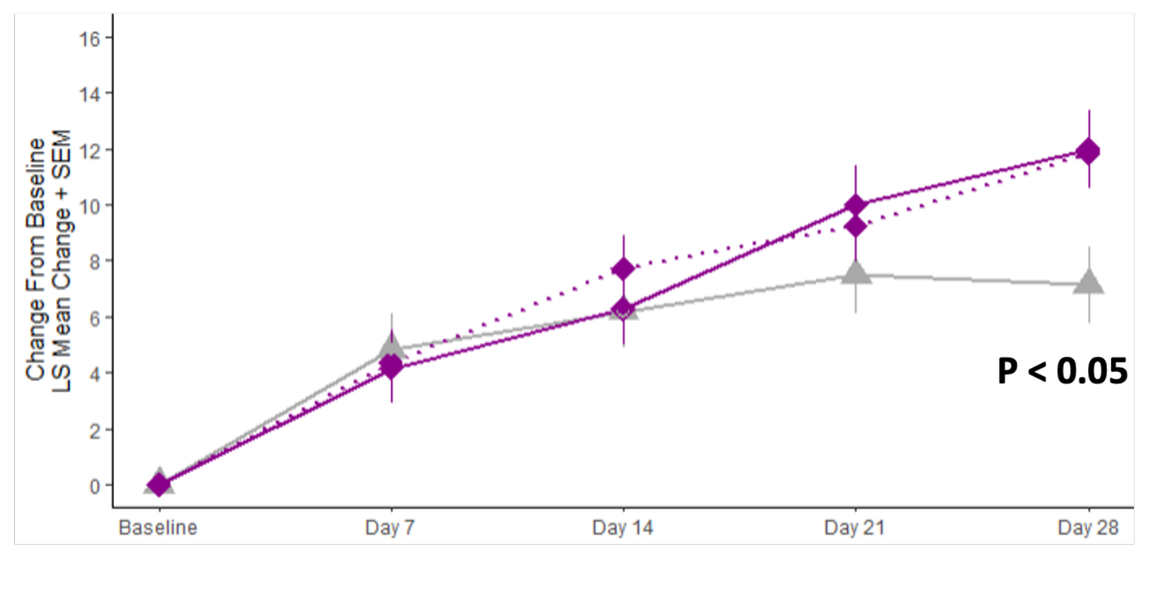
P < 0.05

Who are the VMB Pos Patients?

229 Patients were VBM Positive based on their screening session speech.

179 Patients were identified as VBM Negative.

VBM Positive patients were younger (37 ± 10; F = 15.67, p < 0.004) compared to VBM Negative patients (41 ± 11; F = xx.xx, p < 0.xxx), but didn't differ in sex or treatment assignment.

VBM Positive people had:

- More severe negative symptoms at baseline (F = 29.43, p < 0.001, d = 0.94).
- Slightly less severe positive and anxiety-depression symptoms at baseline (F's = 12.33 & 3.86, p's < 0.001 & < 0.05, d's = 0.40 & 0.11).

VBM Positive people showed a distinct signature in negative symptoms at baseline, characterized by more severe social avoidance (G16), blunted affect (N1), emotional withdrawal N2), poor rapport (N3), social withdrawal (N4), lack of spontaneity & flow of conversation (N6), and stereotyped thinking (N7).

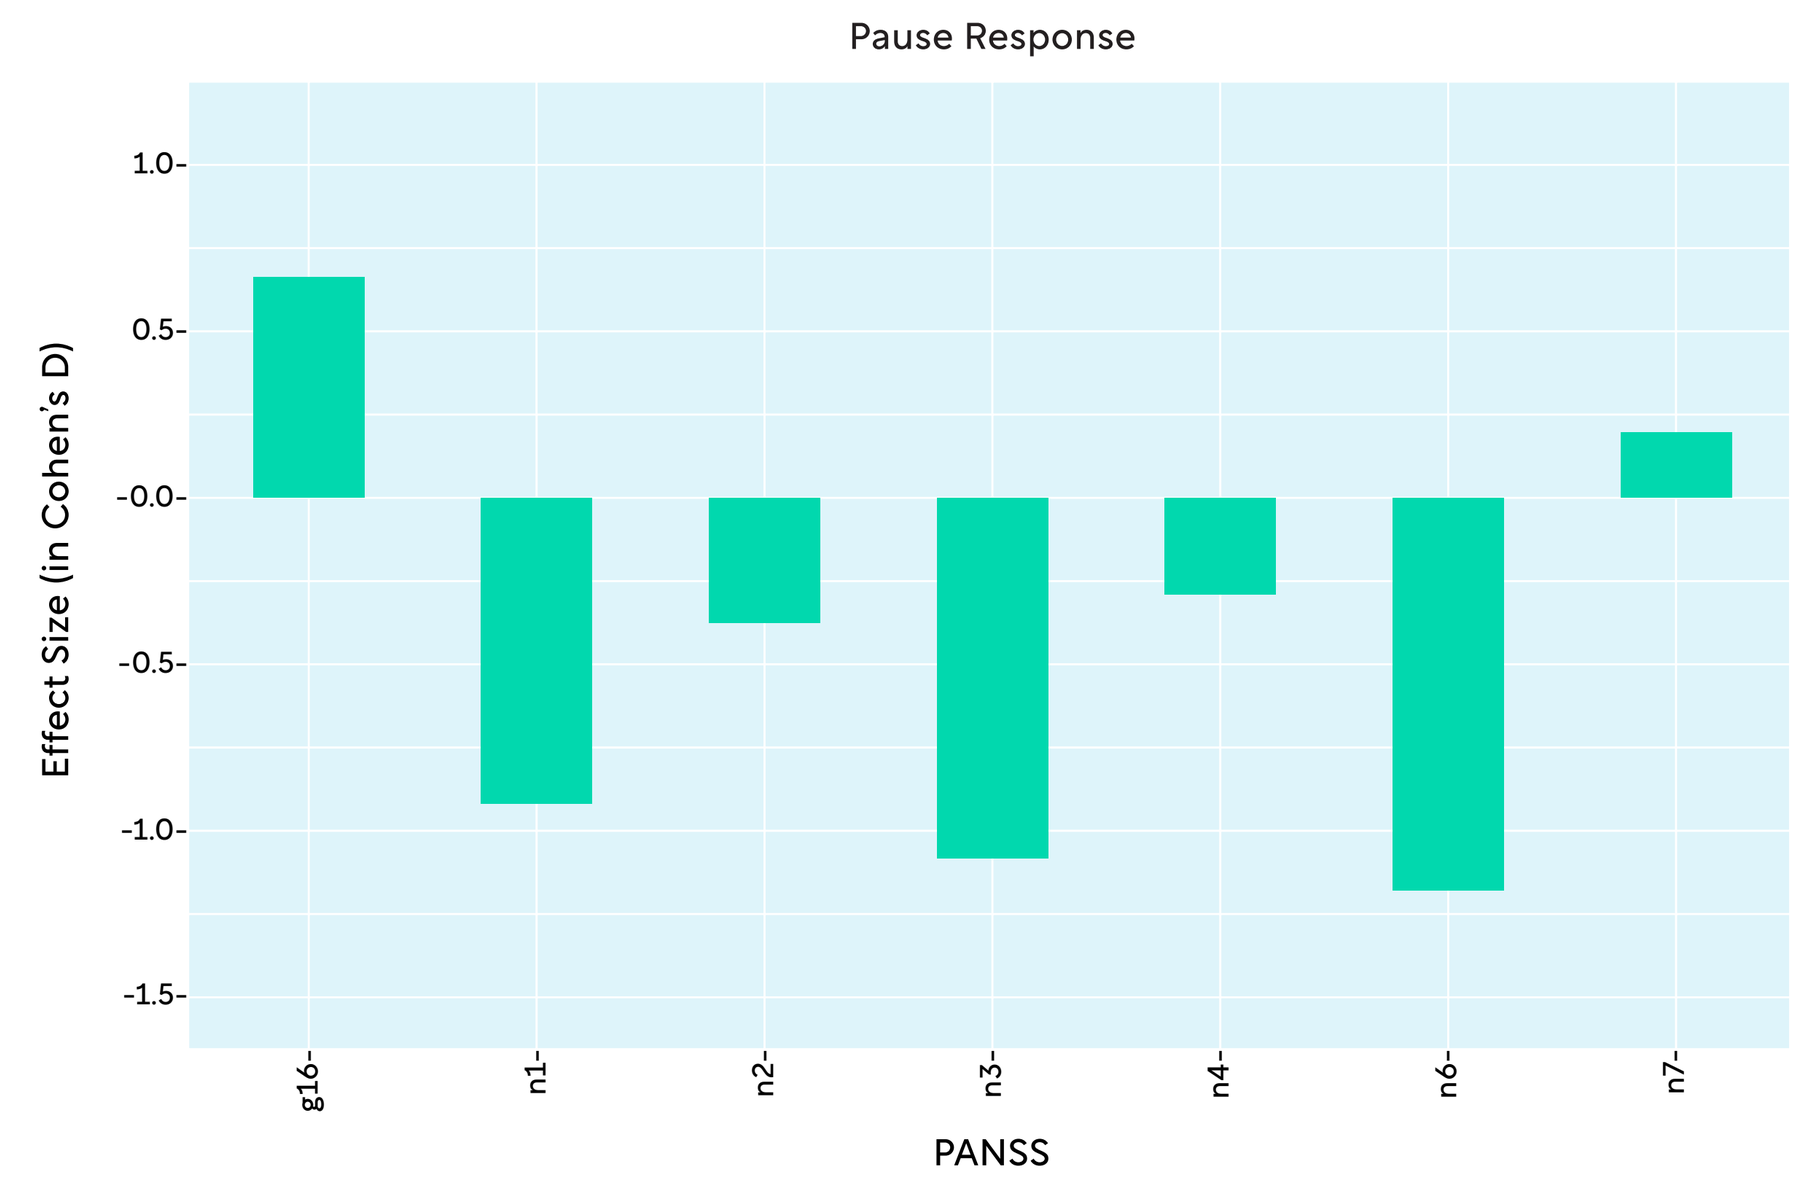
Pause Response
Effect Size (in Cohen's D)
PANSS
g16
n1
n2
n3
n4
n6
n7

During screening, VBM Positive people took 500ms longer to respond than VBM Negative people (1.5 sec. versus 1 sec; F = 42.79, p < 0.001).

During their screening interviews, VBM Positive people talked much less, at a slower pace, and had shorter interviews with fewer turns.

VBM Positive people showed difficulty engaging in the clinical interview.

Financial Acknowledgements

This project reflects a collaboration by Reviva Pharmaceuticals, Quantic Innovation, Louisiana State University, WCG and the Psychiatric Research Institute at the University of Arkansas for Medical Sciences. SB & LB are employees of Reviva Pharmaceuticals, Inc. ASC & BK are employees of Quantic Innovation, Inc, and MO & KT are employees of WCG.

References

Siegel, J. S., Cohen, A. S., Szabo, S. T., Tomioka, S., Opler, M., Kirkpatrick, B., & Hopkins, S. (2024). Enrichment using speech latencies improves treatment effect size in a clinical trial of bipolar depression. Psychiatry Research, 116105.